The N-terminal domain of HARS is a novel NRP2 ligand and can regulate NRP2-dependent macrophage function

Navatha Shree Sharma[1], Samikshan Dutta[1], Steve Crampton[2], Sanna Rosengren[2] and Kaustubh Datta[1]

[1]Department of Biochemistry and Molecular biology, University of Nebraska at Medical Cancer, Omaha, Nebraska, USA. [2] aTyr Pharma, INC, San Diego, CA, USA.

Correspondence: Kaustubh.datta@unmc.edu, srosengren@atyrpharma.com

Abstract

Tumor-associated macrophages (TAM) are associated with regulation of antitumor immune responses, and neuropilin-2 (NRP2), a pleiotropic receptor with an emerging role in immune responses, has recently been demonstrated to be important for this activity (Roy et al, Cancer Res. 2018). Deletion of NRP2 results in impaired clearance of apoptotic tumor cells through reduced efferocytosis, which plays a role in tumor promotion. We now show that the N-terminal domain of the histidyl-tRNA synthetase (HARS) is a specific binding partner of NRP2, and can regulate the phagocytic function of NRP2 in macrophages. The HARS N-terminal domain, which is found only in higher eukaryotes, is conserved among splice variants of HARS and has evolved to regulate immune cell engagement. Incubation of macrophages with recombinant HARS N-terminal domain, had no effect on phagocytic uptake, but significantly impaired the maturation of phagosomes in a dose dependent manner. This phenotype mimics that of the NRP2 knockout, suggesting pharmacological intervention with this agent to modulate NRP2 driven biology may be possible.

Background

- Tumor-associated macrophages (TAMs) are one of the most abundant immune cells in the tumor stroma.
- TAMs play a multifaceted role in promoting tumor growth and angiogenesis by releasing growth factors and inhibiting immune surveillance by clearance of apoptotic tumor cells within the tumor microenvironment via efferocytosis in an immunologically silent manner, thus maintaining an immunosuppressed microenvironment.
- High numbers of TAMs correlate with poor patient outcome in cancers such as pancreatic cancer (PC) which makes TAMs a highly desirable therapeutic target.
- NRP2, a non-tyrosine kinase receptor is expressed by TAMs and reported to regulate their efferocytotic function in the tumor microenvironment (Roy S *et al*, Can. Res 2018).

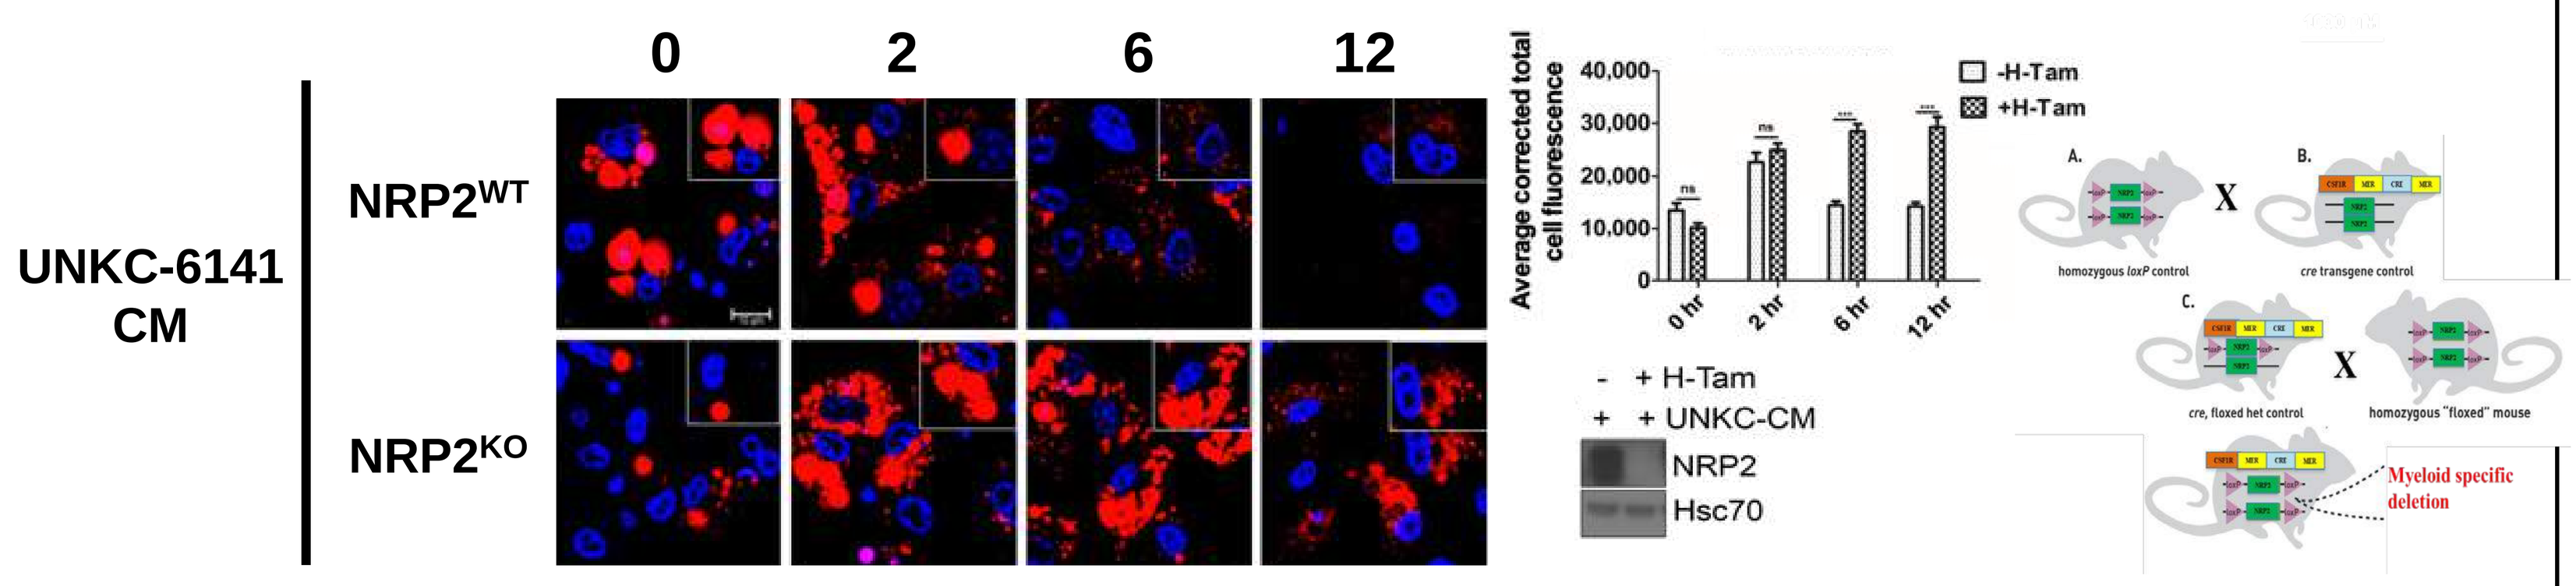
0
2
6
12
UNKC-6141 CM
NRP2WT
NRP2KO
Average corrected total cell fluorescence
-H-Tam
+H-Tam
- + H-Tam
+ + UNKC-CM
NRP2
Hsc70
Myeloid specific deletion

- Studies with subcutaneous tumor implantation of UNKC-6141 (mouse pancreatic tumor cell line) showed that depletion of NRP2 in macrophages caused a decrease in tumor burden, increased necrosis and CD8+ T-cell infiltration (Roy S *et al*, Can. Res. 2018).

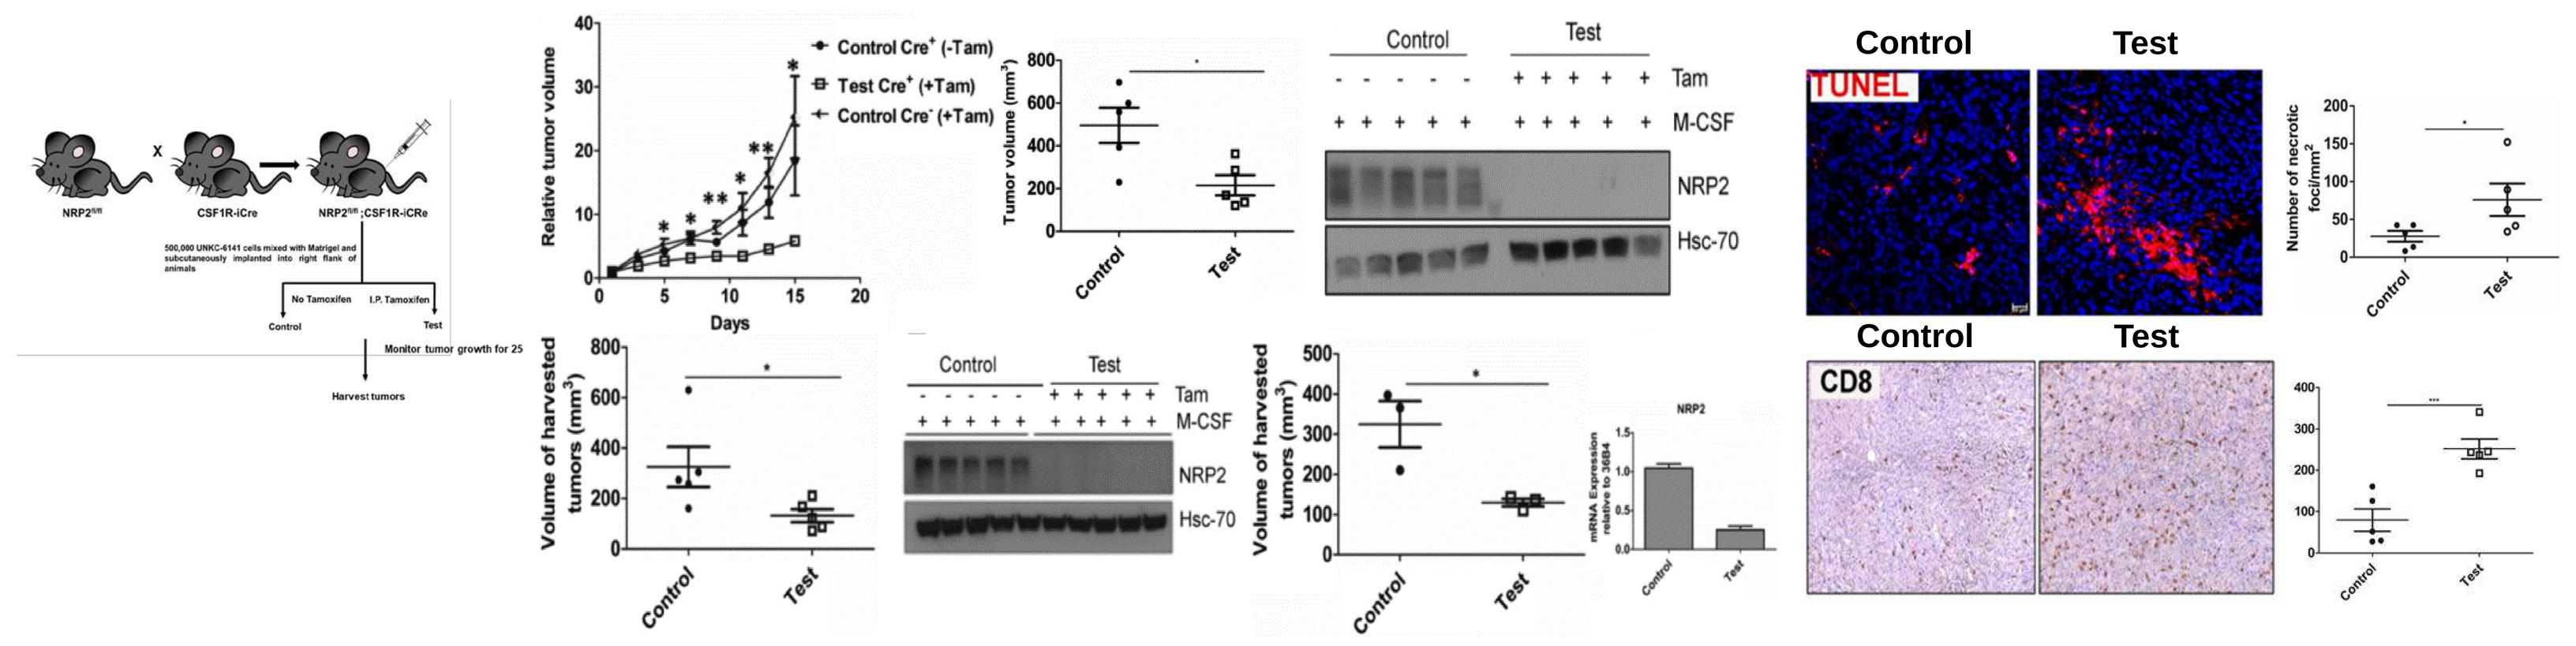
Control
Test
TUNEL
CD8
Relative tumor volume
Days
Volume of harvested tumors (mm3)
Tam
M-CSF
NRP2
Hsc-70

- Together, our published literature demonstrates that **TAM-derived NRP2 plays a crucial role in tumor promotion through efferocytosis and immune suppression, suggesting that it might be a novel target for the development of effective immunotherapy.**
- tRNA synthetases are a class of intracellular proteins crucial to protein synthesis. Proteins derived from the histidyl-tRNA synthetase (HARS) gene are found extracellularly and are thought to play a role in controlling the set point of key cells in the immune system to ensure appropriate control of immune responses.
- aTYR Pharma INC is developing HARS-derived proteins that have immunomodulatory functions. Of them, ATYR1923 (iMod-Fc) is being evaluated in a Phase 1b/2a clinical trial of sarcoidosis.
- iMod, the N-terminal domain of HARS, has immunomodulatory properties, and the iMod-Fc construct was generated to enhance the pharmacokinetic profile of the molecule.
- **NRP2, but not NRP1, was identified as a specific binding partner of iMod-Fc in a receptor screen of >4500 human transmembrane receptors.**
- Binding was confirmed by multiple techniques such as SPR, ELISA, and binding to cells transfected to express NRP2.

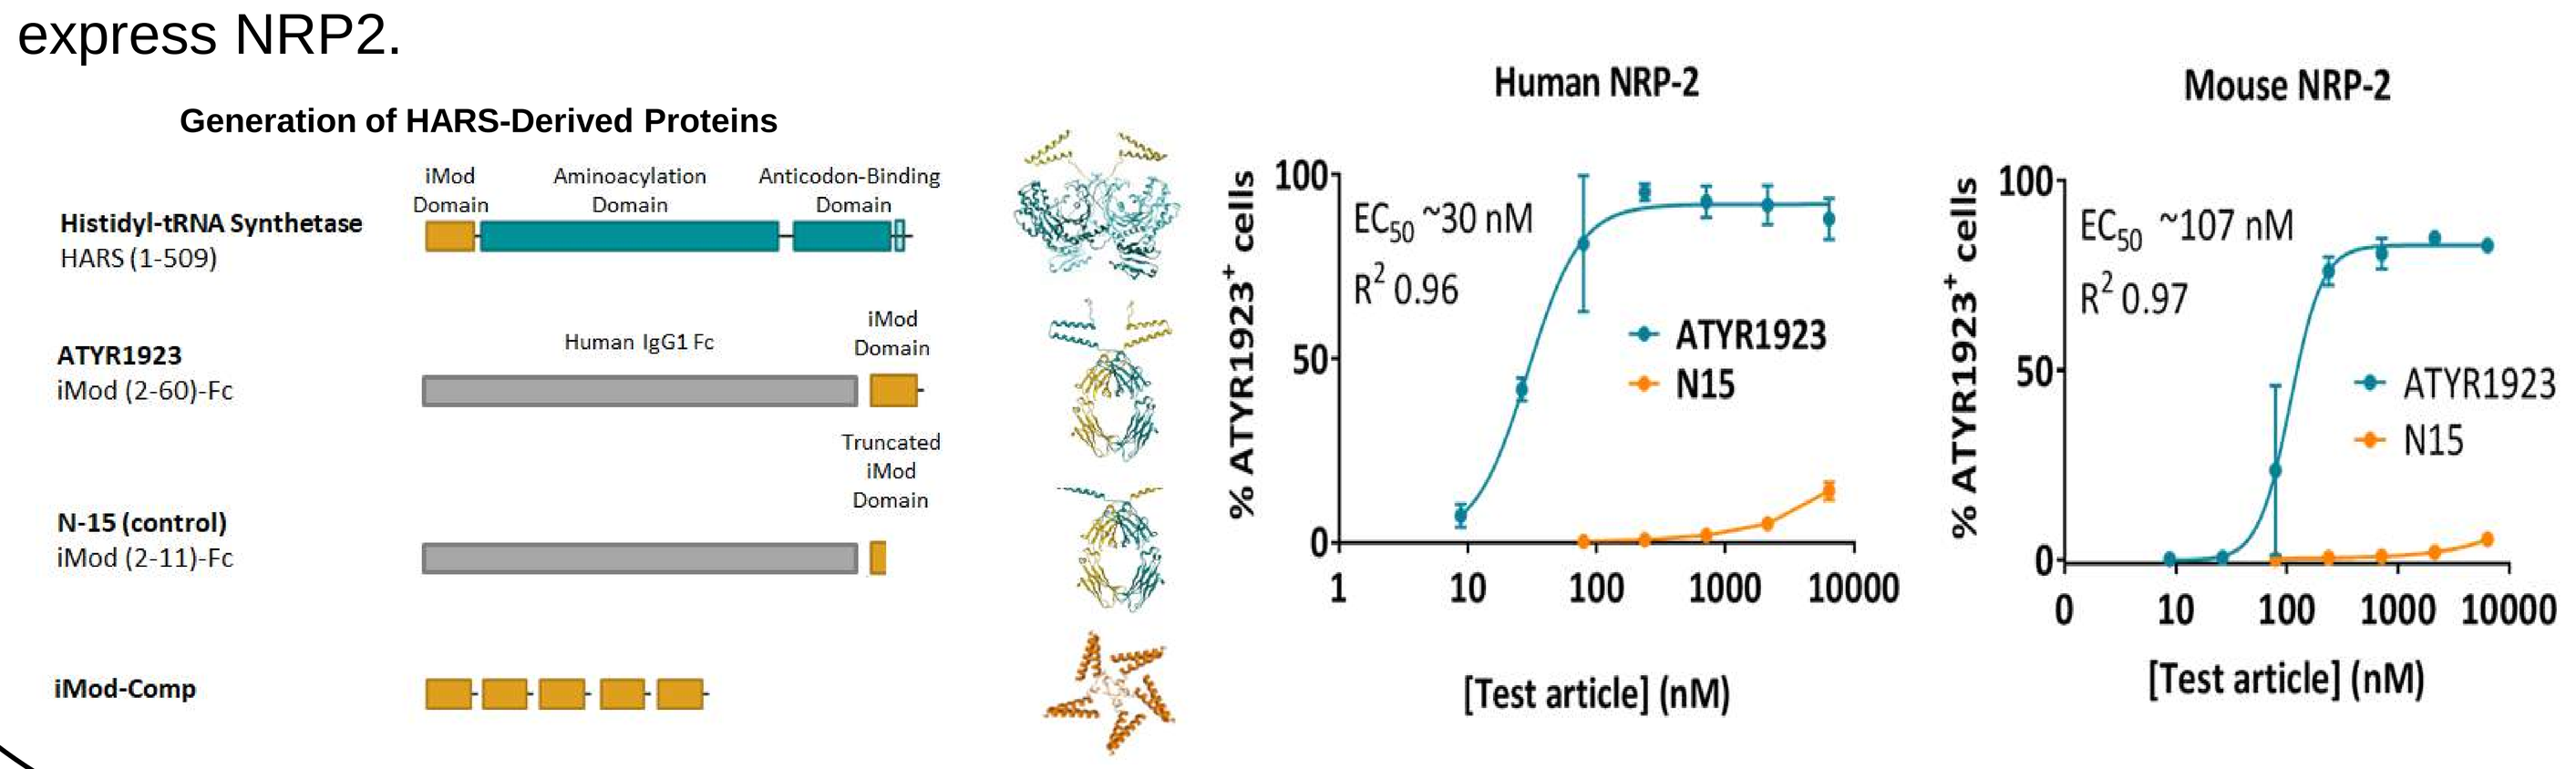
Generation of HARS-Derived Proteins
Histidyl-tRNA Synthetase HARS (1-509)
ATYR1923 iMod (2-60)-Fc
N-15 (control) iMod (2-11)-Fc
iMod-Comp
Human NRP-2
EC50 ~30 nM
R2 0.96
Mouse NRP-2
EC50 ~107 nM
R2 0.97
ATYR1923
N15
% ATYR1923+ cells
[Test article] (nM)

Objective

To determine whether iMod-Fc interferes with macrophage phagocytic function via inhibition of NRP2.

Methodology

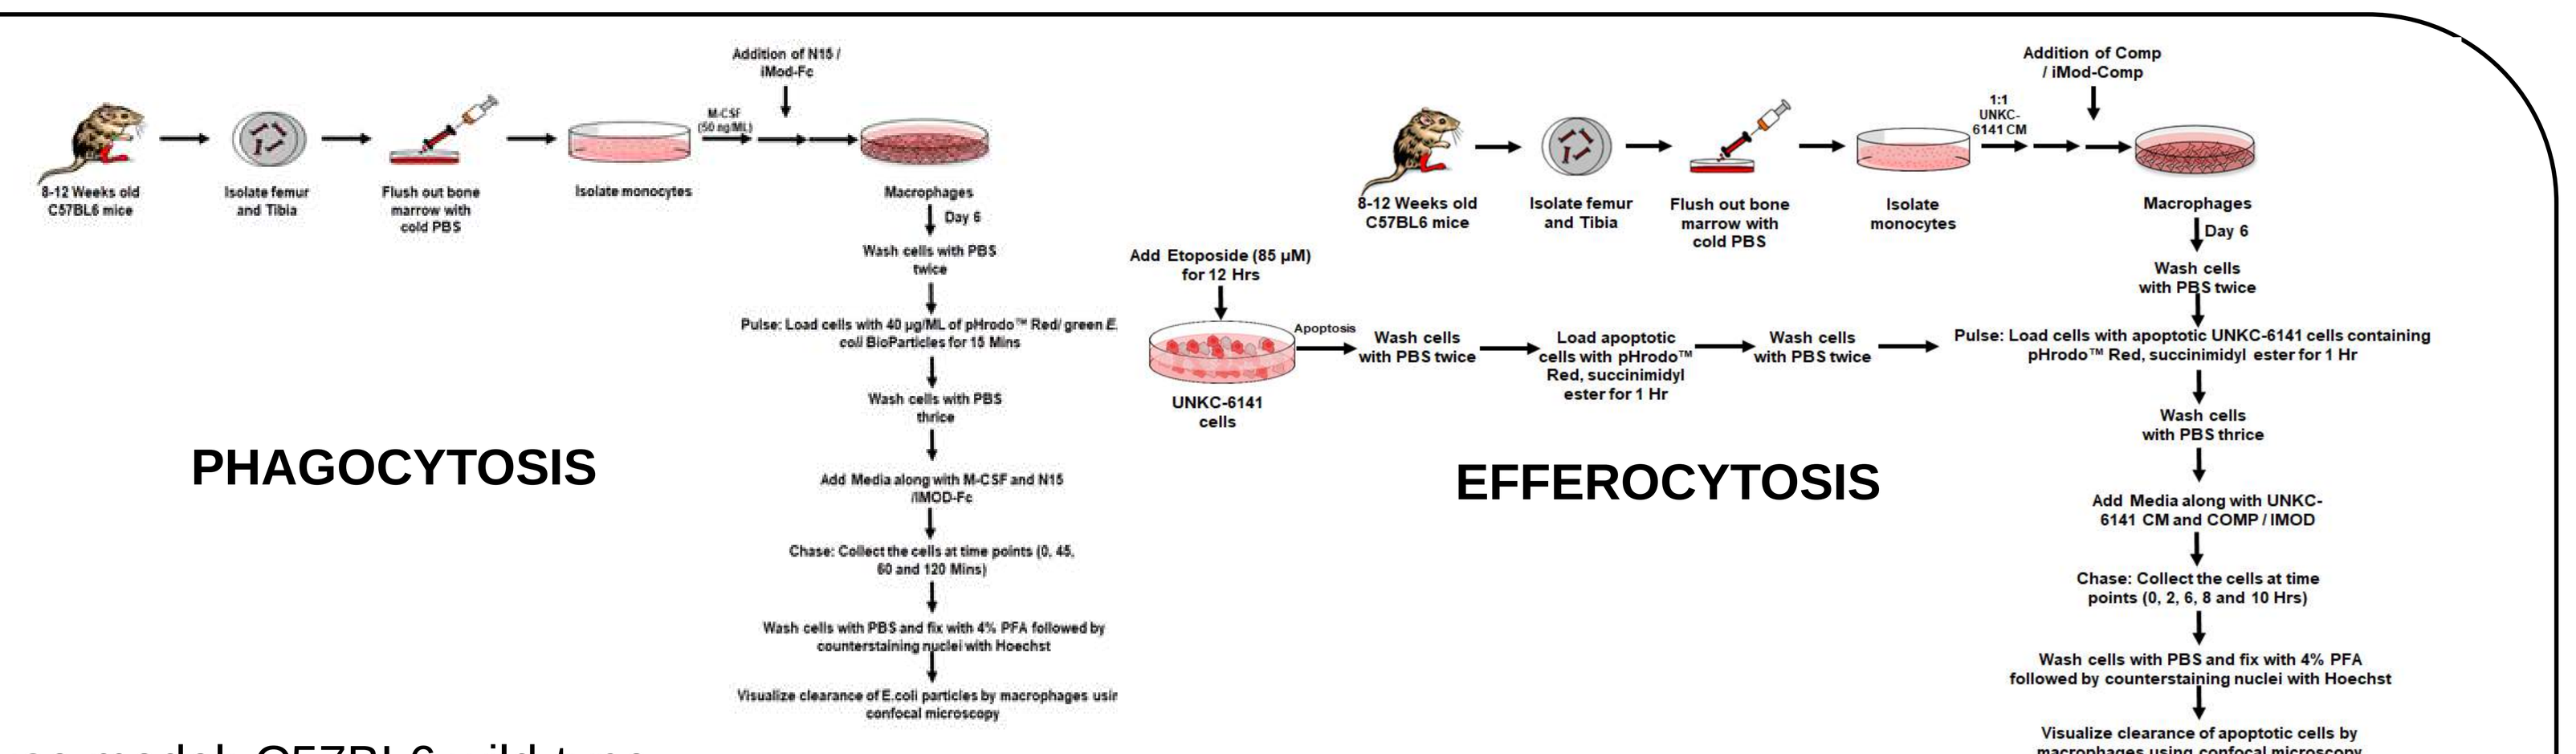
PHAGOCYTOSIS
EFFEROCYTOSIS

- Mouse model: C57BL6 wild-type
- To differentiate into macrophages, mouse monocytes were cultured with either murine M-CSF or 1:1 UNKC-6141 CM for 6 days.
- iMod-Fc or iMod-Comp was added according to the experiment on specified days along with respective N15/ Comp controls.
- Immunofluorescence was used to monitor phagocytosis/ efferocytosis.

Results

NRP2 is essential for macrophage interaction with and internalization of iMod

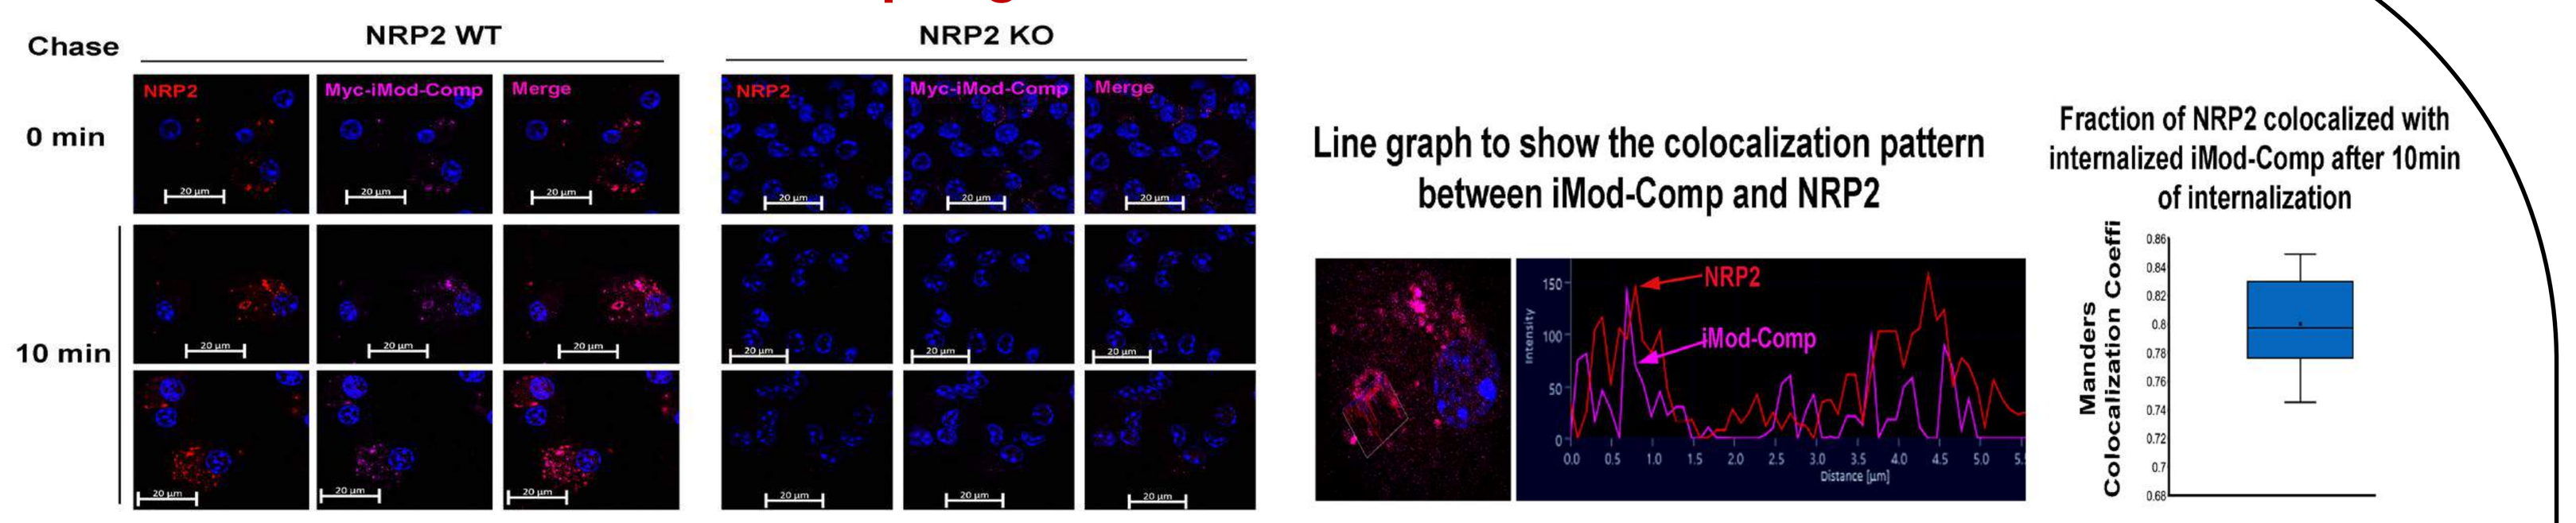
Chase
NRP2 WT
NRP2 KO
0 min
10 min
Line graph to show the colocalization pattern between iMod-Comp and NRP2
Fraction of NRP2 colocalized with internalized iMod-Comp after 10min of internalization
Manders Colocalization Coeffi

iMod-Fc blocks macrophage phagosome maturation in a dose- and time-dependent manner

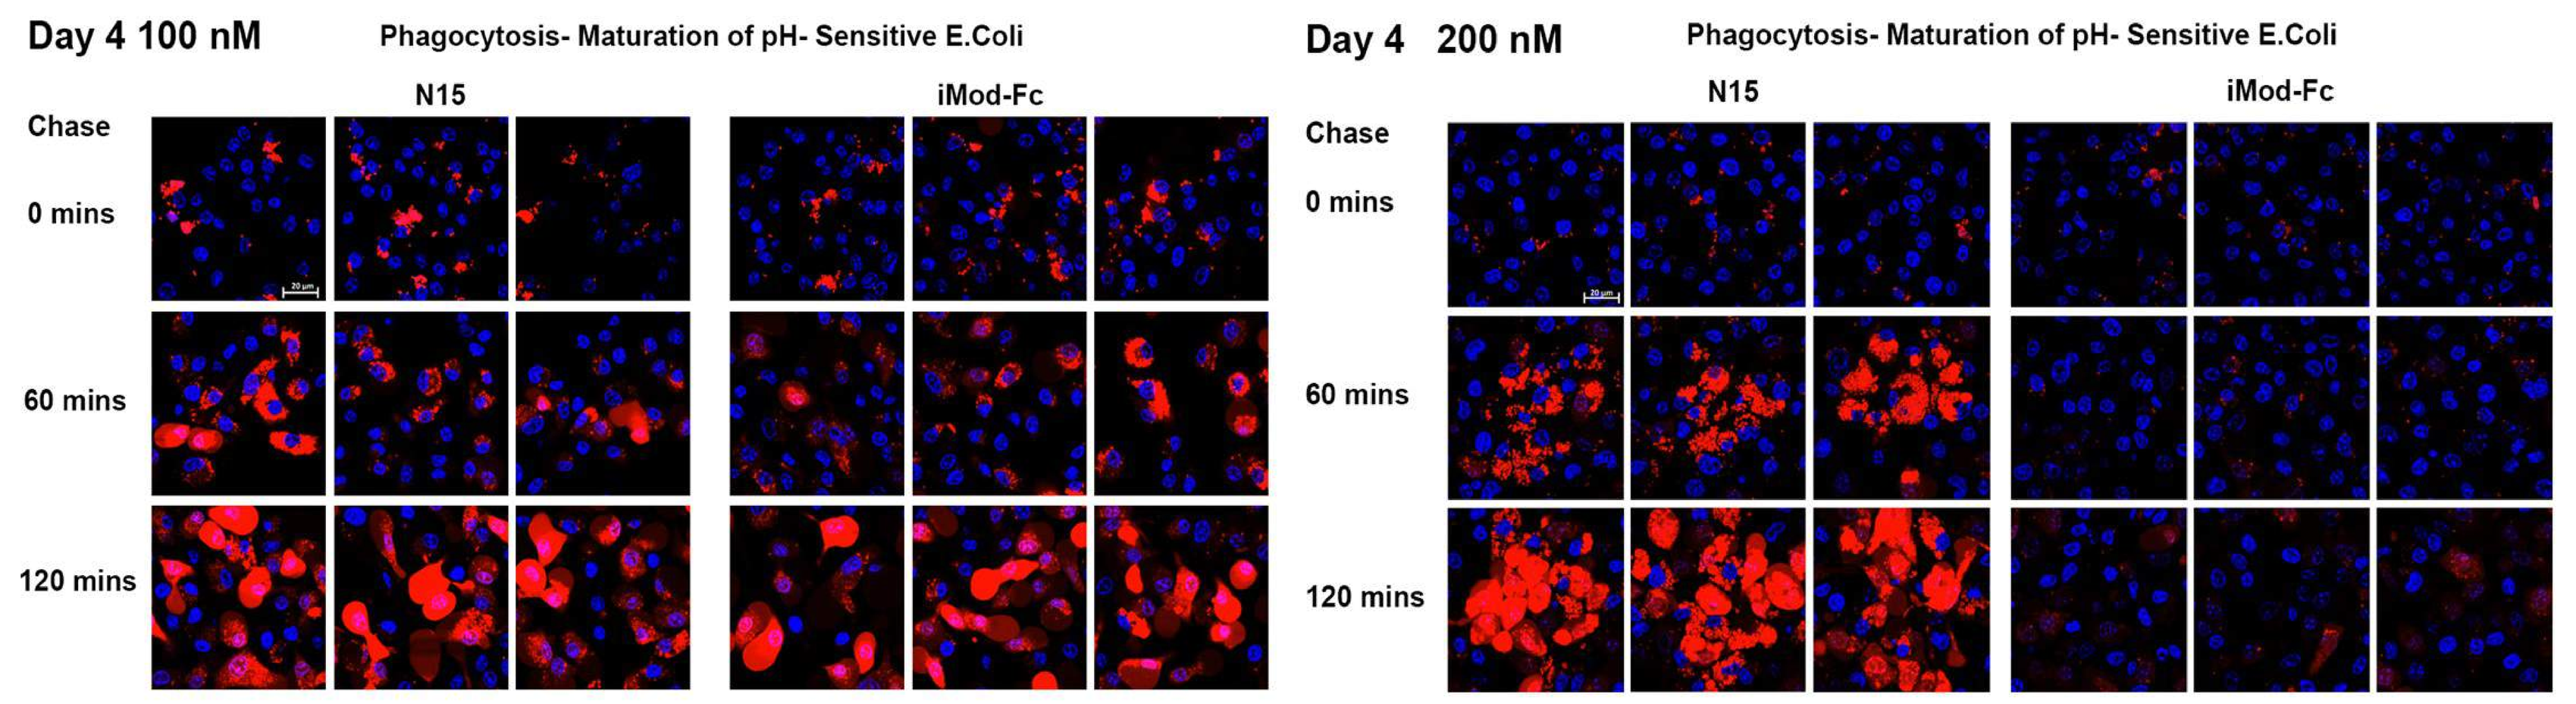
Day 4 100 nM
Phagocytosis- Maturation of pH- Sensitive E.Coli
Day 4 200 nM
N15
iMod-Fc
Chase
0 mins
60 mins
120 mins

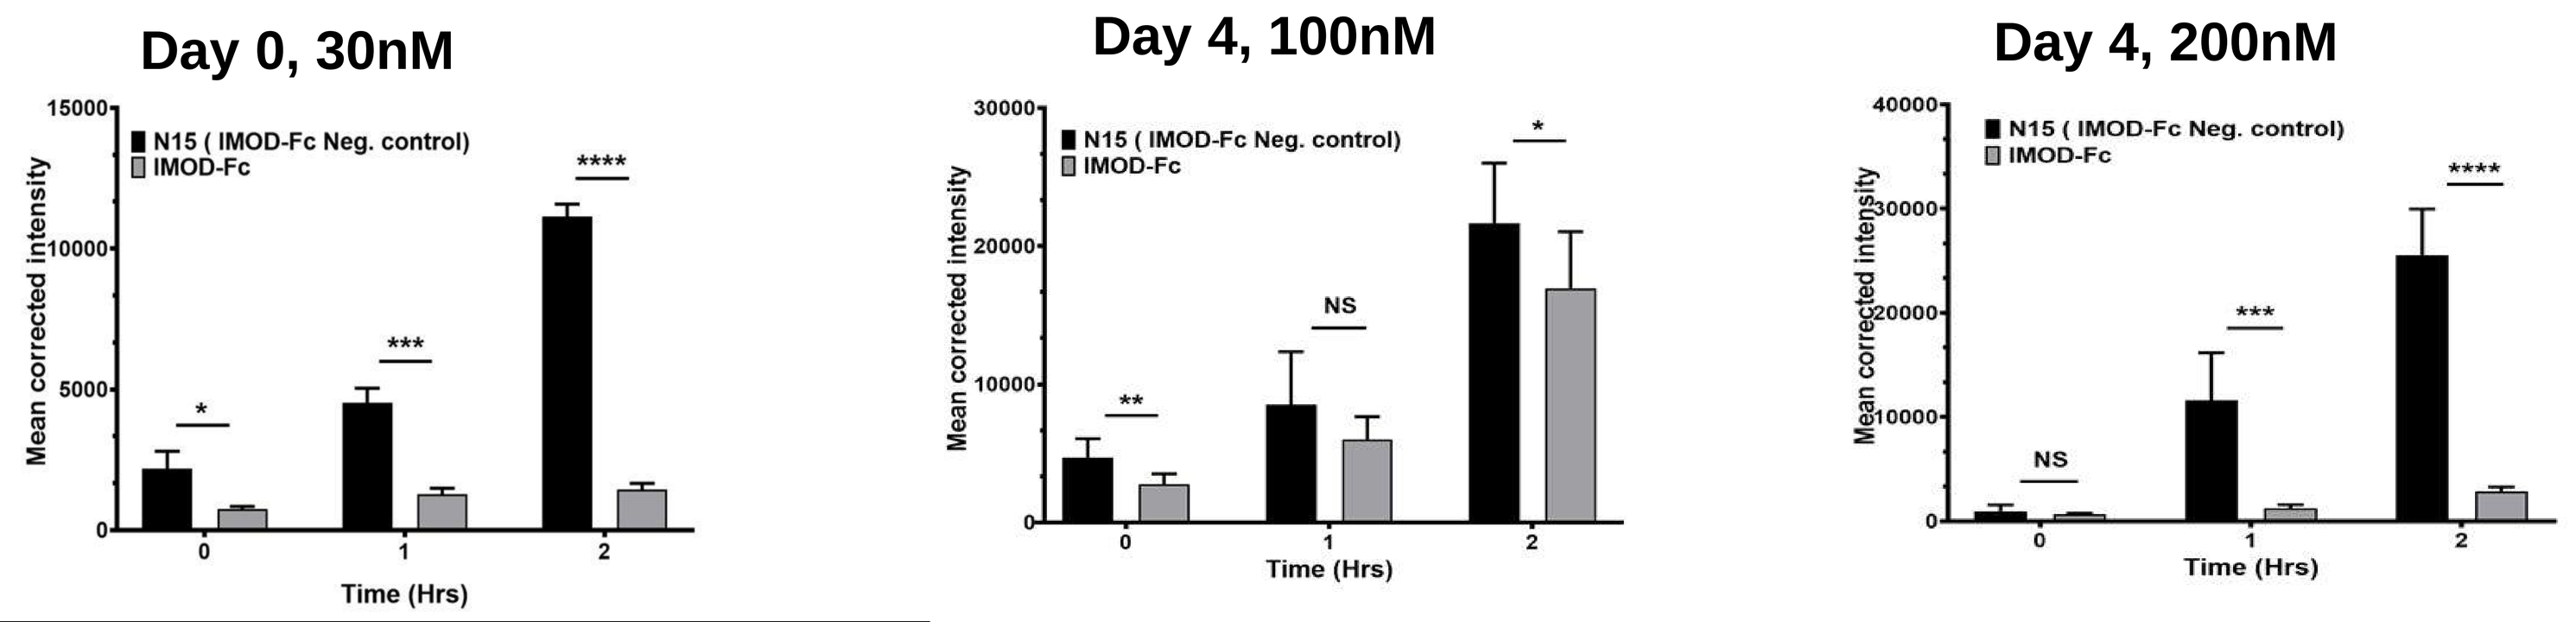
Day 0, 30nM
Day 4, 100nM
Day 4, 200nM
N15 (IMOD-Fc Neg. control)
IMOD-Fc
Mean corrected intensity
Time (Hrs)

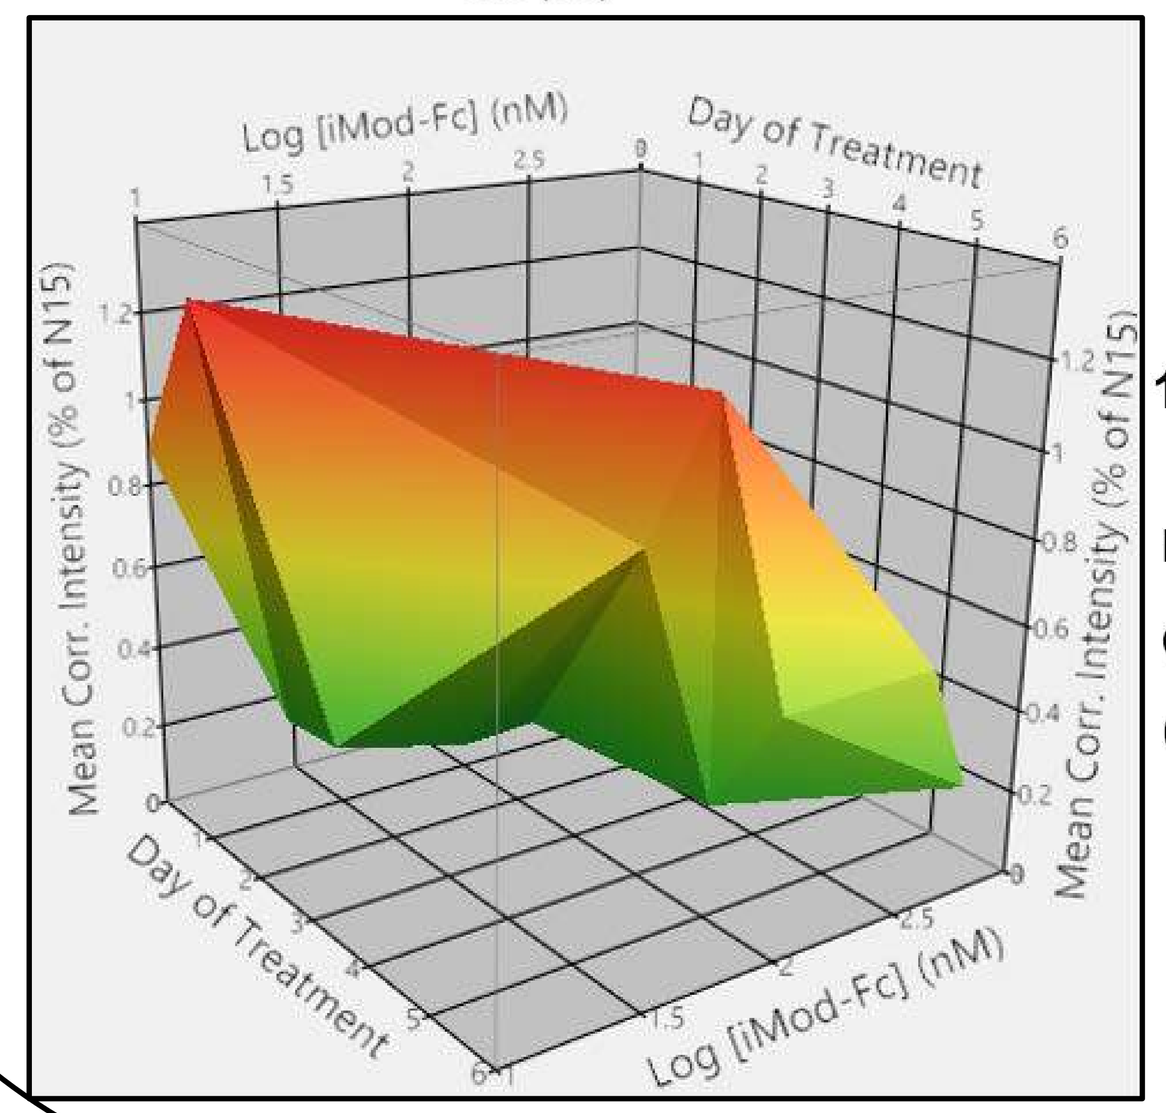
Log [iMod-Fc] (nM)
Day of Treatment
Mean Corr. Intensity (% of N15)

Phagocytotic uptake of pH-insensitive E.Coli particles is not affected

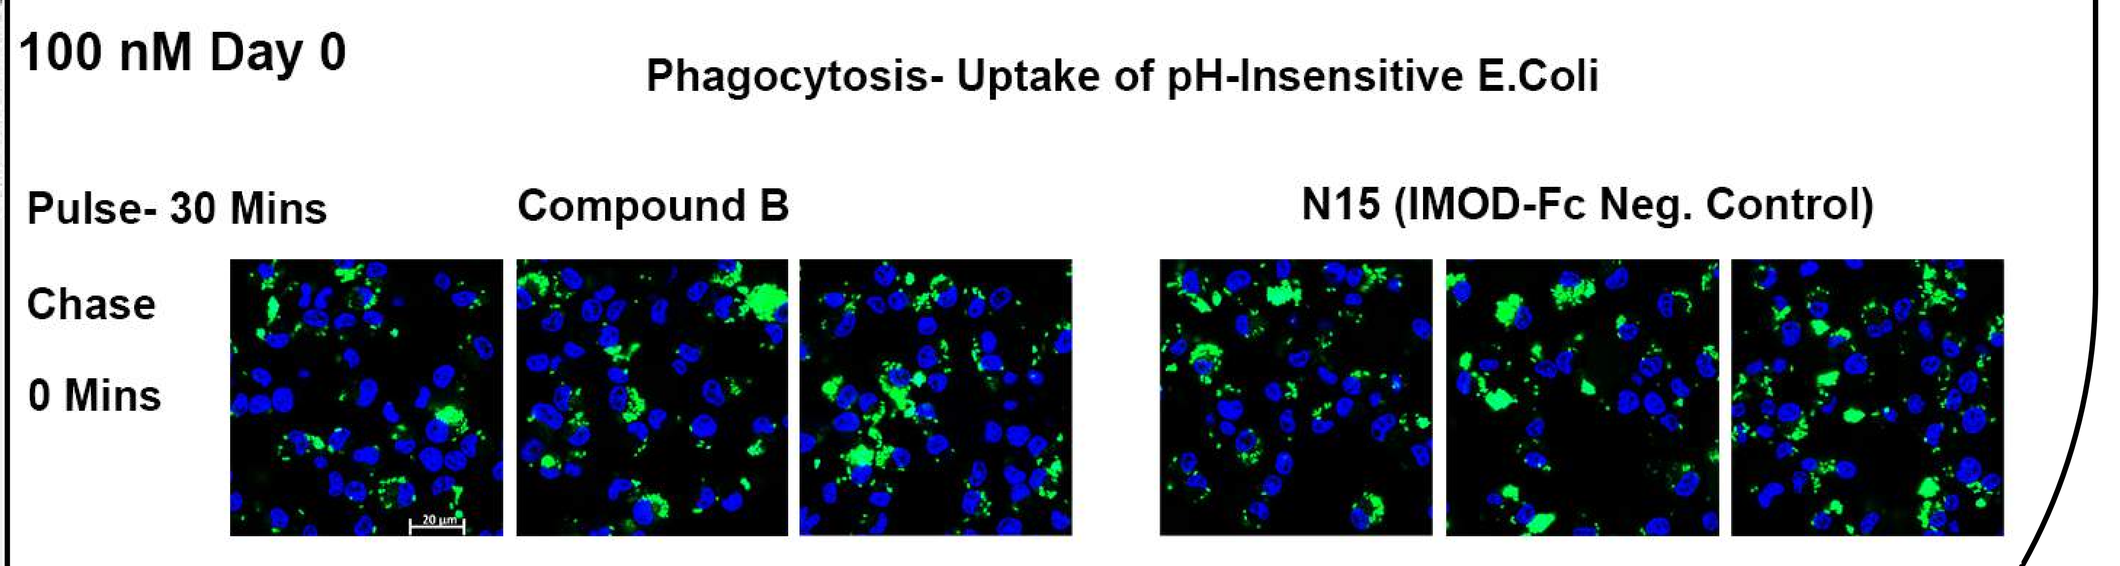
100 nM Day 0
Phagocytosis- Uptake of pH-Insensitive E.Coli
Pulse- 30 Mins
Chase
0 Mins
Compound B
N15 (IMOD-Fc Neg. Control)

Results

iMOD alters the resolution of macrophage efferocytosis in a dose-dependent manner

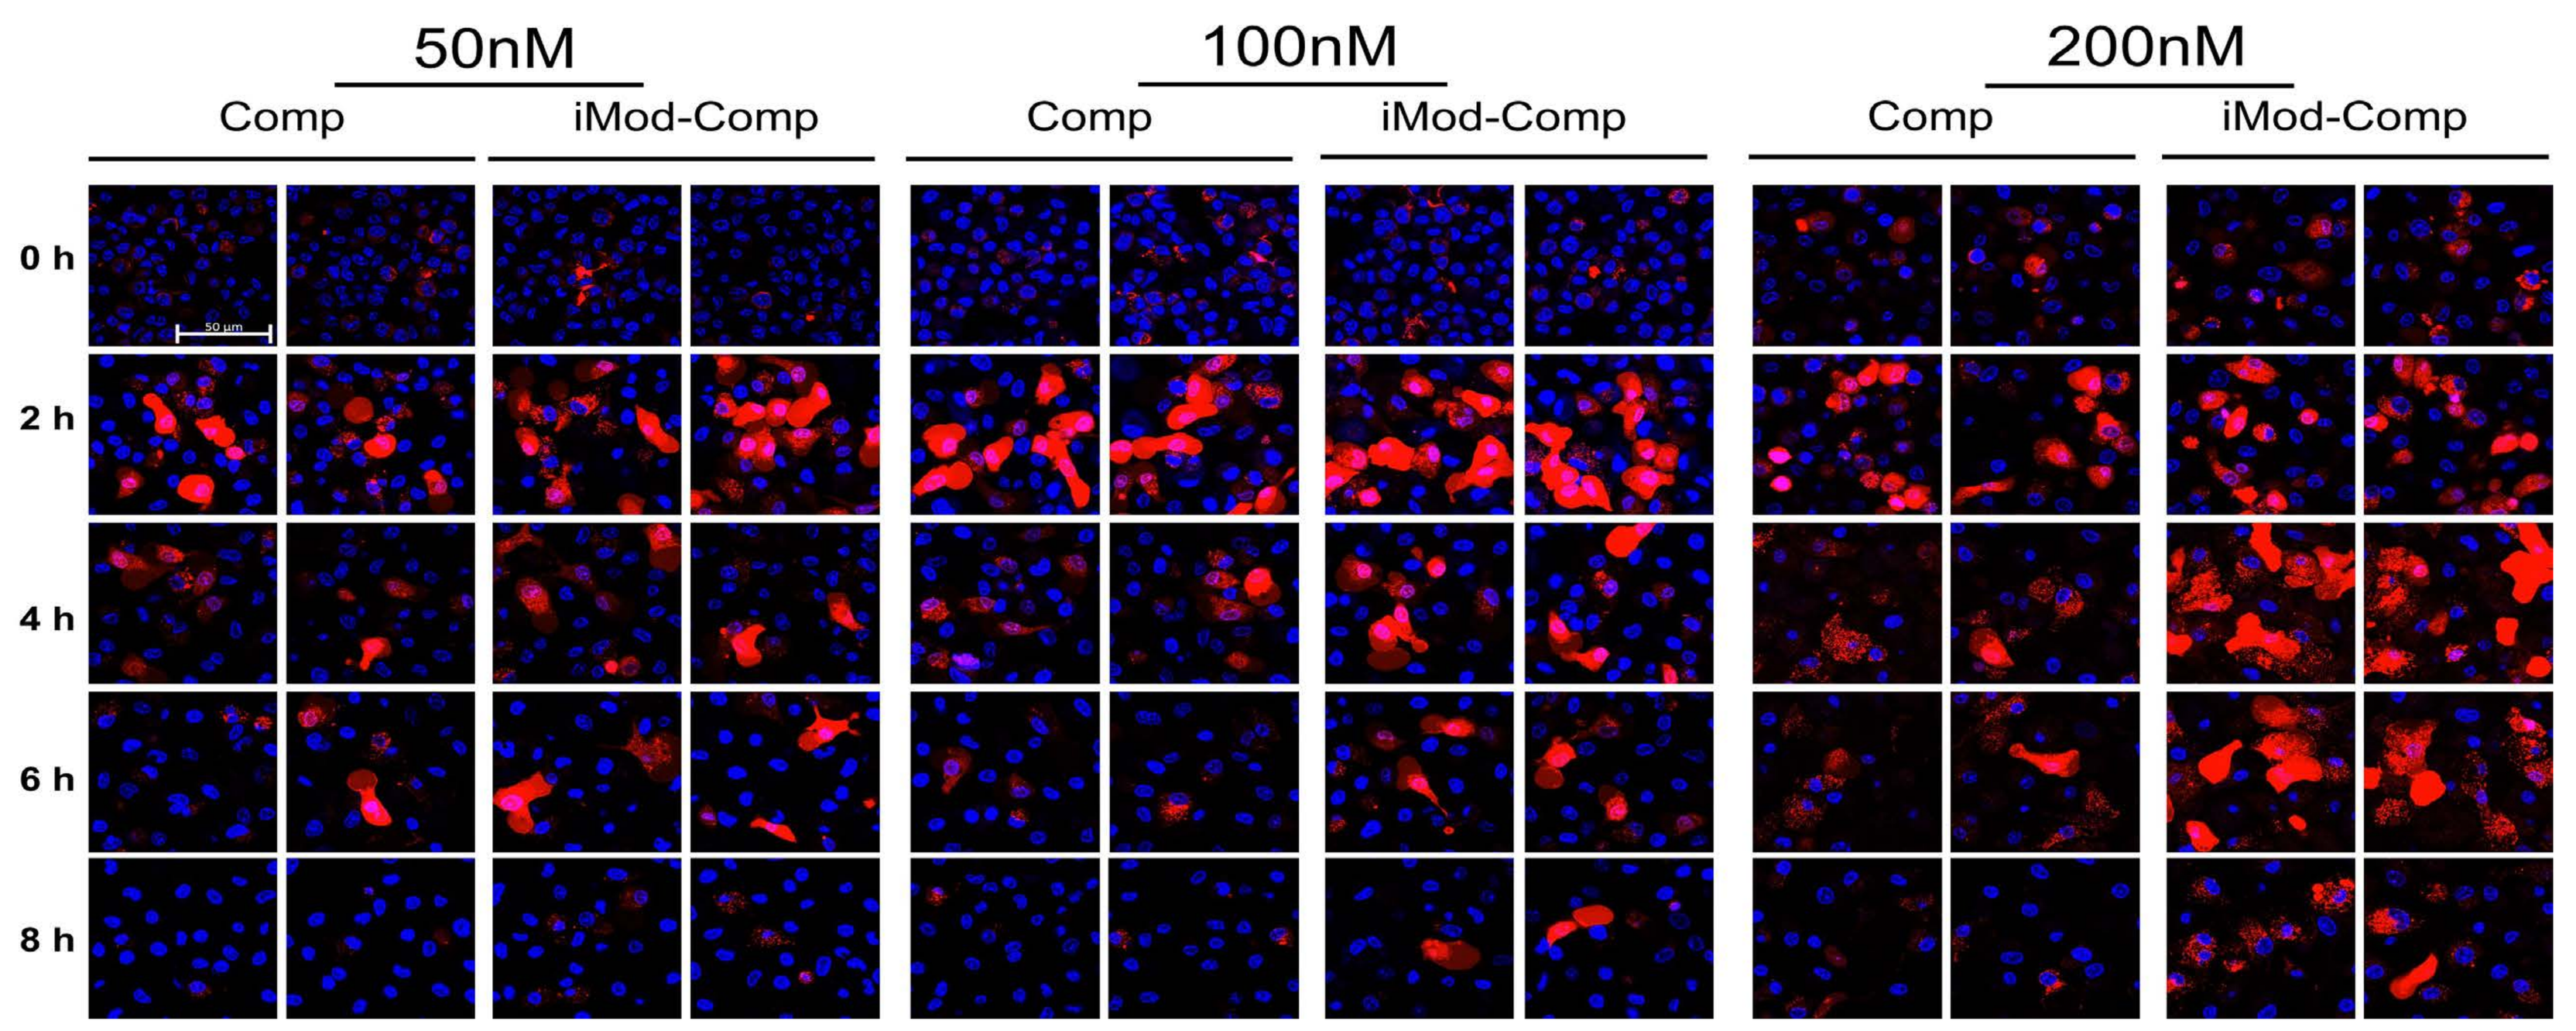
50nM
100nM
200nM
Comp
iMod-Comp
0 h
2 h
4 h
6 h
8 h

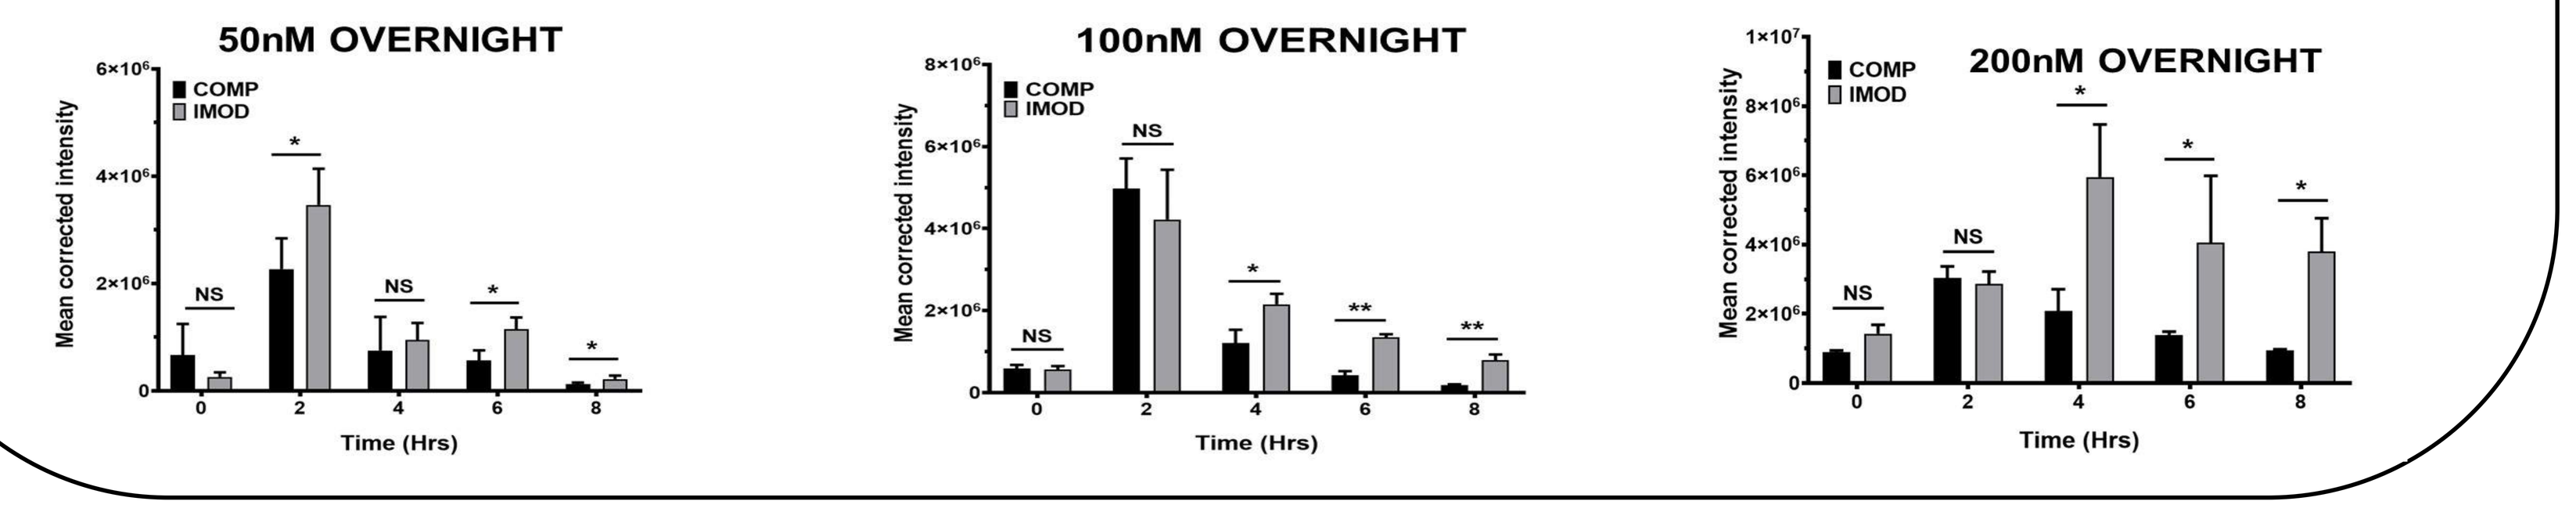
50nM OVERNIGHT
100nM OVERNIGHT
200nM OVERNIGHT
COMP
IMOD
Mean corrected intensity
Time (Hrs)

Conclusions

- The N-terminal HARS domain, iMod, interacts with NRP2 and is internalized by macrophages.
- iMod-Fc blocks phagocytosis in a dose- and time-dependent manner in macrophages.
- In the macrophage efferocytosis assay, iMod inhibits the clearance of apoptotic tumor cells.
- These results coincide with earlier findings using NRP2 knockouts, suggesting that iMod acts via NRP2.

References

- E. Mertsching, J. Ampudia, R. Adams, S. Rosengren, L. Nangle, J. Mendlein, A. Cubitt, F. Ramsdell, K. Ogilvie, and D. King. Identification of a T cell immunomodulatory domain in histidyl-tRNA transferase. Presented at the annual meeting of the American Association of Immunologists. J. Immunol**.** 2018 May 200 (1 Supplement) 112.3
- Roy S, Bag AK, Dutta S, Polavaram NS, Islam R, Schellenburg S, Banwait J, Guda C, Ran S, Hollingsworth MA, Singh RK, Talmadge JE, Muders MH, Batra SK, Datta K. Macrophage-Derived Neuropilin-2 Exhibits Novel Tumor-Promoting Functions. Cancer Res. 2018 Oct 1;78(19):5600-5617.

iMod-Fc (ATYR1923) is covered by PCT Patent publication WO2014145050, entitled HISTIDYL-TRNA SYNTHETASE-FC CONJUGATES.

Acknowledgements

This work is supported by the following funding: aTYR Pharma INC and NIH grant **CA140432, CA182435A** (K Datta).