ATYR1923 Specifically Binds To Neuropilin-2, A Novel Therapeutic Target For The Treatment Of Immune-mediated Diseases

Z. Xu[1], Y. Chong[1], S. Crampton[1], R. Adams[1], K. Rauch[1], S. Paz[1], D. Chu[1], L. Zhai[2], L. Burman[1], J. Ampudia[1], S. Rosengren[1], D. King[1], L. Nangle[1]

1. aTyr Pharma, San Diego, California, USA 2. Hong Kong University of Science and Technology, Hong Kong, Hong Kong

Abstract

INTRODUCTION: We have previously discovered that a splice variant of the histidyl-tRNA synthetase (HARS) gene has an immuno-modulatory effect. ATYR1923 is a fusion protein comprised of this splice variant, which aTyr termed iMod, fused to human IgG1 Fc. ATYR1923 inhibits cytokines and chemokines involved in the regulation of inflammatory and fibrotic responses and reduces inflammation and fibrosis in animal models of interstitial lung diseases (ILD), a group of immune-mediated lung disorders that can result in lung fibrosis. ATYR1923 is being evaluated in a Phase 1b/2a study in patients with pulmonary sarcoidosis, a major form of ILD. We sought to characterize the molecular basis for ATYR1923's immuno-modulatory properties.

RESULTS: Cell microarray screening technology was used to assess ATYR1923 binding to a library of >4500 human plasma membrane proteins, and identified the membrane-bound splice isoforms of Neuropilin-2 (NRP2) as specific, selective hits. Notably, no binding was detected for the related and structurally similar receptor Neuropilin-1 (NRP1). Specific binding of ATYR1923 to NRP2 was confirmed by surface plasmon resonance (SPR) and also by flow cytometry analysis of HEK293 cells over-expressing NRP2. Furthermore, ATYR1923 also bound to cells that endogenously express NRP2 on the surface (such as A549, THP-1 polarized M1 macrophages).

To map the binding sites of ATYR1923 on NRP2, we constructed a number of deletion mutants of the extracellular region of NRP2, and the binding was narrowed to the a2b1b2 fragment that encompasses binding contacts for vascular endothelial growth factors (VEGFs) and semaphorins. Domain swapping studies in which individual domains of cell-surface expressed NRP2 were replaced with the corresponding, structurally similar domain from NRP1 demonstrated that b1 is the key domain for binding, while the a2 and b2 domains likely also provide contacts with ATYR1923. In addition, we solved the crystal structure of the protein complex of ATYR1923 with an anti-iMod antibody that blocks its binding to NRP2, demonstrating the loop region within iMod to be the key interaction site for ATYR1923 with NRP2.

CONCLUSIONS: ATYR1923 specifically and selectively binds to NRP2 on the cell surface. ATYR1923 has previously demonstrated potent immunomodulatory activity *in vitro* and *in vivo*. NRP2 is a pleiotropic cell surface receptor known to be expressed on a number of different immune cell types and may play a key role in regulating inflammatory responses. These findings indicate that modulation of the NRP2 signaling pathway with ATYR1923 could be a novel therapeutic approach to immune-mediated diseases such as pulmonary sarcoidosis and other interstitial lung diseases.

Introduction

Figure 1. Extracellular tRNA Synthetase Biology and Development of ATYR1923 as A Therapeutic Candidate

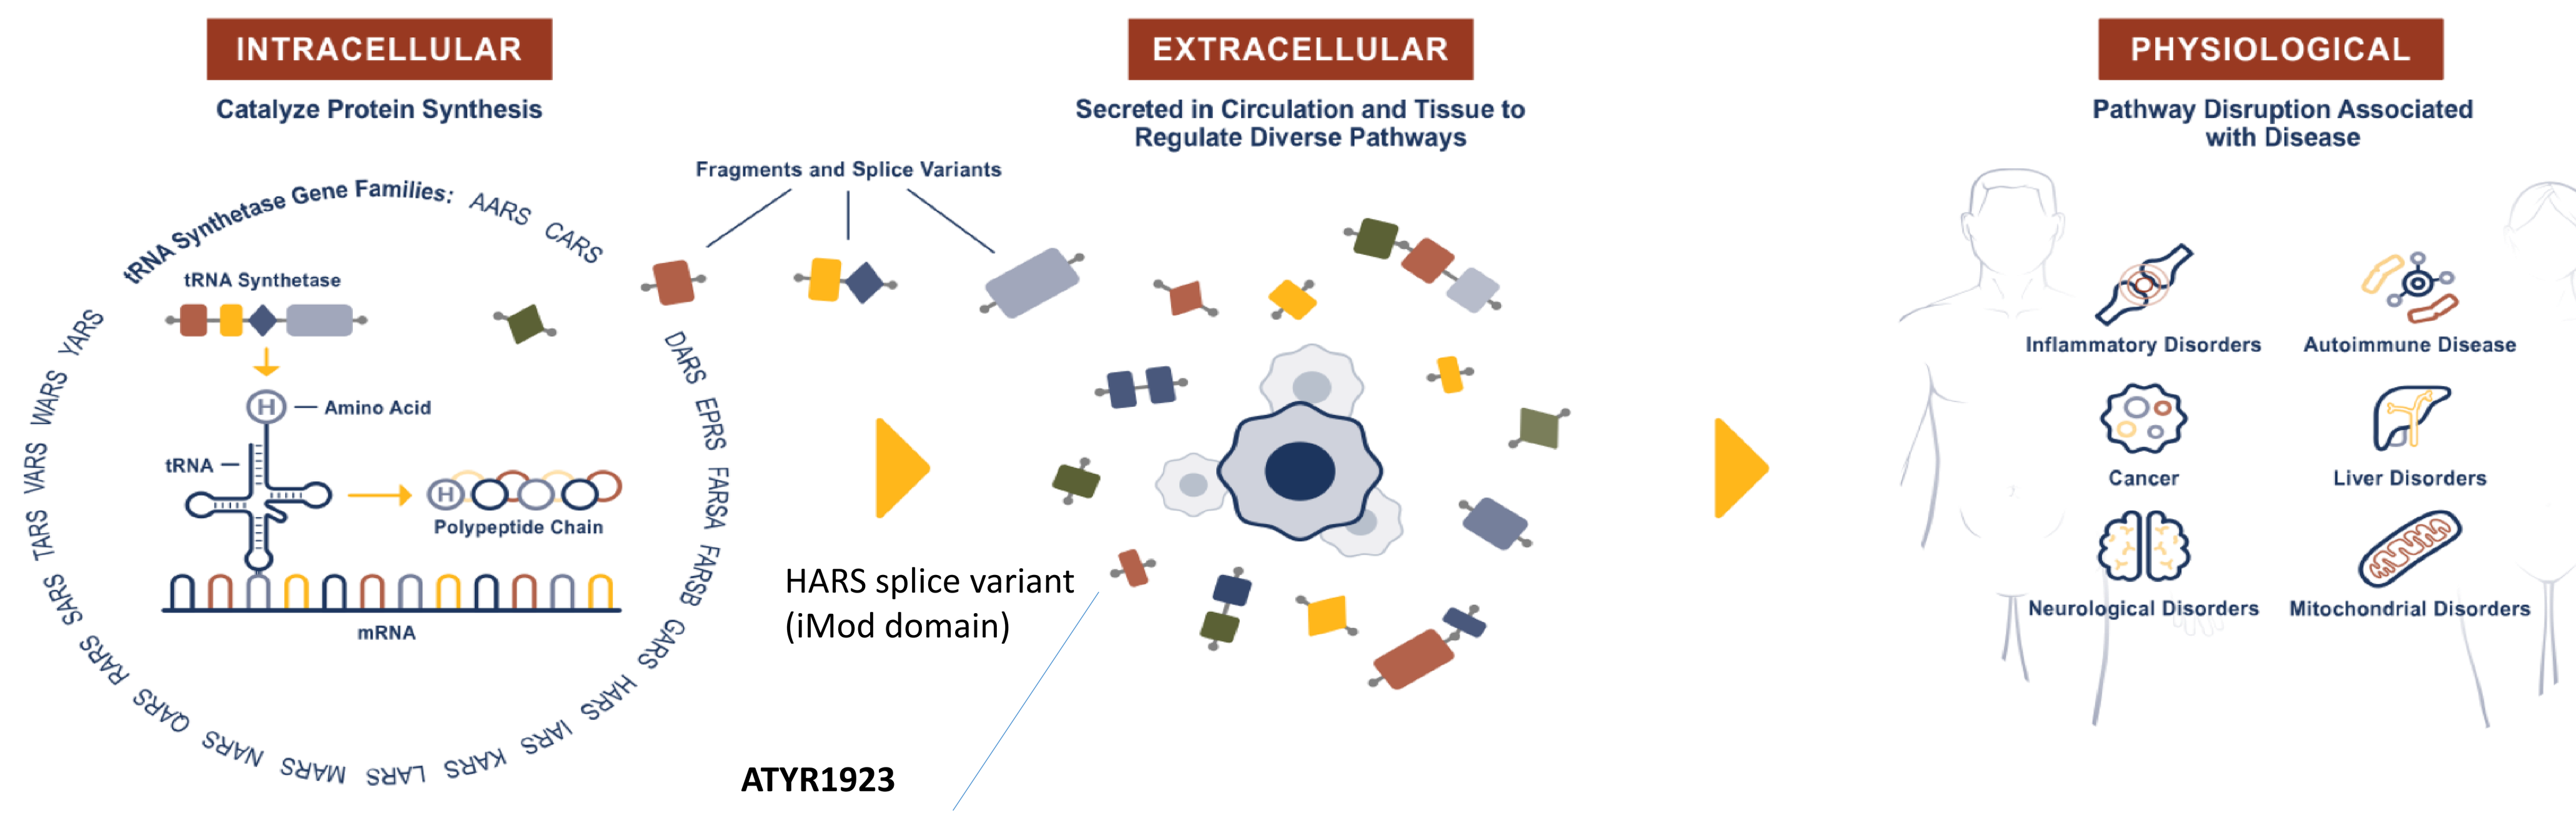
INTRACELLULAR
Catalyze Protein Synthesis
tRNA Synthetase Gene Families:
tRNA Synthetase
Amino Acid
tRNA
Polypeptide Chain
mRNA
EXTRACELLULAR
Secreted in Circulation and Tissue to Regulate Diverse Pathways
Fragments and Splice Variants
HARS splice variant (iMod domain)
PHYSIOLOGICAL
Pathway Disruption Associated with Disease
Inflammatory Disorders
Autoimmune Disease
Cancer
Liver Disorders
Neurological Disorders
Mitochondrial Disorders

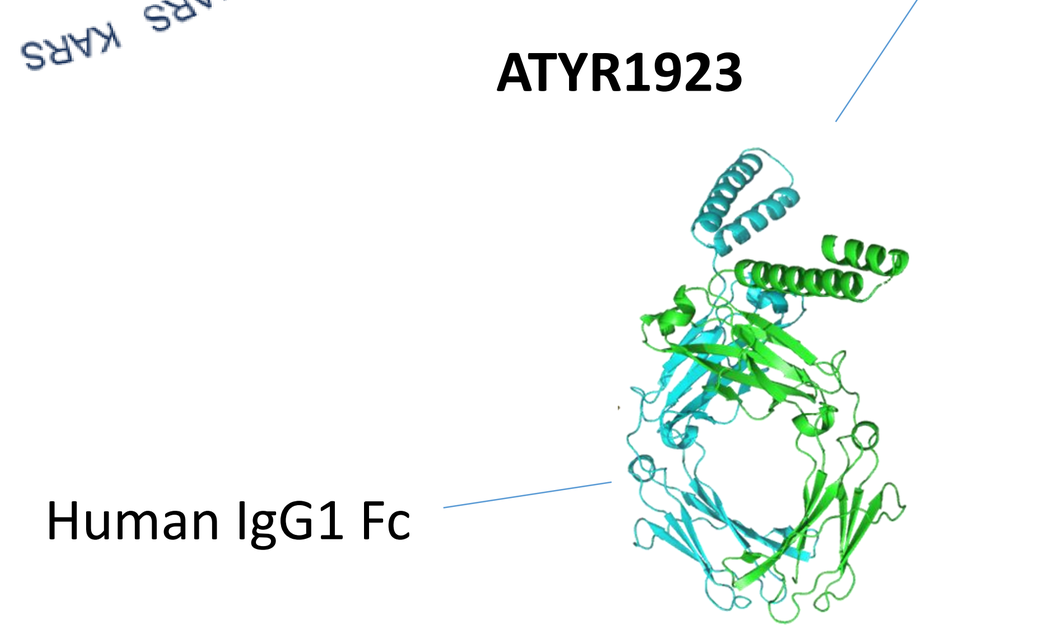
ATYR1923
Human IgG1 Fc

Therapeutic candidate: ATYR1923
Composition: iMod-Fc (HARS iMod domain fused with human IgG1 Fc)
Function: Regulate immune responses
Target disease: Interstitial lung disease
Status: Phase 1a/2b clinical study in pulmonary sarcoidosis patients ongoing

Results

Figure 2. Screening of Human Membrane Proteins at Retrogenix and Identification of NRP2 As a Specific ATYR1923-Binding Partner

A

Expression vectors encoding >4500 full-length human membrane proteins and GFP controls were arrayed

HEK-293 cells grown over the expression vectors, allowing expression of >4500 human membrane proteins in distinct slide locations

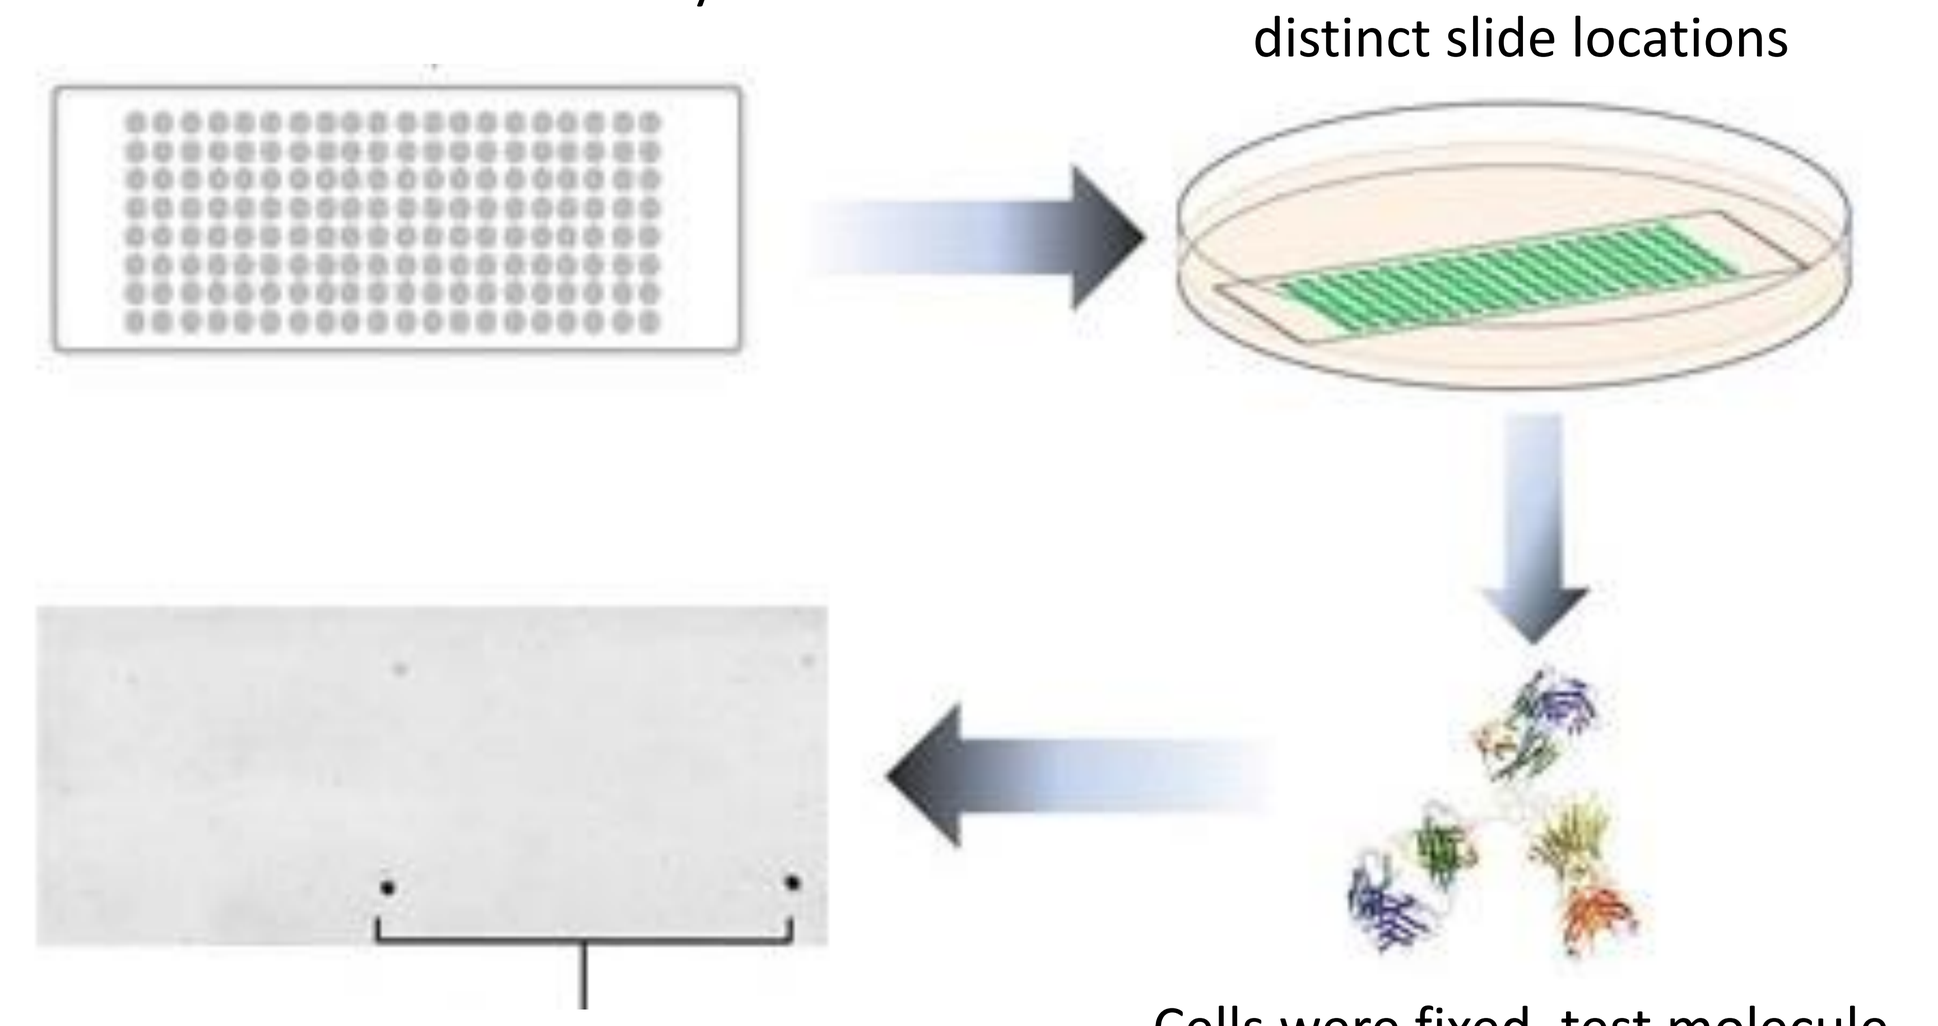

Binding partner identified using fluorescent detection.

Cells were fixed, test molecule (ATYR1923 pre-complexed with AF647 labeled anti-human Fc) was applied

B

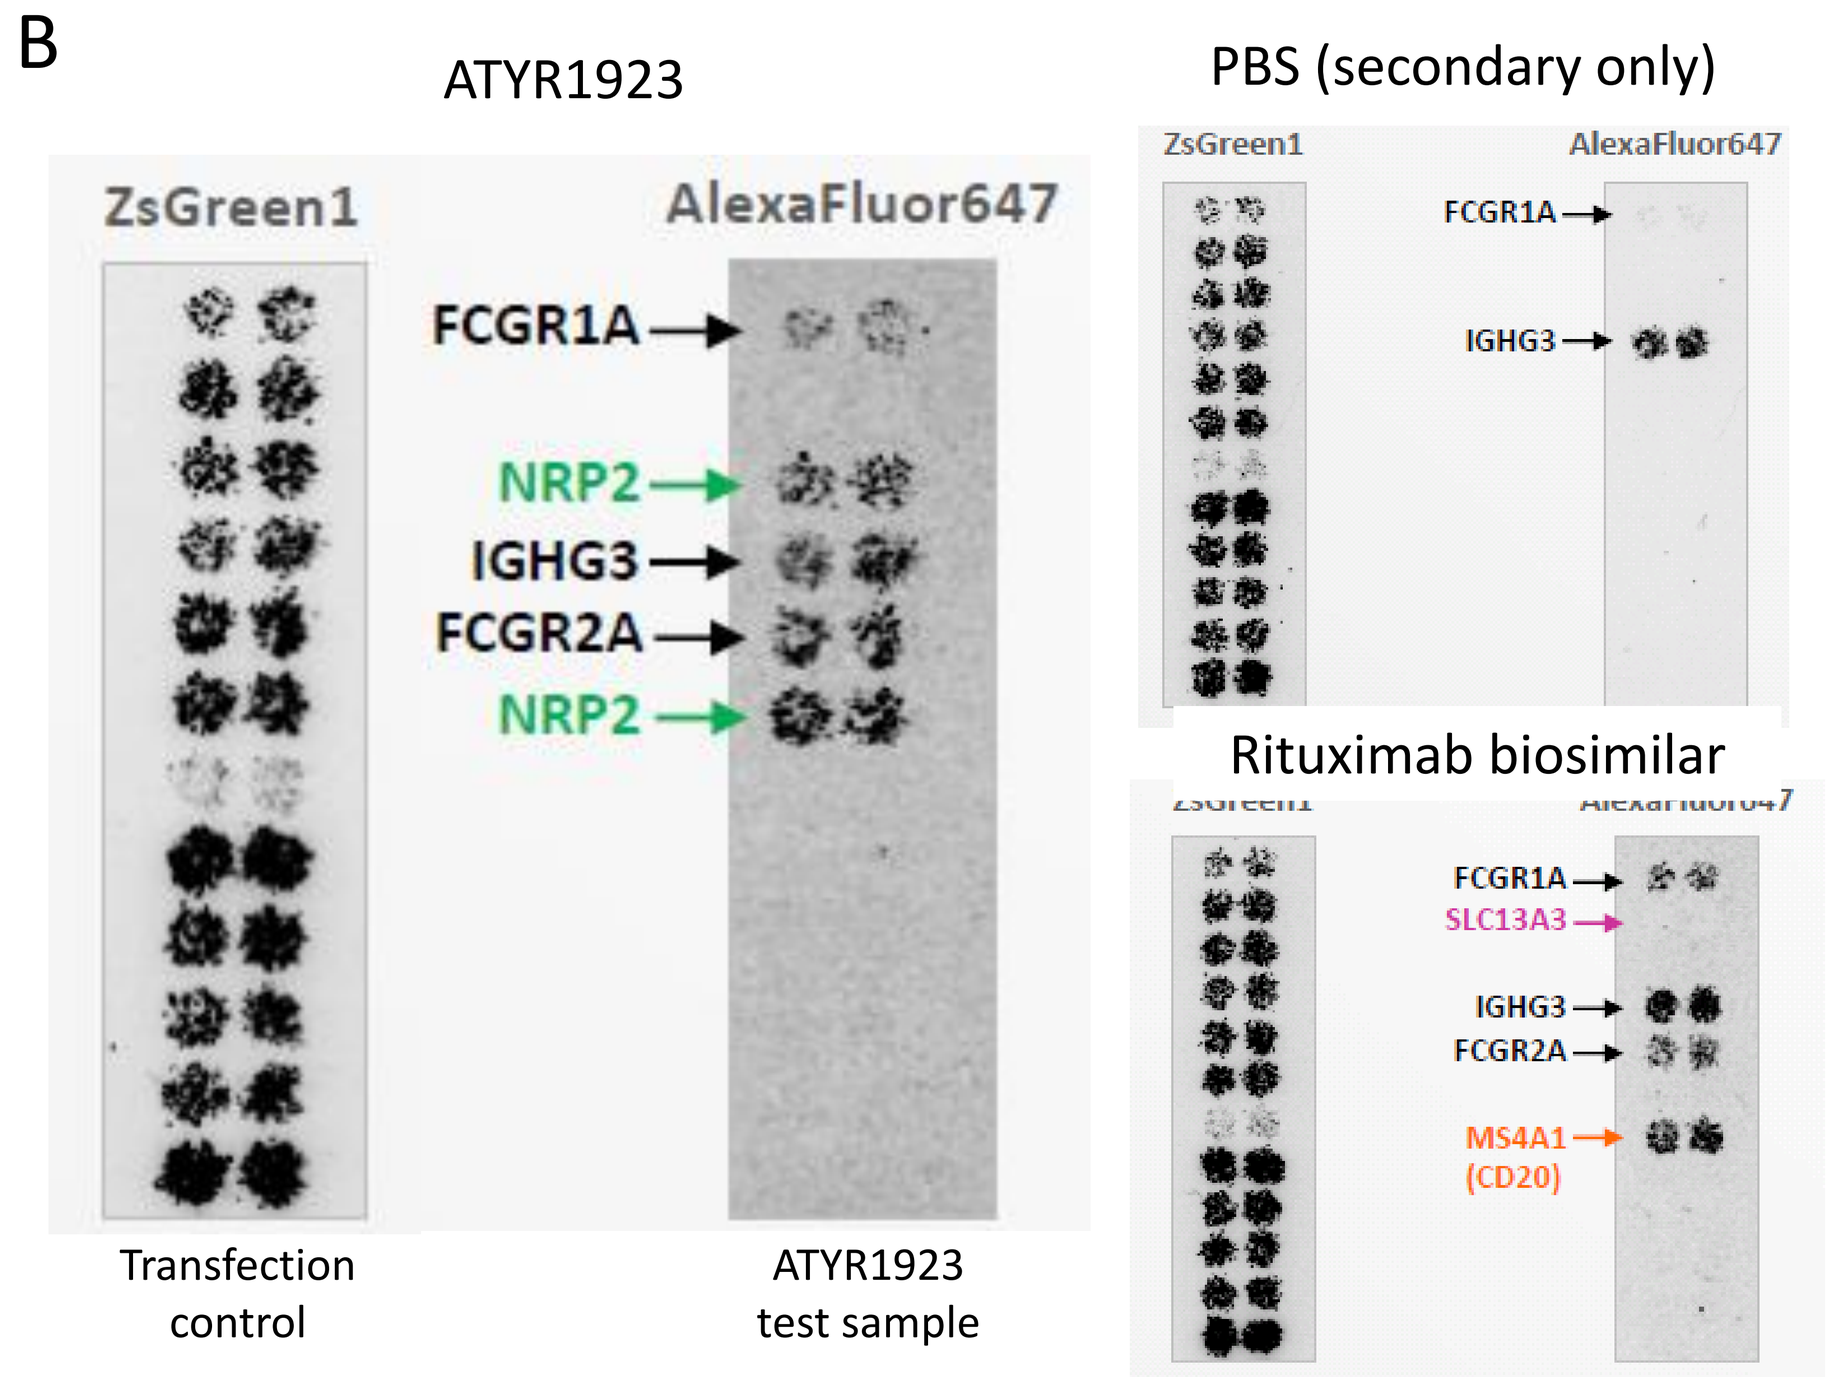
ATYR1923
ZsGreen1
AlexaFluor647
FCGR1A
NRP2
IGHG3
FCGR2A
NRP2
Transfection control
ATYR1923 test sample
PBS (secondary only)
Rituximab biosimilar
SLC13A3
MS4A1 (CD20)

NRP2 (both 2 isoforms NRP2a and NRP2b) were specific hits for ATYR1923

PBS (secondary only) was the negative control, while Rituximab (known to bind CD20) was a positive control

- Regtrogenix screened >4500 human membrane proteins (Methodology shown in Fig. 2A).
- NRP2 was identified as a specific binding partner for ATYR1923 (Fig. 2B).
- Notably, no binding was detected for the related and structurally similar receptor NRP1.

Figure 3. ATYR1923 Specifically Binds to NRP2 but Not NRP1

A

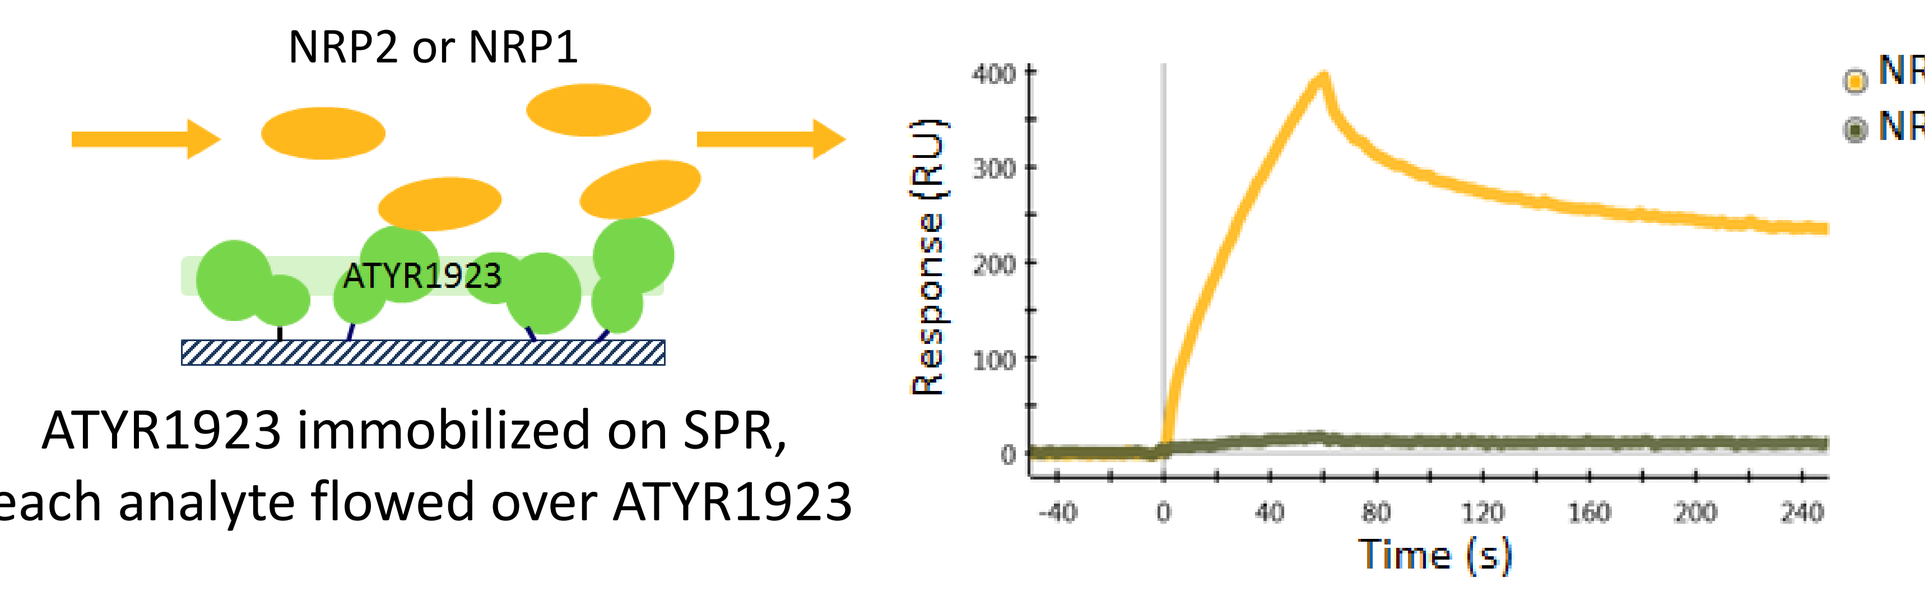
NRP2 or NRP1
ATYR1923
Response (RU)
Time (s)
NRP2
NRP1

ATYR1923 immobilized on SPR, each analyte flowed over ATYR1923

B

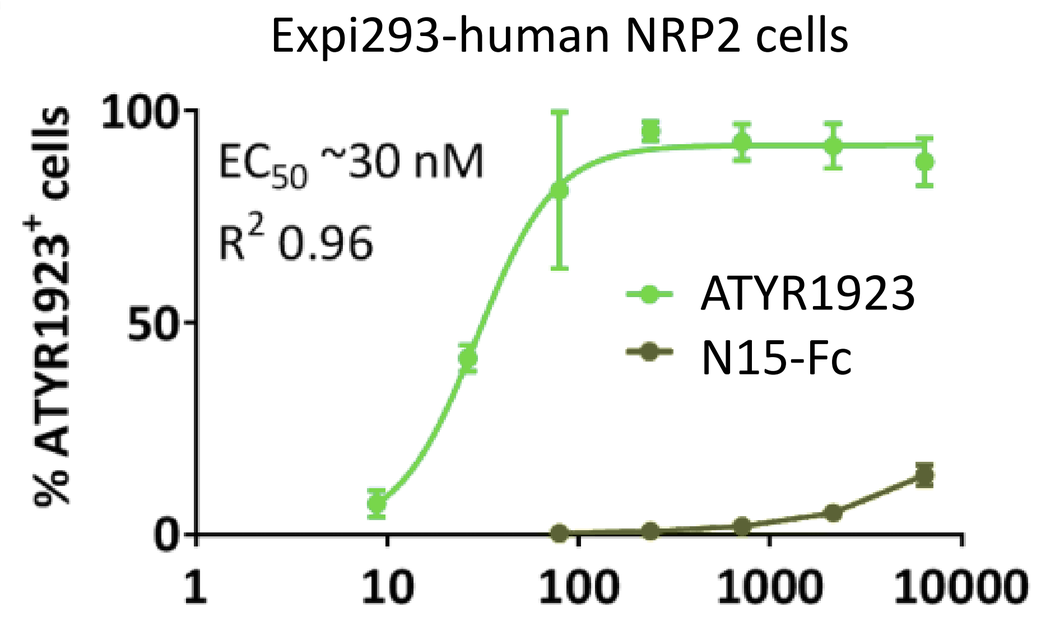
Expi293-human NRP2 cells
% ATYR1923+ cells
EC50 ~30 nM
R2 0.96
ATYR1923
N15-Fc
Protein conc (nM)

Cell surface staining by ATYR1923 or control N15-Fc, followed with flow cytometry analysis

-N15: N-terminal 15aa of HARS

- ATYR1923 binds to NRP2 but not NRP1 by SPR measurement (Fig. 3A)
- ATYR1923 binds to NRP2 overexpressed on Expi293 cells (Fig. 3B).
- NRP2 is a pleiotropic receptor with important functional roles in cardiovascular, nervous and immune systems (Guo HF et al., 2015)

Figure 4. ATYR1923 Binds to Cells That Express Endogenous NRP2

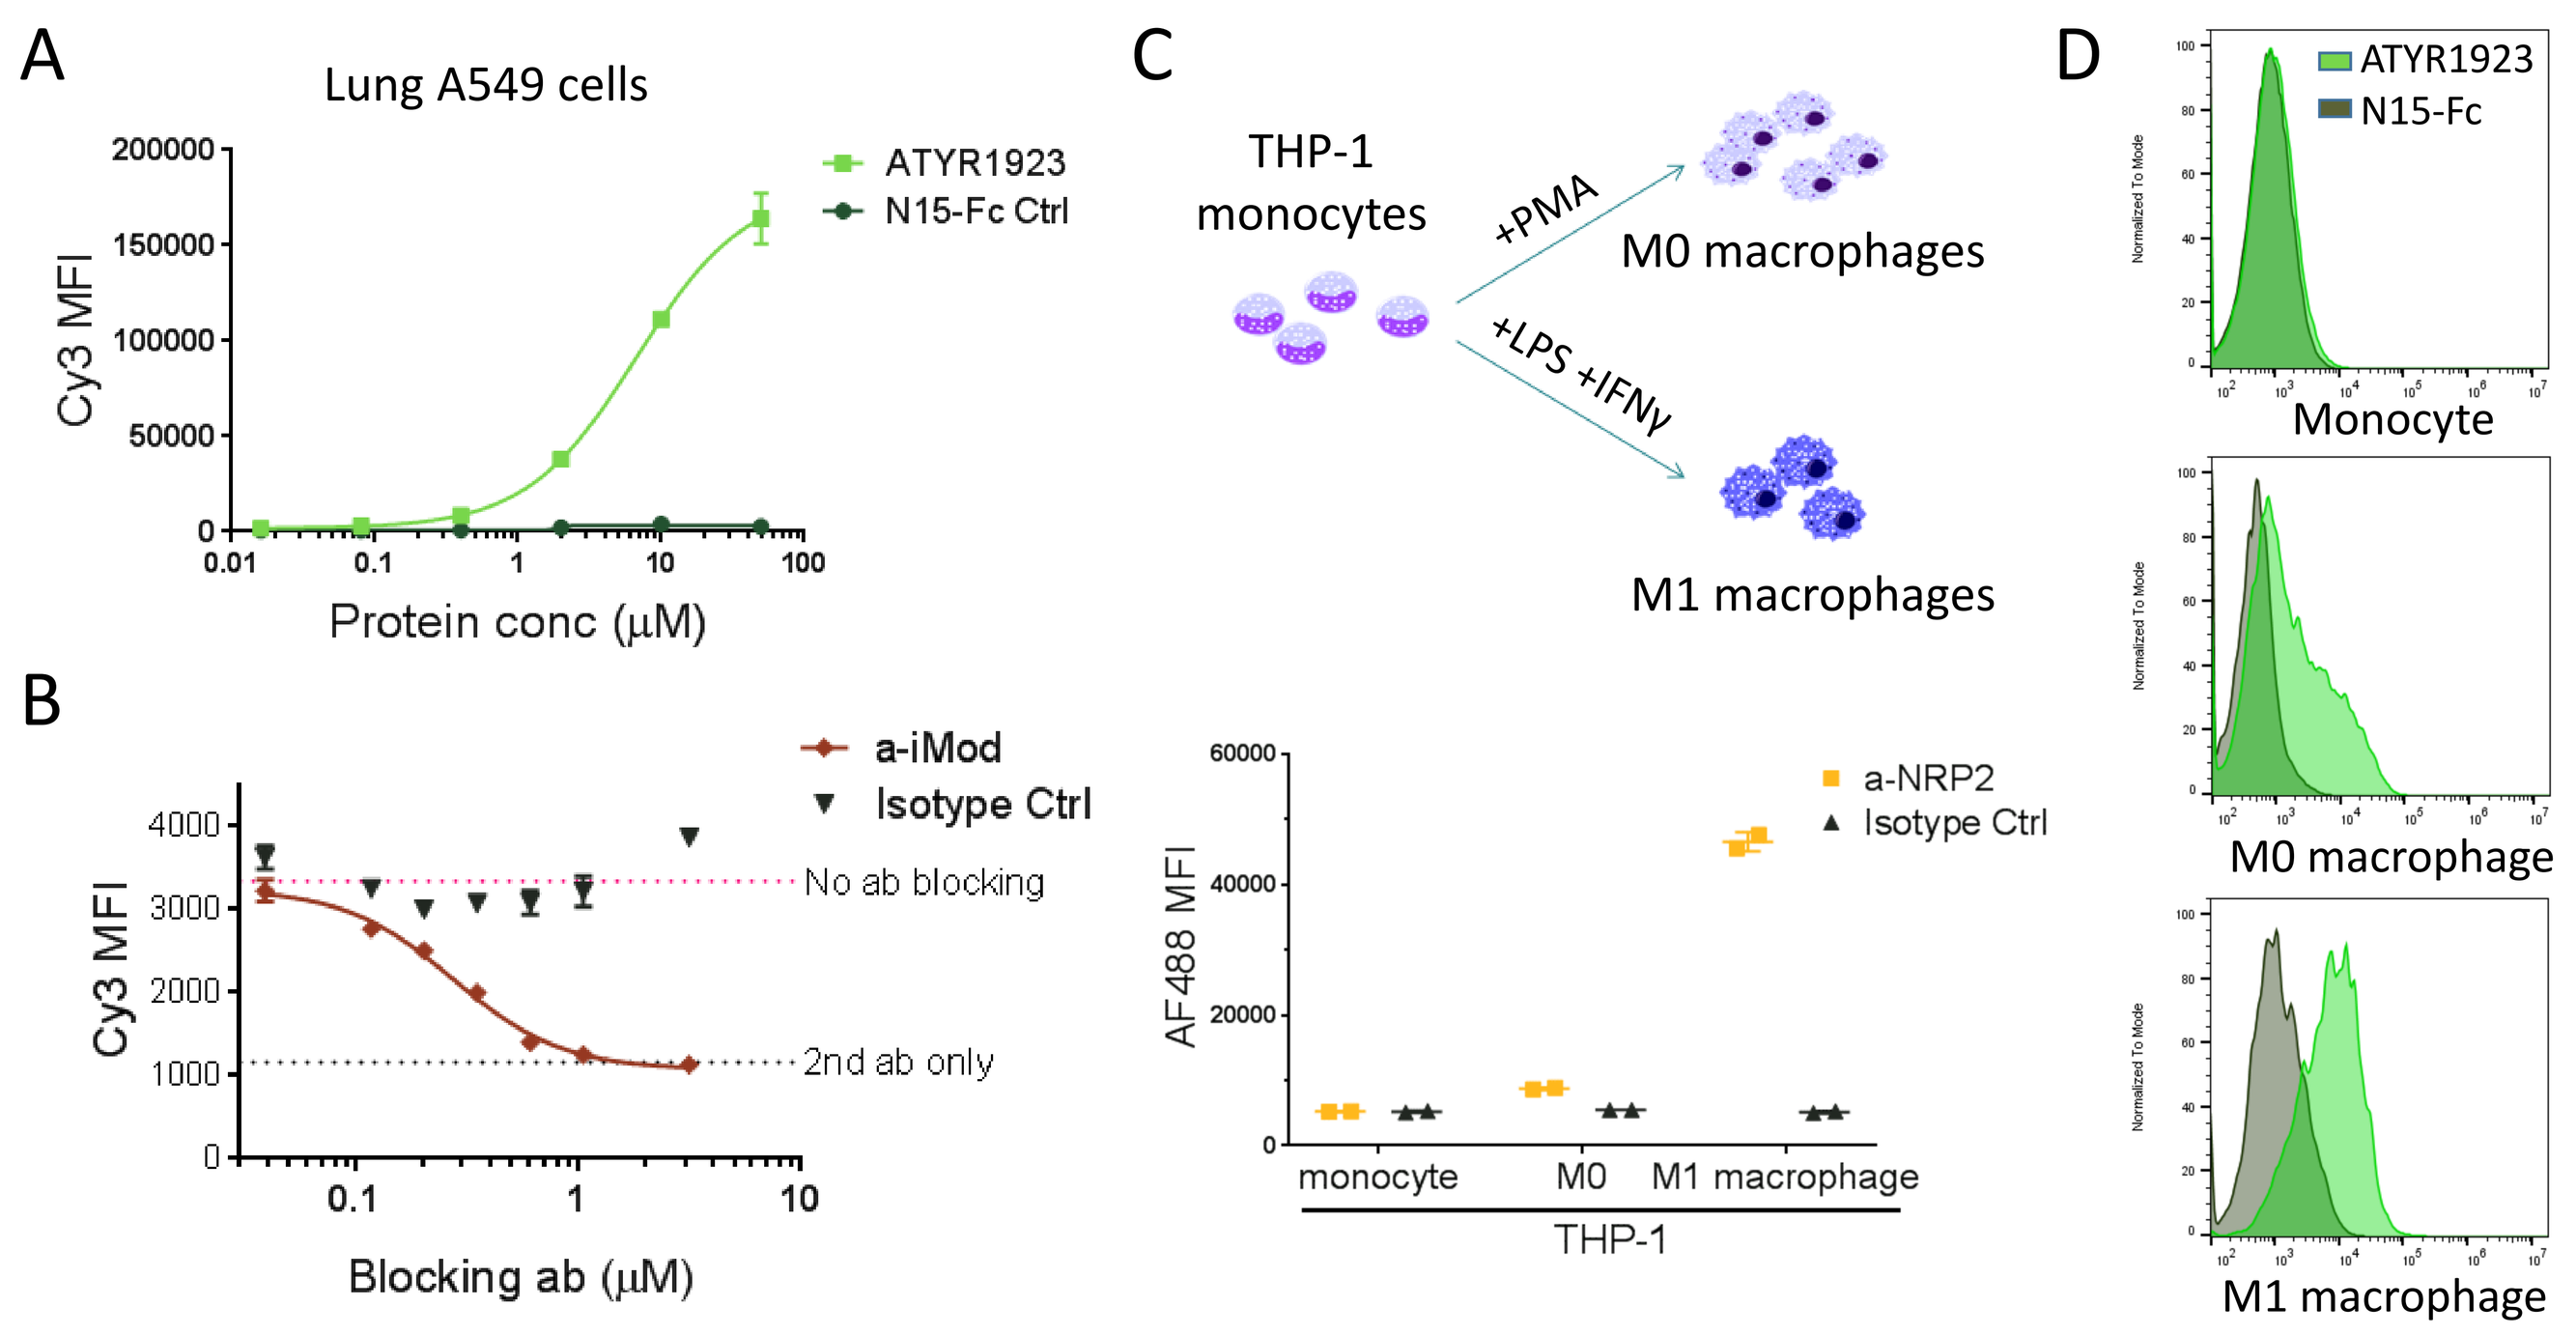
A
Lung A549 cells
Cy3 MFI
ATYR1923
N15-Fc Ctrl
Protein conc (μM)
B
a-iMod
Isotype Ctrl
No ab blocking
2nd ab only
Blocking ab (μM)
C
THP-1 monocytes
+PMA
M0 macrophages
+LPS +IFNγ
M1 macrophages
AF488 MFI
a-NRP2
Isotype Ctrl
monocyte
M0
M1 macrophage
THP-1
D
ATYR1923
N15-Fc
Monocyte
M0 macrophage
M1 macrophage

- ATYR1923 binds to lung A549 cells known to express endogenous NRP2 (Fig. 4A), which is entirely blocked by an anti-iMod antibody (Fig. 4B)
- Cell surface NRP2 level was up-regulated by the THP-1 differentiation, especially on M1 macrophages (Fig. 4C).
- ATYR1923 binds to THP-1 macrophages, which is in accordance with the up-regulation of surface NRP2 level (Fig. 4D)

Figure 5. Delineation of ATYR1923 Binding Region on NRP2 at a2b1b2 Domains

A

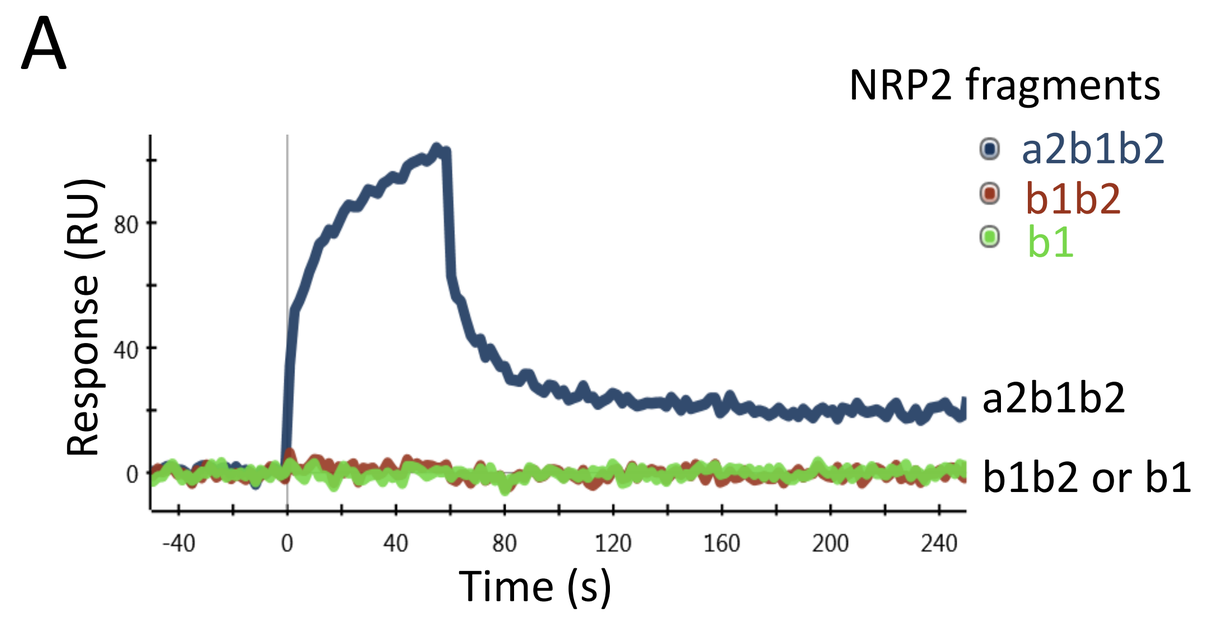
NRP2 fragments
a2b1b2
b1b2
b1
Response (RU)
Time (s)
a2b1b2
b1b2 or b1

ATYR1923 immobilized on SPR via NHS chemistry, 150 nM of each analyte protein was flowed over ATYR1923

B

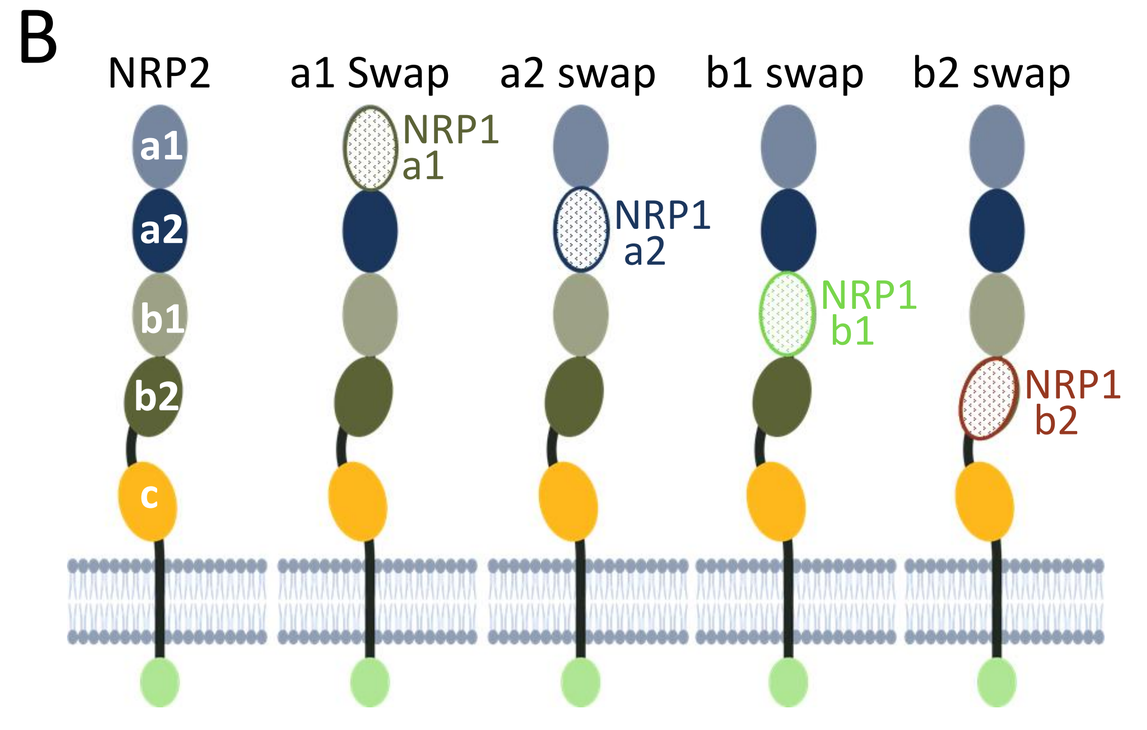
NRP2
a1 Swap
a2 swap
b1 swap
b2 swap
NRP1 a1
NRP1 a2
NRP1 b1
NRP1 b2

Human NRP2 extracellular domains were individually swapped with the corresponding, structurally similar domain of NRP1

C

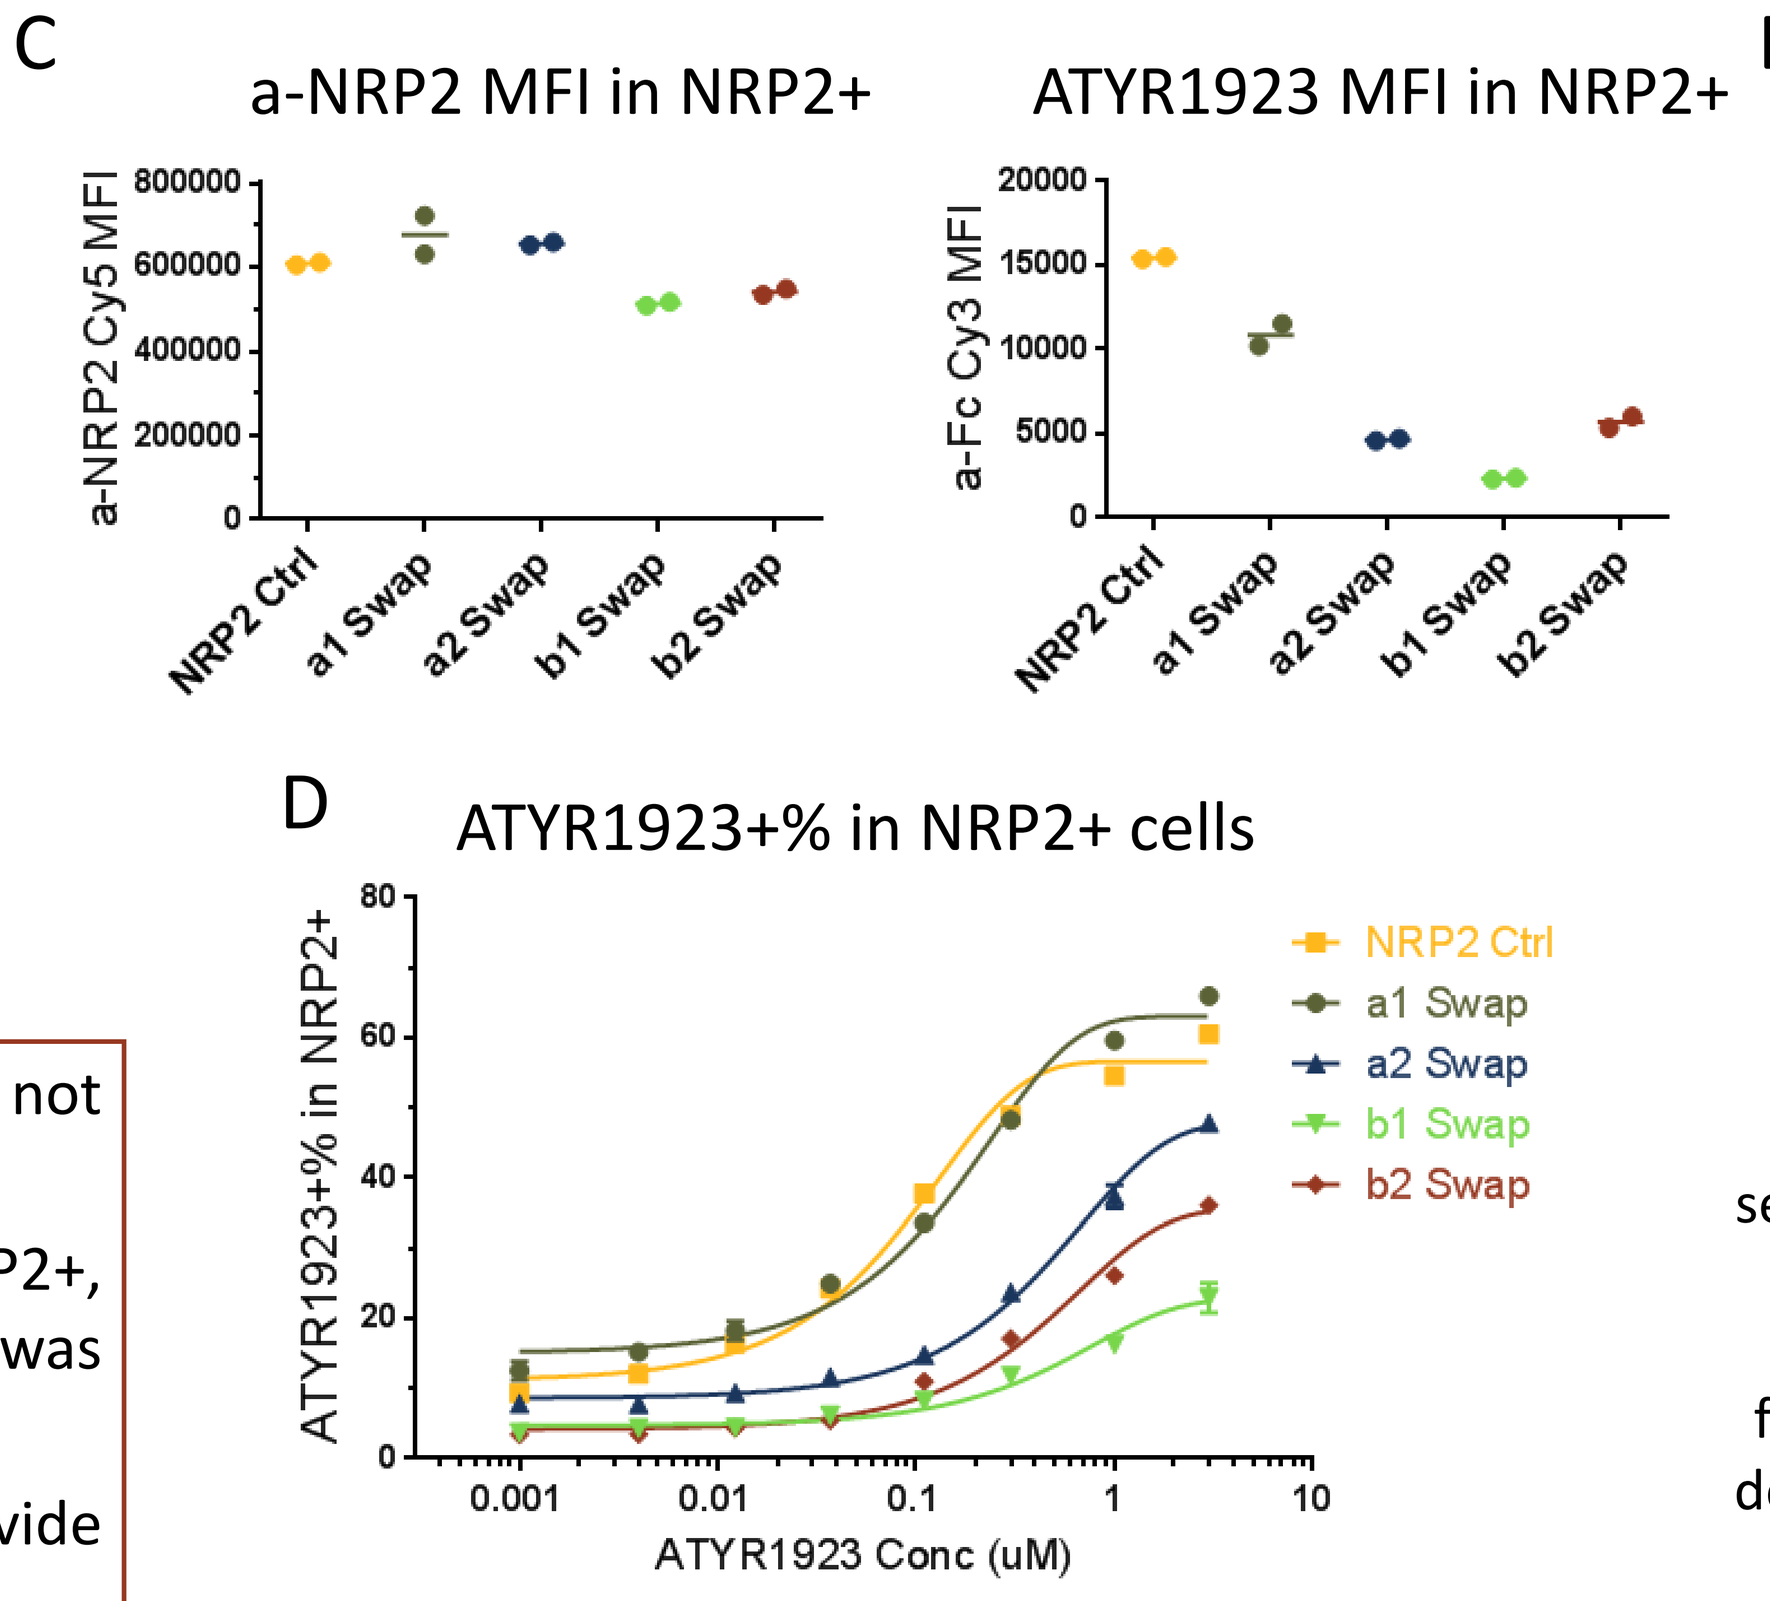
a-NRP2 MFI in NRP2+
ATYR1923 MFI in NRP2+
a-NRP2 Cy5 MFI
a-Fc Cy3 MFI
NRP2 Ctrl
a1 Swap
a2 Swap
b1 Swap
b2 Swap
D
ATYR1923+% in NRP2+ cells
ATYR1923+% in NRP2+
ATYR1923 Conc (uM)

Cell surface staining by an a-NRP2 antibody targeting the C-domain of NRP2, as well as ATYR1923 detected by a-Fc Cy3 followed with flow analysis

E

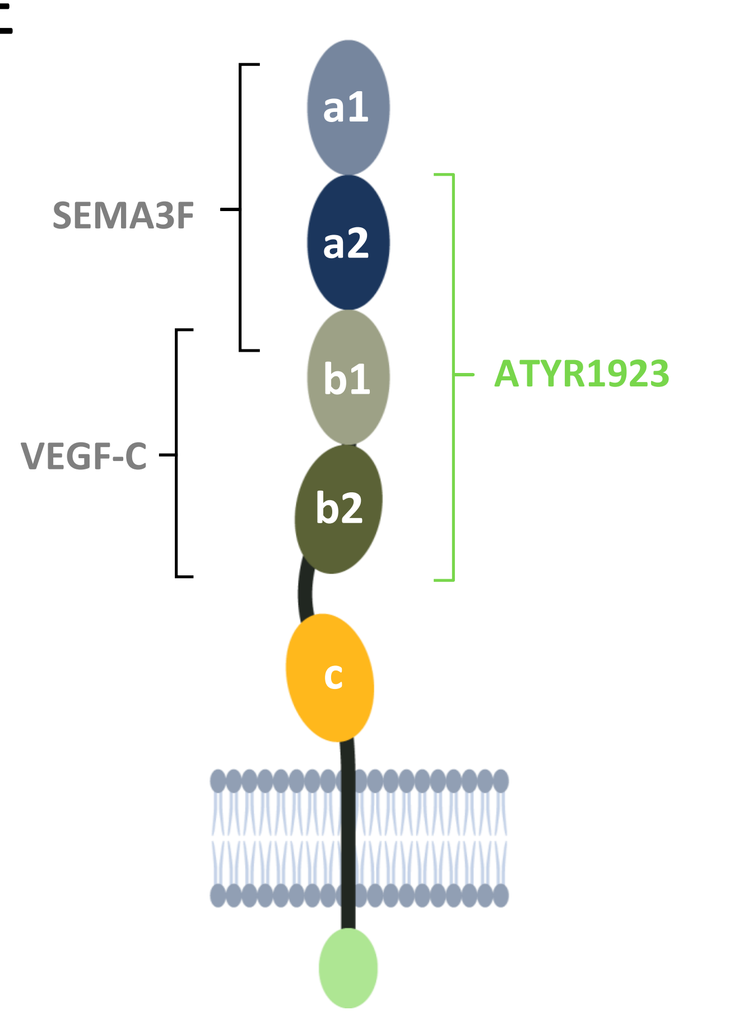
SEMA3F
VEGF-C
ATYR1923
a1
a2
b1
b2
c

VEGF (e.g. VEGF-C) and semaphorins (e.g. Sema3F) are known ligands of NRP2 that influence important cellular functions including lymphatic development and regulation of inflammatory responses

- NRP2 a2b1b2 fragment binds to ATYR1923, but b1b2 or b1 domain alone does not interact with ATYR1923 by SPR analysis (Fig 5A, while b1b2 binds VEGF-C well).
- Cells overexpressing the wildtype or domain-swapped NRP2 were gated as NRP2+, which showed a comparable NRP2 surface level, while ATYR1923 binding was considerably reduced by the a2, b1 or b2 domain swap (Fig. 5B & 5C)
- NRP2 b1 is the key domain for binding, while a2 and b2 domains likely also provide contacts with ATYR1923 (Fig. 5C & 5D)
- NRP2 a2b1b2 domains encompass binding contacts for VEGF and semaphorins (Fig. 5E)

Figure 6. Complex Structure of iMod with a Blocking Antibody

A

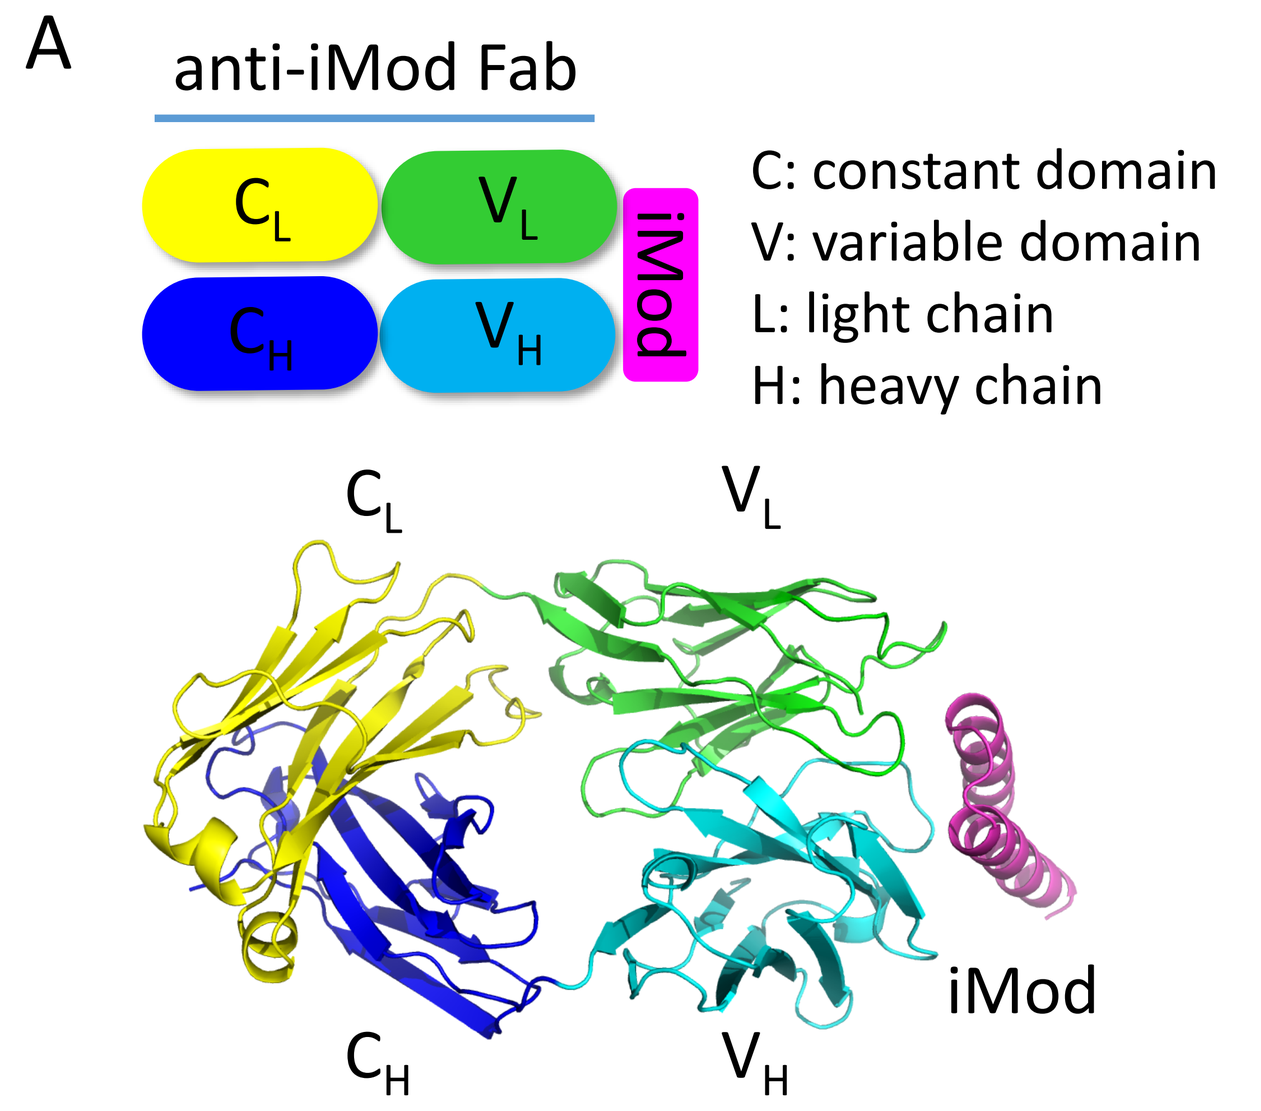
anti-iMod Fab
C_L
V_L
C_H
V_H
iMod
C: constant domain
V: variable domain
L: light chain
H: heavy chain

Schematic and crystal structure of iMod in complex with the antigen-binding fragment (Fab) of an anti-iMod antibody that blocks iMod binding to NRP2 (Fig . 4B).

B

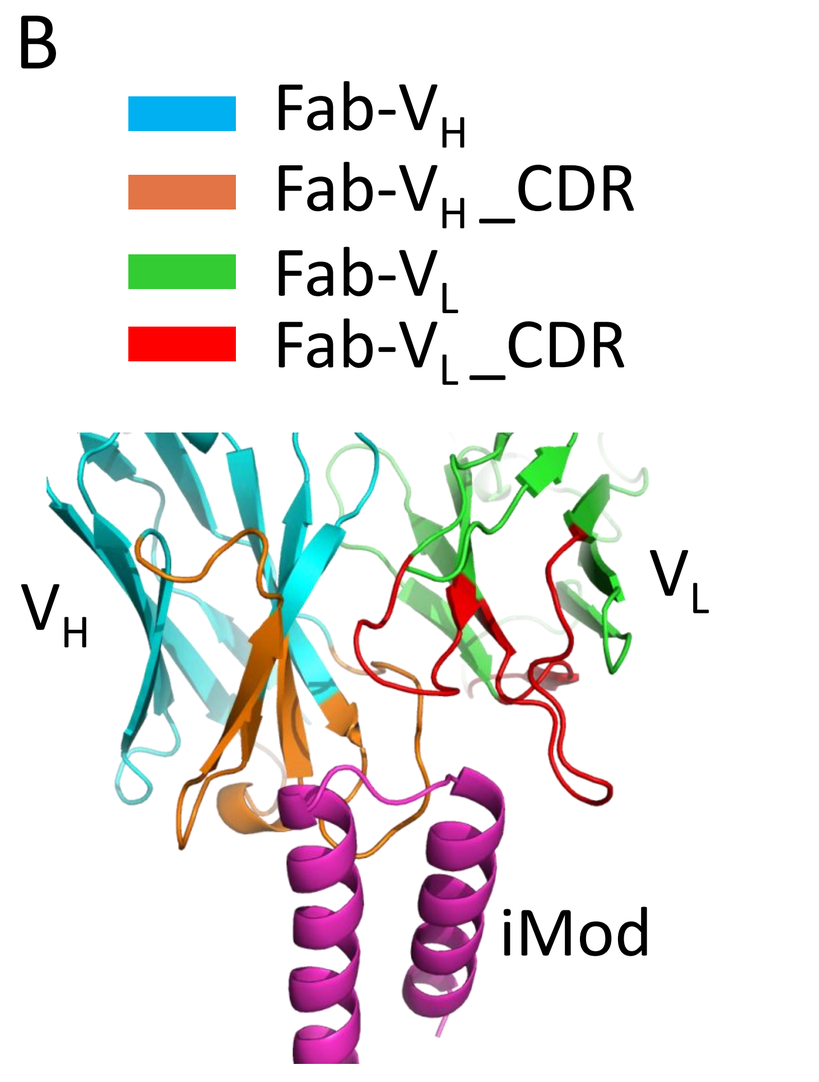
Fab-V_H
Fab-V_H_CDR
Fab-V_L
Fab-V_L_CDR
V_H
V_L
iMod

Enlarged contacting region of the complex (CDR: complementarity-determining region)

- iMod domain of ATYR1923 adopts a helix-turn-helix structure
- The middle loop of iMod interacts with NRP2 (Fig. 6A & 6B)

Conclusions

- **NRP2, a pleiotropic cell surface receptor that is expressed and may play a key role in inflammatory responses, was identified as a specific binding target for ATYR1923**
- **NRP2 extracellular a2b1b2 domains encompass binding sites for ATYR1923**
- **ATYR1923 binds to overexpressed and endogenous NRP2 on human cell surface**
- **The middle loop region of iMod domain in ATYR1923 interacts with NRP2**

References

1. Burkett C, et al. (2019) ATYR1923 modulates the inflammatory response in experimental models of interstitial lung disease. *American Thoracic Society*, Poster #10533
2. Sharma NS, et al. (2019) The N-terminal domain of HARS is a novel NRP2 ligand and can regulate NRP2-dependent macrophage function. *American Association for Cancer Research*.
3. Ogilvie K, et al. (2018) Preclinical characterization of ATYR1923 (iMod.Fc), an immune-modulatory therapeutic with potentially broad application in interstitial lung diseases. *American Thoracic Society*, Poster #A1064/917
4. Guo HF, Vander Kooi CW. (2015) Neuropilin Functions as an Essential Cell Surface Receptor. JBC 290: 29120-29126.
5. Lo WS, et al. (2014) Human tRNA synthetase catalytic nulls with diverse functions. *Science.* 345(6194):328-32.
6. Zhou JJ, et al. (2014) Secreted histidyl-tRNA synthetase splice variants elaborate major epitopes for autoantibodies in inflammatory myositis. *J Biol Chem,* 289(28):19269-75.

Acknowledgement

- Scientists at Retrogenix™ who conducted the screening experiment for ATYR1923.
- This work was supported by aTyr Pharma Inc.