ATYR2810, a Neuropilin-2 antibody, selectively blocks the NRP2/VEGFR signaling axis and sensitizes aggressive cancers to chemotherapy

Yeeting E. Chong[1], Zhiwen Xu[1], Hira Lal Goel[2], Alison G. Barber[1], Christoph Burkart[1], Luke Burman[1], Kaitlyn Rauch[1], Justin Rahman[1], Arthur M. Mercurio[2], Leslie A. Nangle[1*]

1. aTyr Pharma, 2. University of Massachusetts Medical School, *Contact: lnangle@atyrpharma.com

Abstract

INTRODUCTION: Neuropilin-2 (NRP2) is a cell surface receptor that can interact with multiple ligands and co-receptors to influence their functional roles, among which are vascular endothelial growth factors (VEGFs). High NRP2 expression or concurrent NRP2 and VEGF expression is associated with worse outcomes in many forms of cancer. For example, a growing body of evidence has shown that the increased expression of NRP2 is associated with aggressive breast tumors, including triple-negative breast cancer (TNBC), and that NRP2/VEGF signaling contributes to chemotherapy resistance and tumor recurrence. However, a major challenge to exploit NRP2 as a therapeutic target against cancer has been the lack of highly selective anti-human NRP2 monoclonal antibodies (mAbs) that specifically block NRP2/VEGF signaling. aTyr Pharma has generated a panel of high quality, high affinity, NRP2 mAbs that are being developed for the clinical management of diseases that involve NRP2 signaling, including aggressive solid tumors.

RESULTS: We have characterized the aTyr-generated NRP2 mAbs in binding, ligand blocking and epitope mapping studies. A specific anti-human NRP2 mAb, ATYR2810, was shown to bind to the b1 domain of NRP2, which encompasses the VEGF binding site. Mutational studies indicated that ATYR2810 binds to the opening of the VEGF binding pocket, thus blocking ligand access. ATYR2810 selectively blocked VEGF-induced dimerization of NRP2 with VEGF receptors, but did not affect Semaphorin 3F (Sema3F)-induced dimerization of NRP2 and PlexinA1, demonstrating the specificity of ATYR2810 for blocking the NRP2/VEGF pathway.

In certain *in vitro* and *in vivo* cancer models, ATYR2810 administered in combination with widely used anti-cancer therapeutics, including chemotherapy, increased the anti-tumor effects of the therapeutic agents. To identify biomarkers that can predict the tumor response to ATYR2810, we further characterized TNBC cell lines and patient-derived organoids (PDOs) that were responsive or non-responsive to ATYR2810 treatment *in vitro*, as well as TNBC cell line xenografts and TNBC patient-derived xenografts (PDXs) *in vivo*. In some responsive TNBC models, ATYR2810 down-regulated a number of critical epithelial-mesenchymal transition (EMT) and cancer stem cells (CSC) genes, which may be a mechanism that mediates its anti-tumor effects.

CONCLUSIONS: These results demonstrate the selectivity of ATYR2810 in blocking NRP2/VEGF signaling, and the efficacy of ATYR2810 in combination with anti-cancer therapeutics in *in vitro* and *in vivo* cancer models. The data suggest that in certain tumors, ATYR2810, by blocking NRP2 function, may inhibit EMT and CSC properties thereby rendering the tumors more sensitive to the treatment regimens. Therefore, the targeting of NRP2/VEGF signaling by ATYR2810 may provide a potential therapeutic option with improved efficacy in aggressive cancers.

Introduction

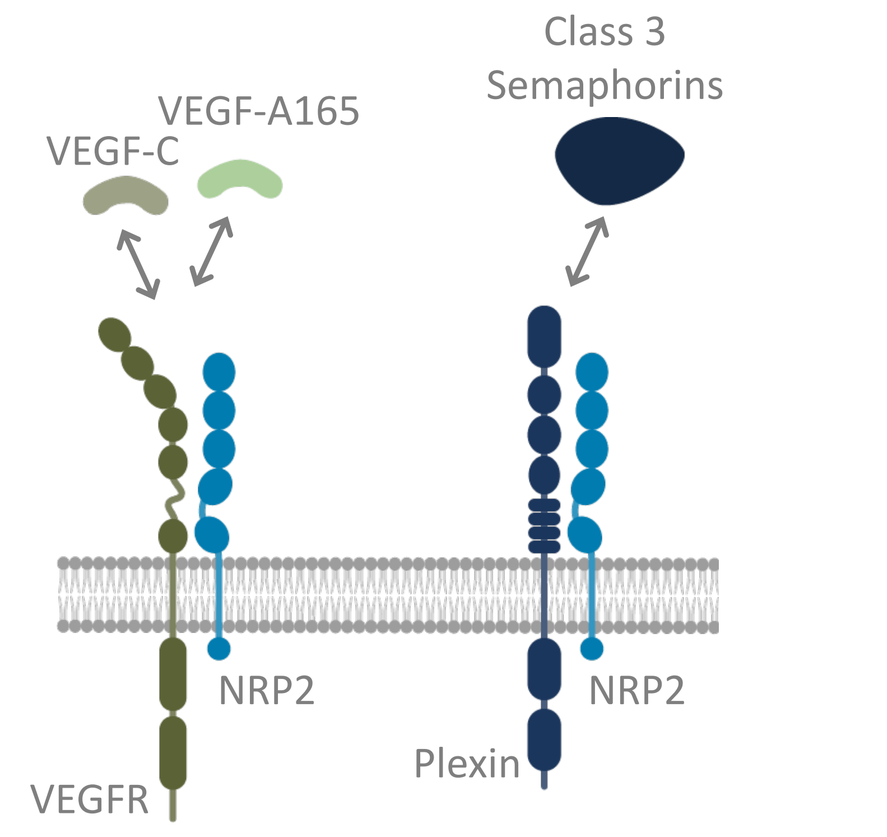
Class 3
Semaphorins
VEGF-A165
VEGF-C
NRP2
NRP2
Plexin
VEGFR

- NRP2 is a multi-functional receptor that exerts its cellular effects by dimerizing with co-receptors, the most well characterized of which are VEGFRs and Plexins, upon binding of ligands such as VEGF and Class 3 Semaphorins.
- High expression of NRP2 has been correlated with poor prognosis in several cancer types, including TNBCs [Ref1].
- NRP2/VEGF signaling has been shown to promote stem-like traits in breast cancer cells, and is associated with proliferation and survival of CSCs [Ref2].
- NRP2 expression is enriched in TNBC PDOs that are chemo-resistant as compared to patient-derived organoids that are chemo-sensitive [Ref3].

NRP2 expression in breast carcinoma tissue

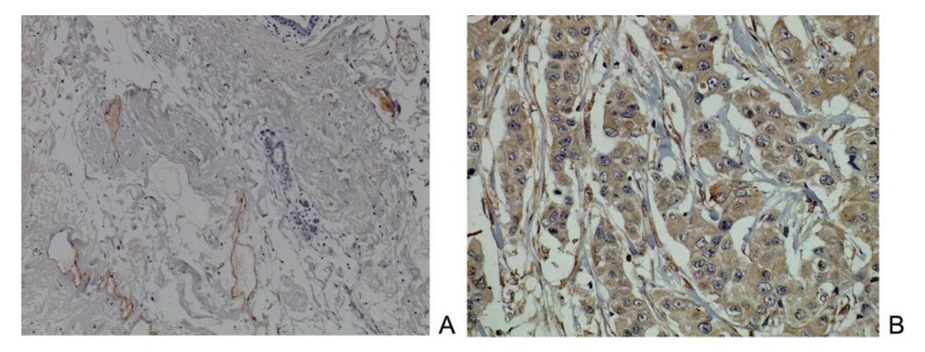

Normal breast epithelium | Breast carcinoma tissue

Results

Figure 1: ATYR2810 is a specific and high affinity antibody against NRP2

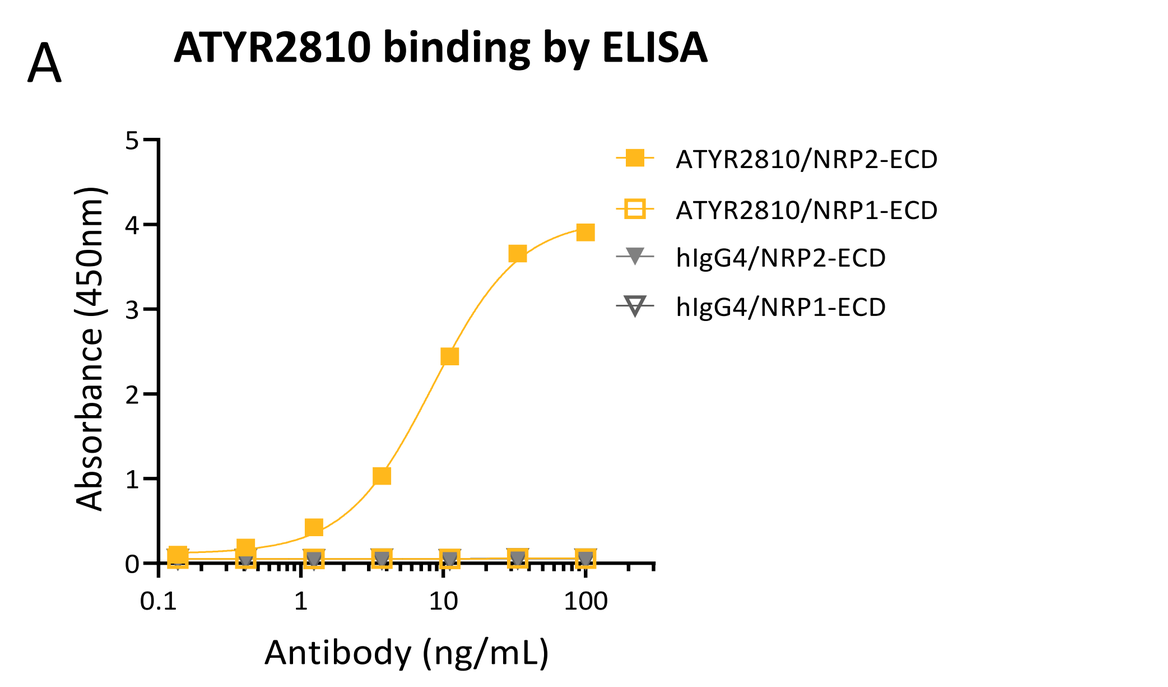
A
ATYR2810 binding by ELISA
ATYR2810/NRP2-ECD
ATYR2810/NRP1-ECD
hIgG4/NRP2-ECD
hIgG4/NRP1-ECD
Absorbance (450nm)
Antibody (ng/mL)

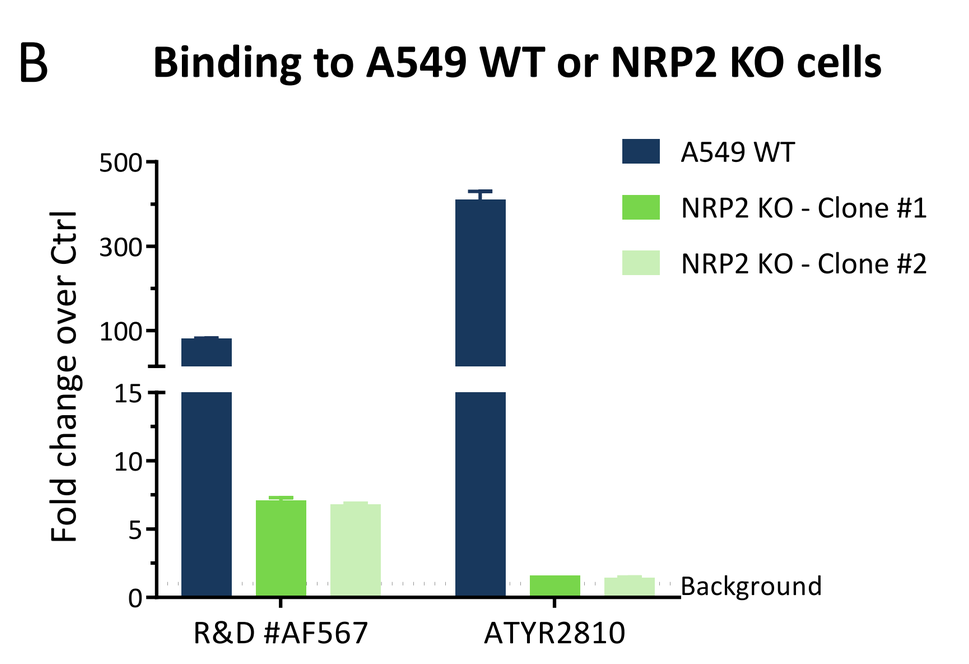
B
Binding to A549 WT or NRP2 KO cells
A549 WT
NRP2 KO - Clone #1
NRP2 KO - Clone #2
Fold change over Ctrl
Background
R&D #AF567
ATYR2810

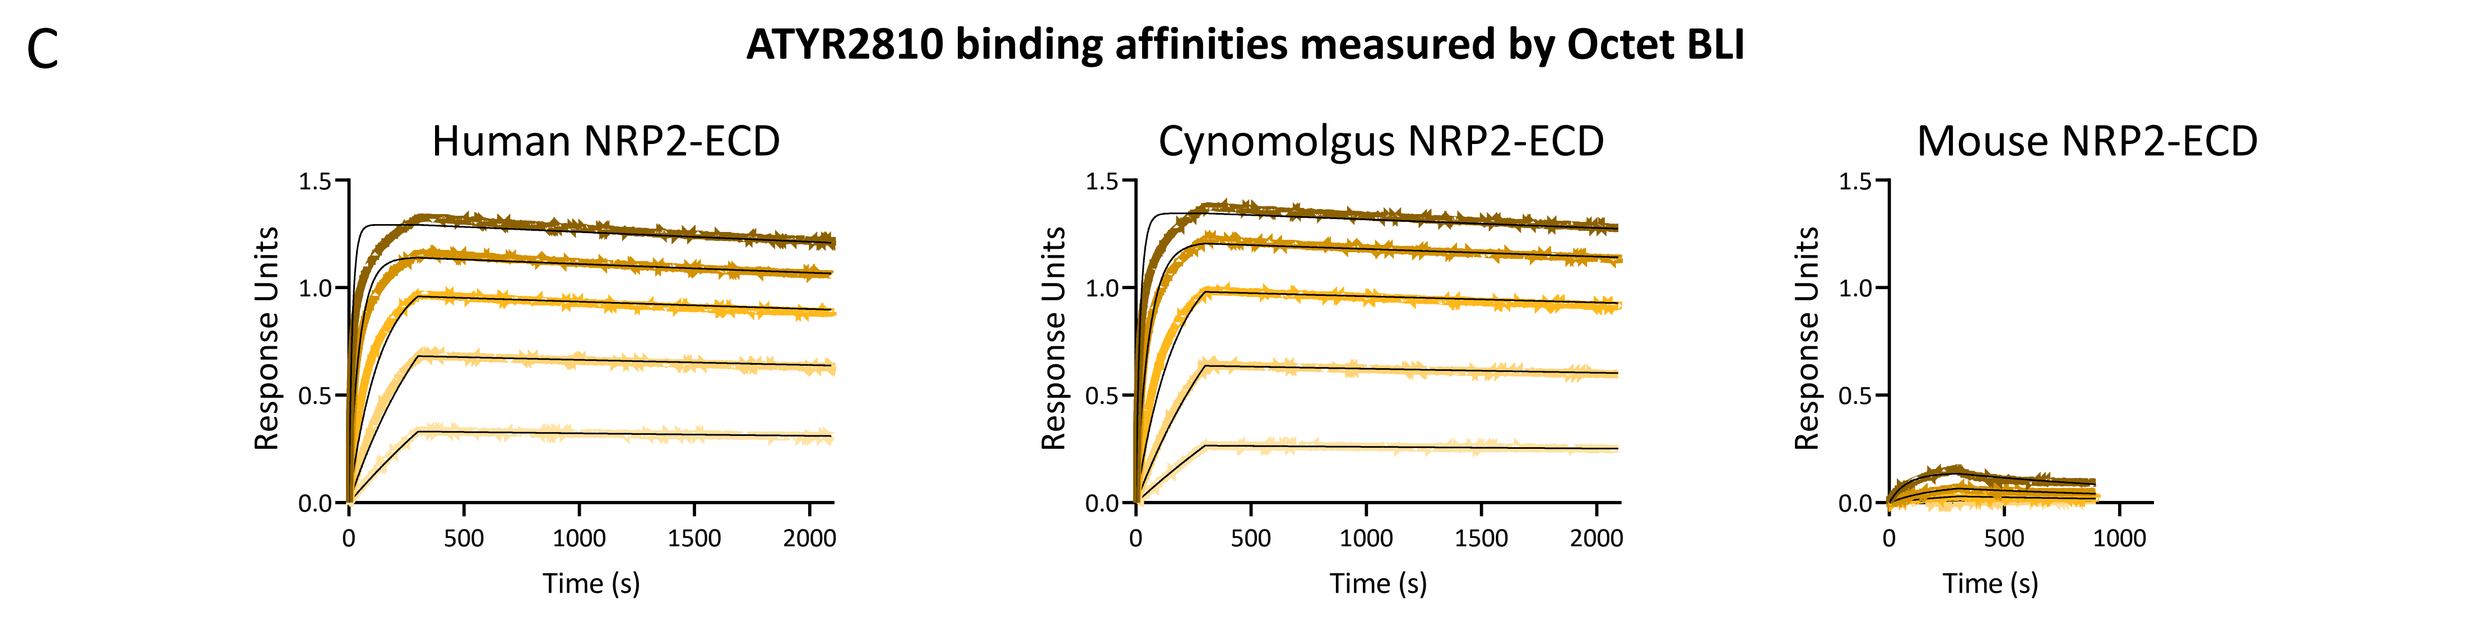
C
ATYR2810 binding affinities measured by Octet BLI
Human NRP2-ECD
Cynomolgus NRP2-ECD
Mouse NRP2-ECD
Response Units
Time (s)

	Human NRP2-ECD	Cyno NRP2-ECD	Mouse NRP2-ECD
k_a ($M^{-1}s^{-1}$)	$4.39x10^5$	$3.70x10^5$	$8.44x10^4$
k_d (s^{-1})	$3.70x10^{-5}$	$3.05x10^{-5}$	$7.68x10^{-4}$
K_D (pM)	84	87	9099

- ATYR2810 binds to recombinant human NRP2 extracellular domain (ECD), but not to the homologous NRP1-ECD in ELISA.
- ATYR2810 binds to A549 wild-type (WT) cells that express endogenous NRP2, but not to NRP2 knockout (KO) A549 clonal cells. ATYR2810 shows better sensitivity and specificity than a commercial a-NRP2 antibody (R&D #AF567).
- ATYR2810 demonstrates high affinity binding against human and cynomolgus NRP2 but very weak binding to mouse NRP2.

Figure 2: ATYR2810 binds to the VEGF binding pocket of NRP2

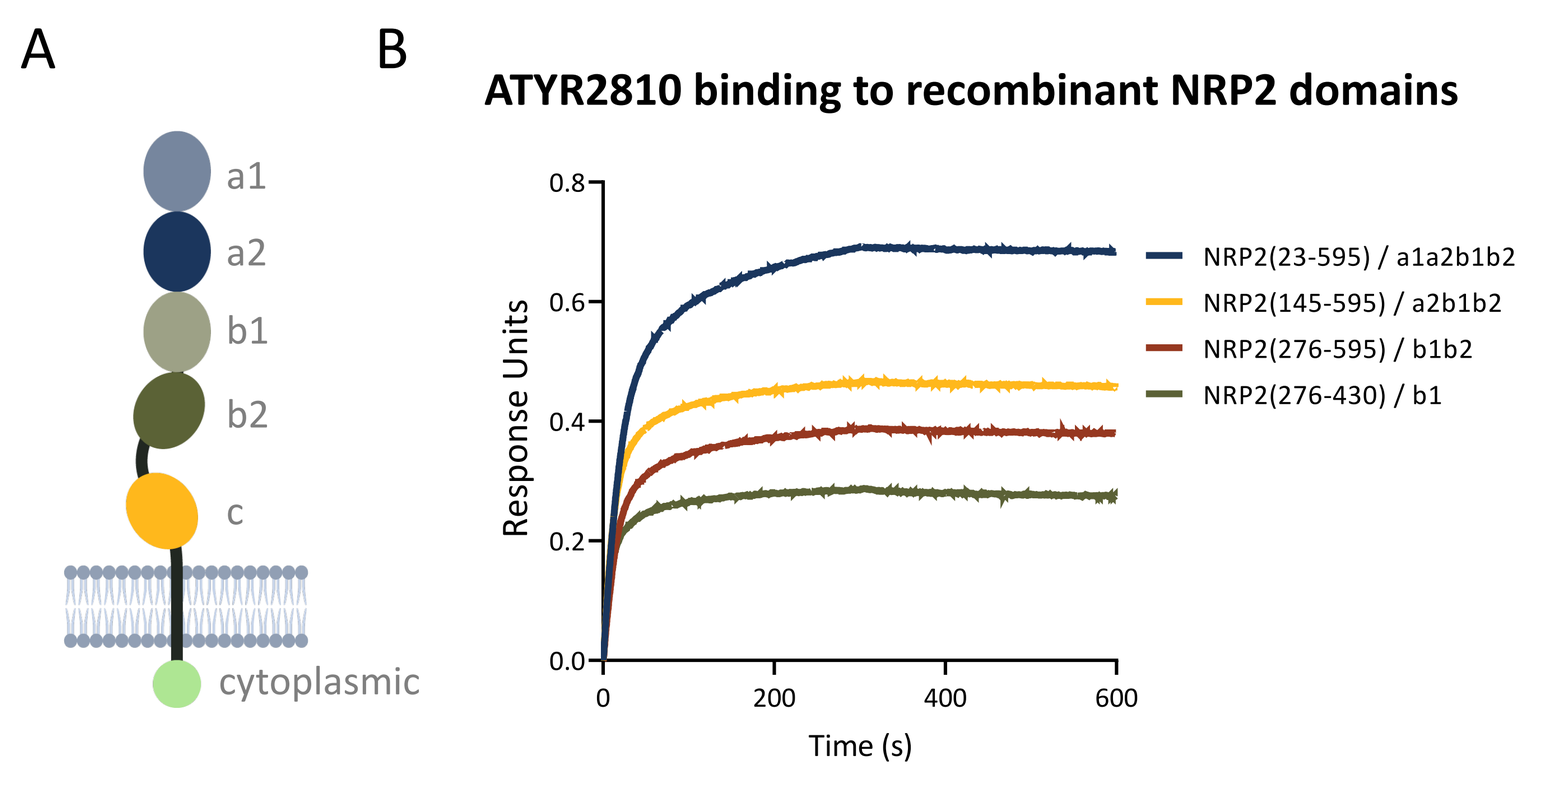
A
a1
a2
b1
b2
c
cytoplasmic
B
ATYR2810 binding to recombinant NRP2 domains
NRP2(23-595) / a1a2b1b2
NRP2(145-595) / a2b1b2
NRP2(276-595) / b1b2
NRP2(276-430) / b1
Response Units
Time (s)

C

```
                                     299
Human NRP2   CNVPLGMESGRIANEQISASSTYSDGRWTP  306
Mouse NRP2   CNVPLGMESGRIANEQISASSTFSDGRWTP  306

Human NRP2   QQSRLHGDDNGWTPNLDSNKEYLQVDLRFL  336
Mouse NRP2   QQSRLHGDDNGWTPNLDSNKEYLQVDLRFL  336

                             354
Human NRP2   TMLTAIATQGAISRETQNGYYVKSYKLEVS  366
Mouse NRP2   TMLTAIATQGAISRETQKGYYVKSYKLEVS  366

                             383
Human NRP2   TNGEDWMVYRHGKNHKVFQANNDATEVVLN  396
Mouse NRP2   TNGEDWMVYRHGKNHKIFQANNDATEVVLN  396

                400    407      416
Human NRP2   KLHAPLLTRFVRIRPQTWHSGIALRLELFG  426
Mouse NRP2   KLHMPLLTRFIRIRPQTWHLGIALRLELFG  426

Human NRP2   CRVT                            430
Mouse NRP2   CRVT                            430
```

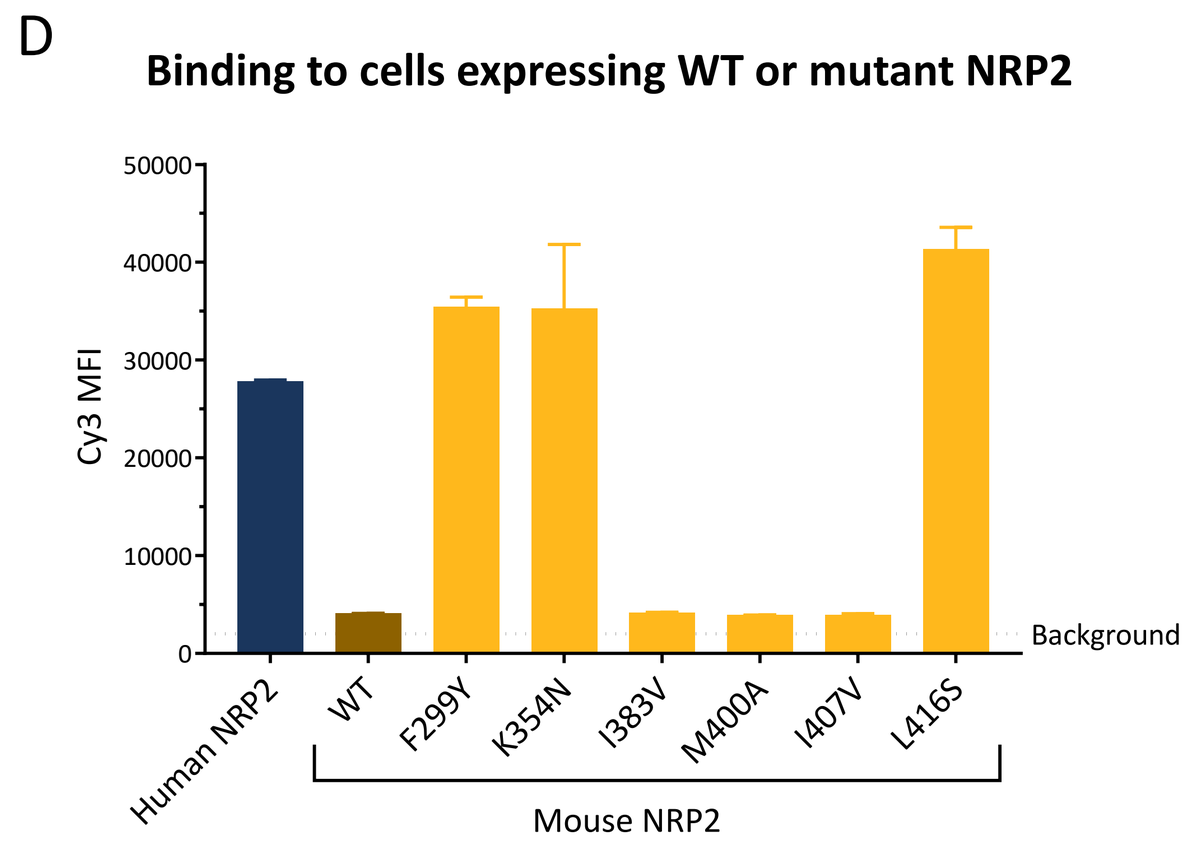
D
Binding to cells expressing WT or mutant NRP2
Cy3 MFI
Background
Human NRP2
WT
F299Y
K354N
I383V
M400A
I407V
L416S
Mouse NRP2

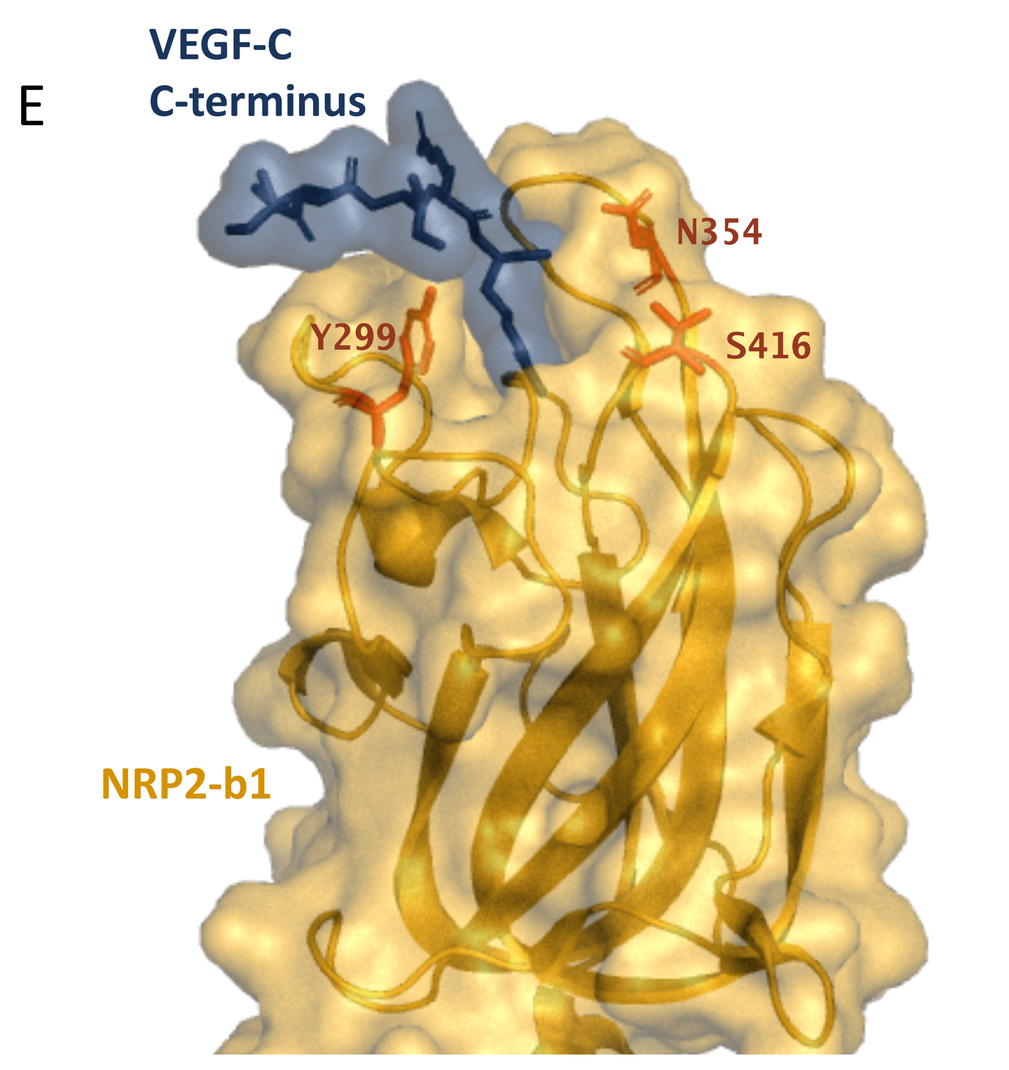
E
VEGF-C
C-terminus
N354
Y299
S416
NRP2-b1

- ATYR2810 binds to the b1 domain of human NRP2, within amino acid residues 276-430.
- Mouse and human NRP2 differ by 6 amino acids in the b1 domain. Each of these residues in mouse NRP2 were individually mutated to the corresponding residue from human NRP2. Binding of ATYR2810 to these mutant mouse NRP2 proteins was assayed by flow cytometry.
- Mutation of residues 299, 354, and 416 in mouse NRP2 to their human counterparts resulted in a gain of ATYR2810 binding towards the mutant mouse NRP2. Mutation of residues 383, 400 and 407 had no effect.
- Based on previous structural studies[Ref4], these three amino acids surround a binding pocket for the C-terminal tail of VEGF. Binding of ATYR2810 at this location is likely to block binding of VEGF to NRP2.

Results

Figure 3: ATYR2810 selectively blocks dimerization of NRP2 with VEGFR

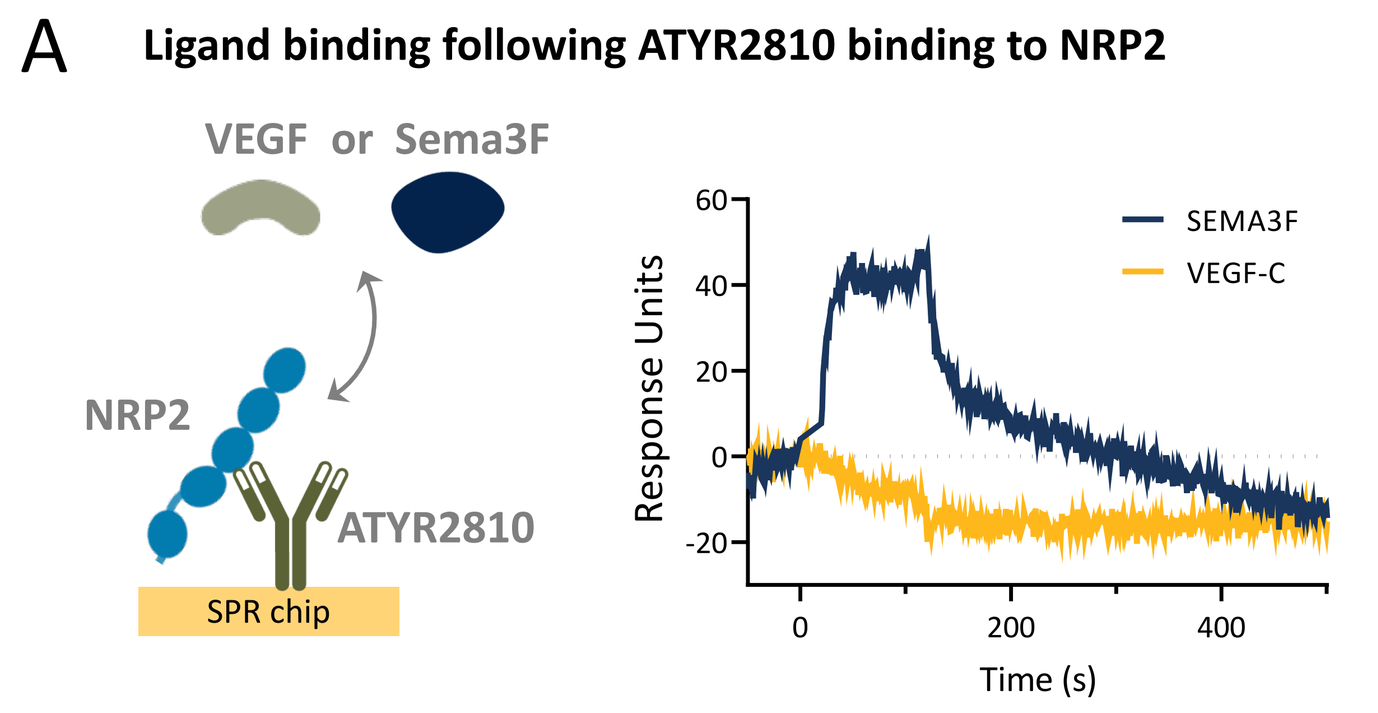
A
Ligand binding following ATYR2810 binding to NRP2
VEGF or Sema3F
NRP2
ATYR2810
SPR chip
SEMA3F
VEGF-C
Response Units
Time (s)

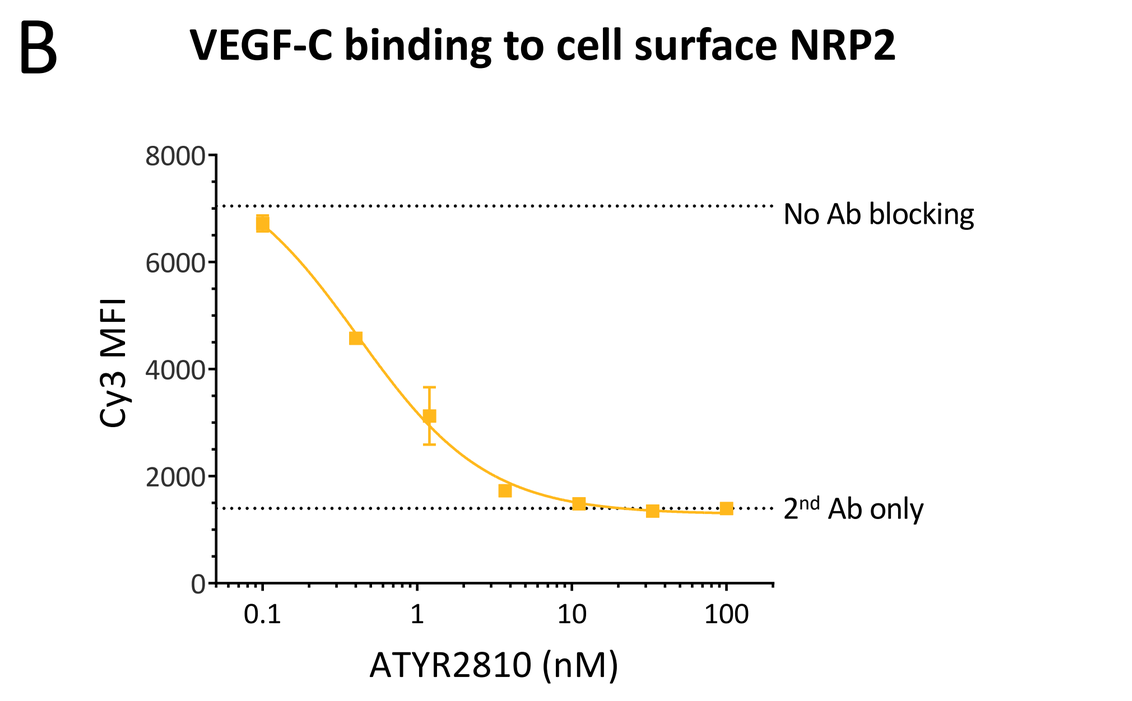
B
VEGF-C binding to cell surface NRP2
No Ab blocking
2nd Ab only
Cy3 MFI
ATYR2810 (nM)

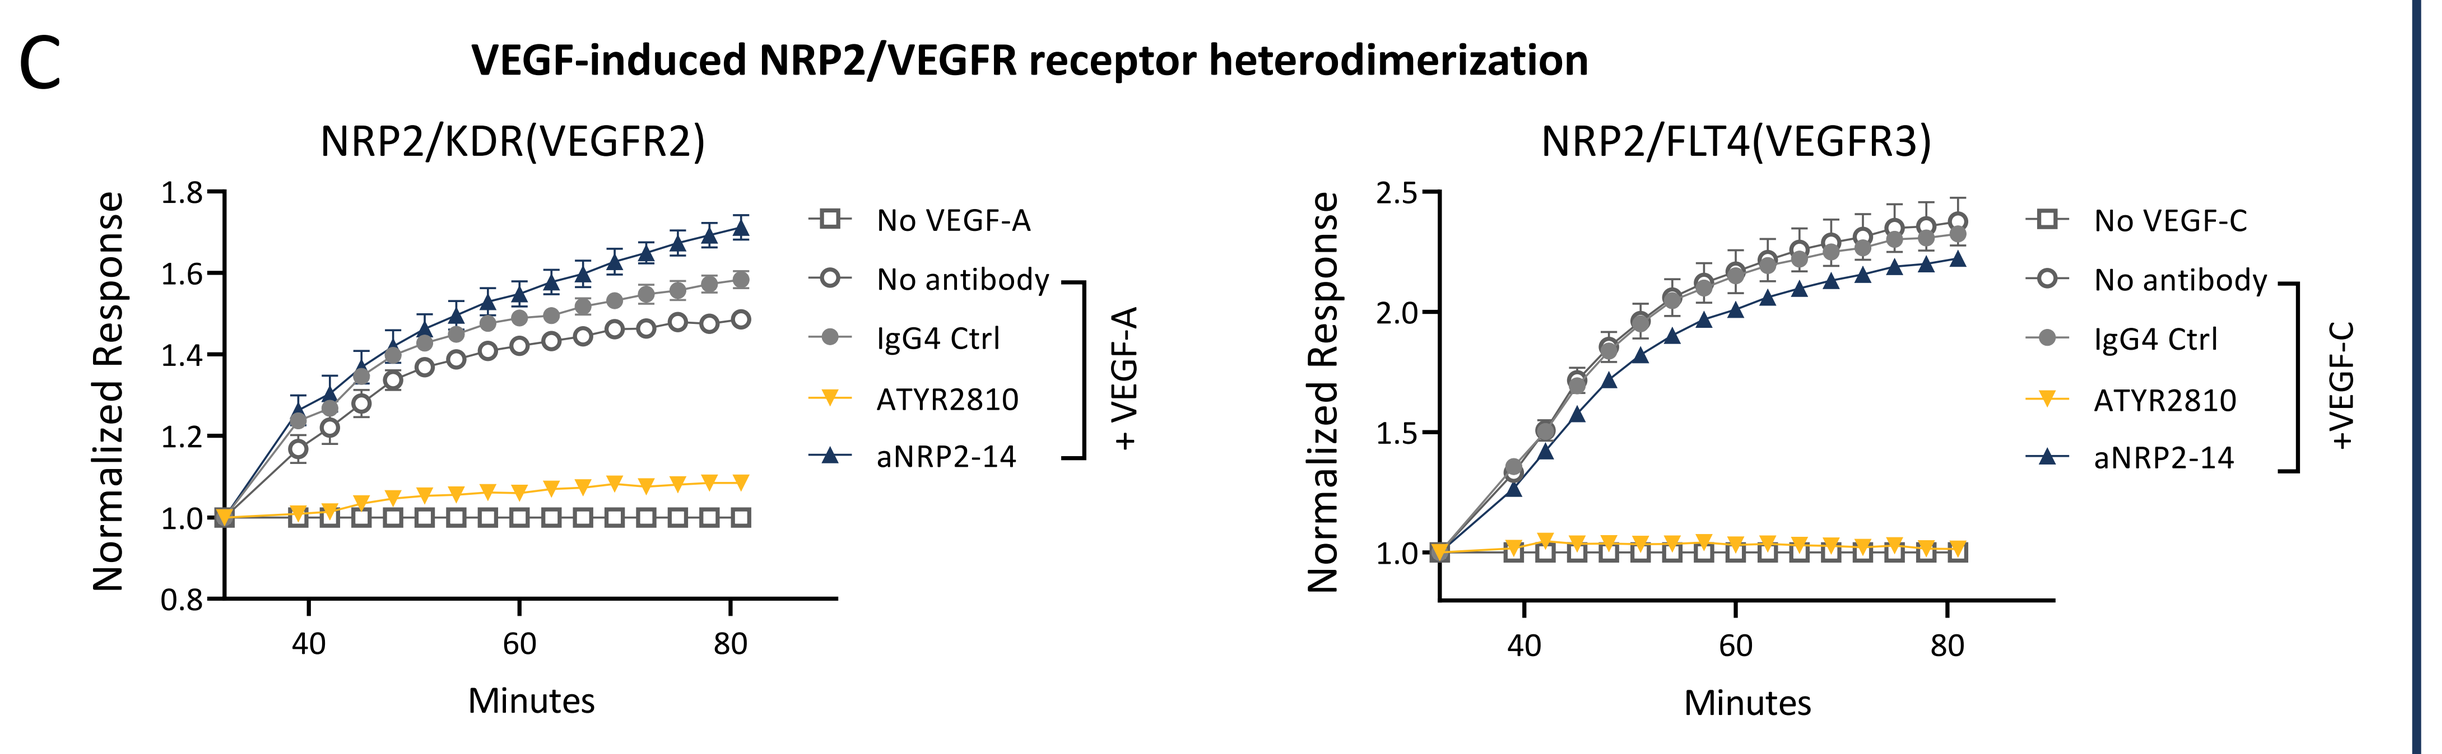
C
VEGF-induced NRP2/VEGFR receptor heterodimerization
NRP2/KDR(VEGFR2)
NRP2/FLT4(VEGFR3)
No VEGF-A
No antibody
IgG4 Ctrl
ATYR2810
aNRP2-14
+ VEGF-A
No VEGF-C
+VEGF-C
Normalized Response
Minutes

- ATYR2810 blocks VEGF-C binding to NRP2-ECD, but does not block Sema3F binding to NRP2-ECD.
- ATYR2810 entirely blocks VEGF-C binding to Expi293 cells overexpressing NRP2 on the cell surface.
- ATYR2810 completely blocks VEGF-A induced NRP2/KDR (VEGFR2) dimerization and VEGF-C induced NRP2/FLT4 (VEGFR3) dimerization. Blocking in these assays is not observed with a Sema3F-blocking antibody (aNRP2-14). ATYR2810 does not have any effect on Sema3F induced NRP2/PlexinA1 dimerization [Ref5].

Figure 4: ATYR2810 in combination with chemotherapy enhances *in vivo* efficacy in Hs578T TNBC xenograft model

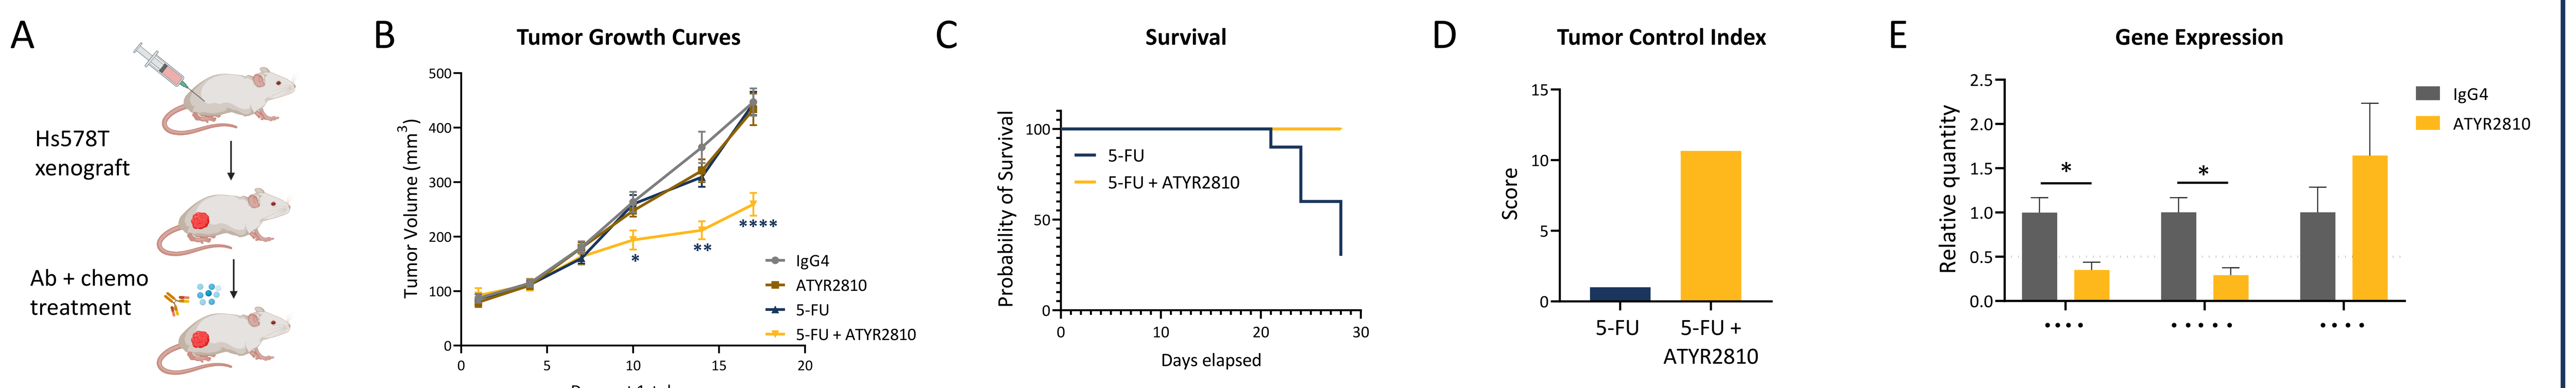
A
Hs578T xenograft
Ab + chemo treatment
B
Tumor Growth Curves
IgG4
ATYR2810
5-FU
5-FU + ATYR2810
Tumor Volume (mm3)
Day post 1st dose
C
Survival
5-FU
5-FU + ATYR2810
Probability of Survival
Days elapsed
D
Tumor Control Index
Score
5-FU
5-FU + ATYR2810
E
Gene Expression
IgG4
ATYR2810
Relative quantity

- Effects of ATYR2810 in combination with 5-fluorouracil (5-FU) chemotherapy on tumor growth were determined in a TNBC Hs578T xenograft model.
- ATYR2810 + 5-FU combination therapy enhanced efficacy in tumor growth inhibition and improved survival compared to 5-FU monotherapy in this model.
- ATYR2810 down-regulated gene expression of EMT (ZEB1) and CSC (NANOG) markers, and up-regulated epithelial (CDH1/E-cadherin) markers in tumor samples from the Hs578T xenograft model.

Figure 5: ATYR2810 down-regulates EMT and CSC markers in TNBC patient-derived organoids

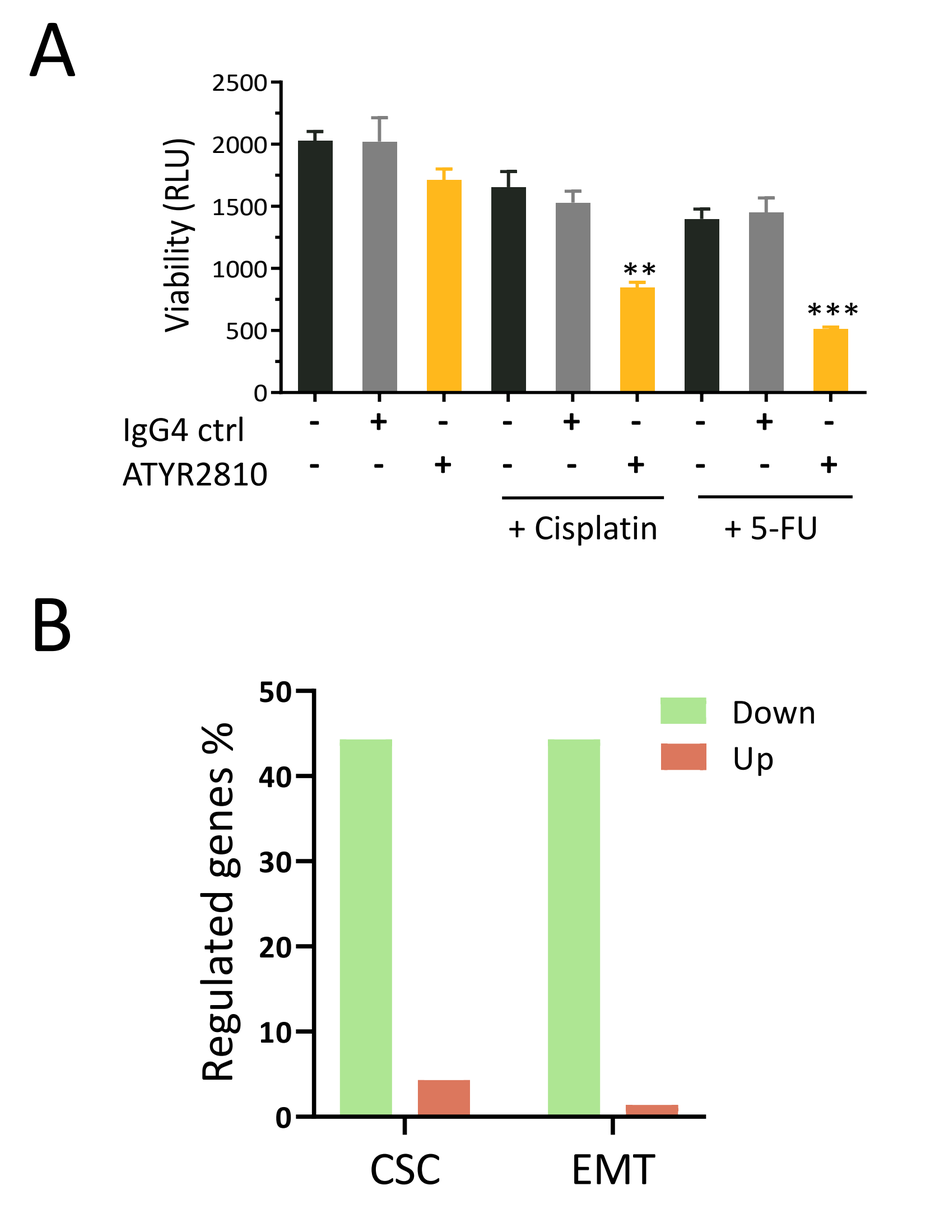
A
Viability (RLU)
IgG4 ctrl
ATYR2810
+ Cisplatin
+ 5-FU
B
Regulated genes %
Down
Up
CSC
EMT

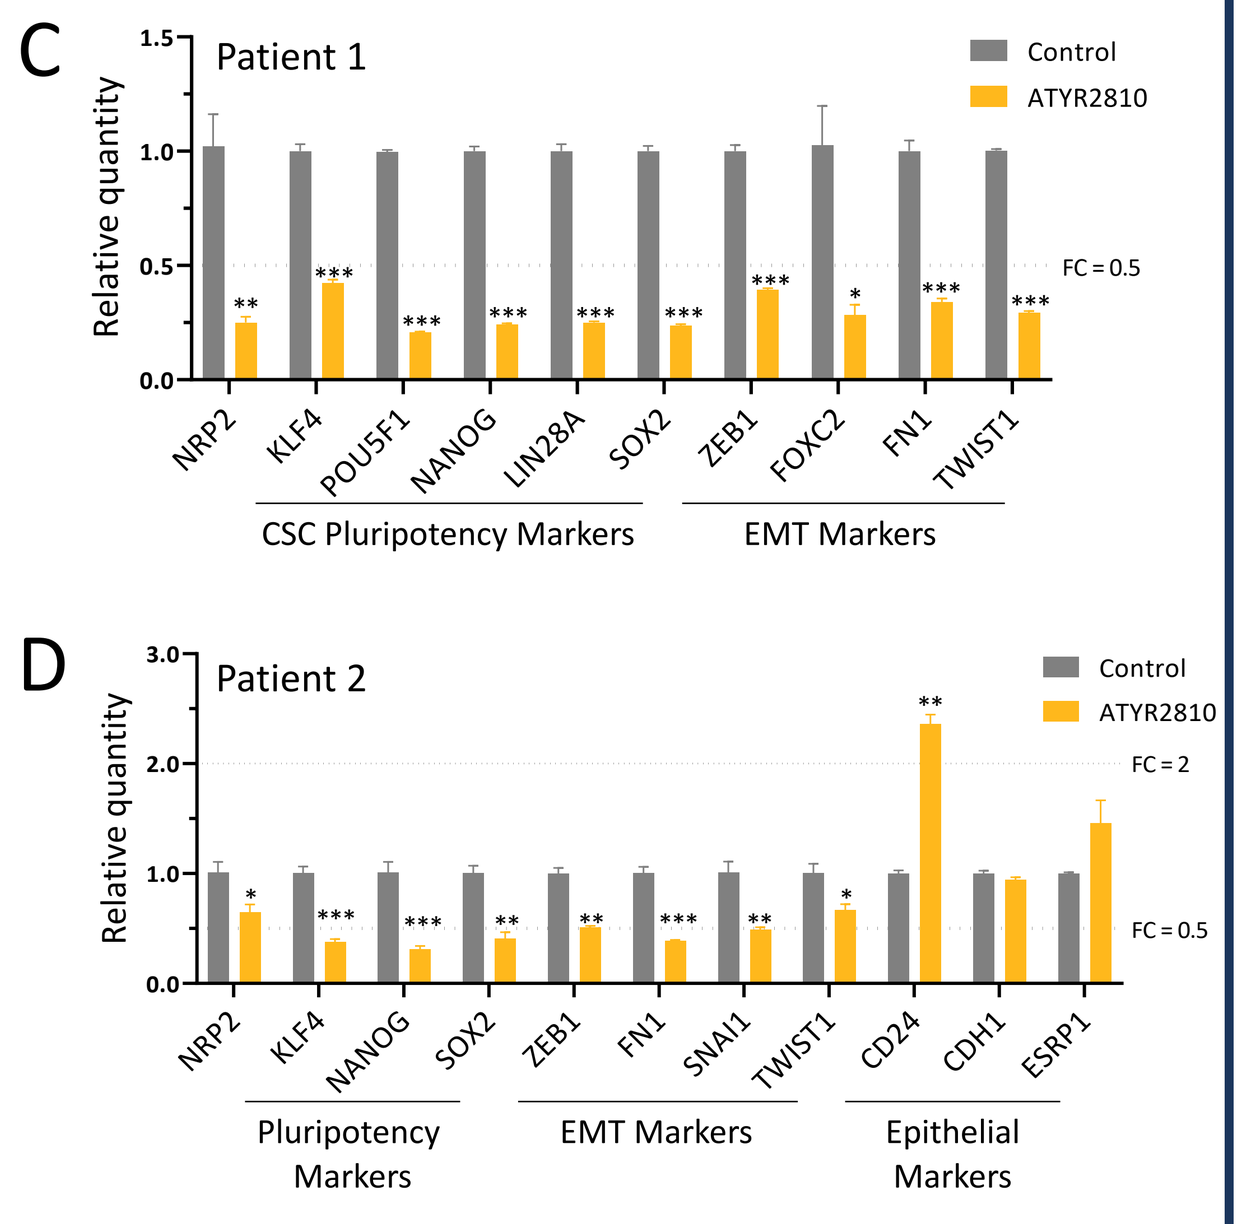
C
Patient 1
Control
ATYR2810
Relative quantity
FC = 0.5
NRP2
KLF4
POU5F1
NANOG
LIN28A
SOX2
ZEB1
FOXC2
FN1
TWIST1
CSC Pluripotency Markers
EMT Markers
D
Patient 2
Control
ATYR2810
FC = 2
FC = 0.5
NRP2
KLF4
NANOG
SOX2
ZEB1
FN1
SNAI1
TWIST1
CD24
CDH1
ESRP1
Pluripotency Markers
EMT Markers
Epithelial Markers

- ATYR2810 enhanced chemosensitivity of TNBC patient-derived organoids.
- ATYR2810 down-regulated CSC and EMT markers in TNBC patient-derived organoids. 80 genes in each of CSC and EMT pathways were analyzed by Fluidigm, and nearly half of markers with detectable expression levels were down-regulated >2-fold by ATYR2810 compared to control.
- ATYR2810 down-regulated a number of critical CSC pluripotency markers and EMT markers in organoids derived from both TNBC Patient 1 and Patient 2, and up-regulated certain epithelial markers (e.g. CD24) in organoids derived from TNBC Patient 2.

Figure 6: ATYR2810 in combination with chemotherapy enhances efficacy in TNBC patient-derived xenografts

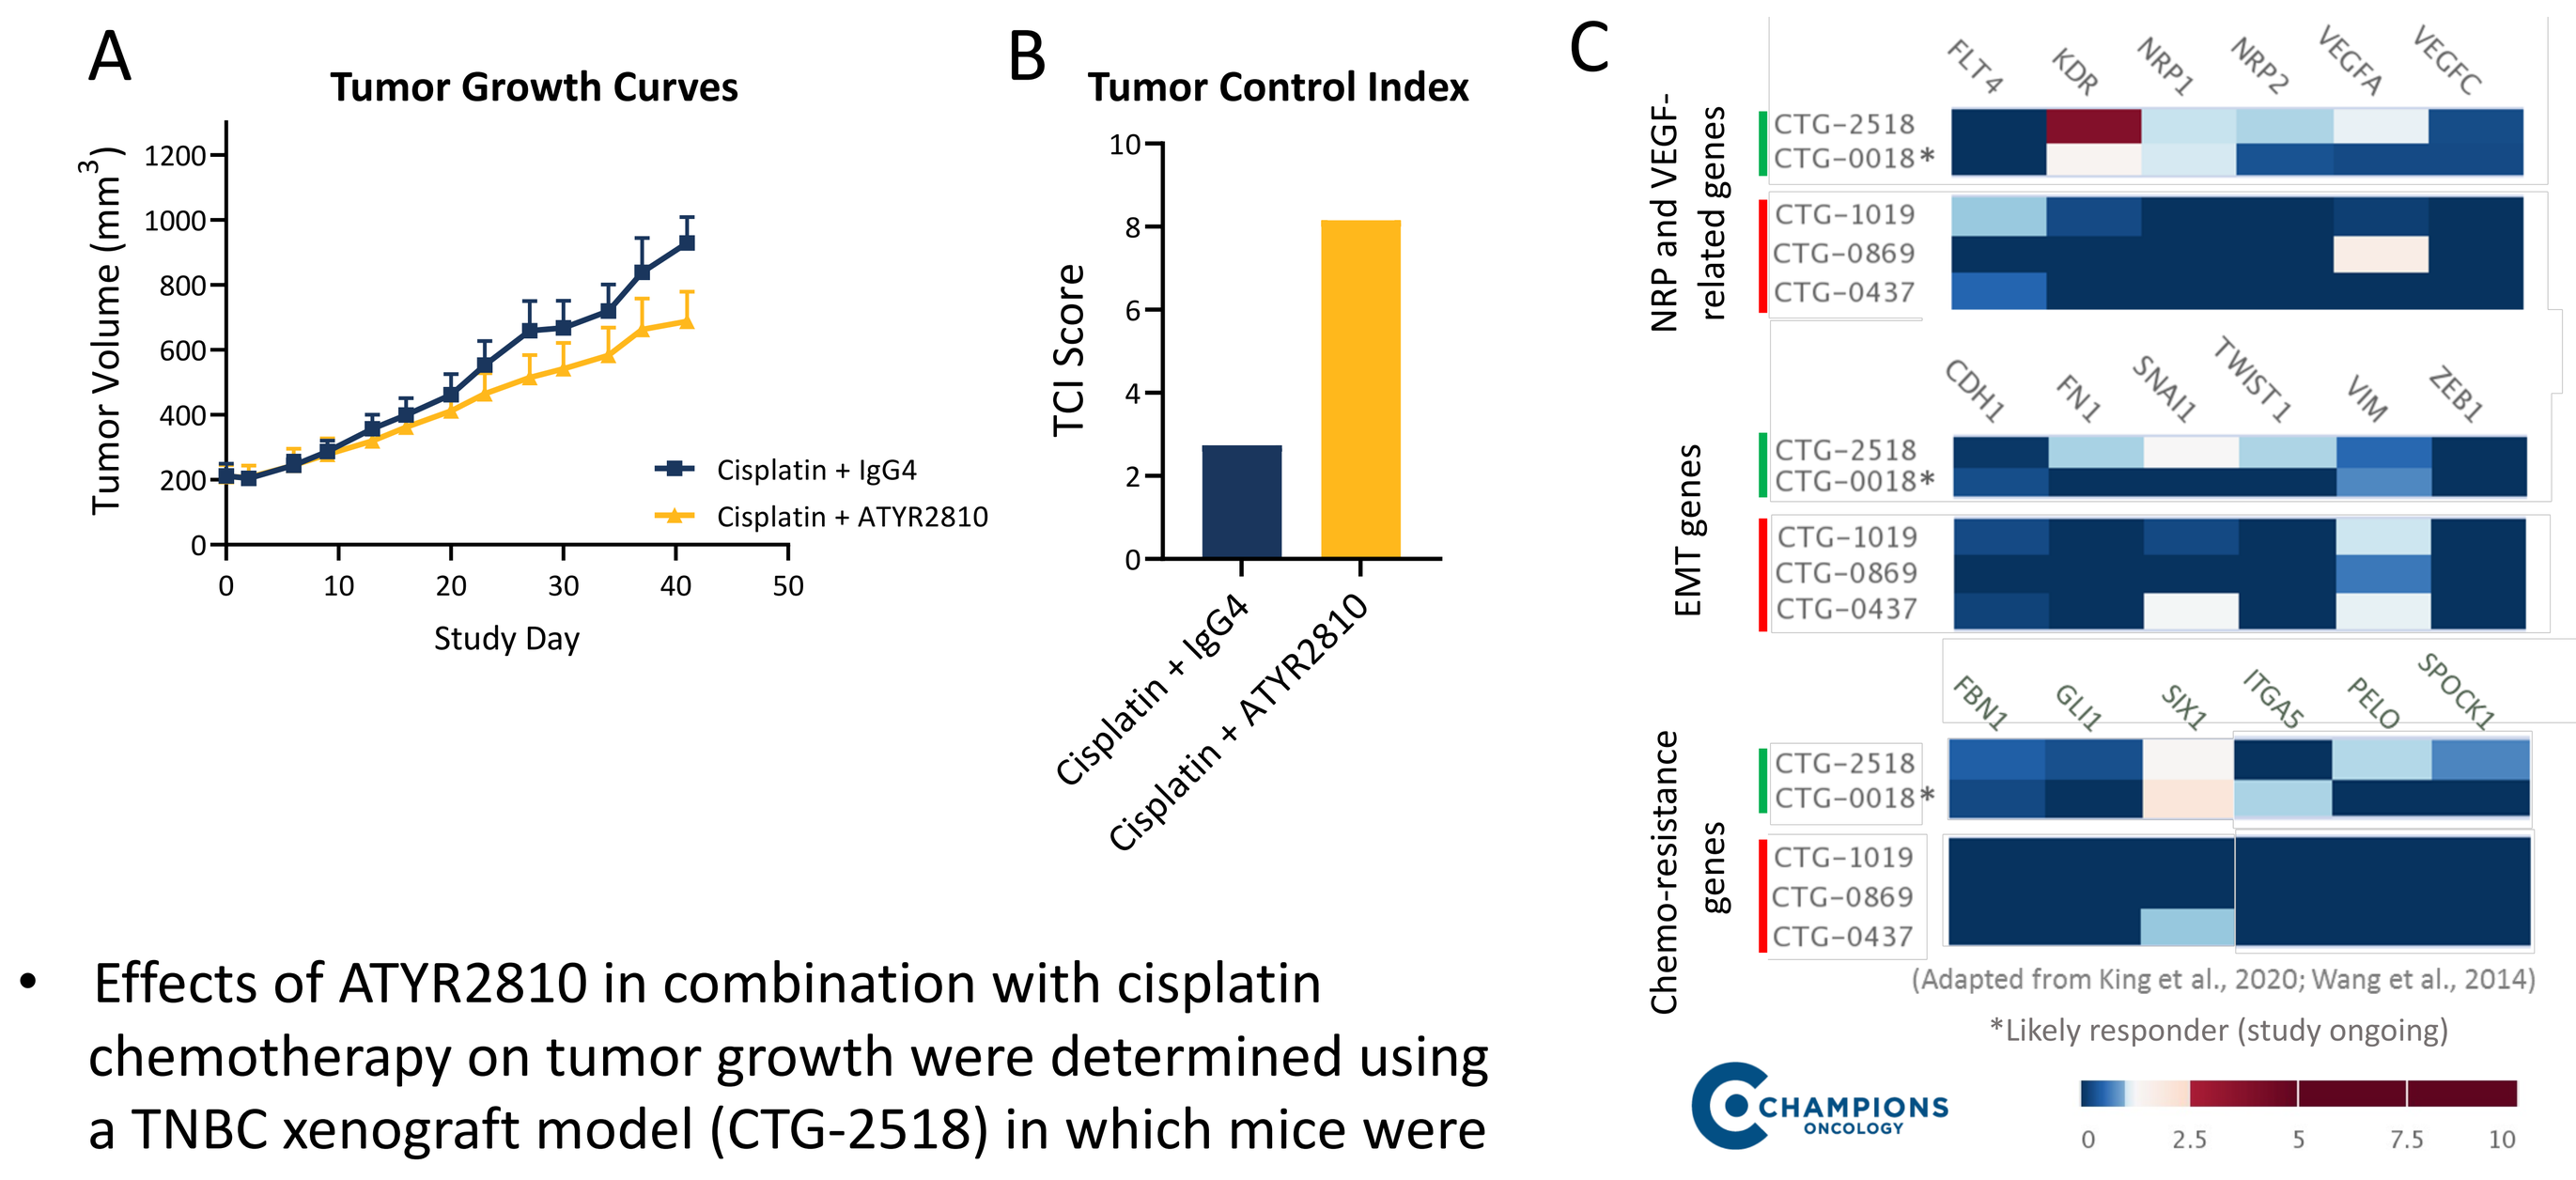
A
Tumor Growth Curves
Tumor Volume (mm3)
Study Day
Cisplatin + IgG4
Cisplatin + ATYR2810
B
Tumor Control Index
TCI Score
Cisplatin + IgG4
Cisplatin + ATYR2810
C
NRP and VEGF-related genes
FLT4
KDR
NRP1
NRP2
VEGFA
VEGFC
CTG-2518
CTG-0018*
CTG-1019
CTG-0869
CTG-0437
EMT genes
CDH1
FN1
SNAI1
TWIST1
VIM
ZEB1
Chemo-resistance genes
FBN1
GLI1
SIX1
ITGA5
PELO
SPOCK1
(Adapted from King et al., 2020; Wang et al., 2014)
*Likely responder (study ongoing)
CHAMPIONS ONCOLOGY

- Effects of ATYR2810 in combination with cisplatin chemotherapy on tumor growth were determined using a TNBC xenograft model (CTG-2518) in which mice were inoculated with PDX tumor cells.
- ATYR2810 + cisplatin combination therapy (TCI score of 8.2) enhanced tumor growth inhibition and led to a 3-fold increase in tumor control as compared to cisplatin monotherapy (TCI score of 2.7).
- Prior to treatment, the responsive TNBC PDX models (CTG-2518 and CTG-0018) showed increased expression of several NRP and VEGF-related genes as well as several EMT and chemo-resistance genes as compared to non-responsive models (CTG-1019, CTG-0869 and CTG-0473), suggesting that tumors with high expression of these genes may be responsive to ATYR2810 combination treatment.

Conclusions

- ATYR2810 is a highly specific and high affinity antibody against human NRP2, and binds to the b1 domain of NRP2 in a manner than blocks VEGF ligand binding.
- ATYR2810 selectively blocks VEGF-induced dimerization of NRP2 with VEGFR, while not affecting Sema3F-induced dimerization of NRP2 with PlexinA1.
- In combination with chemotherapeutics, ATYR2810 displays enhanced efficacy in TNBC xenograft models as compared to the chemotherapy alone.
- Gene expression data from TNBC xenograft samples and TNBC PDOs reveal down-regulation of several CSC and EMT markers following ATYR2810 treatment.
- ATYR2810 blocking of the NRP2/VEGF signaling pathway may sensitize chemo-resistant TNBC models to anti-cancer therapeutics.

References

1. Yasuoka, Hironao *et al*. "Neuropilin-2 expression in breast cancer: correlation with lymph node metastasis, poor prognosis, and regulation of CXCR4 expression", BMC Cancer. 2009; 9(220)
2. Elaimy, Ameer L. *et al*. "VEGF–neuropilin-2 signaling promotes stem-like traits in breast cancer cells by TAZ-mediated repression of the Rac GAP β2-chimaerin", Sci Signal. 2018; 11(528)
3. Xu, Zhiwen *et al*. "A domain-specific antibody to NRP2 down-regulated epithelial-mesenchymal transition genes and enhanced efficacy of standard-of-care therapeutics for aggressive breast cancer", 2021 AACR Virtual Annual Meeting (Abstract #5316, Poster #LB095)
4. Parker, Matthew W. *et al*. "Structural Basis for VEGF-C Binding to Neuropilin-2 and Sequestration by a Soluble Splice Form", Structure. 2015; 23(4) PDB ID: 4QDQ
5. Nangle, Leslie A. *et al*. "Domain-specific Antibodies to Neuropilin-2 Implicate VEGF-C and Not Semaphorin 3F in Breast Cancer Stem Cell Function", 2020 AACR Virtual Annual Meeting II (Abstract #7308, Poster #1785)