Efzofitimod, A Novel Immunomodulator for Pulmonary Sarcoidosis, modulates Patient Inflammatory Responses Through Myeloid Cells

D. Siefker[1], Z. Xu[1], S. Paz[1], M. Ferrer[1], E. Escobedo[1], Y. Chong[1], L. Guy[1], K. Rauch[1], S. Klopp-Savino[1], K. Hamel[1], L. Burman[1], L. Zhai[2], L. Eide[1], C. Polizzi[1], C. Burkart[1], G. Walker[1], R. Adams[1], L. Nangle[1]

[1]aTyr Pharma - San Diego (USA), [2]Pangu/HKUST- Hong Kong (China)

Abstract

Pulmonary sarcoidosis is a complex immune disease characterized by the recruitment and accumulation of circulating monocytes to the lung, where they differentiate into macrophages and co-localize with activated T cells to form granulomas.[1] Steroid therapy is the standard of care to control inflammation. However, chronic steroid use results in serious side effects, and safer, more effective immunomodulatory therapies are needed. Efzofitimod is a novel immunomodulator that selectively binds neuropilin-2 (NRP2), a pleiotropic membrane protein highly expressed on immune cells during lung inflammation and on circulating monocytes and macrophages within granulomas. In a phase 1b/2a clinical trial in pulmonary sarcoidosis patients, efzofitimod demonstrated dose dependent trends of improvement compared to placebo on key efficacy endpoints, including steroid reduction, lung function and patient symptom measures.[2] Efzofitimod also demonstrated dose dependent control of inflammatory biomarkers.[3] We hypothesize that efzofitimod modulates monocyte/macrophage function in pulmonary sarcoidosis patients as a mechanism to reduce inflammation and improve lung function.

To investigate immunomodulatory functions of efzofitimod on monocytes, peripheral blood mononuclear cells (PBMC) from pulmonary sarcoidosis patients or healthy donors were evaluated for surface marker expression by flow cytometry and cytokine/chemokine secretion by immunoassay. CD14+ monocytes from healthy donors were differentiated into macrophages in the presence of efzofitimod and evaluated for surface marker expression, as well as cytokine/chemokine expression and secretion. We found that NRP2, the binding partner to efzofitimod, is highly expressed in lung granulomas from pulmonary sarcoidosis patients and colocalizes with the macrophage markers CD68 and CD163. Circulating monocytes from pulmonary sarcoidosis patients have elevated NRP2 compared to those from healthy donors, and exhibit a more inflammatory phenotype. For example, pulmonary sarcoidosis patient monocytes secrete a greater amount of pro-inflammatory cytokines/chemokines including TNFα, IL-6, MIP-1α, and MCP-1. They produce less activation marker CD80 upon LPS stimulation compared to healthy donors and show dysregulation of the MCP-1/CCR2 signaling axis. In primary monocyte-derived macrophages, efzofitimod affects macrophage morphology and reduces proinflammatory gene expression and CCR2, the receptor for MCP-1.

These results demonstrate that efzofitimod modulates expression of surface receptors and pro-inflammatory cytokines/chemokines in myeloid cells, expanding our understanding of efzofitimod's mechanism of action.

tRNA synthetase drug discovery platform

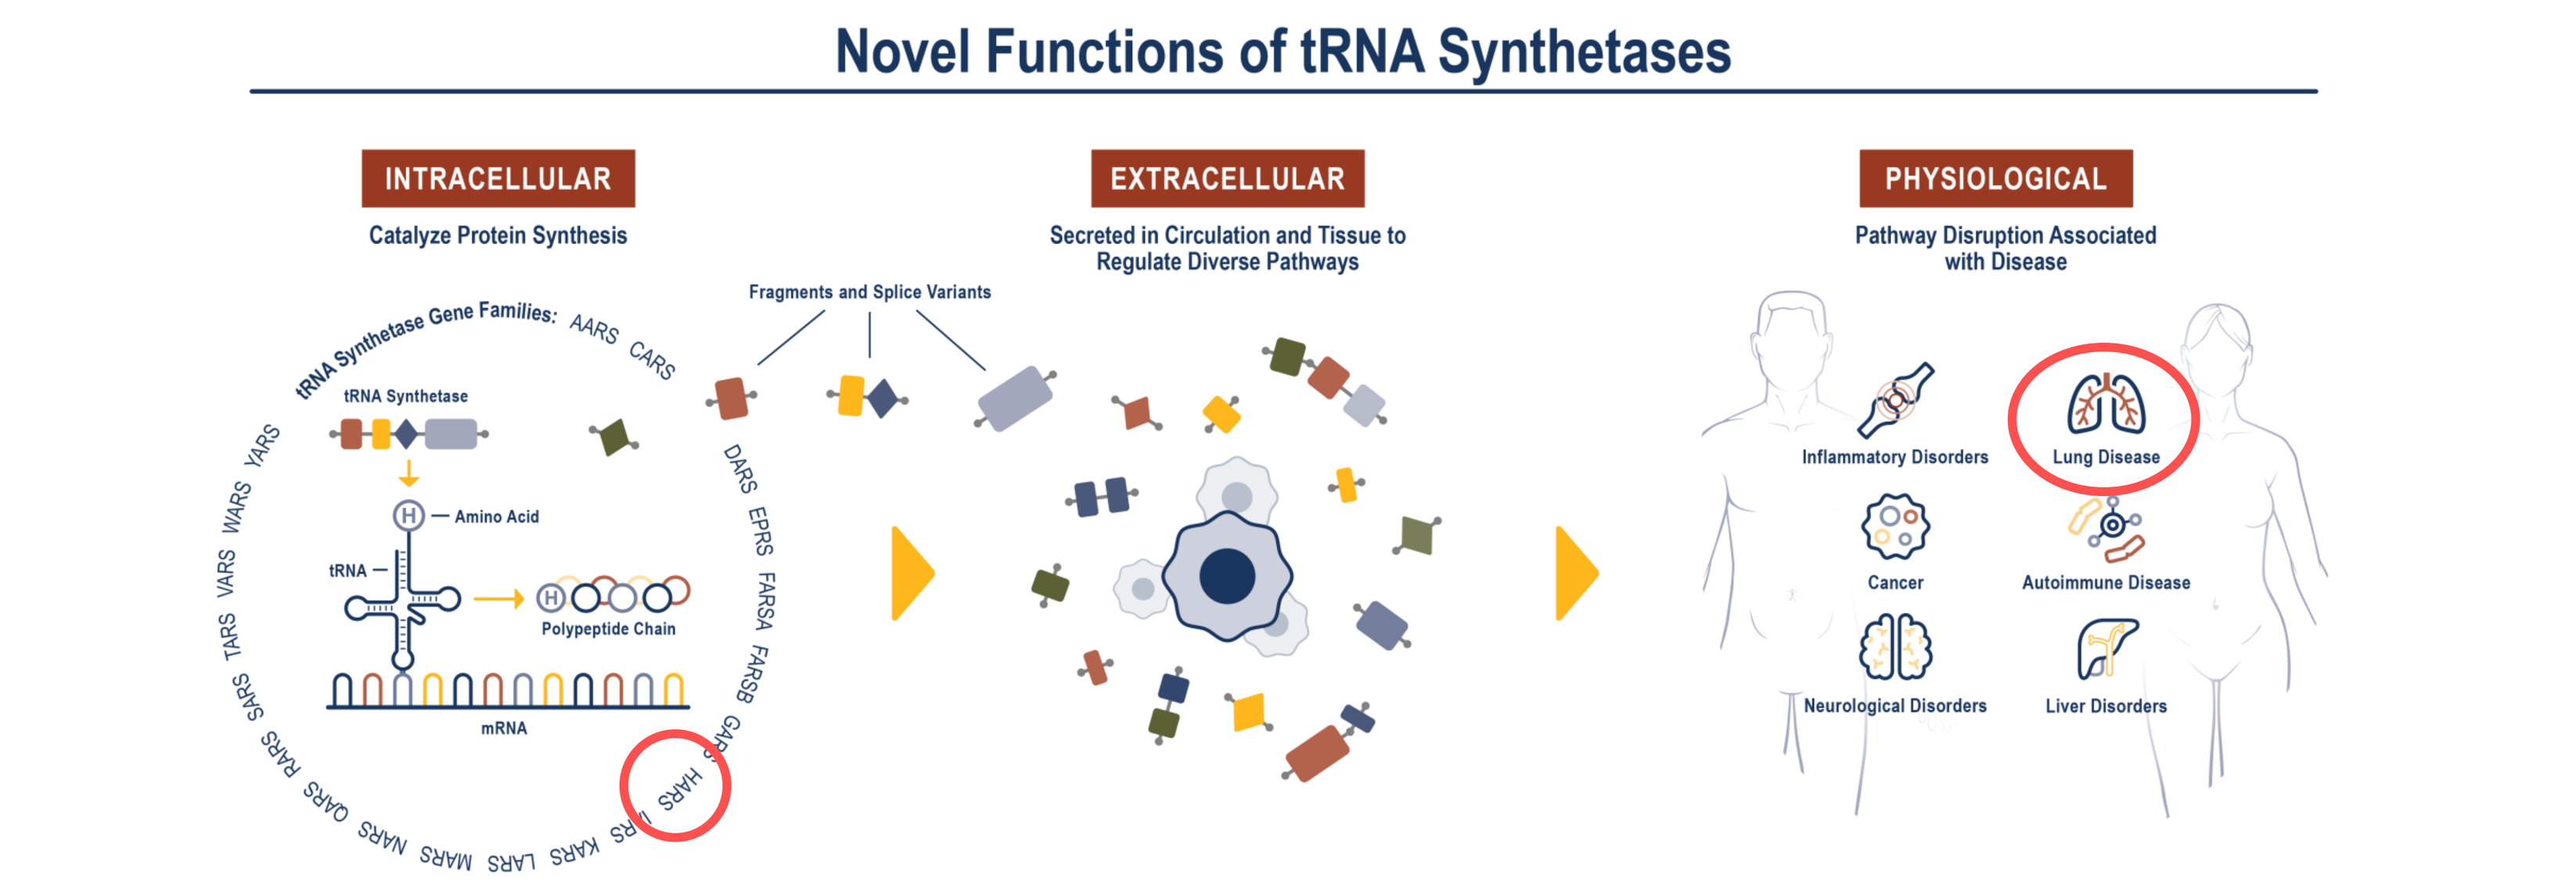

Novel Functions of tRNA Synthetases
INTRACELLULAR
Catalyze Protein Synthesis
EXTRACELLULAR
Secreted in Circulation and Tissue to Regulate Diverse Pathways
PHYSIOLOGICAL
Pathway Disruption Associated with Disease
Fragments and Splice Variants
Inflammatory Disorders
Lung Disease
Cancer
Autoimmune Disease
Neurological Disorders
Liver Disorders

Figure 1. Extracullular tRNA synthetases have non-canonical immuno-modulating properties. The canonical role of tRNA synthetases is to catalyze protein synthesis within the cell. Splice variants and proteolytic fragments of tRNA synthetases have been identified in extracellular spaces, where they have novel functions. aTyr Pharma is engaged in the discovery and development of potential first-in-class medicines based on newly discovered pathways effected by extracellular tRNA synthetases.

Efzofitimod, a novel ligand for Neuropilin-2 (NRP2)

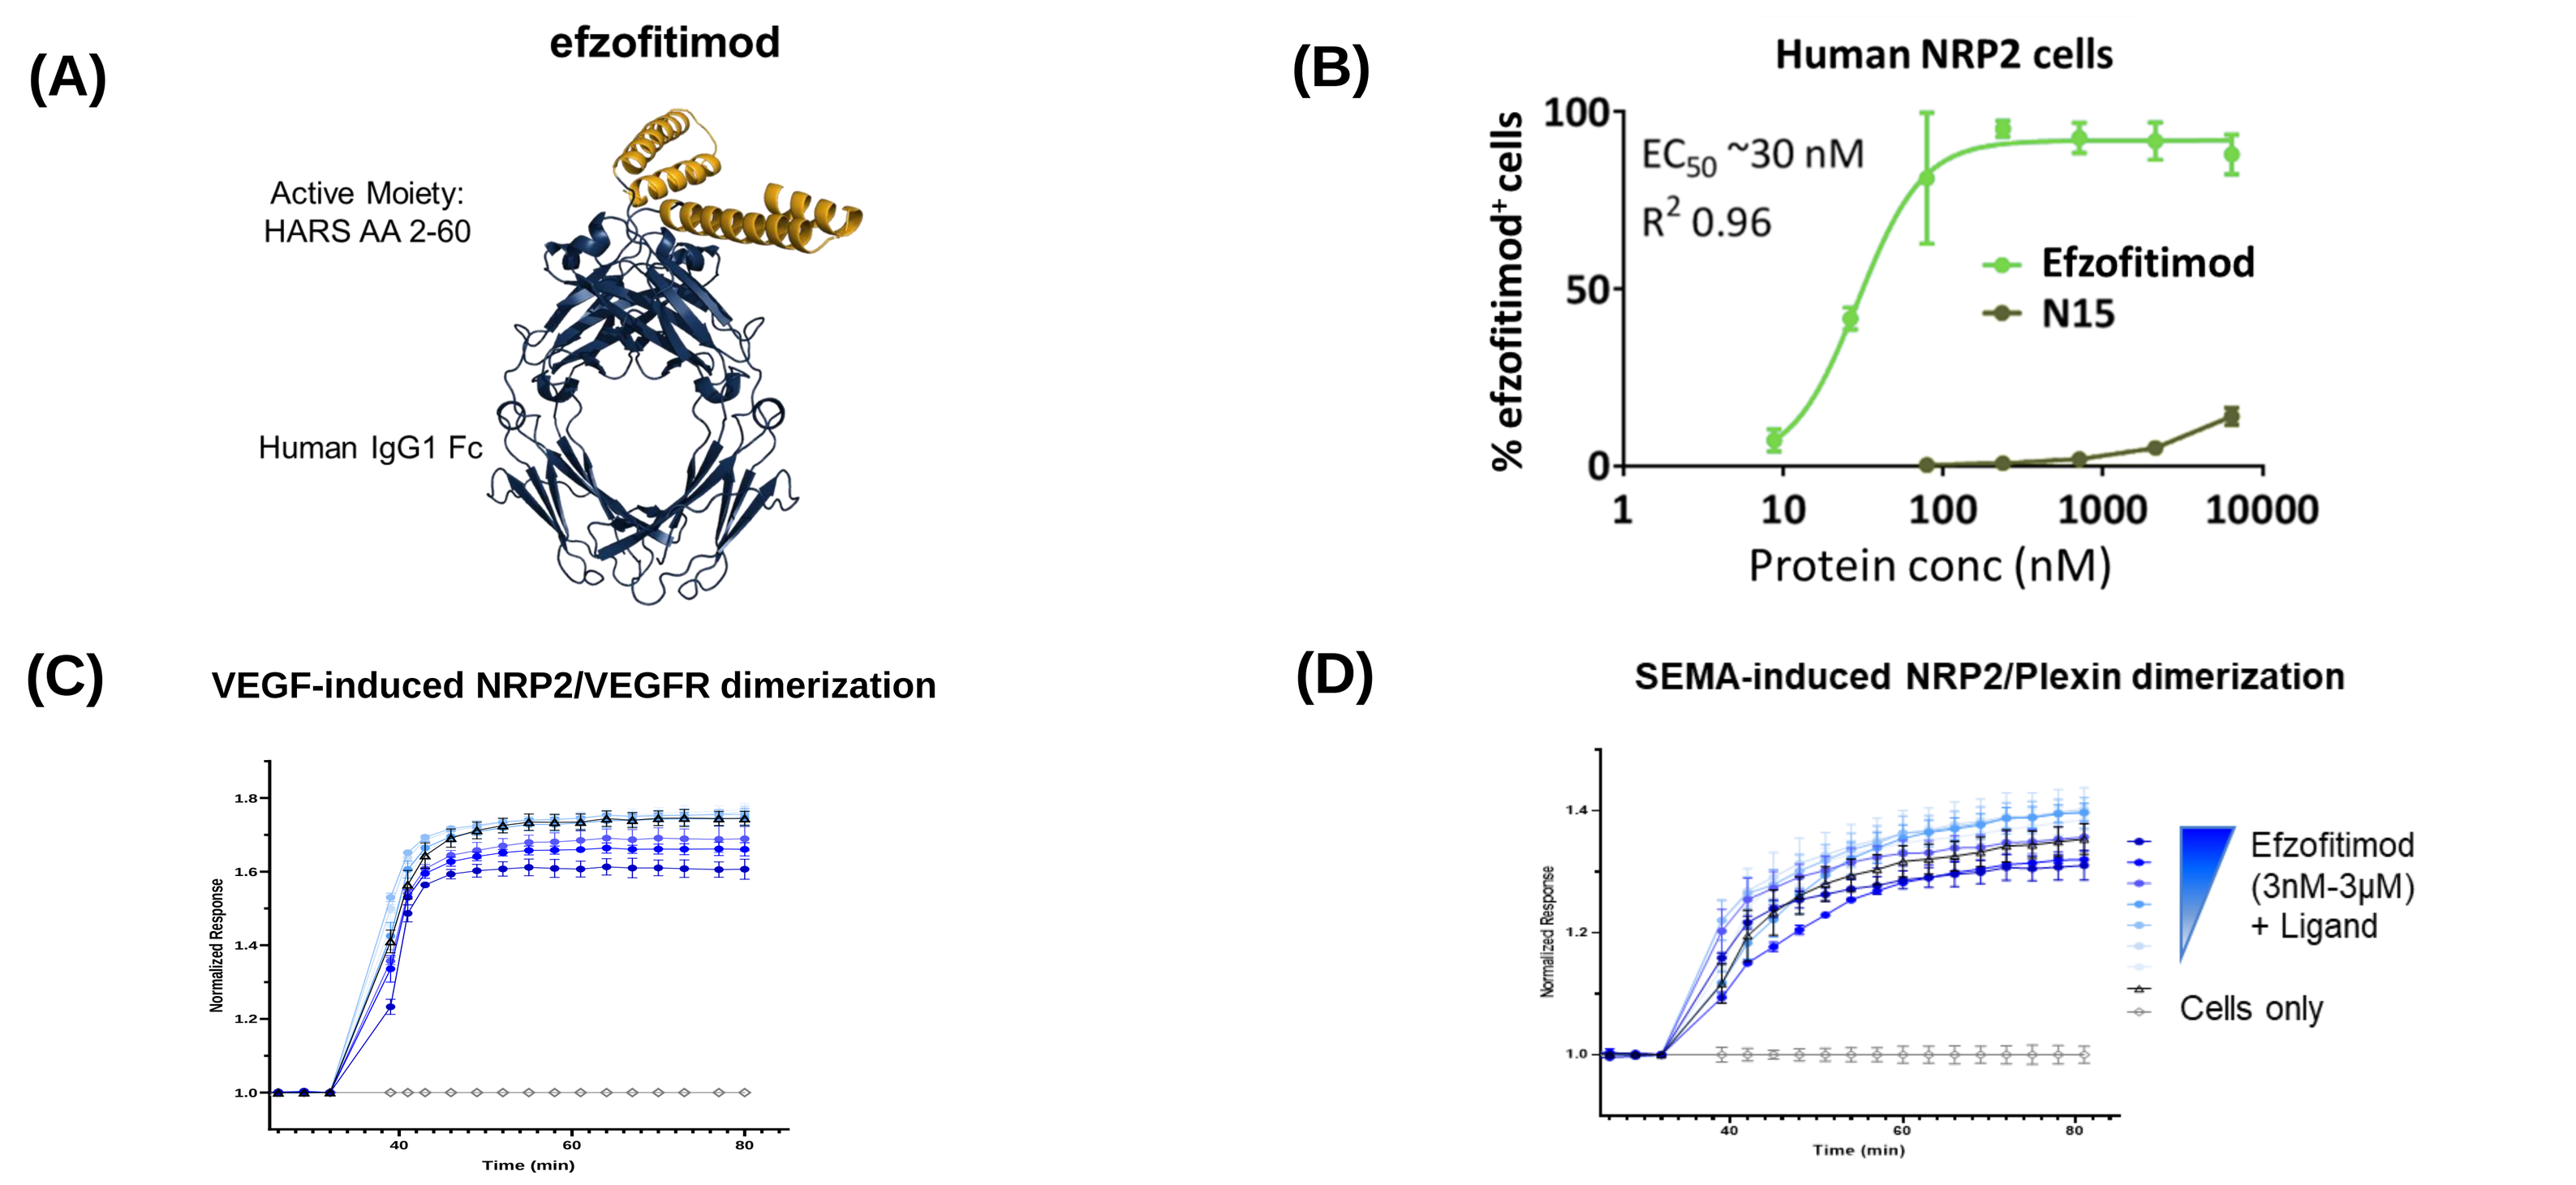

(A) efzofitimod
Active Moiety: HARS AA 2-60
Human IgG1 Fc
(B) Human NRP2 cells
EC_{50} ~30 nM
R^2 0.96
Efzofitimod
N15
% efzofitimod+ cells
Protein conc (nM)
(C) VEGF-induced NRP2/VEGFR dimerization
(D) SEMA-induced NRP2/Plexin dimerization
Efzofitimod (3nM-3μM) + Ligand
Cells only

Figure 2. Efzofitimod binds to NRP2 with nanomolar affinity and does not impact VEGF/SEMA interactions with NRP2 and co-receptors. (A) Efzofitimod is a fusion protein consisting of amino acids 2-60 of Histidyl-tRNA synthetase (HARS) and human IgG1 Fc. (B) Efzofitimod binds to human NRP2 with nanomolar affinity. Efzofitimod binding to NRP2 does not impact VEGF-induced (C) or SEMA-induced (D) interactions of NRP2 with co-receptors.

References:

[1]Broos, EB, et al. Granuloma formation in pulmonary sarcoidosis. *Frontiers in Immunology*. 4,437 (2013)

[2]Baughman, RP, et al. Efzofitimod: A novel anti-inflammatory agent for sarcoidosis. *Sarcoidosis, Vasculitis and Diffuse Lung Diseases*. 40,1 (2023)

[3]Adams, R, et al. Efzofitimod (ATYR1923) treatment reduces pro-inflammatory serum biomarkers in pulmonary sarcoidosis patients. *American Thoracic Society, P560* (2023)

[4]Foerster, et. Al. Manuscript under review

Monocytes from Pulmonary Sarcoidosis Patients Are Phenotypically and Functionally Distinct

NRP2 is expressed on monocyte-derived macrophages and dendritic cells

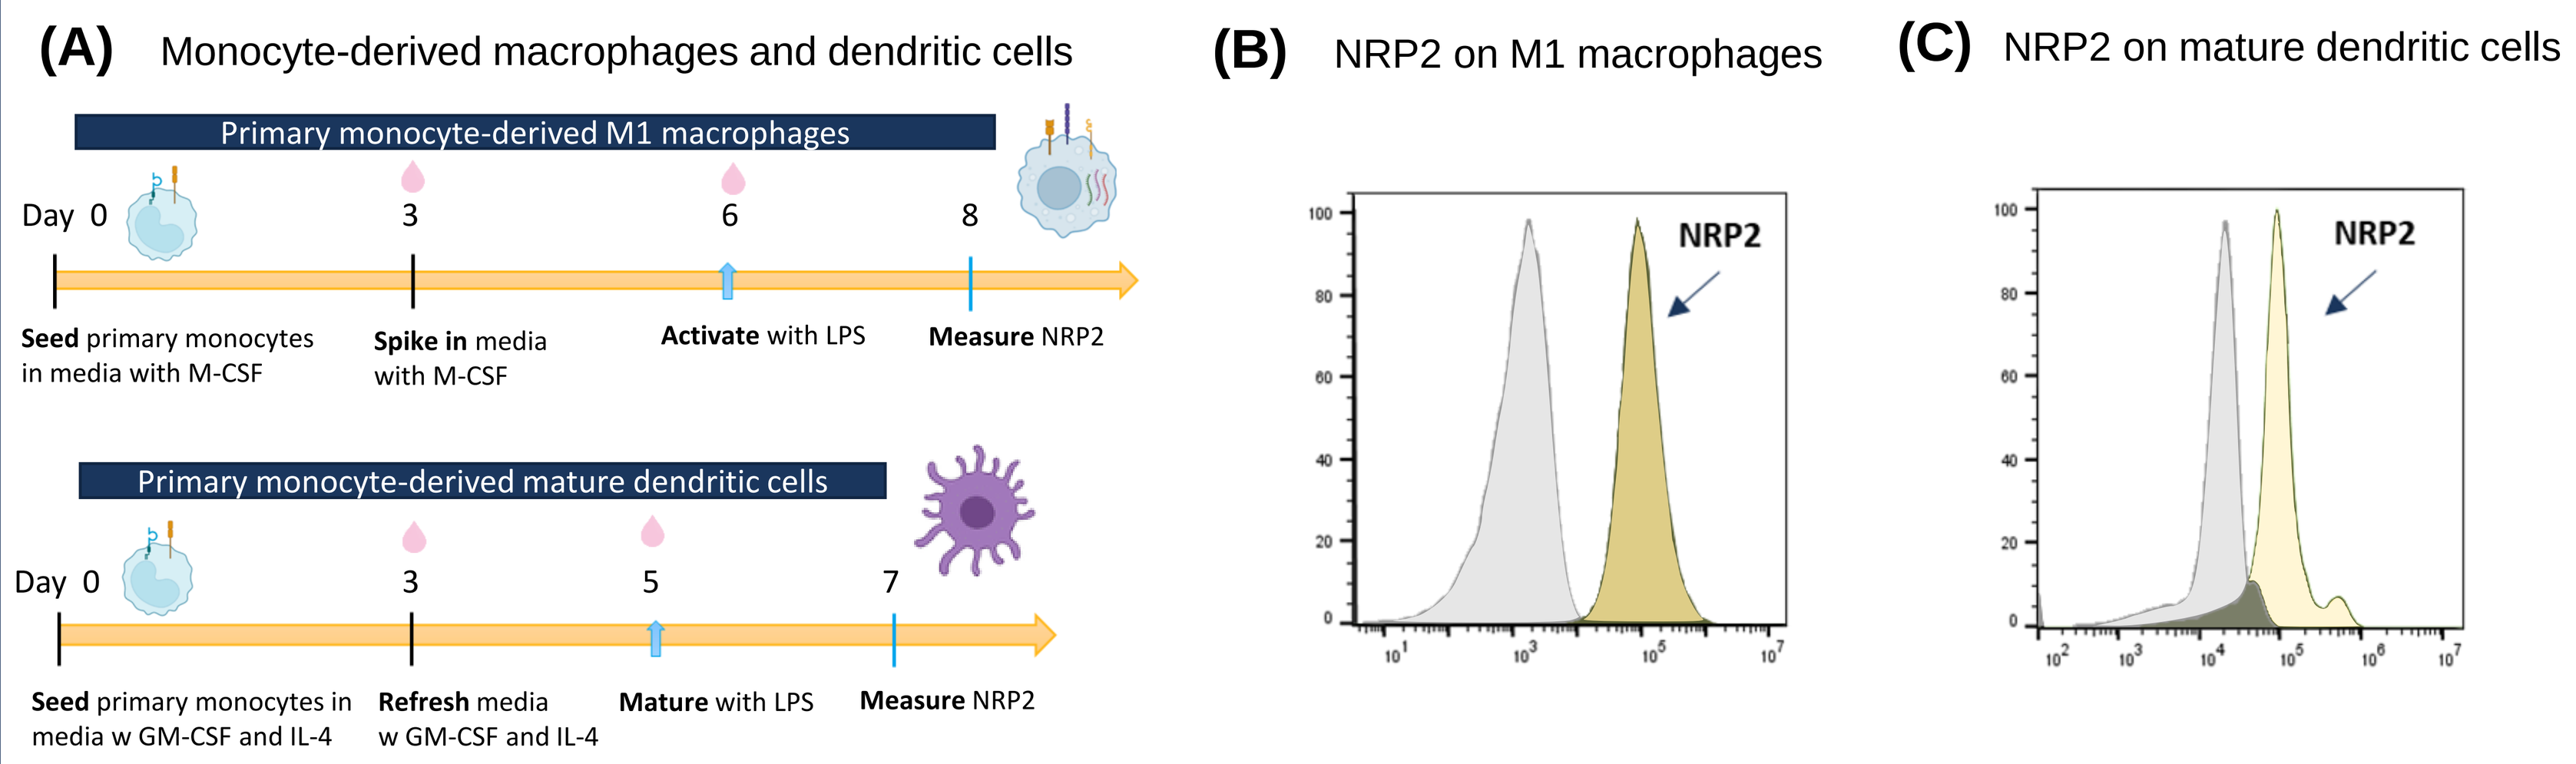

(A) Monocyte-derived macrophages and dendritic cells
Primary monocyte-derived M1 macrophages
Day 0 3 6 8
Seed primary monocytes in media with M-CSF
Spike in media with M-CSF
Activate with LPS
Measure NRP2
Primary monocyte-derived mature dendritic cells
Day 0 3 5 7
Seed primary monocytes in media w GM-CSF and IL-4
Refresh media w GM-CSF and IL-4
Mature with LPS
Measure NRP2
(B) NRP2 on M1 macrophages
(C) NRP2 on mature dendritic cells
NRP2

Figure 3. (A) Schematic showing generation of M1 macrophages and mature dendritic cells from primary human monocytes (B) NRP2 expression by flow cytometry on M1 macrophages. (C) NRP2 expression by flow cytometry on mature dendritic cells.

NRP2 is expressed on circulating monocytes and macrophages in pulmonary sarcoidosis granulomas

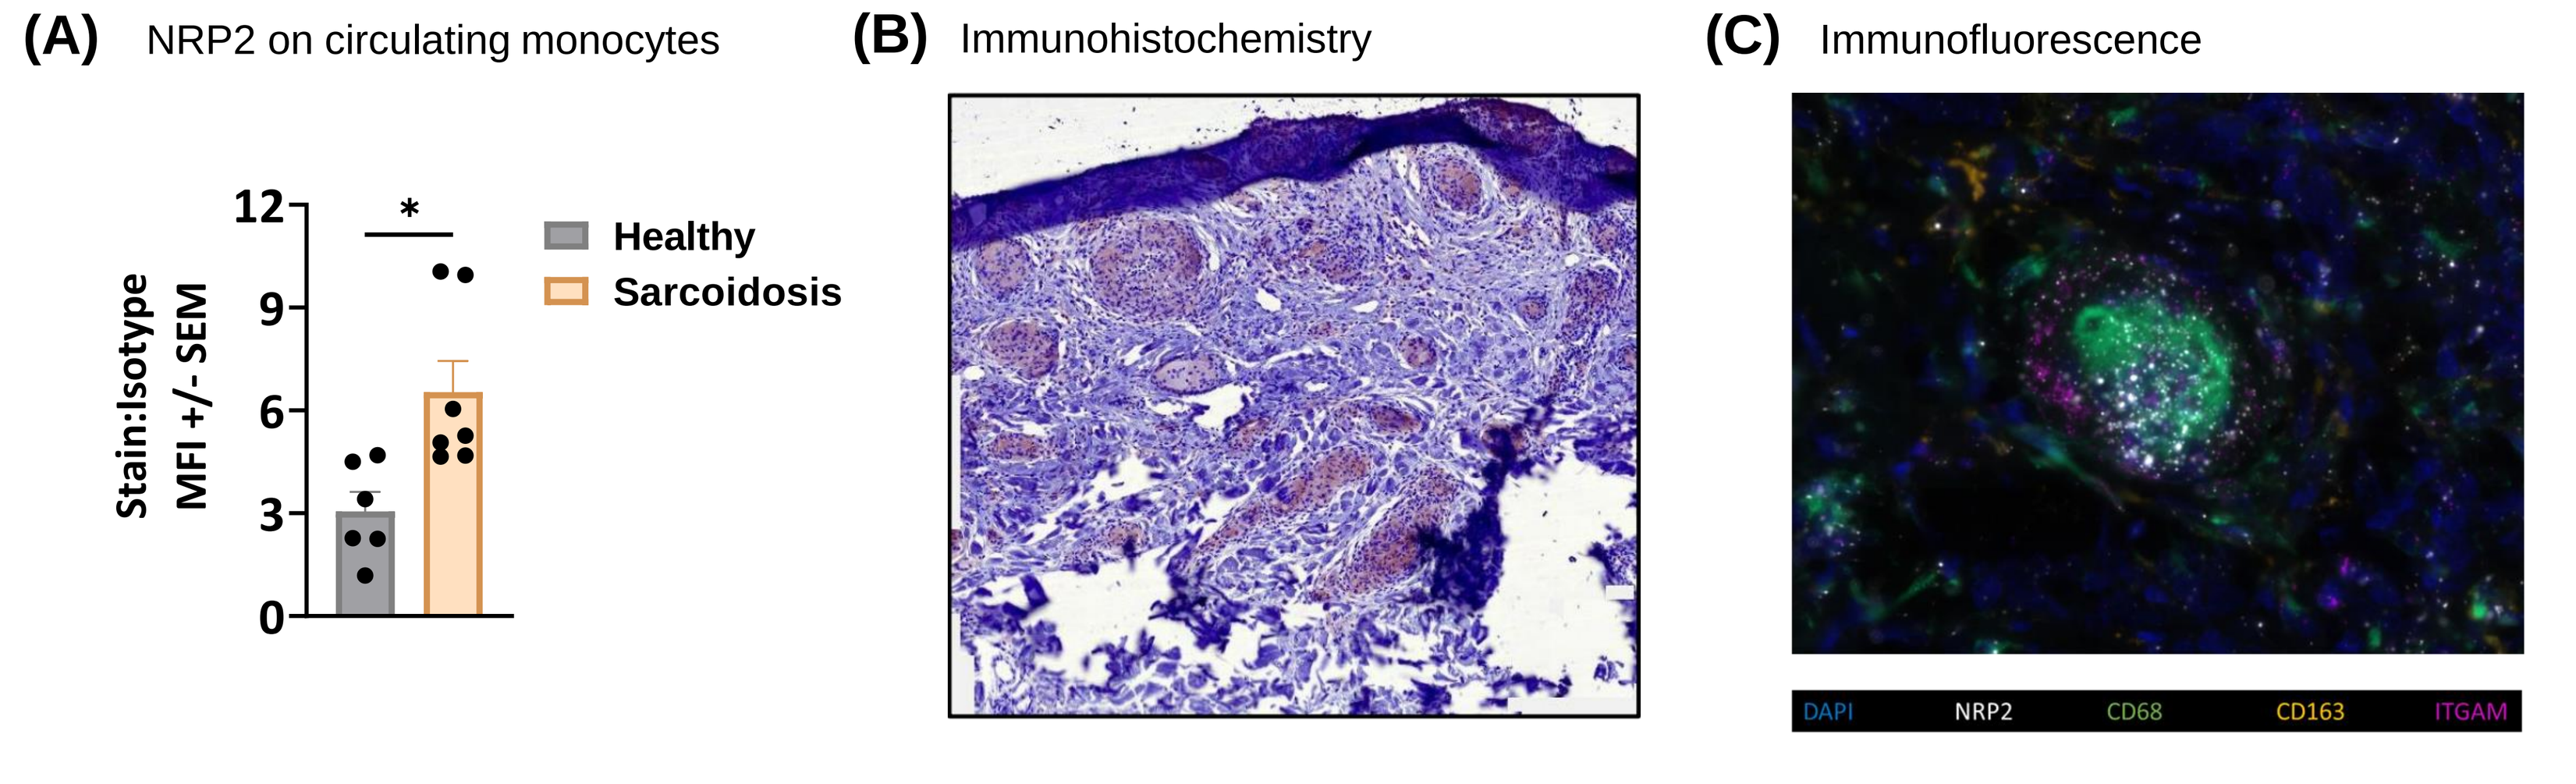

(A) NRP2 on circulating monocytes
(B) Immunohistochemistry
(C) Immunofluorescence
Stain:Isotype MFI +/- SEM
Healthy
Sarcoidosis
DAPI
NRP2
CD68
CD163
ITGAM

Figure 4. High levels of NRP2 are expressed on circulating monocytes in pulmonary sarcoidosis patients and concentrated within myeloid cells of pulmonary granulomas. (A) NRP2 cell surface expression in circulating CD14+ monocytes from healthy donors and pulmonary sarcoidosis patients. (B) NRP2 (brown) is concentrated in granulomas from pulmonary sarcoidosis patients.[4] (C) NRP2 RNA (white dots) is colocalized with macrophage markers CD68 (green) and CD163 (orange) in granulomas from pulmonary sarcoidosis patients.

PBMC from pulmonary sarcoidosis patients are highly inflammatory at baseline

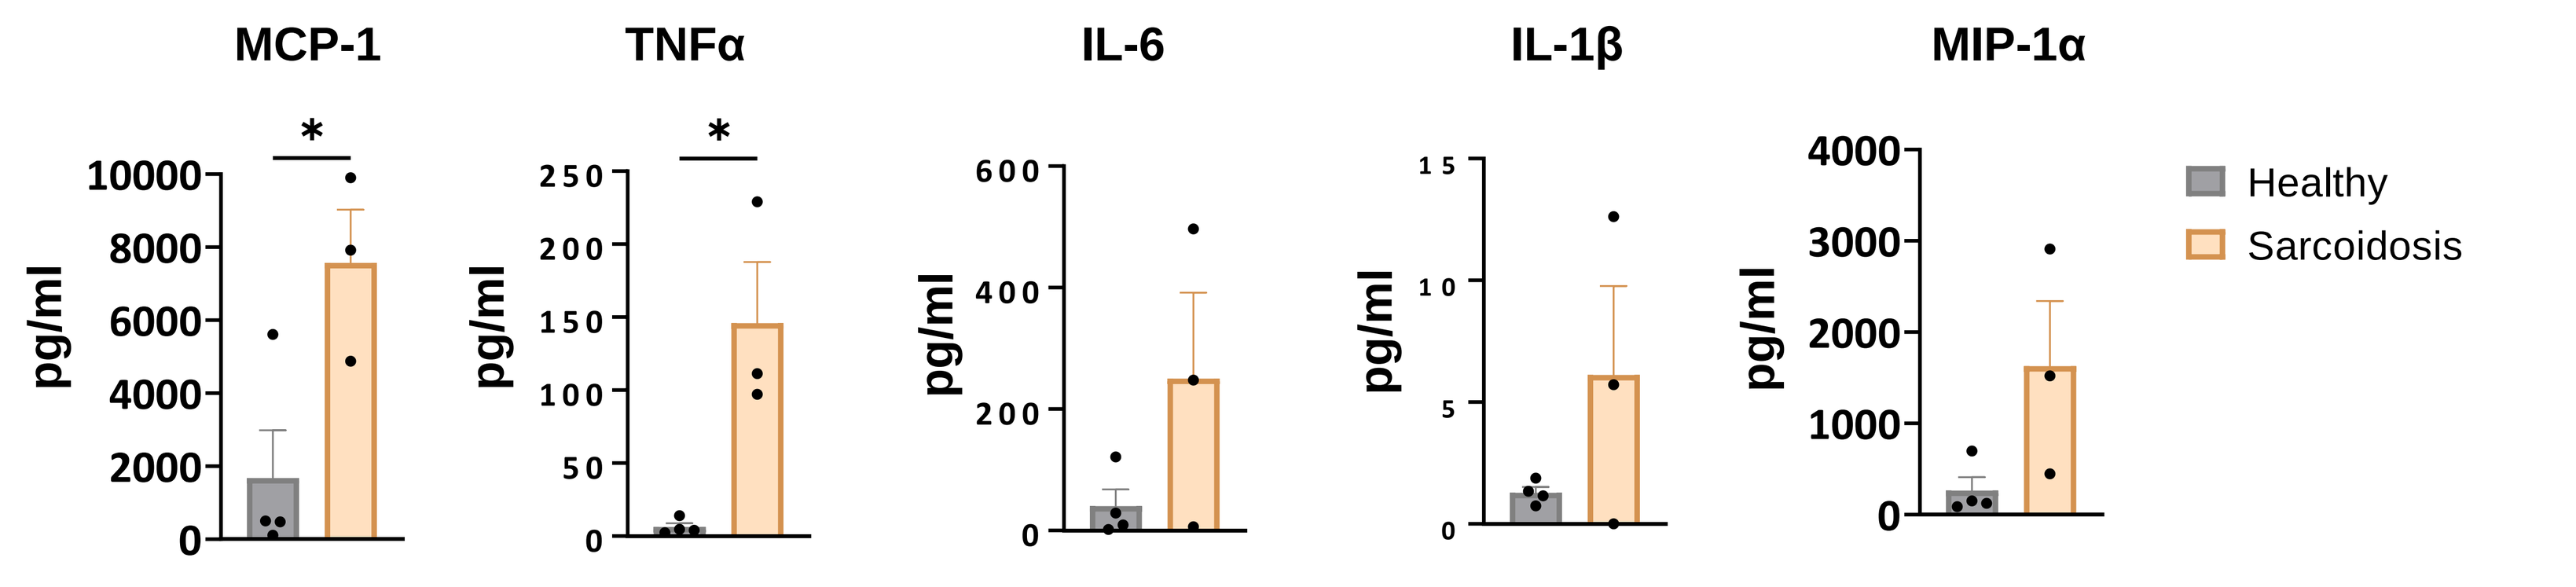

MCP-1
TNFα
IL-6
IL-1β
MIP-1α
pg/ml
Healthy
Sarcoidosis

Figure 5. PBMC from healthy donors and pulmonary sarcoidosis patients were cultured overnight and supernatants were evaluated for secreted cytokines by immunoassay (MesoScale Discovery).

Pulmonary sarcoidosis patients have a dysregulated response to inflammatory stimuli

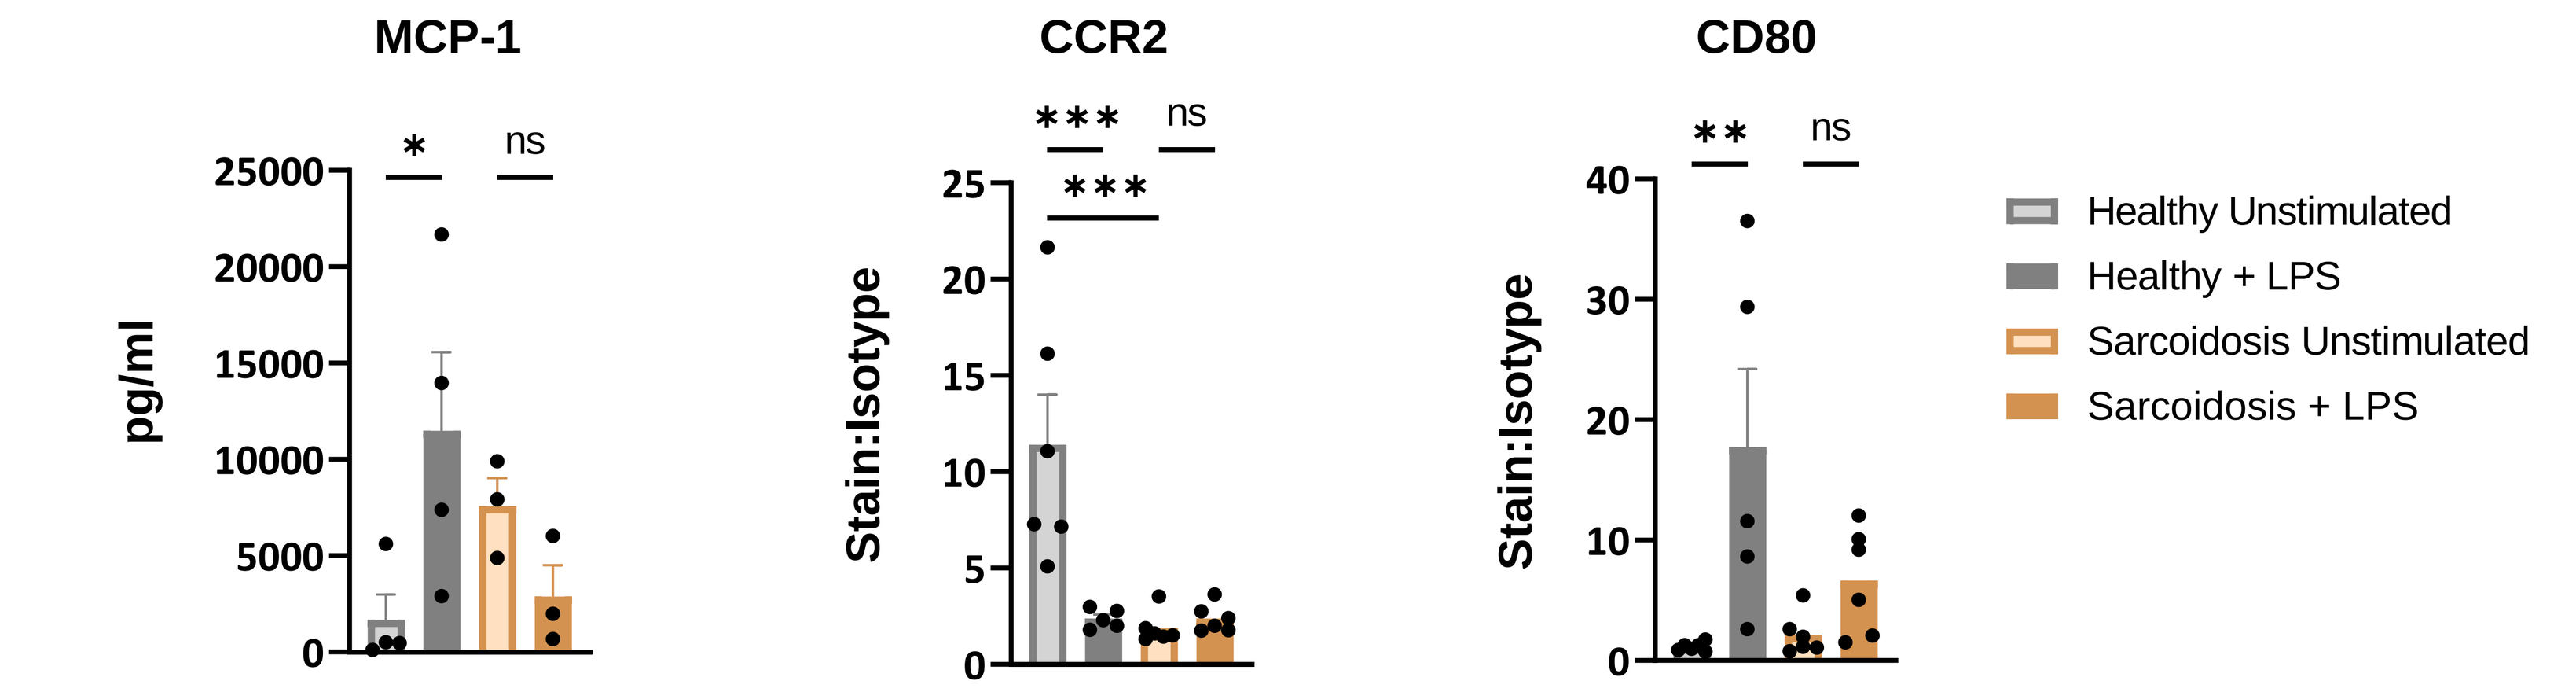

MCP-1
CCR2
CD80
pg/ml
Stain:Isotype
Healthy Unstimulated
Healthy + LPS
Sarcoidosis Unstimulated
Sarcoidosis + LPS

Figure 6. PBMC from healthy donors and pulmonary sarcoidosis patients were cultured overnight with or without 100ng/ml LPS. Supernatants were evaluated for MCP-1 secretion by immunoassay (MesoScale Discovery). CCR2 and CD80 expression on CD14+ monocytes were evaluated by flow cytometry.

Efzofitimod Modulates Immune responses in Myeloid Cells

Efzofitimod effects morphology and downregulates pro-inflammatory pathways

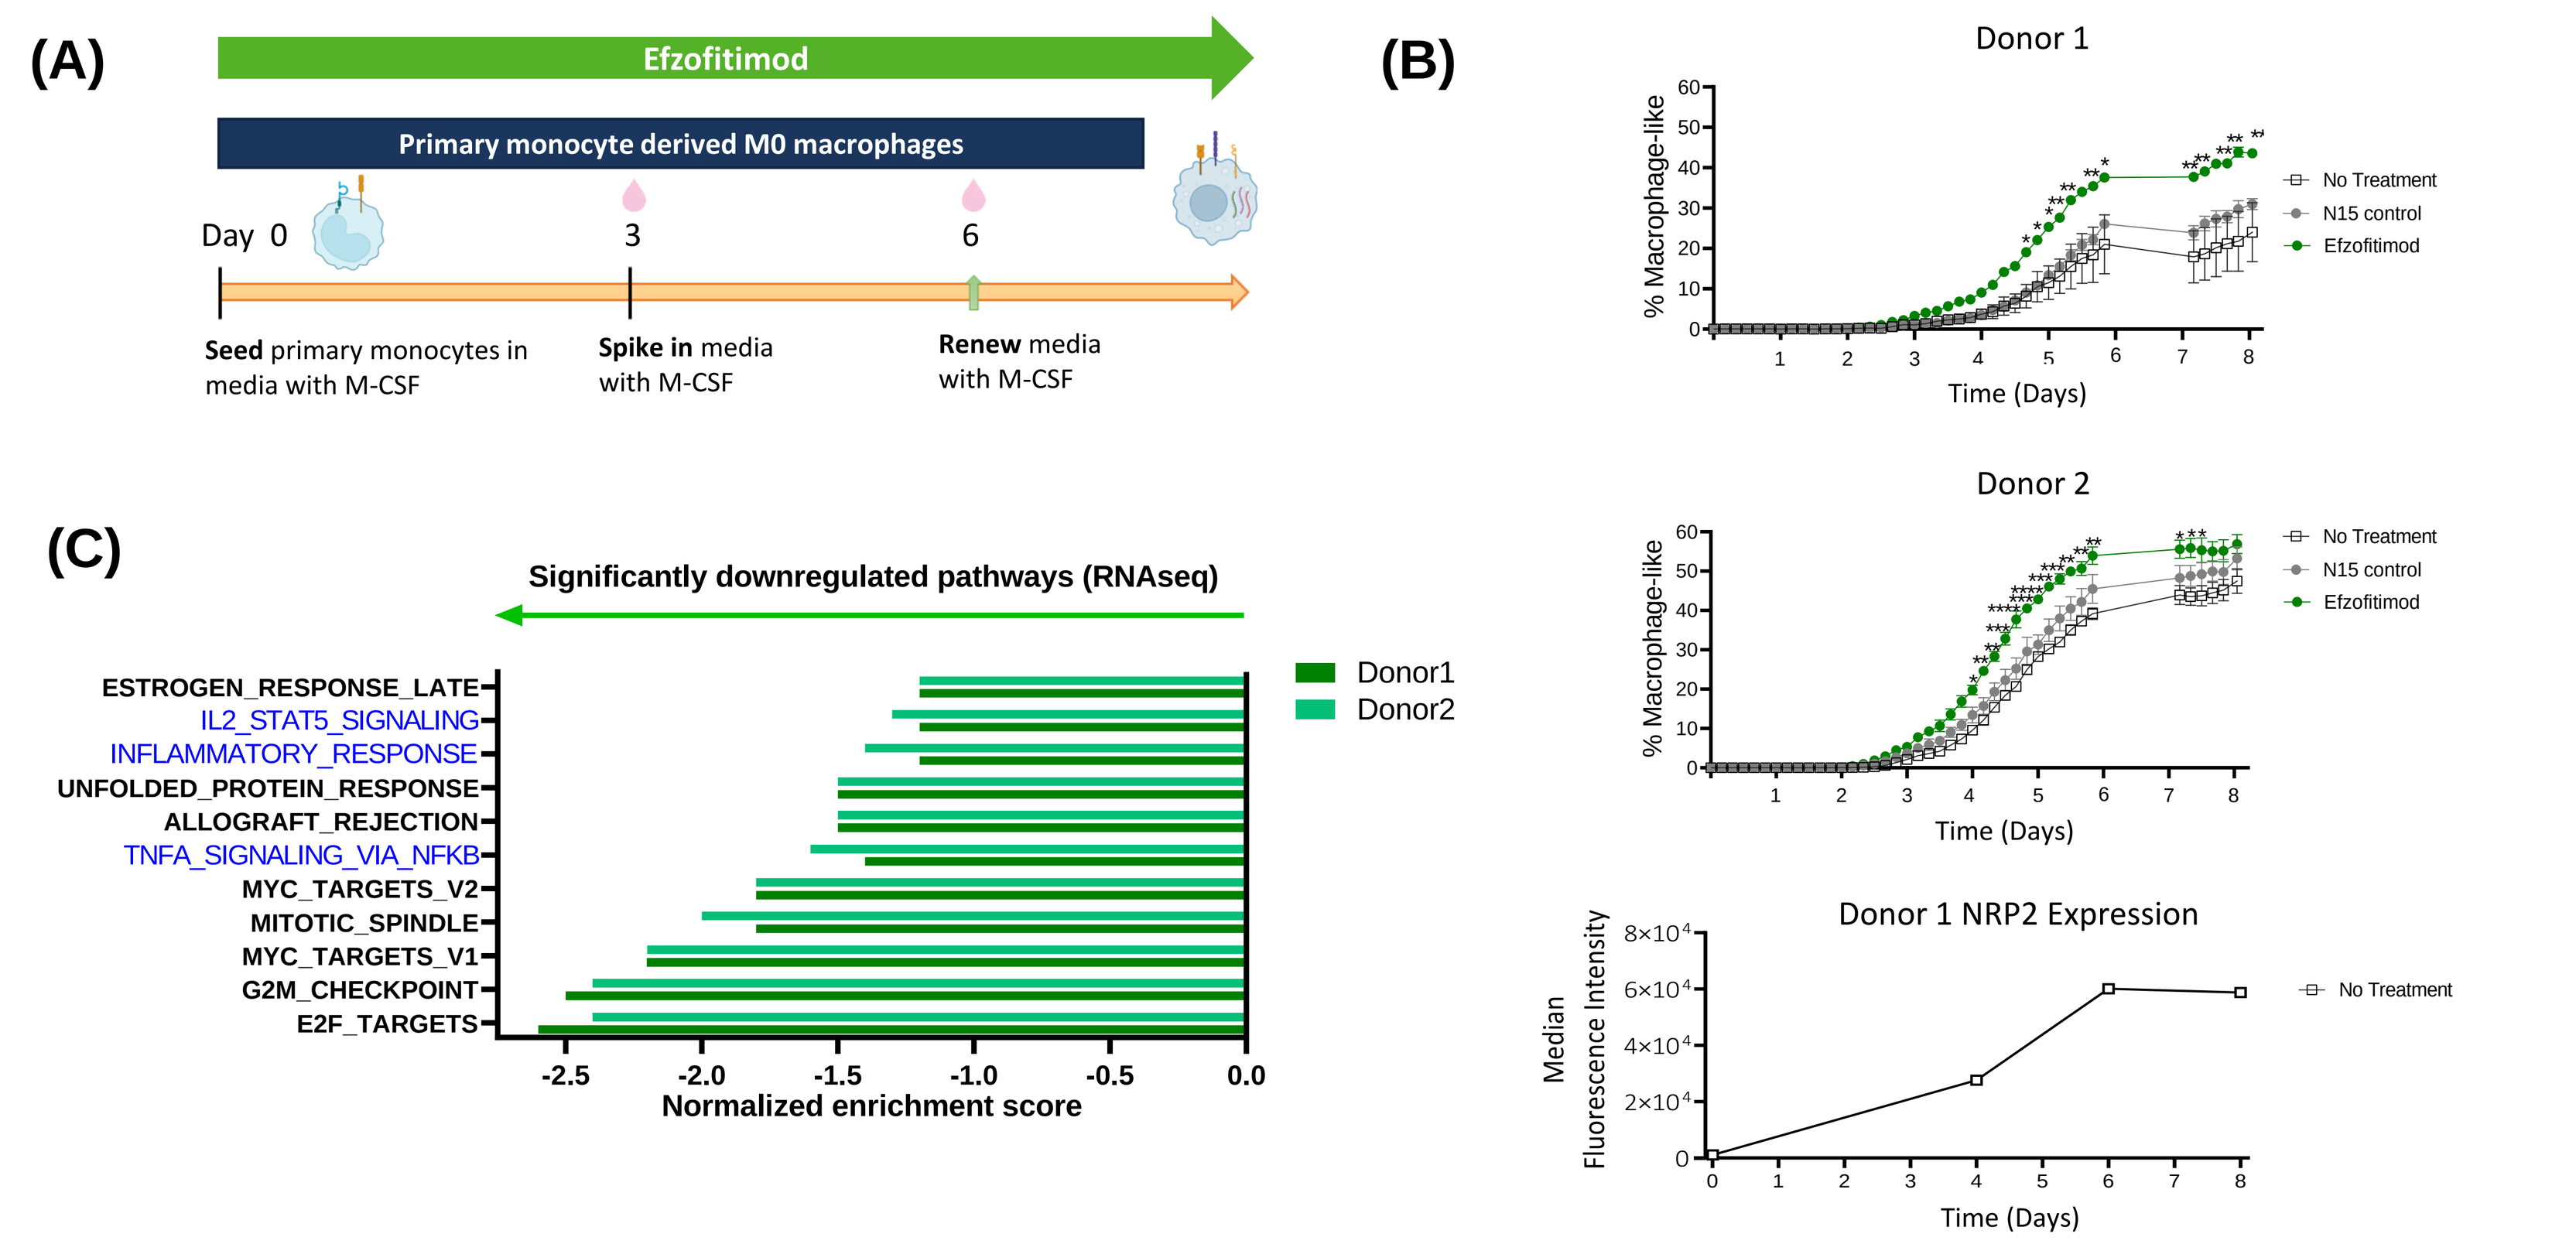

(A)
Efzofitimod
Primary monocyte derived M0 macrophages
Day 0 3 6
Seed primary monocytes in media with M-CSF
Spike in media with M-CSF
Renew media with M-CSF
(B)
Donor 1
Donor 2
% Macrophage-like
Time (Days)
No Treatment
N15 control
Efzofitimod
Donor 1 NRP2 Expression
Median Fluorescence Intensity
(C)
Significantly downregulated pathways (RNAseq)
ESTROGEN_RESPONSE_LATE
IL2_STAT5_SIGNALING
INFLAMMATORY_RESPONSE
UNFOLDED_PROTEIN_RESPONSE
ALLOGRAFT_REJECTION
TNFA_SIGNALING_VIA_NFKB
MYC_TARGETS_V2
MITOTIC_SPINDLE
MYC_TARGETS_V1
G2M_CHECKPOINT
E2F_TARGETS
Donor1
Donor2
Normalized enrichment score

Figure 7. (A) Schematic showing generation of M0 macrophages from primary human monocytes in the presence of 1μM N15 control or 1μM efzofitimod. (B) Morphology change was monitored by images captured every 3 hours by Incucyte (Sartorius). NRP2 expression was measured by flow cytometry at day of seeding (day 0), day 4, day 6, and day 8. (C) RNAseq analysis showing inflammatory pathways downregulated by efzofitimod in monocyte-derived M0 macrophages.

Efzofitimod regulates the MCP-1 (CCL2) / CCR2 monocyte chemotaxis pathway

(A) Summary of efzofitimod's effects

Protein category	Protein name	Protein function	Efzofitimod clinical data - serum biomarkers	Gene expression by RNA-seq/ Fluidigm/qPCR Donor1	Gene expression Donor2	Cell surface receptor staining Donor1	Cell surface receptor staining Donor2	Secreted protein 2 Donors
Cytokines	MCP-1/CCL2	Chemokine for monocytes	Down	Down	Down			Down
	TNF	Inflammatory cytokine	Down	Down	Down			Down
	IL-6	Inflammatory cytokine	Down	n.d.	n.d.			Down
	IL1B	Inflammatory cytokine		Down	Down			Down
	MIG/CXCL9	Inflammatory chemokine		n.d.	n.d.			Down
Receptors	CD14	TLR4 co-receptor		Down	Down	Down	Down	Down
	CCR2	Chemokine receptor		n.d.	n.d.	Down	Down	
	CCR7	Chemokine receptor		Down	Down	Down	Down	
	CXCR4	Chemokine receptor		Down	Down	Down	Down	
	CD206/MRC1	Macrophage marker		-	Down	Down	Down	
	CD28	Co-stimulatory receptor		Down	Down			
	TLR2	Receptor for lipoproteins		Down	Down			
	TLR4	Receptor for LPS		Down	Down			

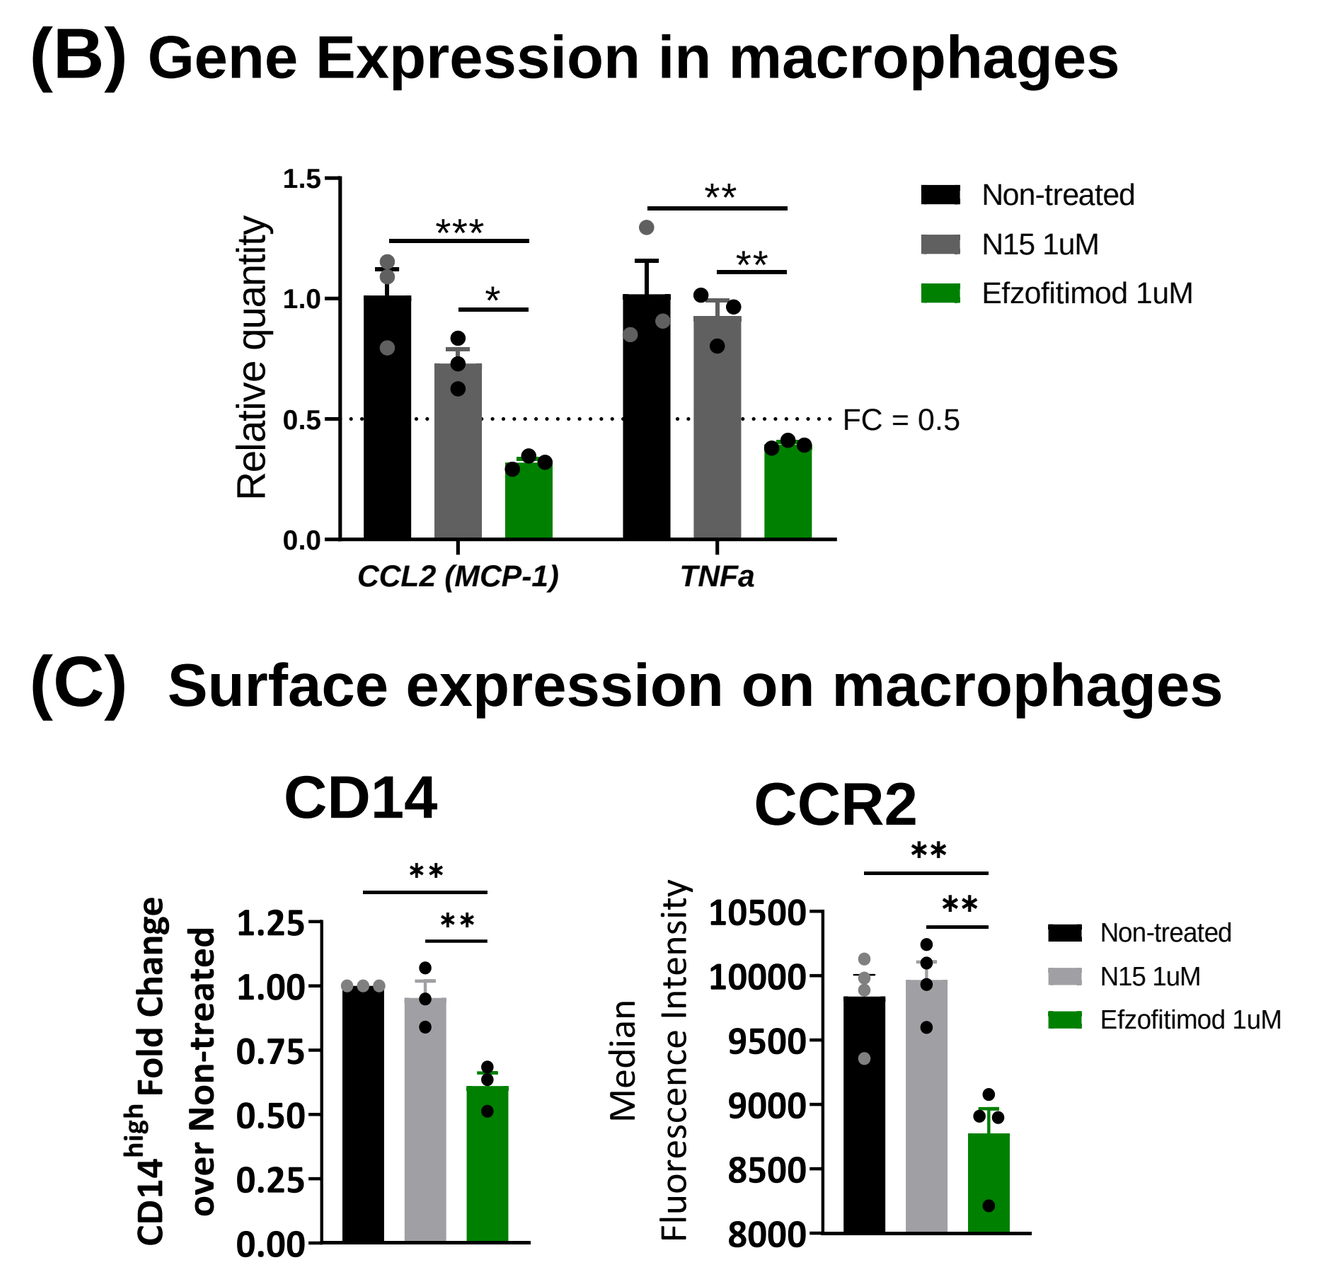

(B) Gene Expression in macrophages
Relative quantity
FC = 0.5
CCL2 (MCP-1)
TNFa
Non-treated
N15 1uM
Efzofitimod 1uM
(C) Surface expression on macrophages
CD14
CCR2
CD14high Fold Change over Non-treated
Median Fluorescence Intensity

Figure 8. Efzofitimod downregulates the chemokine (MCP-1) and receptor (CCR2) involved in monocyte recruitment to sites of inflammation. (A) Relative quantity of CCL2 (MCP-1) and TNFa RNA transcripts in monocyte-derived macrophages. (B) CD14 and CCR2 surface expression on monocyte-derived macrophages.

Conclusions

- Efzotifimod is a novel immunomodulator that has demonstrated clinical proof of concept in a Phase 2a pulmonary sarcoidosis trial
- NRP2, the sole binding partner to efzofitimod, is highly expressed in circulating monocytes and granulomas of sarcoidosis patients
- Efzofitimod promotes macrophage-like morphology of myeloid cells and downregulates inflammatory cytokines and receptors that are dysregulated in sarcoidosis
- Efzofitimod's immunomodulatory effects may apply to other ILDs which share common immune pathologies to sarcoidosis

Proposed Mechanism of Action

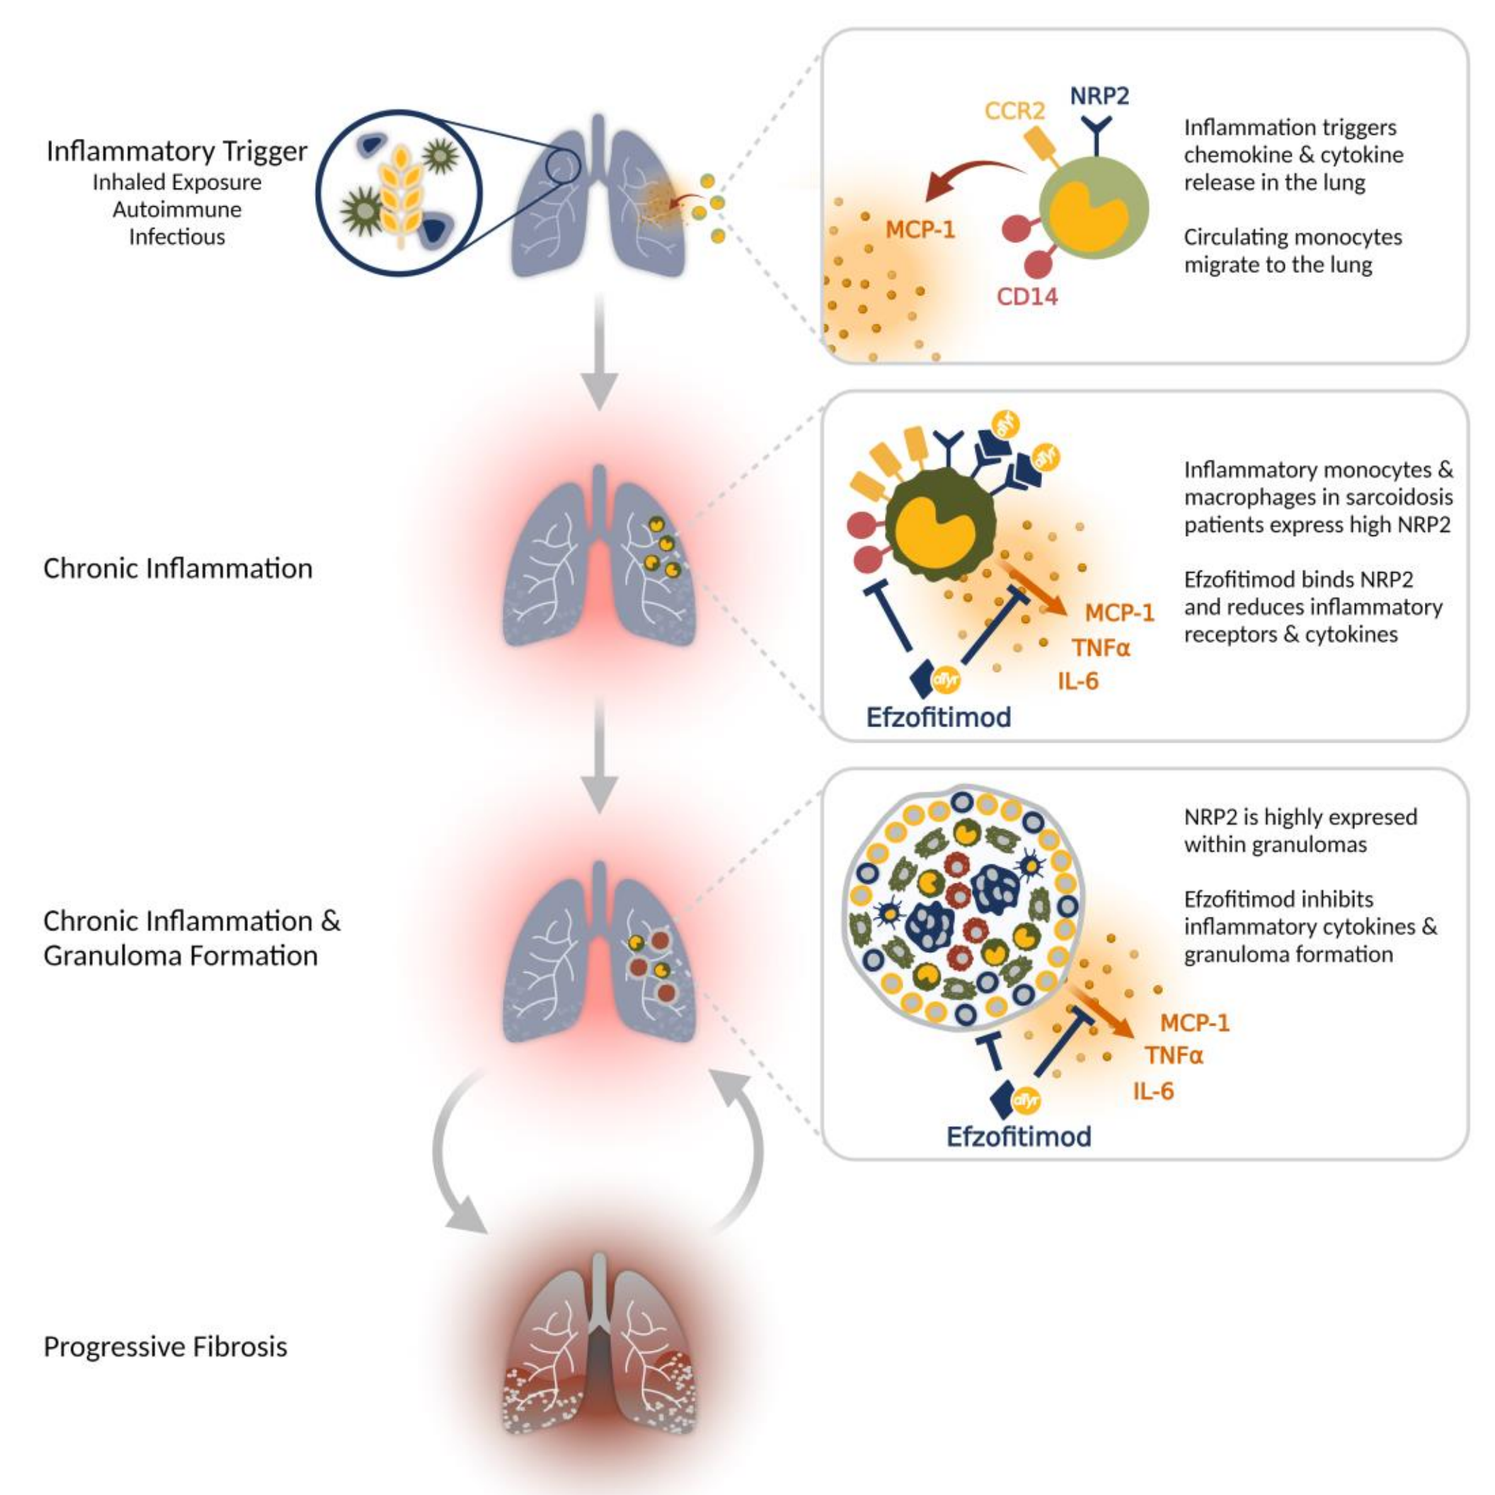

Inflammatory Trigger
Inhaled Exposure
Autoimmune
Infectious
Inflammation triggers chemokine & cytokine release in the lung
Circulating monocytes migrate to the lung
Chronic Inflammation
Inflammatory monocytes & macrophages in sarcoidosis patients express high NRP2
Efzofitimod binds NRP2 and reduces inflammatory receptors & cytokines
Chronic Inflammation & Granuloma Formation
NRP2 is highly expresed within granulomas
Efzofitimod inhibits inflammatory cytokines & granuloma formation
Progressive Fibrosis
Efzofitimod
MCP-1
TNFα
IL-6