Efzofitimod: A Novel Therapeutic Candidate for SSc-ILD

D. Siefker, Z. Xu, C. Burkart, M. Ferrer, C. Polizzi, K. Rauch, S. Klopp-Savino, L. Guy, E. Yu, L. Glendening, Y. Qing, Y. Chong, R. Adams, L. Nangle

aTyr Pharma - San Diego (USA)

Abstract/Aims

Systemic sclerosis-associated interstitial lung disease (SSc-ILD) is an autoimmune disease characterized by inflammation and fibrosis of skin and lungs. Efzofitimod is an immunomodulator that has demonstrated clinical proof-of-concept (POC) in pulmonary sarcoidosis, a related ILD. Efzofitimod binds to neuropilin-2 (NRP2), a membrane protein upregulated at sites of inflammation, most notably on myeloid cells which are involved in the pathology of both sarcoidosis and SSc-ILD. We aimed to explore the therapeutic potential of efzofitimod in animal models of SSc-ILD and by evaluating myeloid cells from SSc-ILD patients. Efzofitimod reduced lung inflammation and fibrosis in multiple animal models of ILD, including SSc-ILD. Similar to sarcoidosis[1], circulating monocytes from SSc-ILD patients expressed higher levels of NRP2 than healthy donors. NRP2 was also highly expressed on macrophages in skin from SSc patients. Efzofitimod downregulated inflammatory cytokines and receptors on primary monocyte-derived macrophages, including CD14, a membrane protein for which high expression is associated with the progression of SSc diseases[2,3]. Given the similarities between SSc-ILD and sarcoidosis and the anti-inflammatory effect of efzofitimod on myeloid cells, together with its anti-inflammatory and anti-fibrotic effects in animals and clinical POC in sarcoidosis, efzofitimod is a promising therapeutic candidate for SSc-ILD with a novel mechanism of action.

tRNA synthetase drug discovery platform

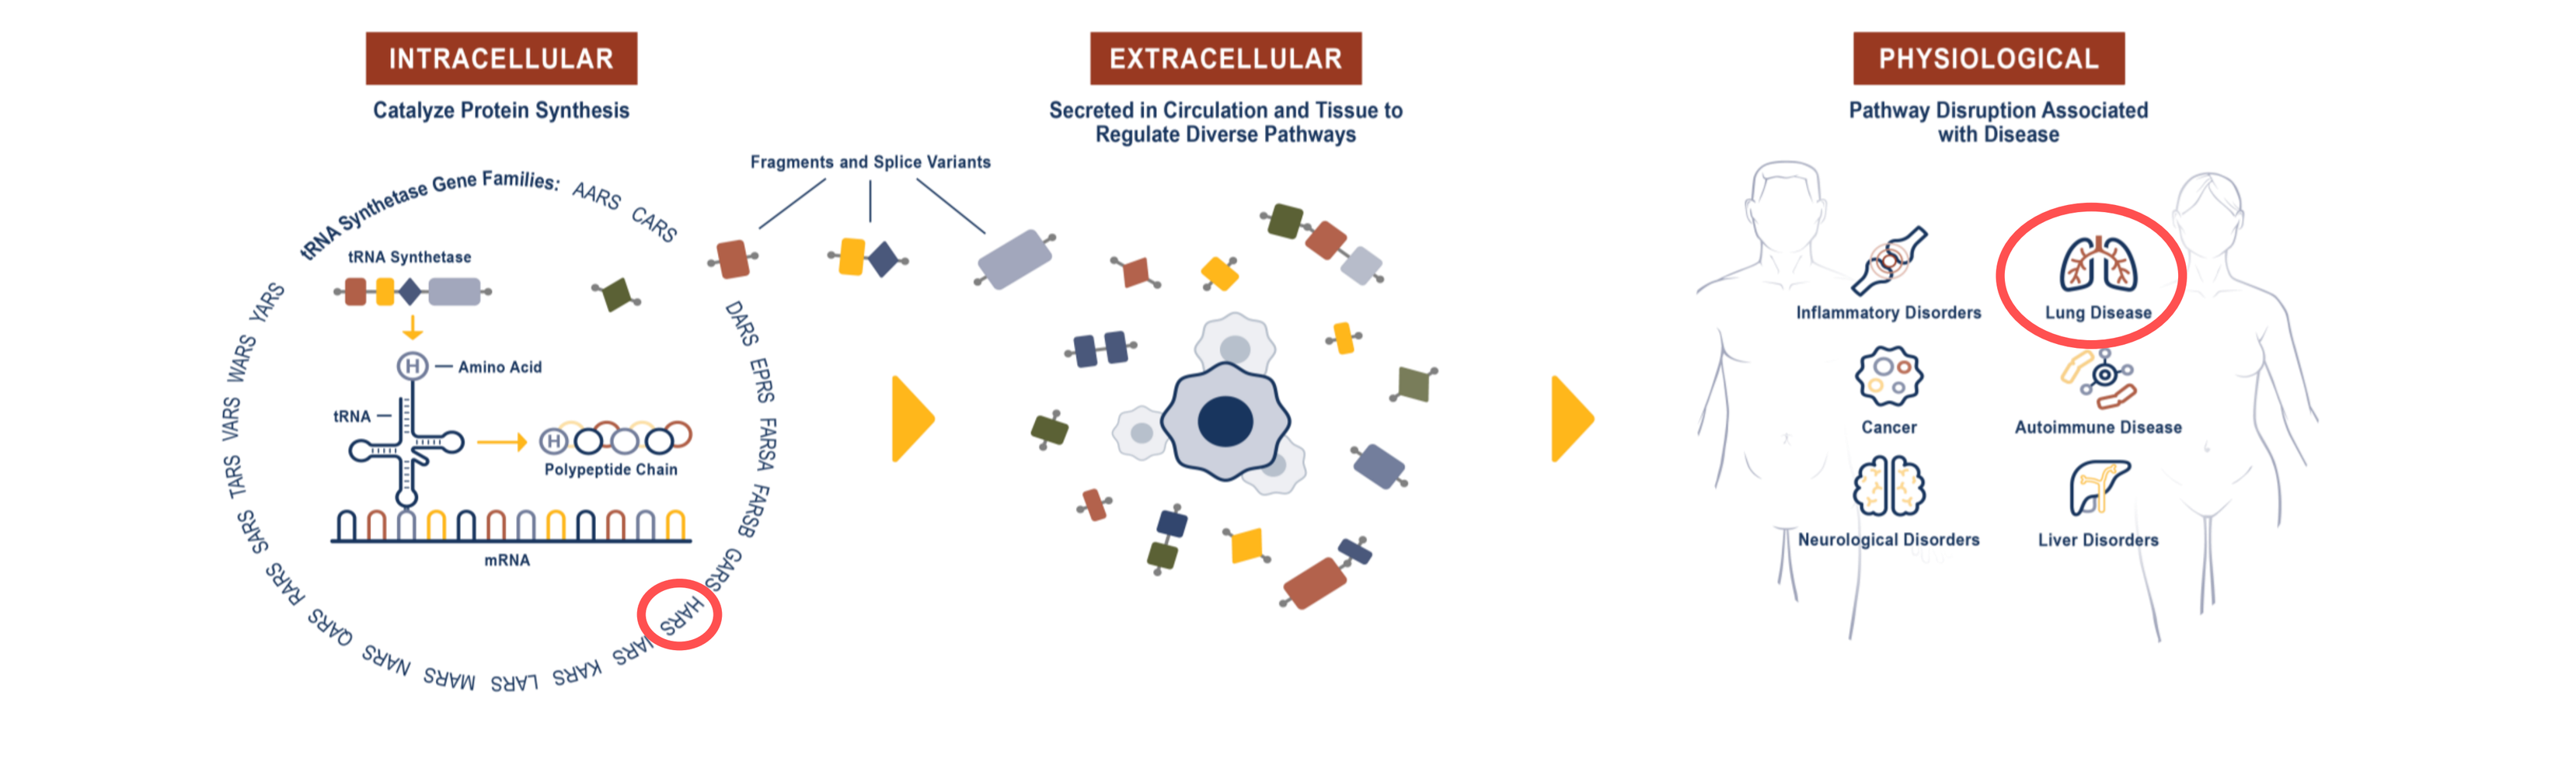
INTRACELLULAR
Catalyze Protein Synthesis
EXTRACELLULAR
Secreted in Circulation and Tissue to Regulate Diverse Pathways
PHYSIOLOGICAL
Pathway Disruption Associated with Disease
Fragments and Splice Variants
tRNA Synthetase Gene Families:
tRNA Synthetase
Amino Acid
tRNA
Polypeptide Chain
mRNA
Inflammatory Disorders
Lung Disease
Cancer
Autoimmune Disease
Neurological Disorders
Liver Disorders

Figure 1. Splice variants and proteolytic fragments of tRNA synthetases have been identified in extracellular spaces, where they have novel functions. aTyr Pharma is engaged in the discovery and development of potential first-in-class medicines based on newly discovered pathways effected by extracellular tRNA synthetases.

Efzofitimod, a novel ligand for Neuropilin-2 (NRP2) receptor

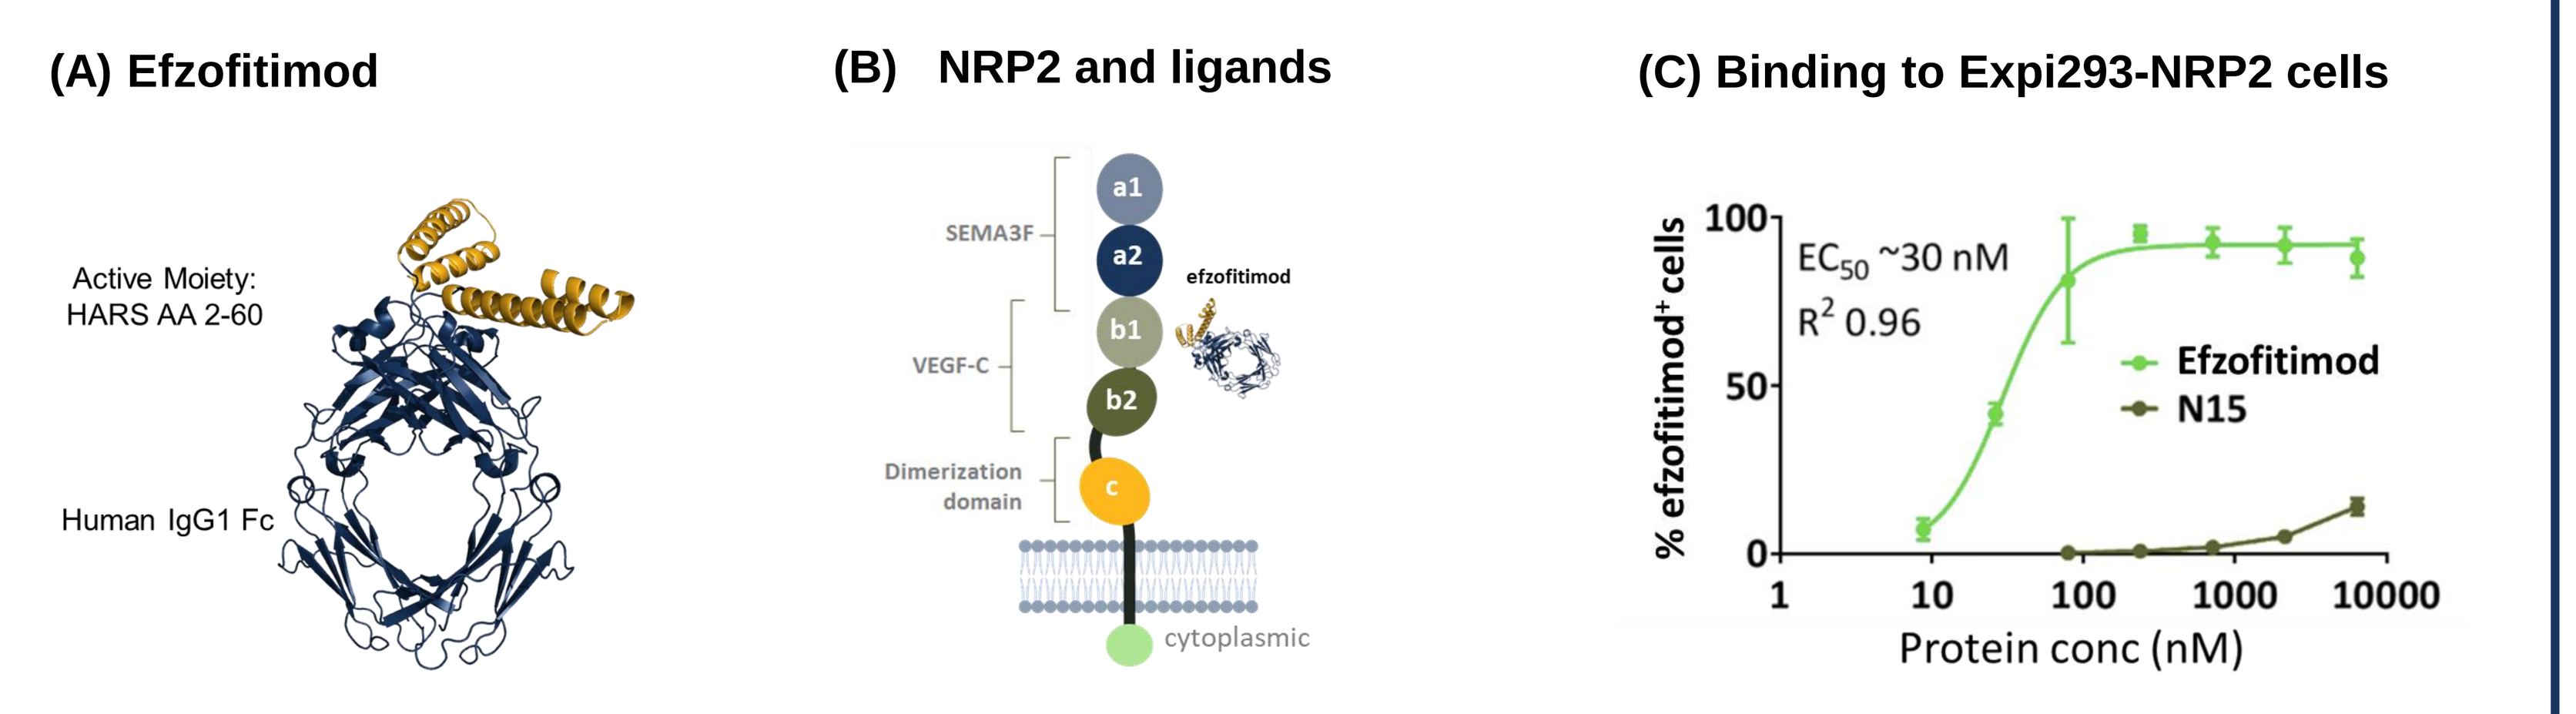
(A) Efzofitimod
Active Moiety: HARS AA 2-60
Human IgG1 Fc
(B) NRP2 and ligands
SEMA3F
VEGF-C
Dimerization domain
a1
a2
b1
b2
c
efzofitimod
cytoplasmic
(C) Binding to Expi293-NRP2 cells
EC50 ~30 nM
R2 0.96
Efzofitimod
N15
% efzofitimod+ cells
Protein conc (nM)

Figure 2. Efzofitimod binds to NRP2 receptor. (A) Efzofitimod is a fusion protein consisting of amino acids 2-60 of Histidyl-tRNA synthetase (HARS) and human IgG1 Fc. (B) Schematic of NRP2 domains and ligands. (C) Efzofitimod binds to Expi293F-NRP2 cells.

Efzofitimod reduces inflammation and fibrosis in rodent models of ILD and improves lung function in ILD patients

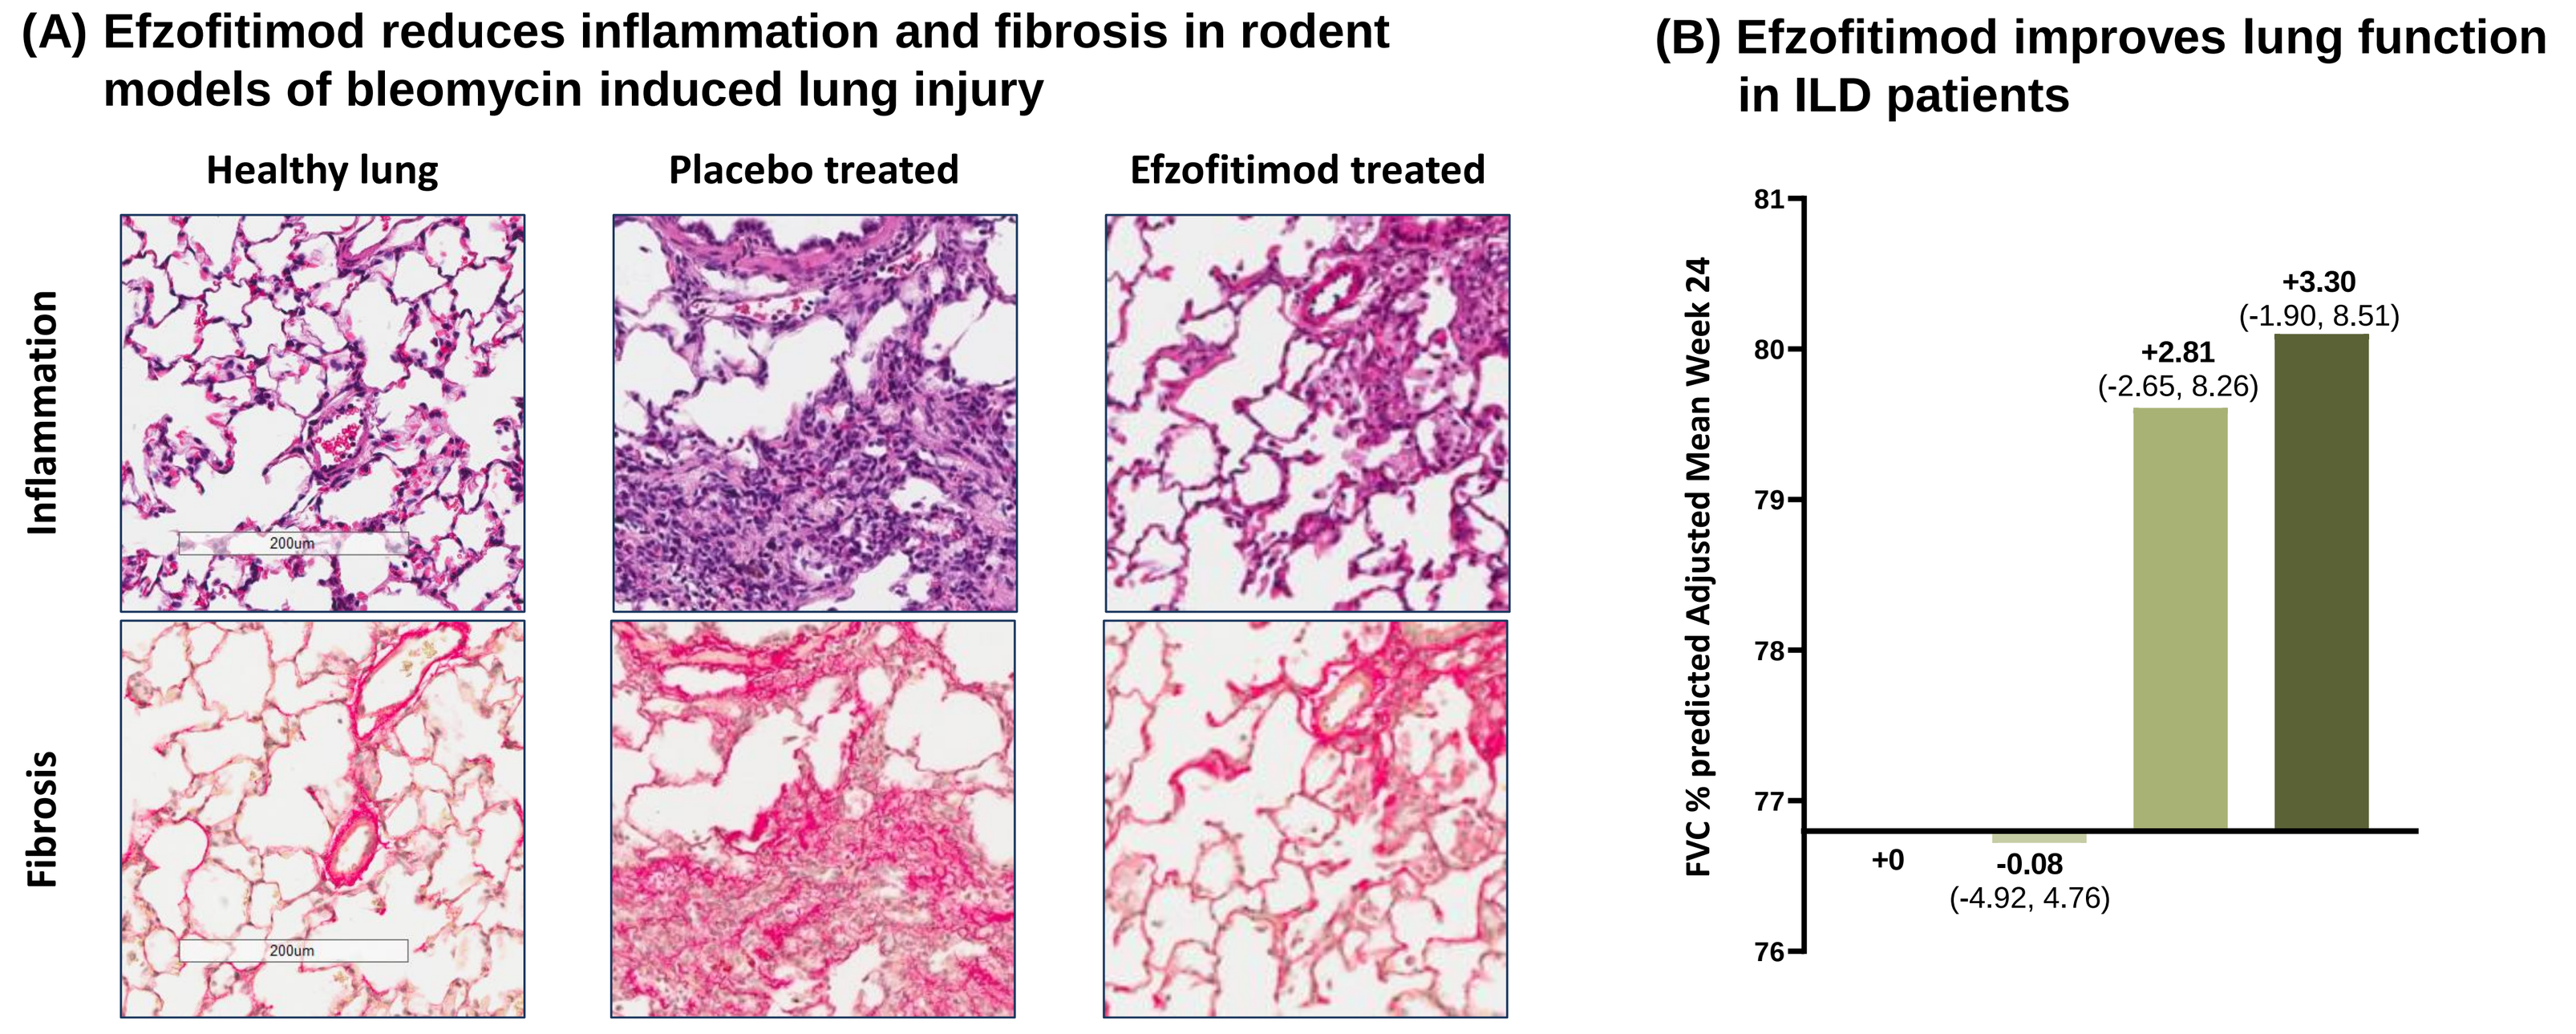
(A) Efzofitimod reduces inflammation and fibrosis in rodent models of bleomycin induced lung injury
Healthy lung
Placebo treated
Efzofitimod treated
Inflammation
Fibrosis
(B) Efzofitimod improves lung function in ILD patients
FVC % predicted Adjusted Mean Week 24
+3.30
(-1.90, 8.51)
+2.81
(-2.65, 8.26)
+0
-0.08
(-4.92, 4.76)

Figure 3. (A) Lungs from a rodent model of bleomycin-induced lung injury were stained with H&E (inflammation) and Picrosirius Red (fibrosis) to evaluate efzofitimod efficacy. (B) In a Phase 1b/2a clinical trial[4] with 37 pulmonary sarcoidosis patients, percent-predicted forced vital capacity improved with 3 and 5mg/kg doses.

Methods and Results

Efzofitimod demonstrated anti-fibrotic effects in lungs of mice in a SSc-ILD model

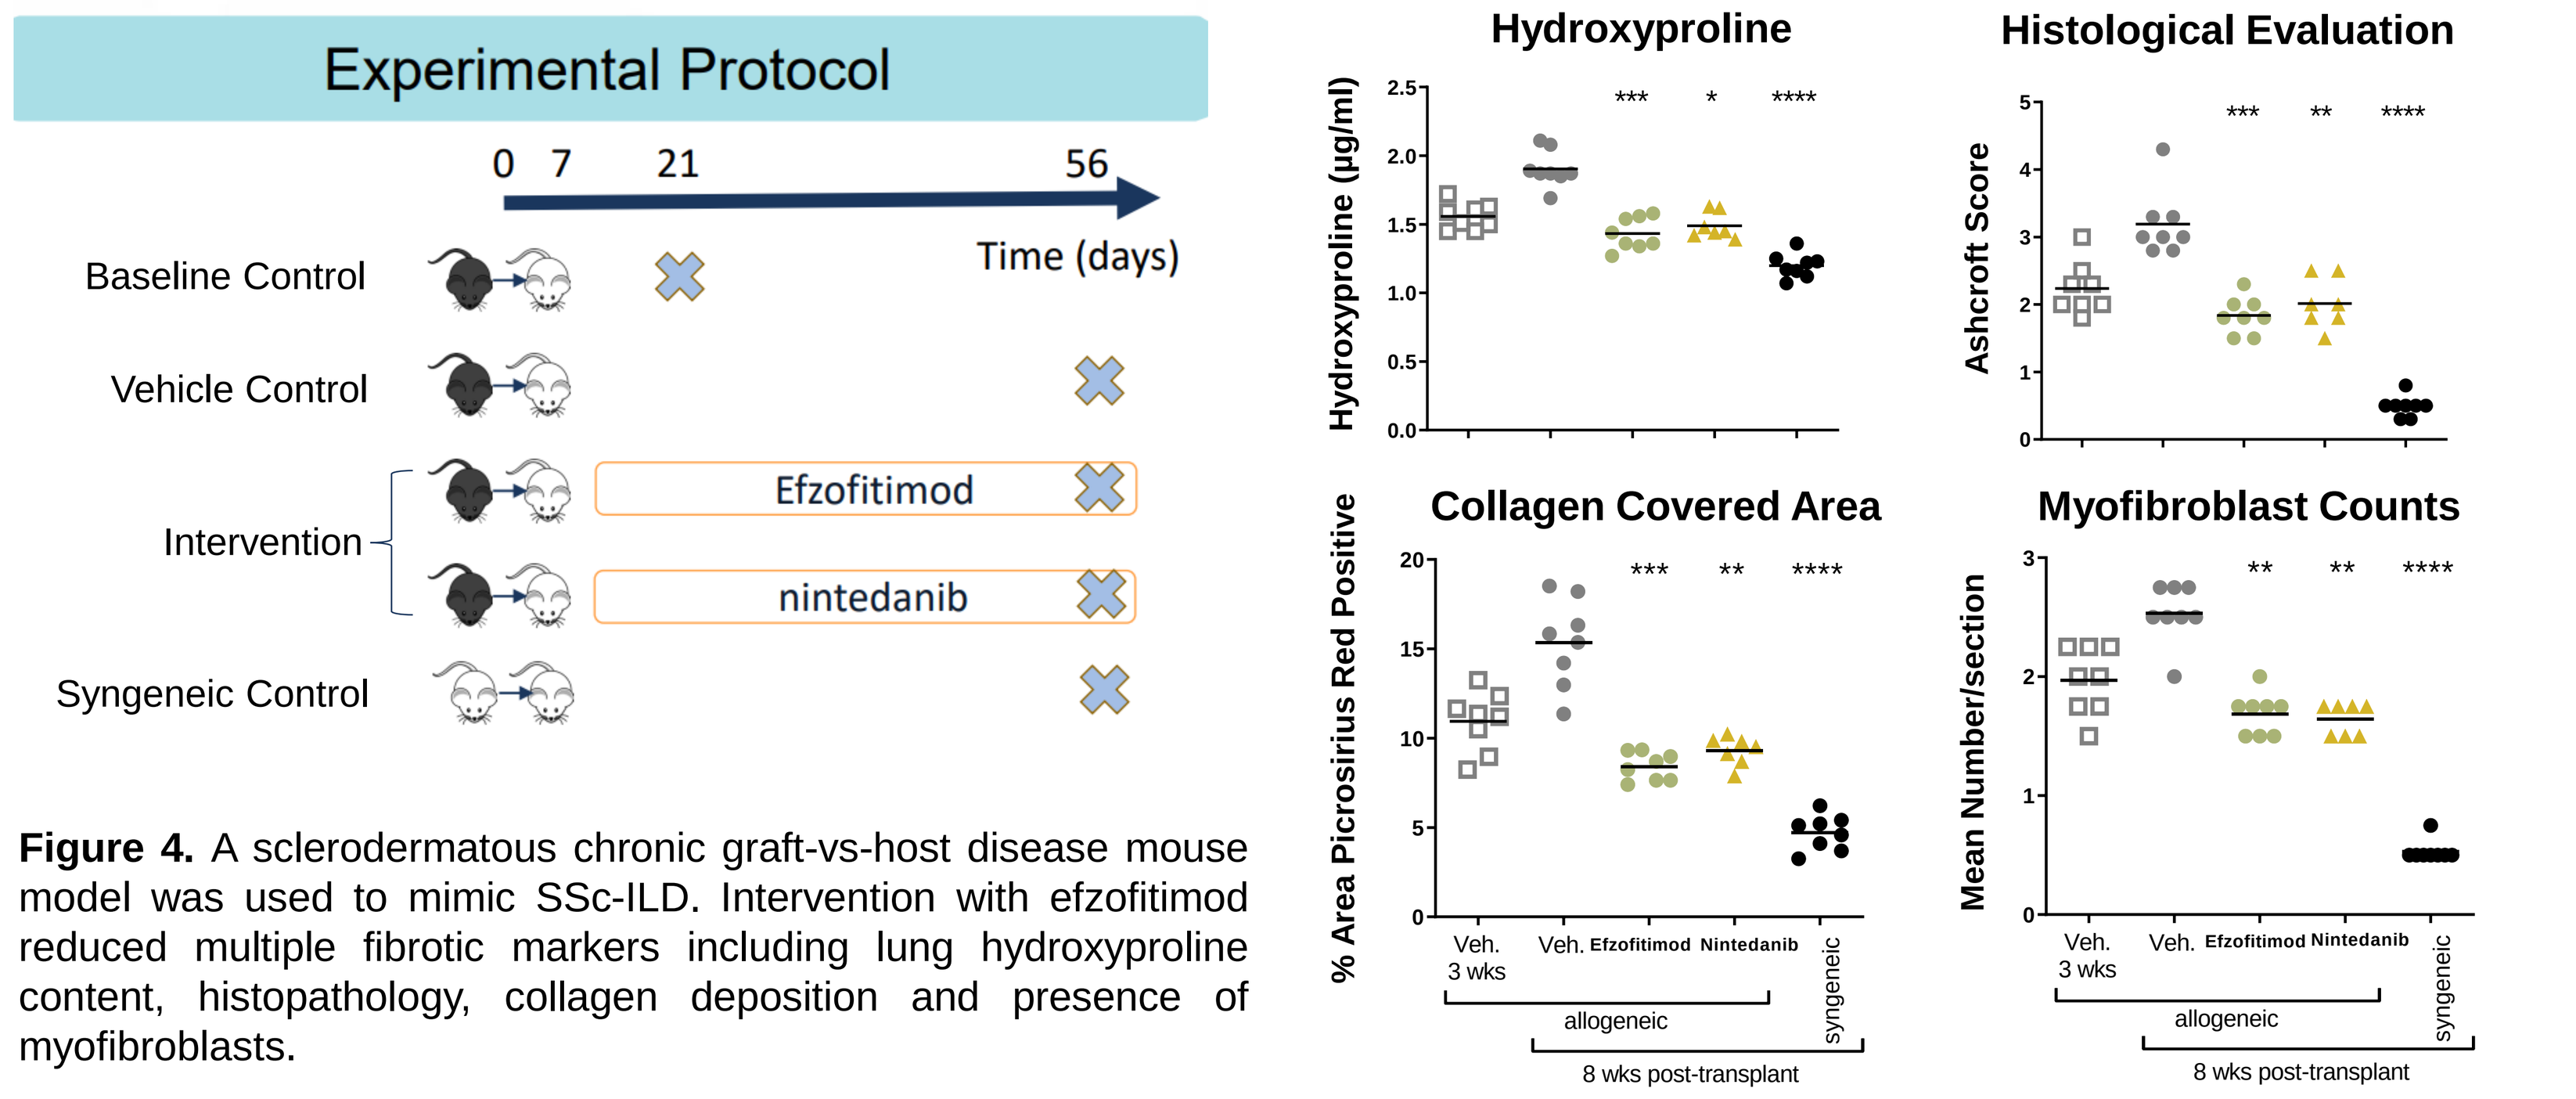
Experimental Protocol
0 7 21 56
Time (days)
Baseline Control
Vehicle Control
Intervention
Efzofitimod
nintedanib
Syngeneic Control
Hydroxyproline
Histological Evaluation
Collagen Covered Area
Myofibroblast Counts
Hydroxyproline (µg/ml)
Ashcroft Score
% Area Picrosirius Red Positive
Mean Number/section
Veh. 3 wks
Veh.
Efzofitimod
Nintedanib
syngeneic
allogeneic
8 wks post-transplant

Figure 4. A sclerodermatous chronic graft-vs-host disease mouse model was used to mimic SSc-ILD. Intervention with efzofitimod reduced multiple fibrotic markers including lung hydroxyproline content, histopathology, collagen deposition and presence of myofibroblasts.

Efzofitimod exhibits anti-inflammatory/anti-fibrotic activities across preclinical ILD models

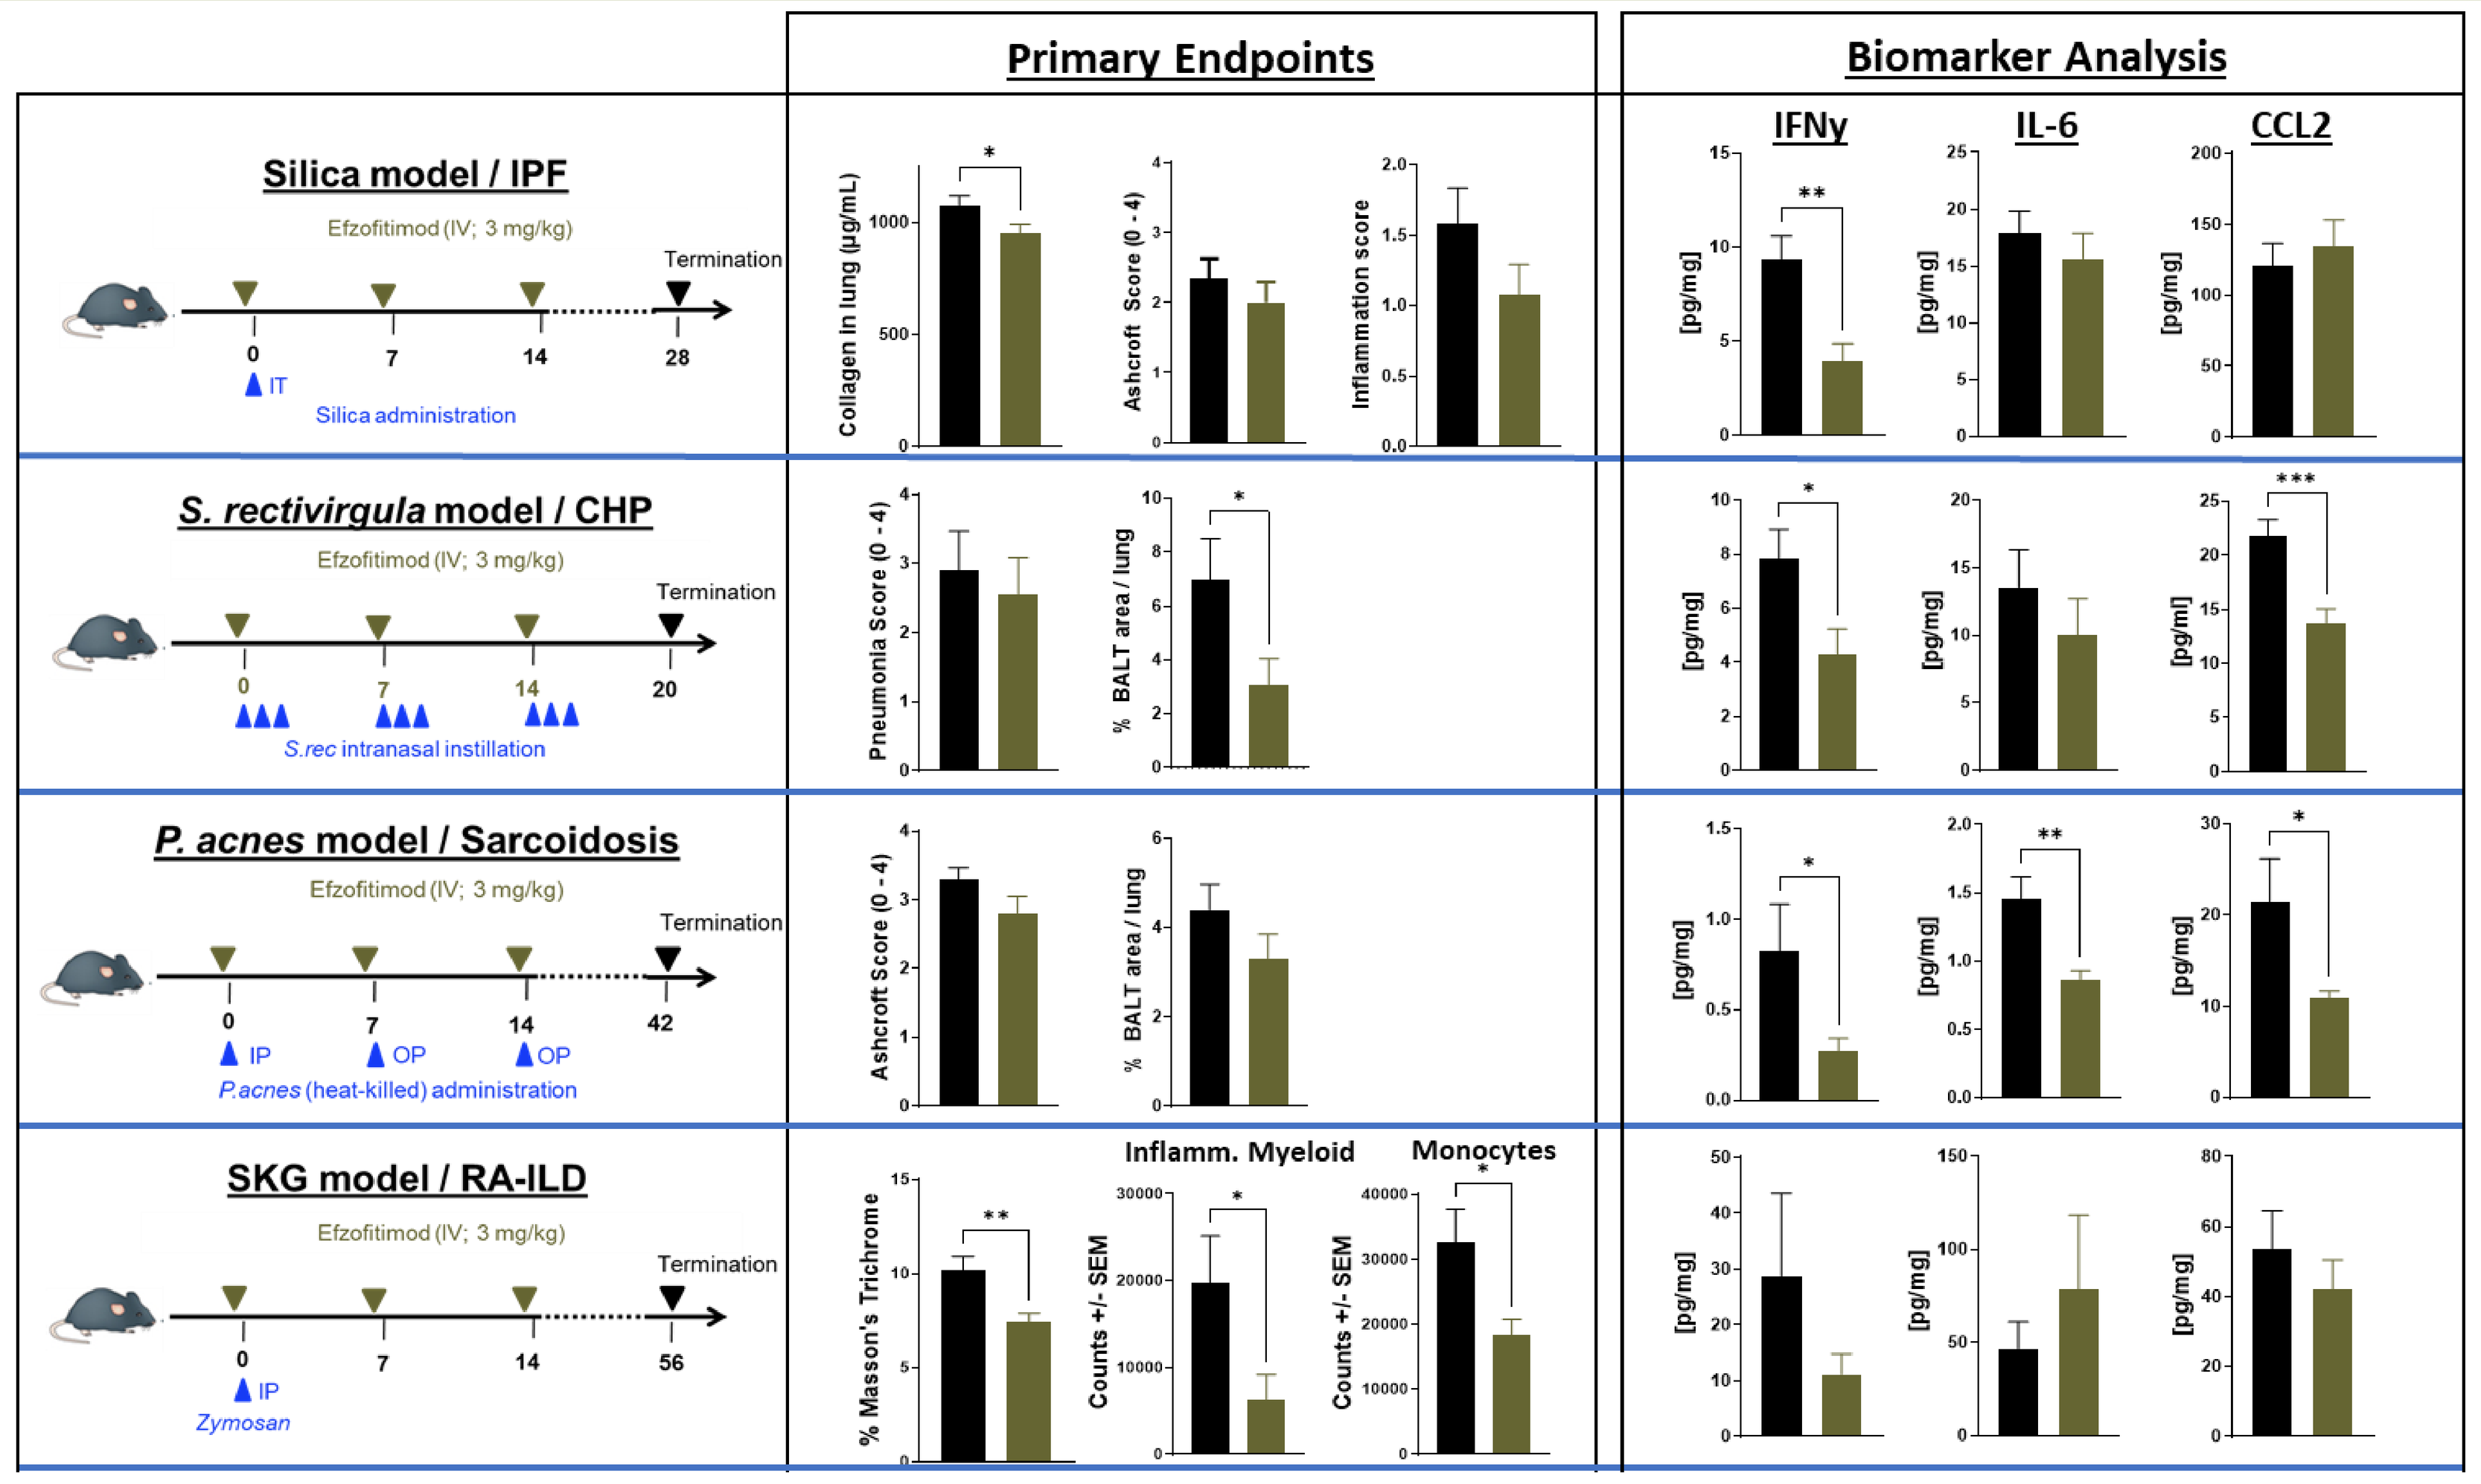
Primary Endpoints
Biomarker Analysis
IFNγ
IL-6
CCL2
Silica model / IPF
Efzofitimod (IV; 3 mg/kg)
Termination
Silica administration
S. rectivirgula model / CHP
S.rec intranasal instillation
P. acnes model / Sarcoidosis
P.acnes (heat-killed) administration
SKG model / RA-ILD
Zymosan
Inflamm. Myeloid
Monocytes

Figure 5. Efzofitimod reduces inflammatory cytokines/chemokines and fibrosis markers in multiple mouse models of ILD including idiopathic pulmonary fibrosis (IPF), chronic hypersensitivity pneumonitis (CHP), pulmonary sarcoidosis, and rheumatoid arthritis-associated ILD (RA-ILD).

NRP2 is highly expressed on activated myeloid cells

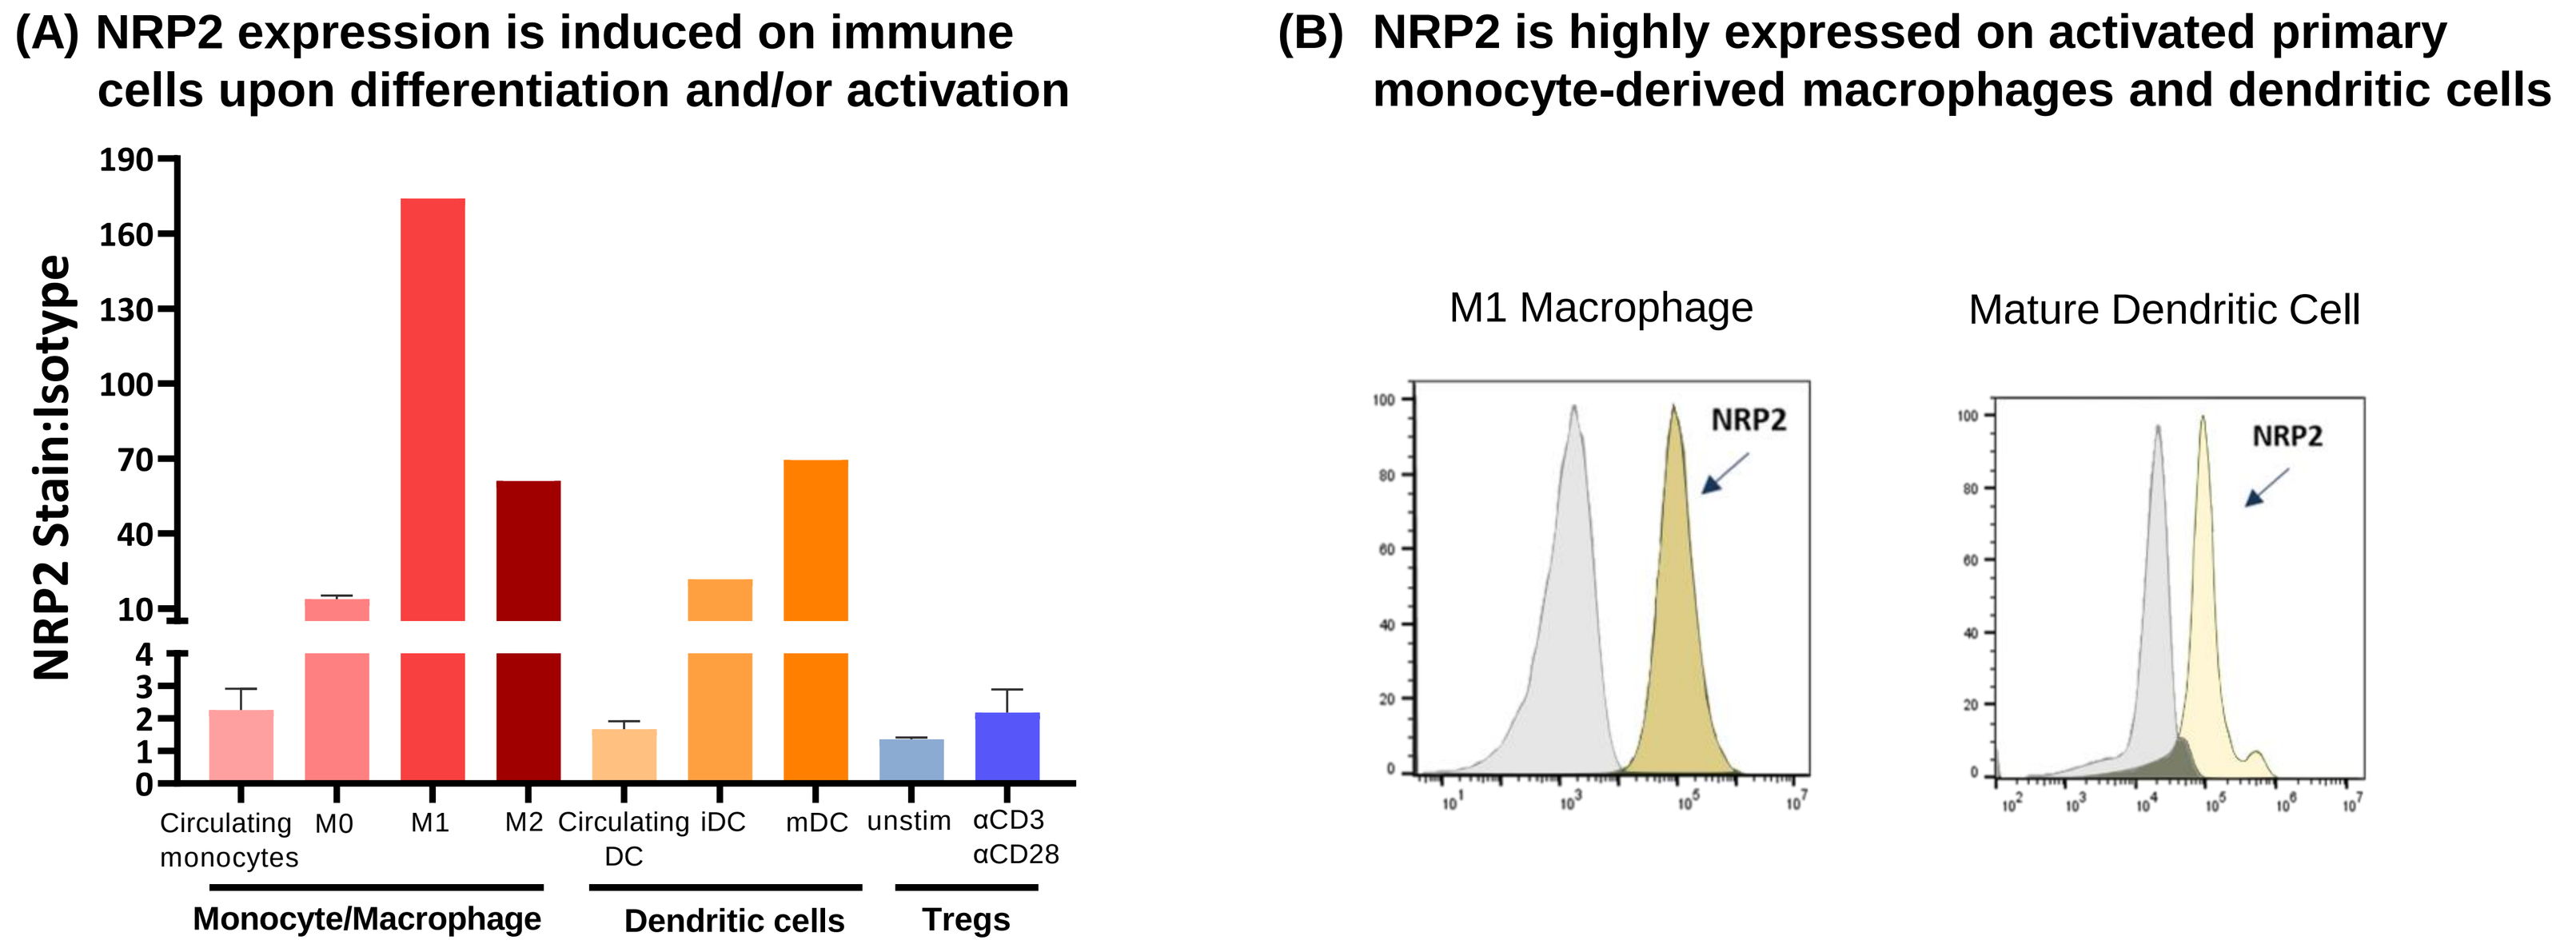
(A) NRP2 expression is induced on immune cells upon differentiation and/or activation
NRP2 Stain:Isotype
Circulating monocytes
M0
M1
M2
Circulating DC
iDC
mDC
unstim
αCD3 αCD28
Monocyte/Macrophage
Dendritic cells
Tregs
(B) NRP2 is highly expressed on activated primary monocyte-derived macrophages and dendritic cells
M1 Macrophage
Mature Dendritic Cell
NRP2

Figure 6. M0, M1 and M2 macrophages or immature DC (iDC) and mature DC (mDC) were generated in vitro from primary monocytes isolated from PBMC, and NRP2 expression was determined by flow cytometry. PBMC were cultured overnight unstimulated or stimulated with anti-CD3 and anti-CD28, and NRP2 expression was determined by flow cytometry on circulating monocytes (CD14+), circulating DC (CD14-CD11c+), and Tregs (CD25+CD127-).

Methods and Results

NRP2 is expressed in circulating monocytes and macrophages in SSc skin

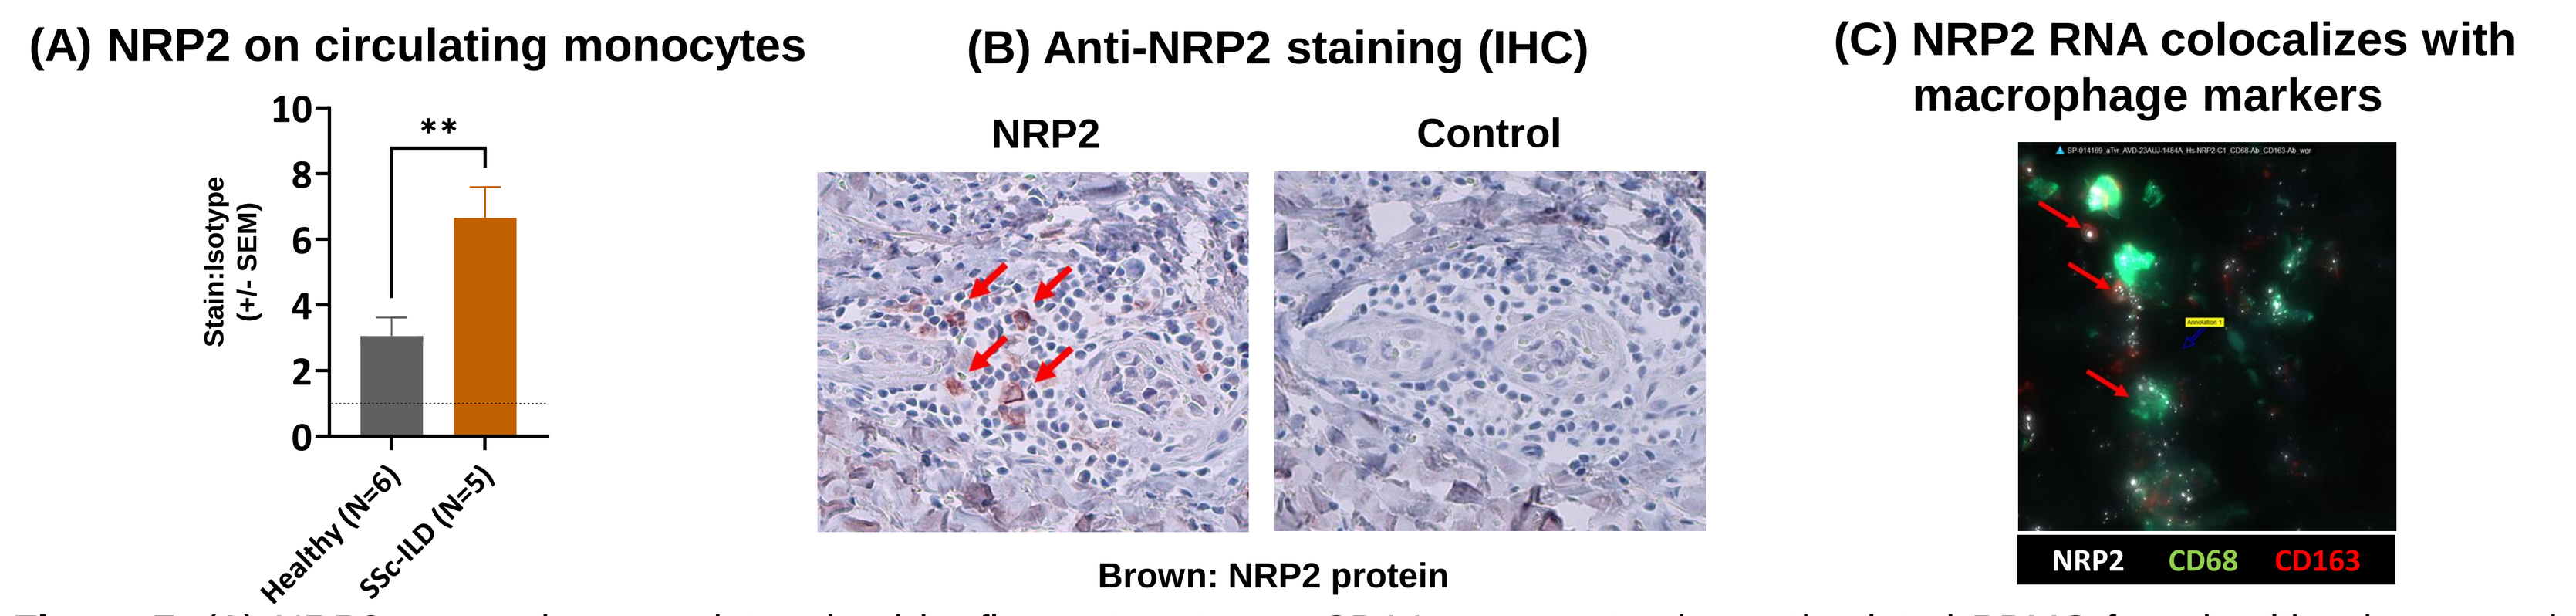
(A) NRP2 on circulating monocytes
Stain:Isotype (+/- SEM)
Healthy (N=6)
SSc-ILD (N=5)
(B) Anti-NRP2 staining (IHC)
NRP2
Control
Brown: NRP2 protein
(C) NRP2 RNA colocalizes with macrophage markers
NRP2
CD68
CD163

Figure 7. (A) NRP2 expression was determined by flow cytometry on CD14+ monocytes in unstimulated PBMC from healthy donors and SSc-ILD patients. (B) NRP2 protein expression (brown) is detectable by IHC in cells with macrophage morphology in skin from SSc-ILD patients. (C) NRP2 RNA* (white spots) shows co-localization with the macrophage markers CD68 (green) and CD163 (red). *RNAscope® technology by ACD.

Efzofitimod alters morphology and reduces inflammatory markers in primary macrophages

(A) Efzofitimod alters morphology of primary monocyte-derived macrophages

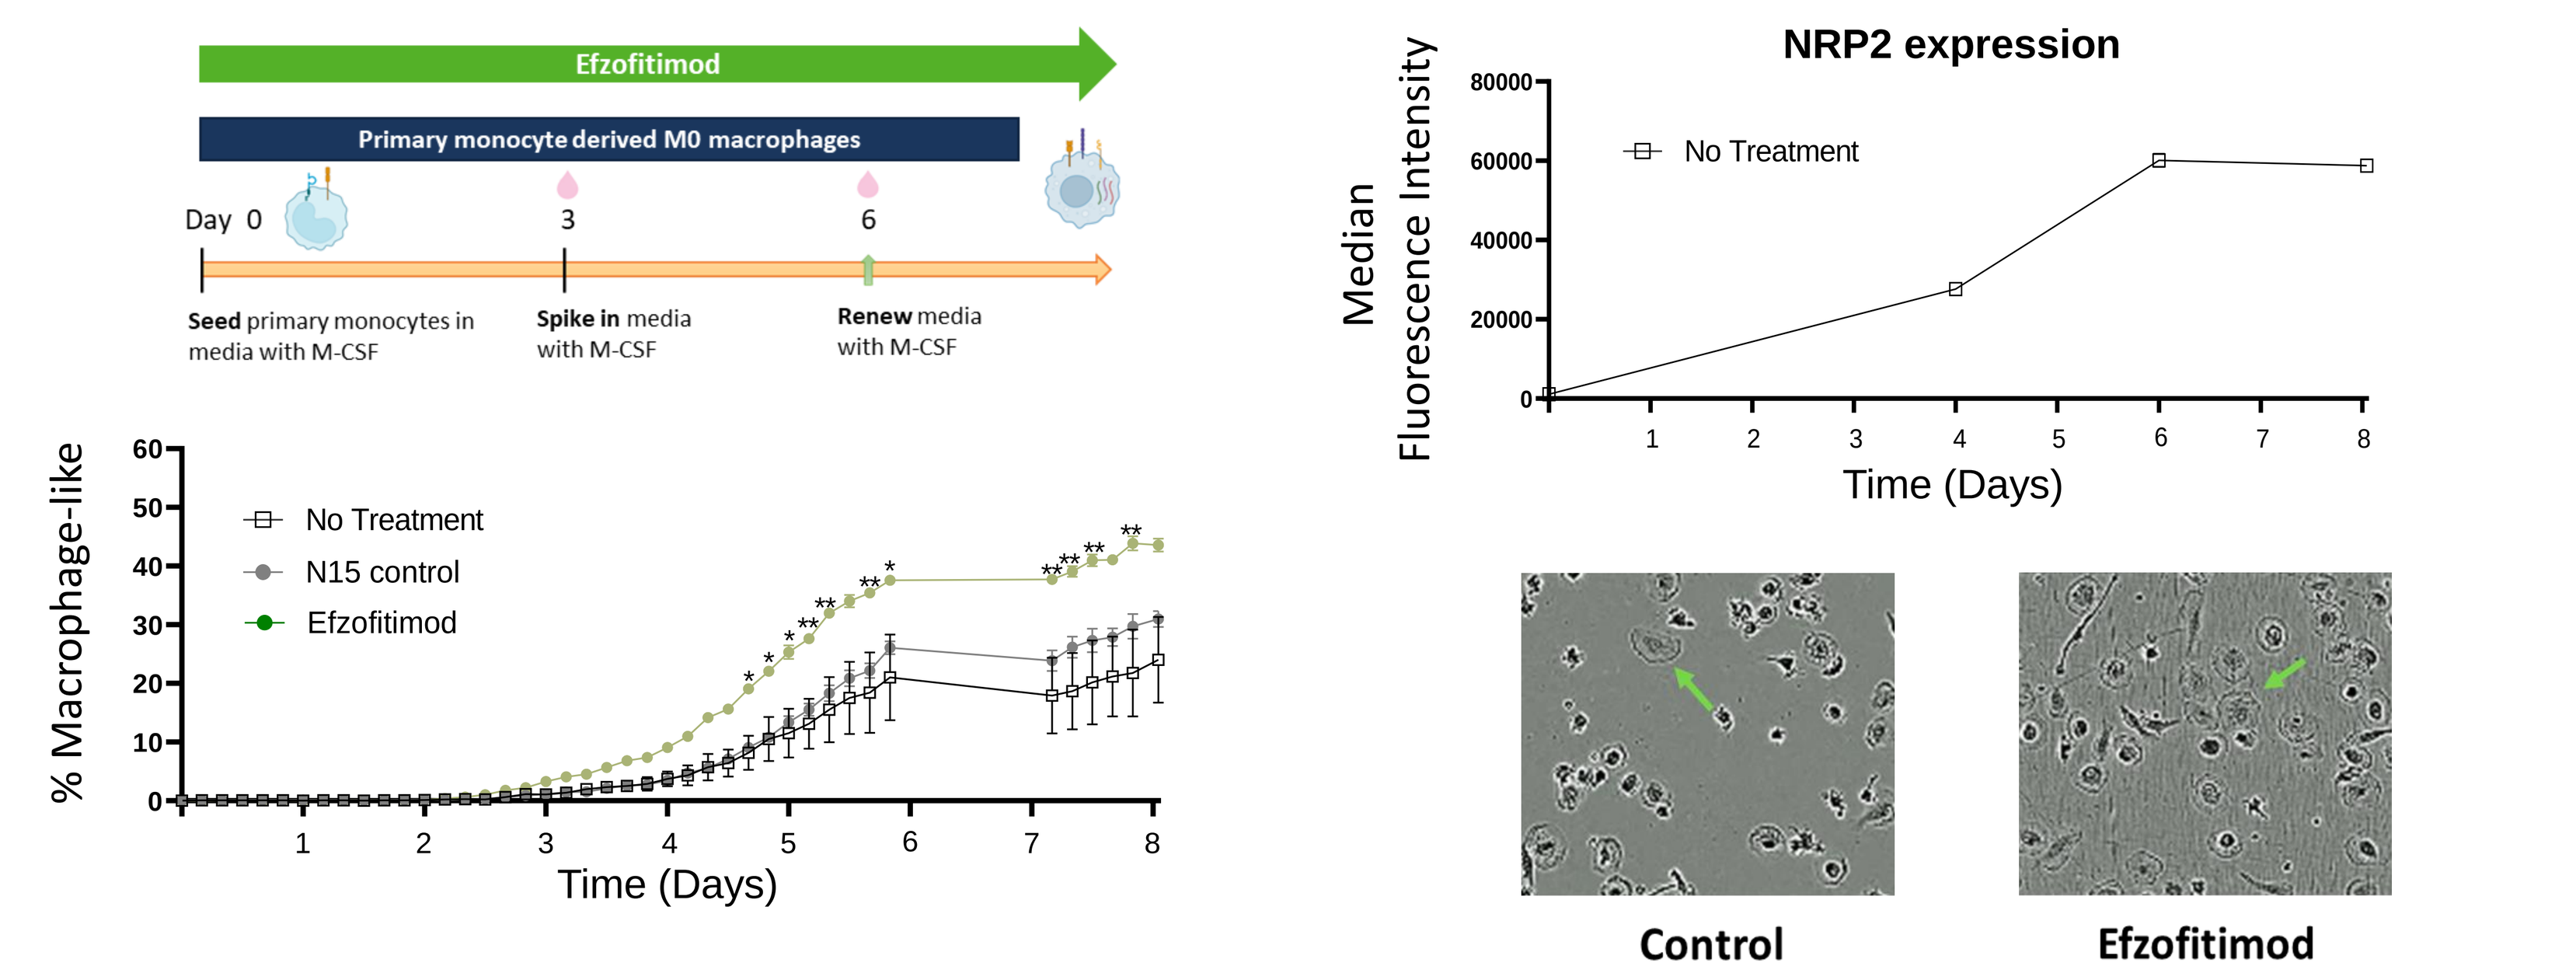
Efzofitimod
Primary monocyte derived M0 macrophages
Day 0
3
6
Seed primary monocytes in media with M-CSF
Spike in media with M-CSF
Renew media with M-CSF
% Macrophage-like
No Treatment
N15 control
Efzofitimod
Time (Days)
NRP2 expression
Median Fluorescence Intensity
Control
Efzofitimod

(B) Efzofitimod reduces inflammatory markers

		Clinical serum biomarkers	DE RNA-seq	DE qPCR	Cell surface receptors	Secreted proteins
Cytokines	CCL2 (MCP-1)	Down	Down	Down		Down
	TNF	Down	Down	Down		Down
	IL-6	Down	n.d.	n.d.		Down
	IL1B		Down			Down
	CXCL9 (MIG)		n.d.	n.d.		Down
Receptors	CD14		Down	Down	Down	Down
	CCR2		n.d.	n.d.	Down	
	CCR7		n.d.	Down	Down	
	CXCR4		-	Down	Down	
	MRC1 (CD206)		-	-	Down	
	CD28		Down	Down		
	TLR2		Down	Down		
	TLR4		-	Down		

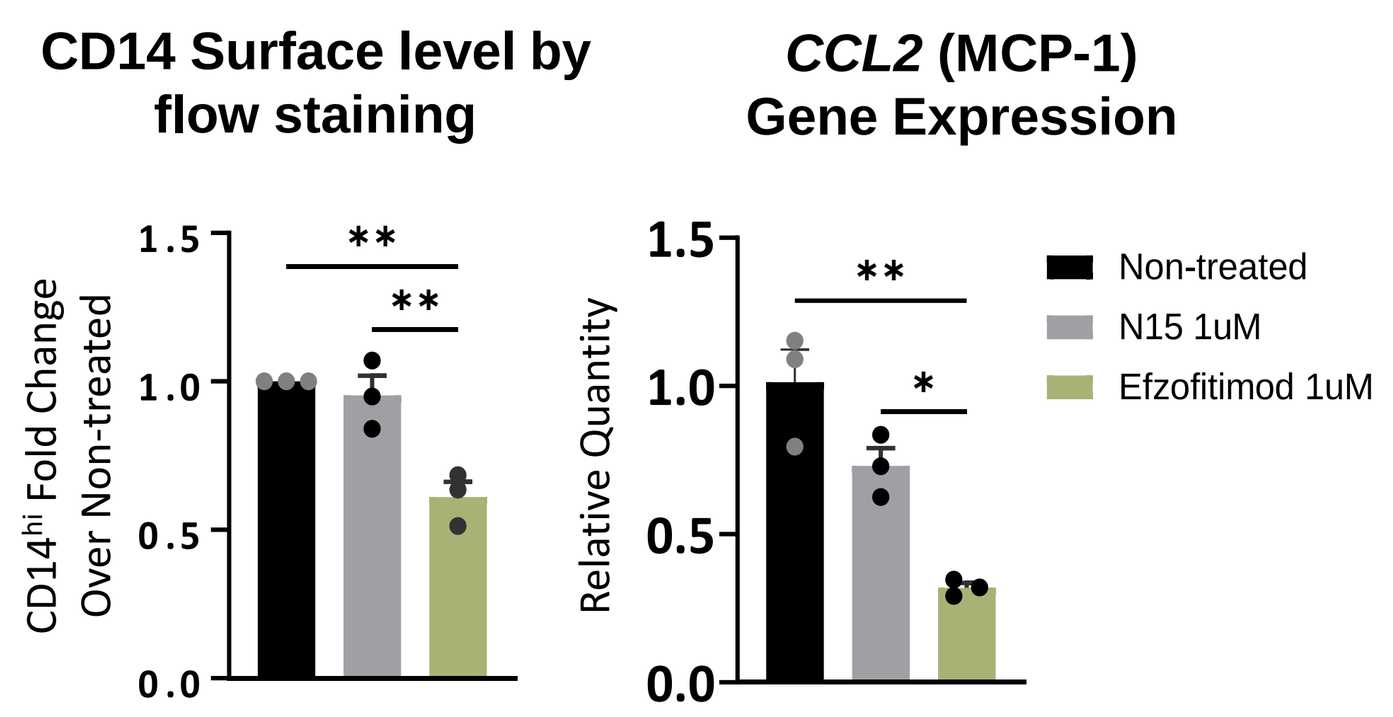
CD14 Surface level by flow staining
CD14hi Fold Change Over Non-treated
CCL2 (MCP-1) Gene Expression
Relative Quantity
Non-treated
N15 1uM
Efzofitimod 1uM

Figure 8. (A) Monocyte-derived macrophages were generated in the presence or absence of 1uM efzofitimod and macrophage-like morphology change was monitored by Incucyte® every 3 hours for 8 days. NRP2 expression was determined by flow cytometry. (B) Efzofitimod reduced surface marker expression and pro-inflammatory gene and protein expression.

Conclusions

- Efzofitimod is a novel immunomodulator that has demonstrated POC in a Phase 1b/2a trial in patients with pulmonary sarcoidosis.
- NRP2, the sole binding partner to efzofitimod, is highly expressed on circulating monocytes and skin of SSc patients.
- Efzofitimod promotes macrophage-like morphology of myeloid cells and downregulates inflammatory cytokines and receptors.
- Efzofitimod has demonstrated efficacy in pre-clinical models of ILD.
- Data support investigation of efzofitimod in SSc-ILD and other ILD's with common immune pathologies.

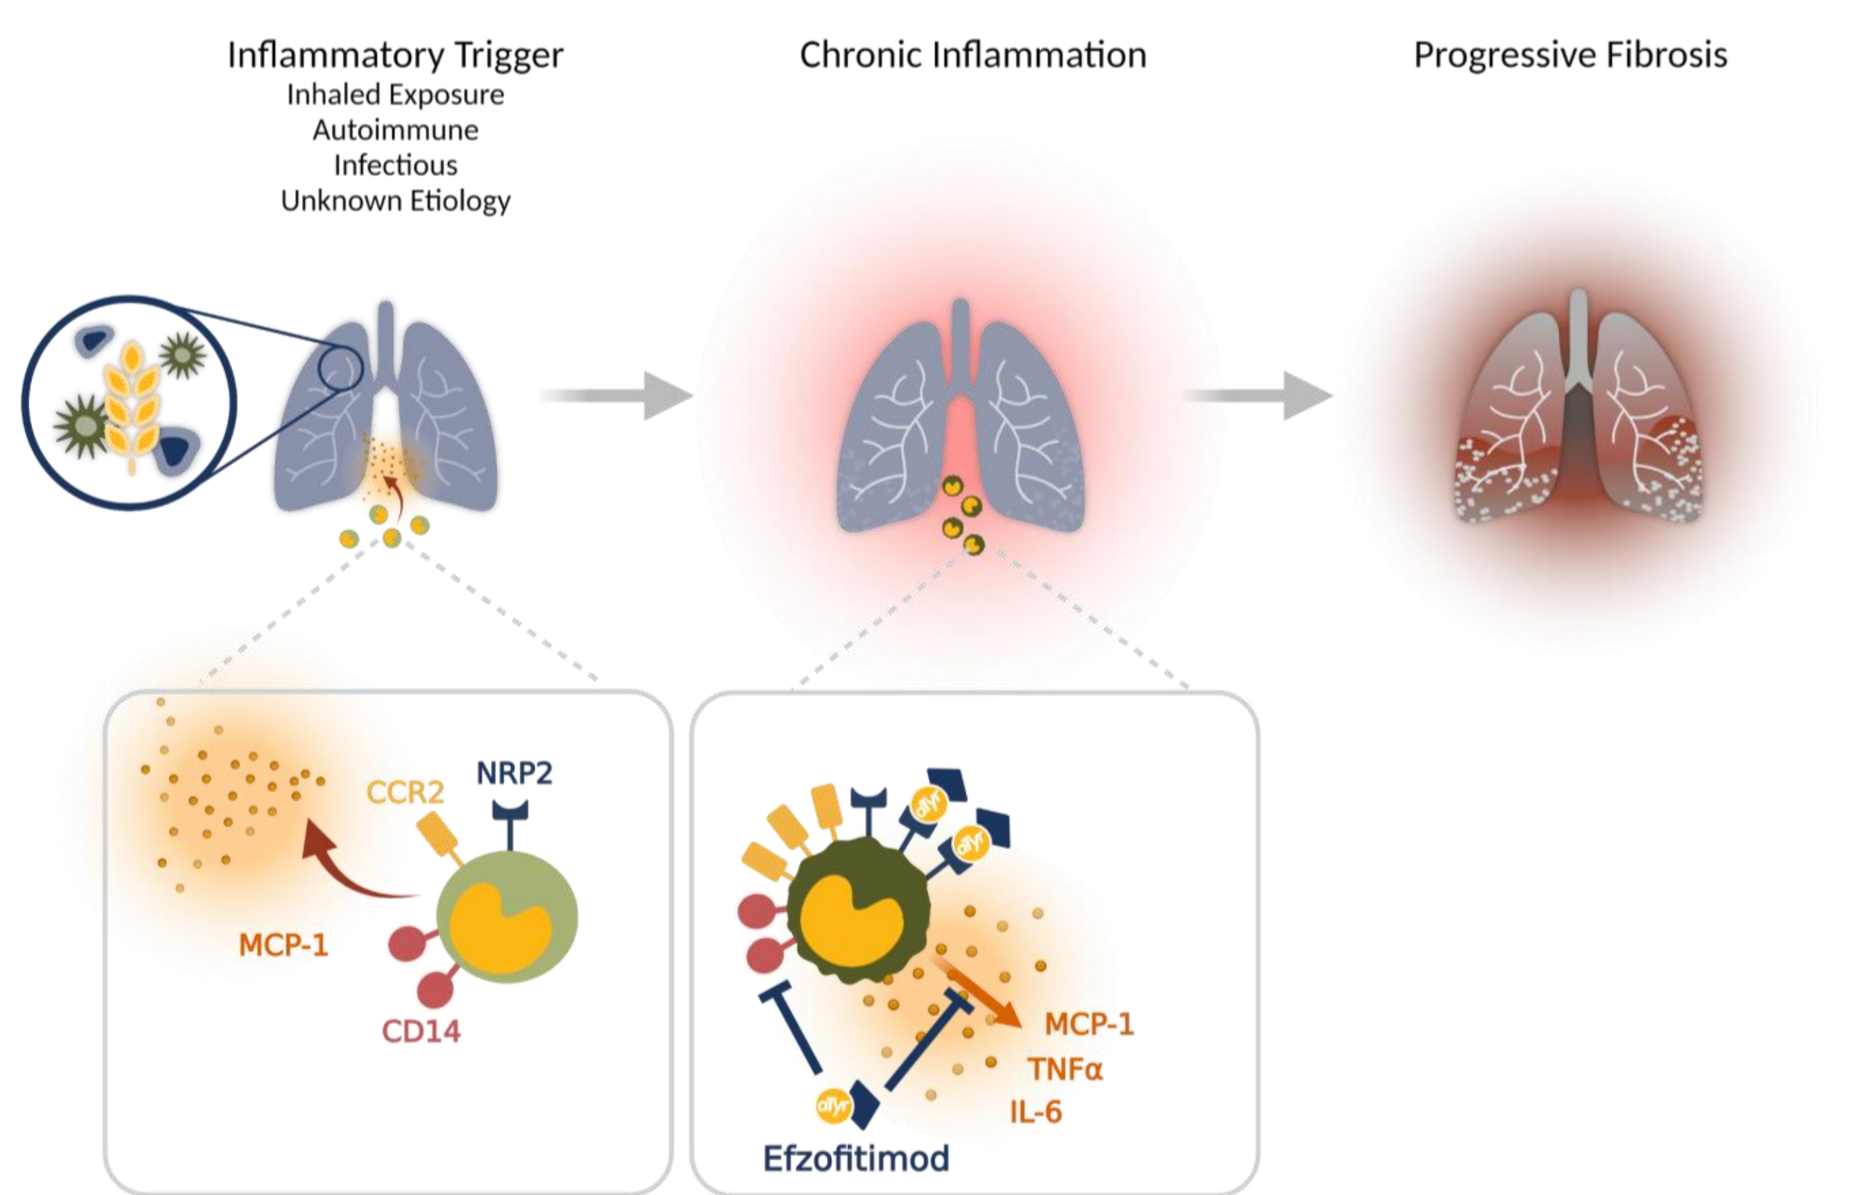
Inflammatory Trigger
Inhaled Exposure
Autoimmune
Infectious
Unknown Etiology
Chronic Inflammation
Progressive Fibrosis
CCR2
NRP2
MCP-1
CD14
MCP-1
TNFα
IL-6
Efzofitimod

Disclosures:
All authors are employees of aTyr Pharma, Inc.
Funding support: This work was supported by aTyr Pharma, Inc.

References:
[1]Baughman, RP, et al. Efzofitimod: A novel anti-inflammatory agent for sarcoidosis. *Sarcoidosis, Vasculitis and Diffuse Lung Diseases*. 40,1 (2023)
[2]Stifano G, et. al. Chronic Toll-like receptor 4 stimulation in skin induces inflammation, macrophage activation, transforming growth factor beta signature gene expression, and fibrosis. *Arthritis Research and Therapy.* 16:R136 (2014)
[3]Stifano G, et. al. Skin gene expression is prognostic for the trajectory of skin disease in patients with diffuse cutaneous systemic sclerosis. *Arthritis Rheumatol.* 70,1 (2018)
[4]Culver D, et. al. Efzofitimod for the Treatment of Pulmonary Sarcoidosis. *Chest. 163* (4) (2023)