#8837

Efzofitimod Is An Immunomodulator of Myeloid Cell Function and Novel Therapeutic Candidate for Interstitial Lung Diseases

David Siefker, Zhiwen Xu, Annie Wang, Sofia Klopp-Savino, Lauren Guy, Christopher Larsen, Luke Burman, Yang Qing, Ryan Adams, Leslie Nangle

aTyr Pharma - San Diego (USA)

Abstract/Aims

Interstitial lung disease (ILD) represents a diverse group of inflammatory and fibrotic conditions with limited treatments available. Myeloid cells are becoming increasingly appreciated as drivers of inflammation and fibrosis in ILD. Efzofitimod is a myeloid cell immunomodulator currently in a Phase 3 clinical trial for pulmonary sarcoidosis. It selectively binds to neuropilin-2 (NRP2), a membrane protein upregulated at sites of inflammation, most notably on myeloid cells, and has been shown to reduce inflammation and fibrosis across various murine ILD models. An efzofitimod Phase 1b/2a trial in pulmonary sarcoidosis patients showed dose dependent trends of improvement in lung function and reduced pro-inflammatory serum biomarkers[1]. Given these promising results, we probed more deeply into the underlying molecular mechanism that is the basis for efzofitimod's activity and therapeutic potential in ILD.

Here, we investigated the mechanism of action of efzofitimod using samples from ILD patients. Expression of target protein NRP2 in ILD patient cells and tissue samples was probed by flow cytometry, public RNA sequencing datasets, immunohistochemistry (IHC) and immunofluorescence techniques. The morphology change of primary monocyte-derived macrophages (MDM) by efzofitimod treatment was monitored using an automated live cell imaging and analysis platform. Transcriptome regulation of macrophages and PBMC from sarcoidosis patients was analyzed by RNA sequencing and gene set enrichment analysis. Results from these studies demonstrate that efzofitimod acts on myeloid cells in ILD patients to diminish inflammatory potential. Given the central role of myeloid cells in the pathology of numerous ILDs and other chronic inflammatory and fibrotic diseases, efzofitimod may have broad therapeutic potential in ILD and beyond.

Evolution of Non-Catalytic Domains of tRNA Synthetases for Novel Functions

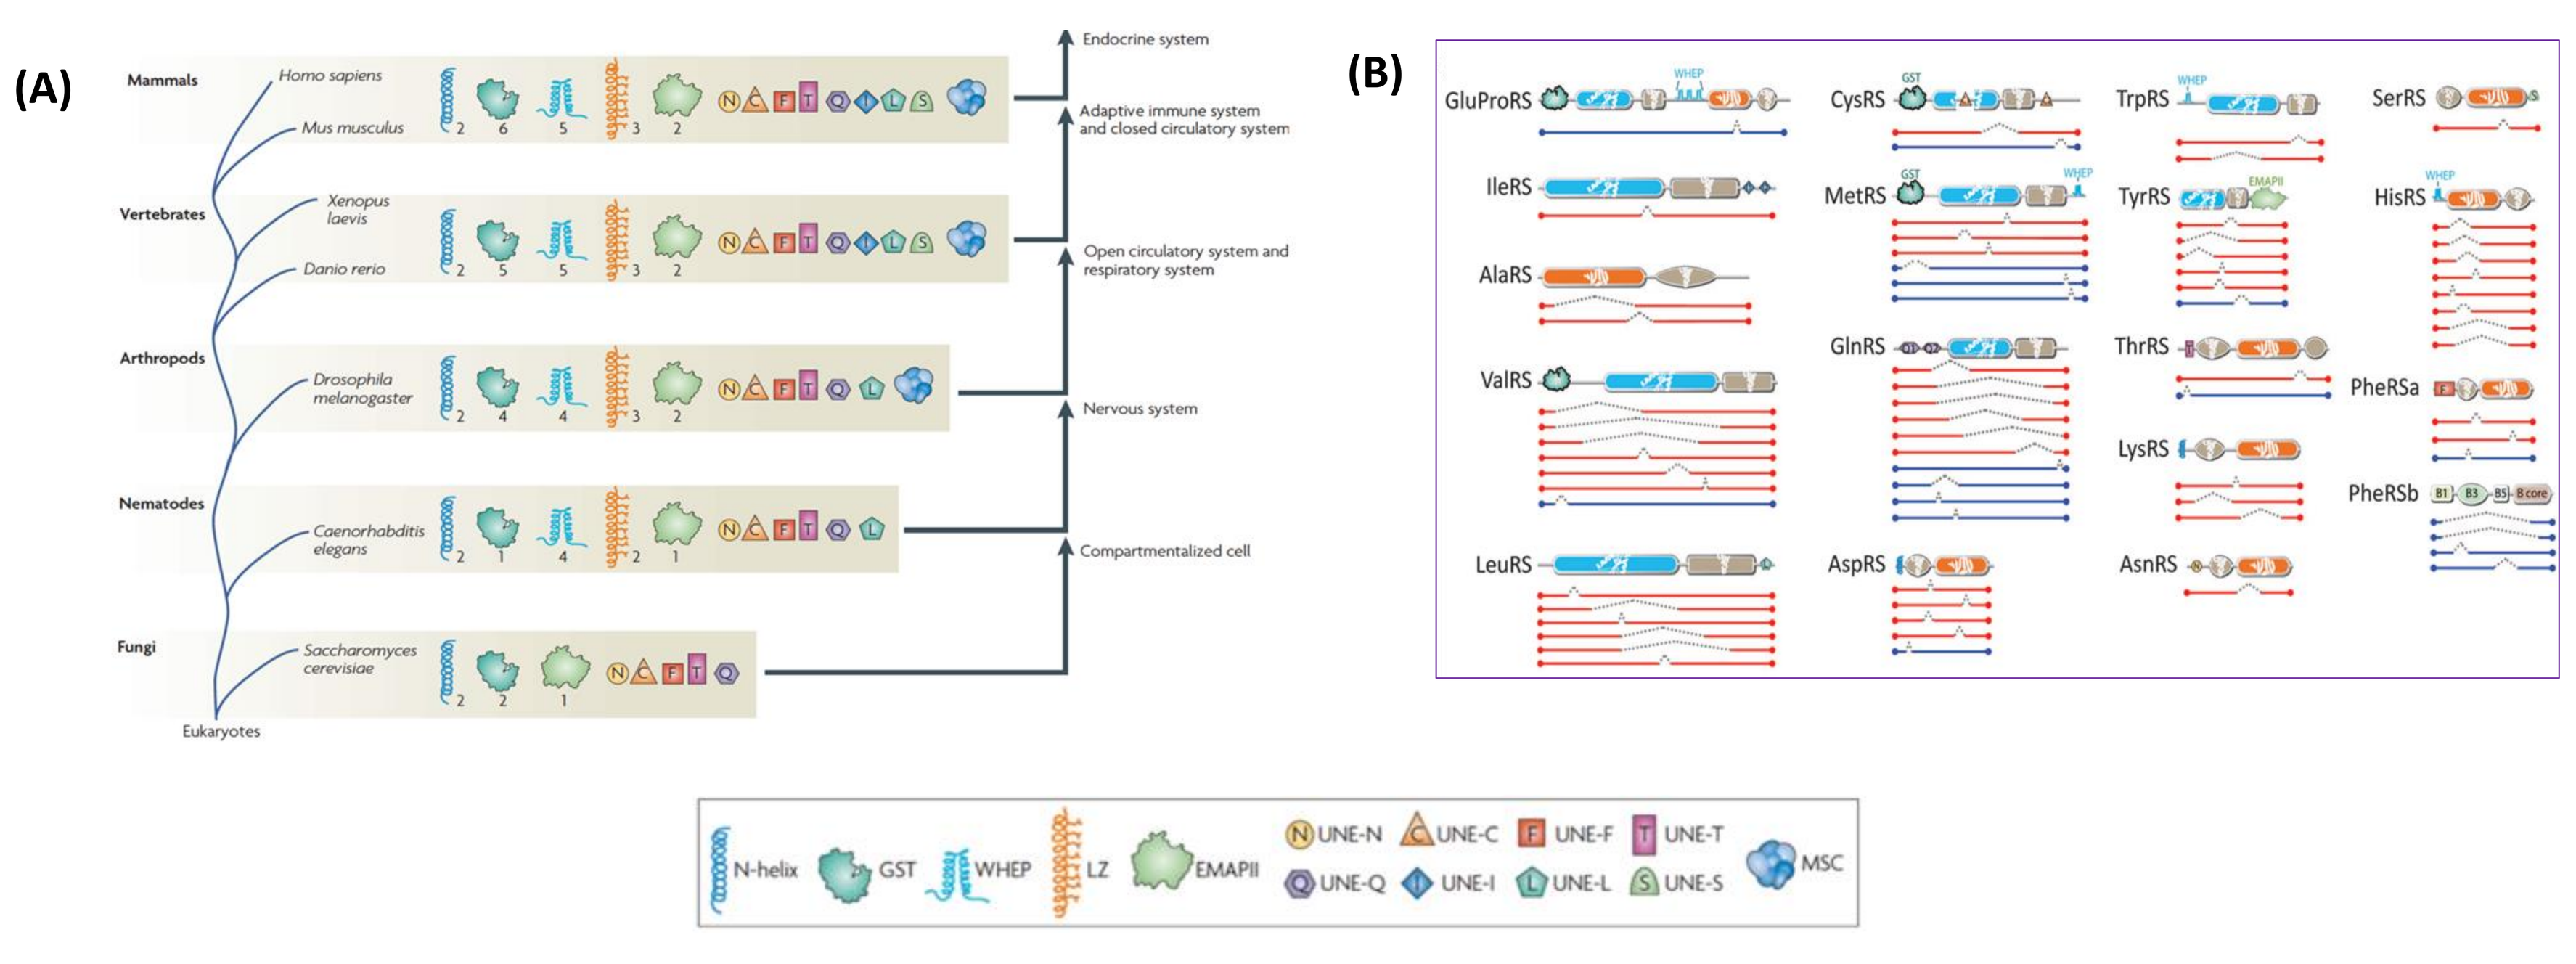
(A)
Mammals
Homo sapiens
Mus musculus
Vertebrates
Xenopus laevis
Danio rerio
Arthropods
Drosophila melanogaster
Nematodes
Caenorhabditis elegans
Fungi
Saccharomyces cerevisiae
Eukaryotes
Endocrine system
Adaptive immune system and closed circulatory system
Open circulatory system and respiratory system
Nervous system
Compartmentalized cell
(B)
GluProRS
CysRS
TrpRS
SerRS
IleRS
MetRS
TyrRS
HisRS
AlaRS
GlnRS
ThrRS
ValRS
PheRSa
LysRS
PheRSb
LeuRS
AspRS
AsnRS
N-helix
GST
WHEP
LZ
EMAPII
UNE-N
UNE-C
UNE-F
UNE-T
UNE-Q
UNE-I
UNE-L
UNE-S
MSC

Figure 1. The catalytic domains of tRNA synthetases are ancient and conserved from bacteria to humans. (A) As life became more complex, tRNA synthetases evolved additional non-catalytic domains with novel functions that coincide with the appearance of higher organ systems[2]. (B) About 80% of tRNA synthetase splice variants do not possess the aminoacylation catalytic domains and are instead liberated novel signaling domains[3].

tRNA Synthetase Drug Discovery Platform

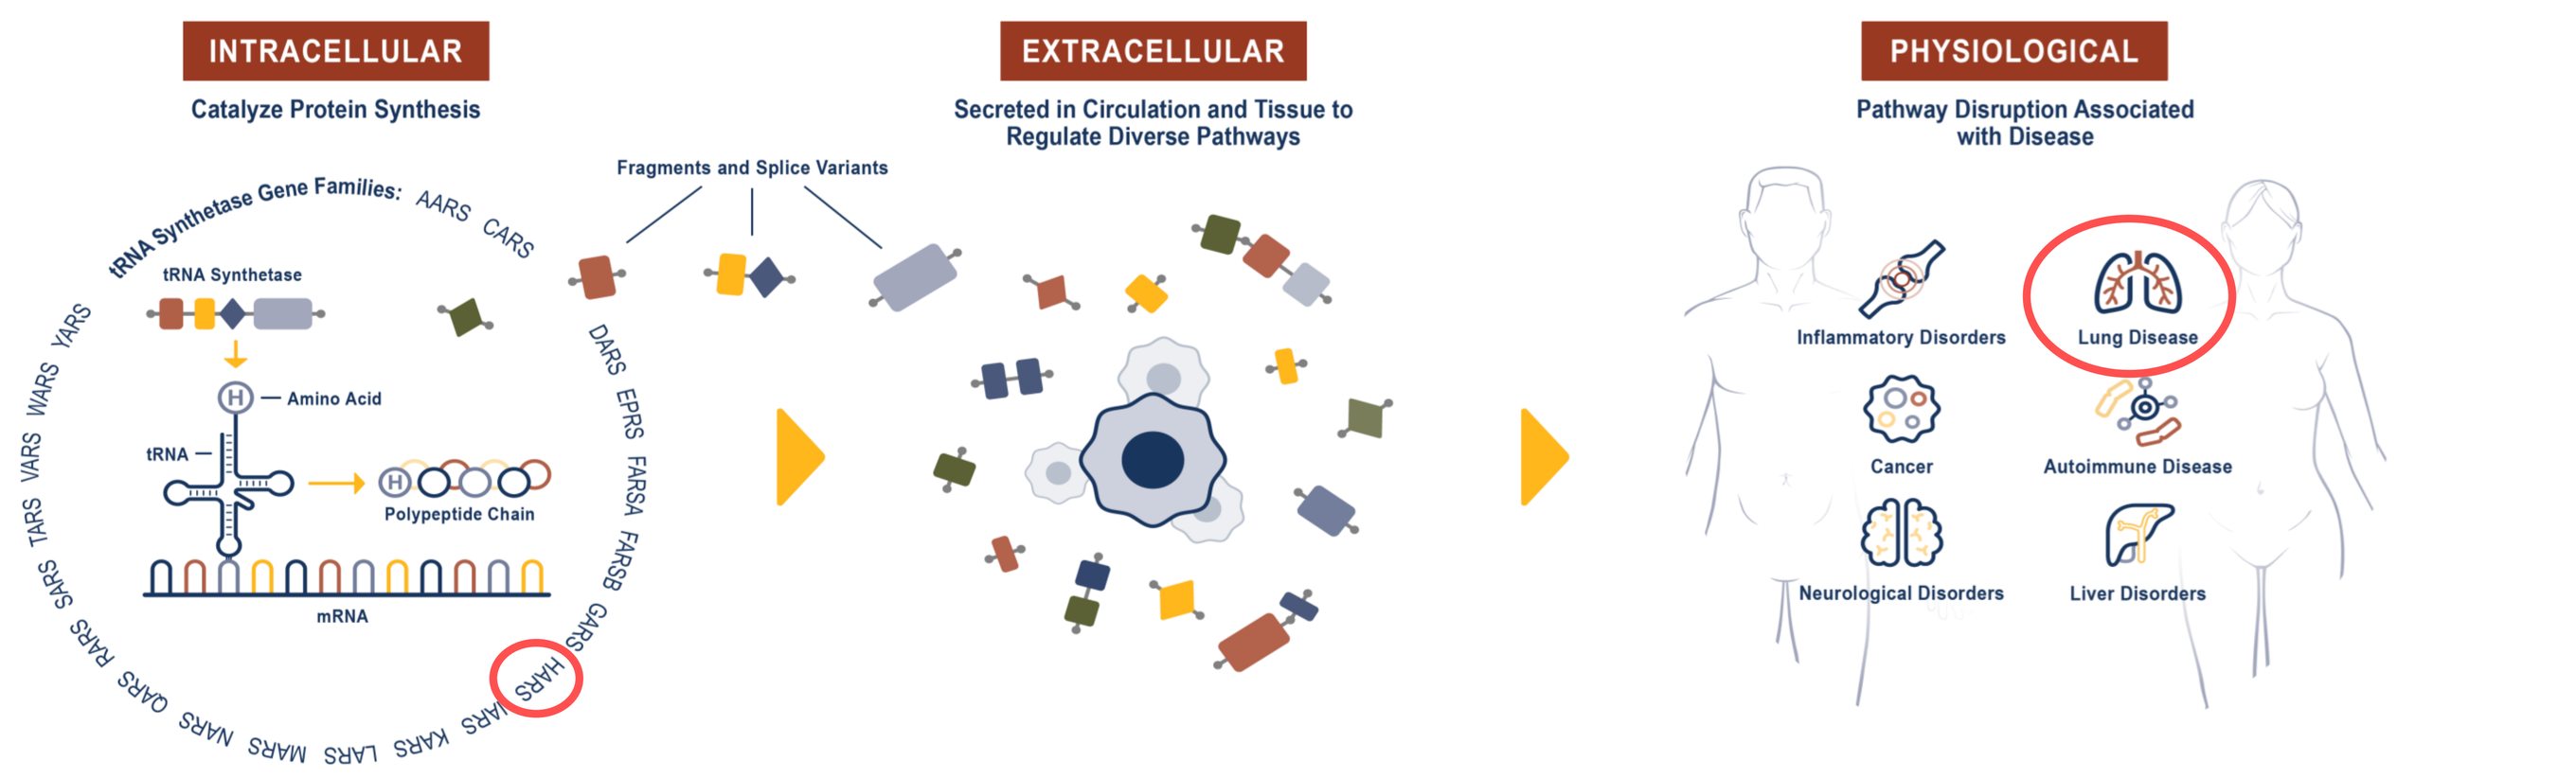
INTRACELLULAR
Catalyze Protein Synthesis
tRNA Synthetase Gene Families:
tRNA Synthetase
Amino Acid
tRNA
Polypeptide Chain
mRNA
Fragments and Splice Variants
EXTRACELLULAR
Secreted in Circulation and Tissue to Regulate Diverse Pathways
PHYSIOLOGICAL
Pathway Disruption Associated with Disease
Inflammatory Disorders
Lung Disease
Cancer
Autoimmune Disease
Neurological Disorders
Liver Disorders

Figure 2. Splice variants and proteolytic fragments of tRNA synthetases have been identified in extracellular spaces, where they have novel functions. aTyr Pharma is engaged in the discovery and development of potential first-in-class medicines based on newly discovered pathways effected by extracellular tRNA synthetases.

Efzofitimod, a Novel Ligand for Neuropilin-2 (NRP2) Receptor

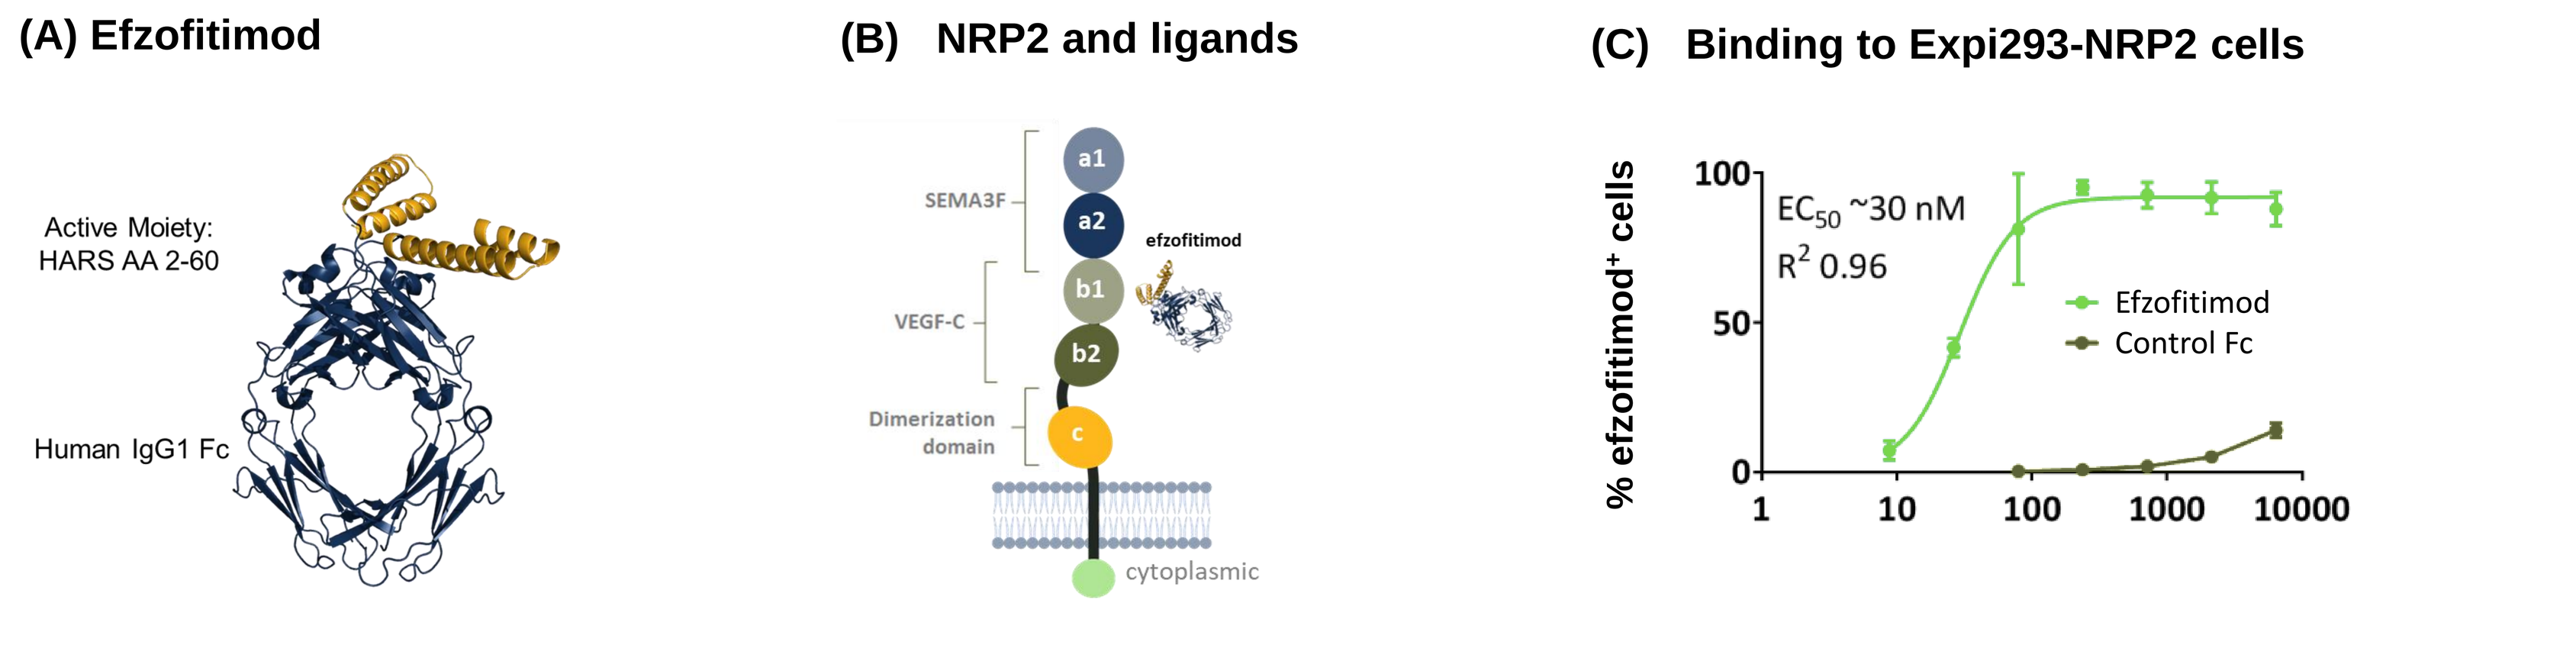
(A) Efzofitimod
Active Moiety: HARS AA 2-60
Human IgG1 Fc
(B) NRP2 and ligands
SEMA3F
VEGF-C
Dimerization domain
a1
a2
b1
b2
c
efzofitimod
cytoplasmic
(C) Binding to Expi293-NRP2 cells
EC50 ~30 nM
R2 0.96
Efzofitimod
Control Fc
% efzofitimod+ cells
100
50
0
1
10
100
1000
10000

Figure 3. Efzofitimod binds to the NRP2 receptor. (A) Efzofitimod is a fusion protein consisting of amino acids 2-60 of histidyl-tRNA synthetase (HARS) and human IgG1 Fc. (B) Schematic of NRP2 domains and ligands. (C) Efzofitimod binds to Expi293F-NRP2 cells.

Methods and Results

NRP2 is Highly Expressed in Circulating Monocytes and Tissue Macrophages from Pulmonary Sarcoidosis and SSc-ILD Patients

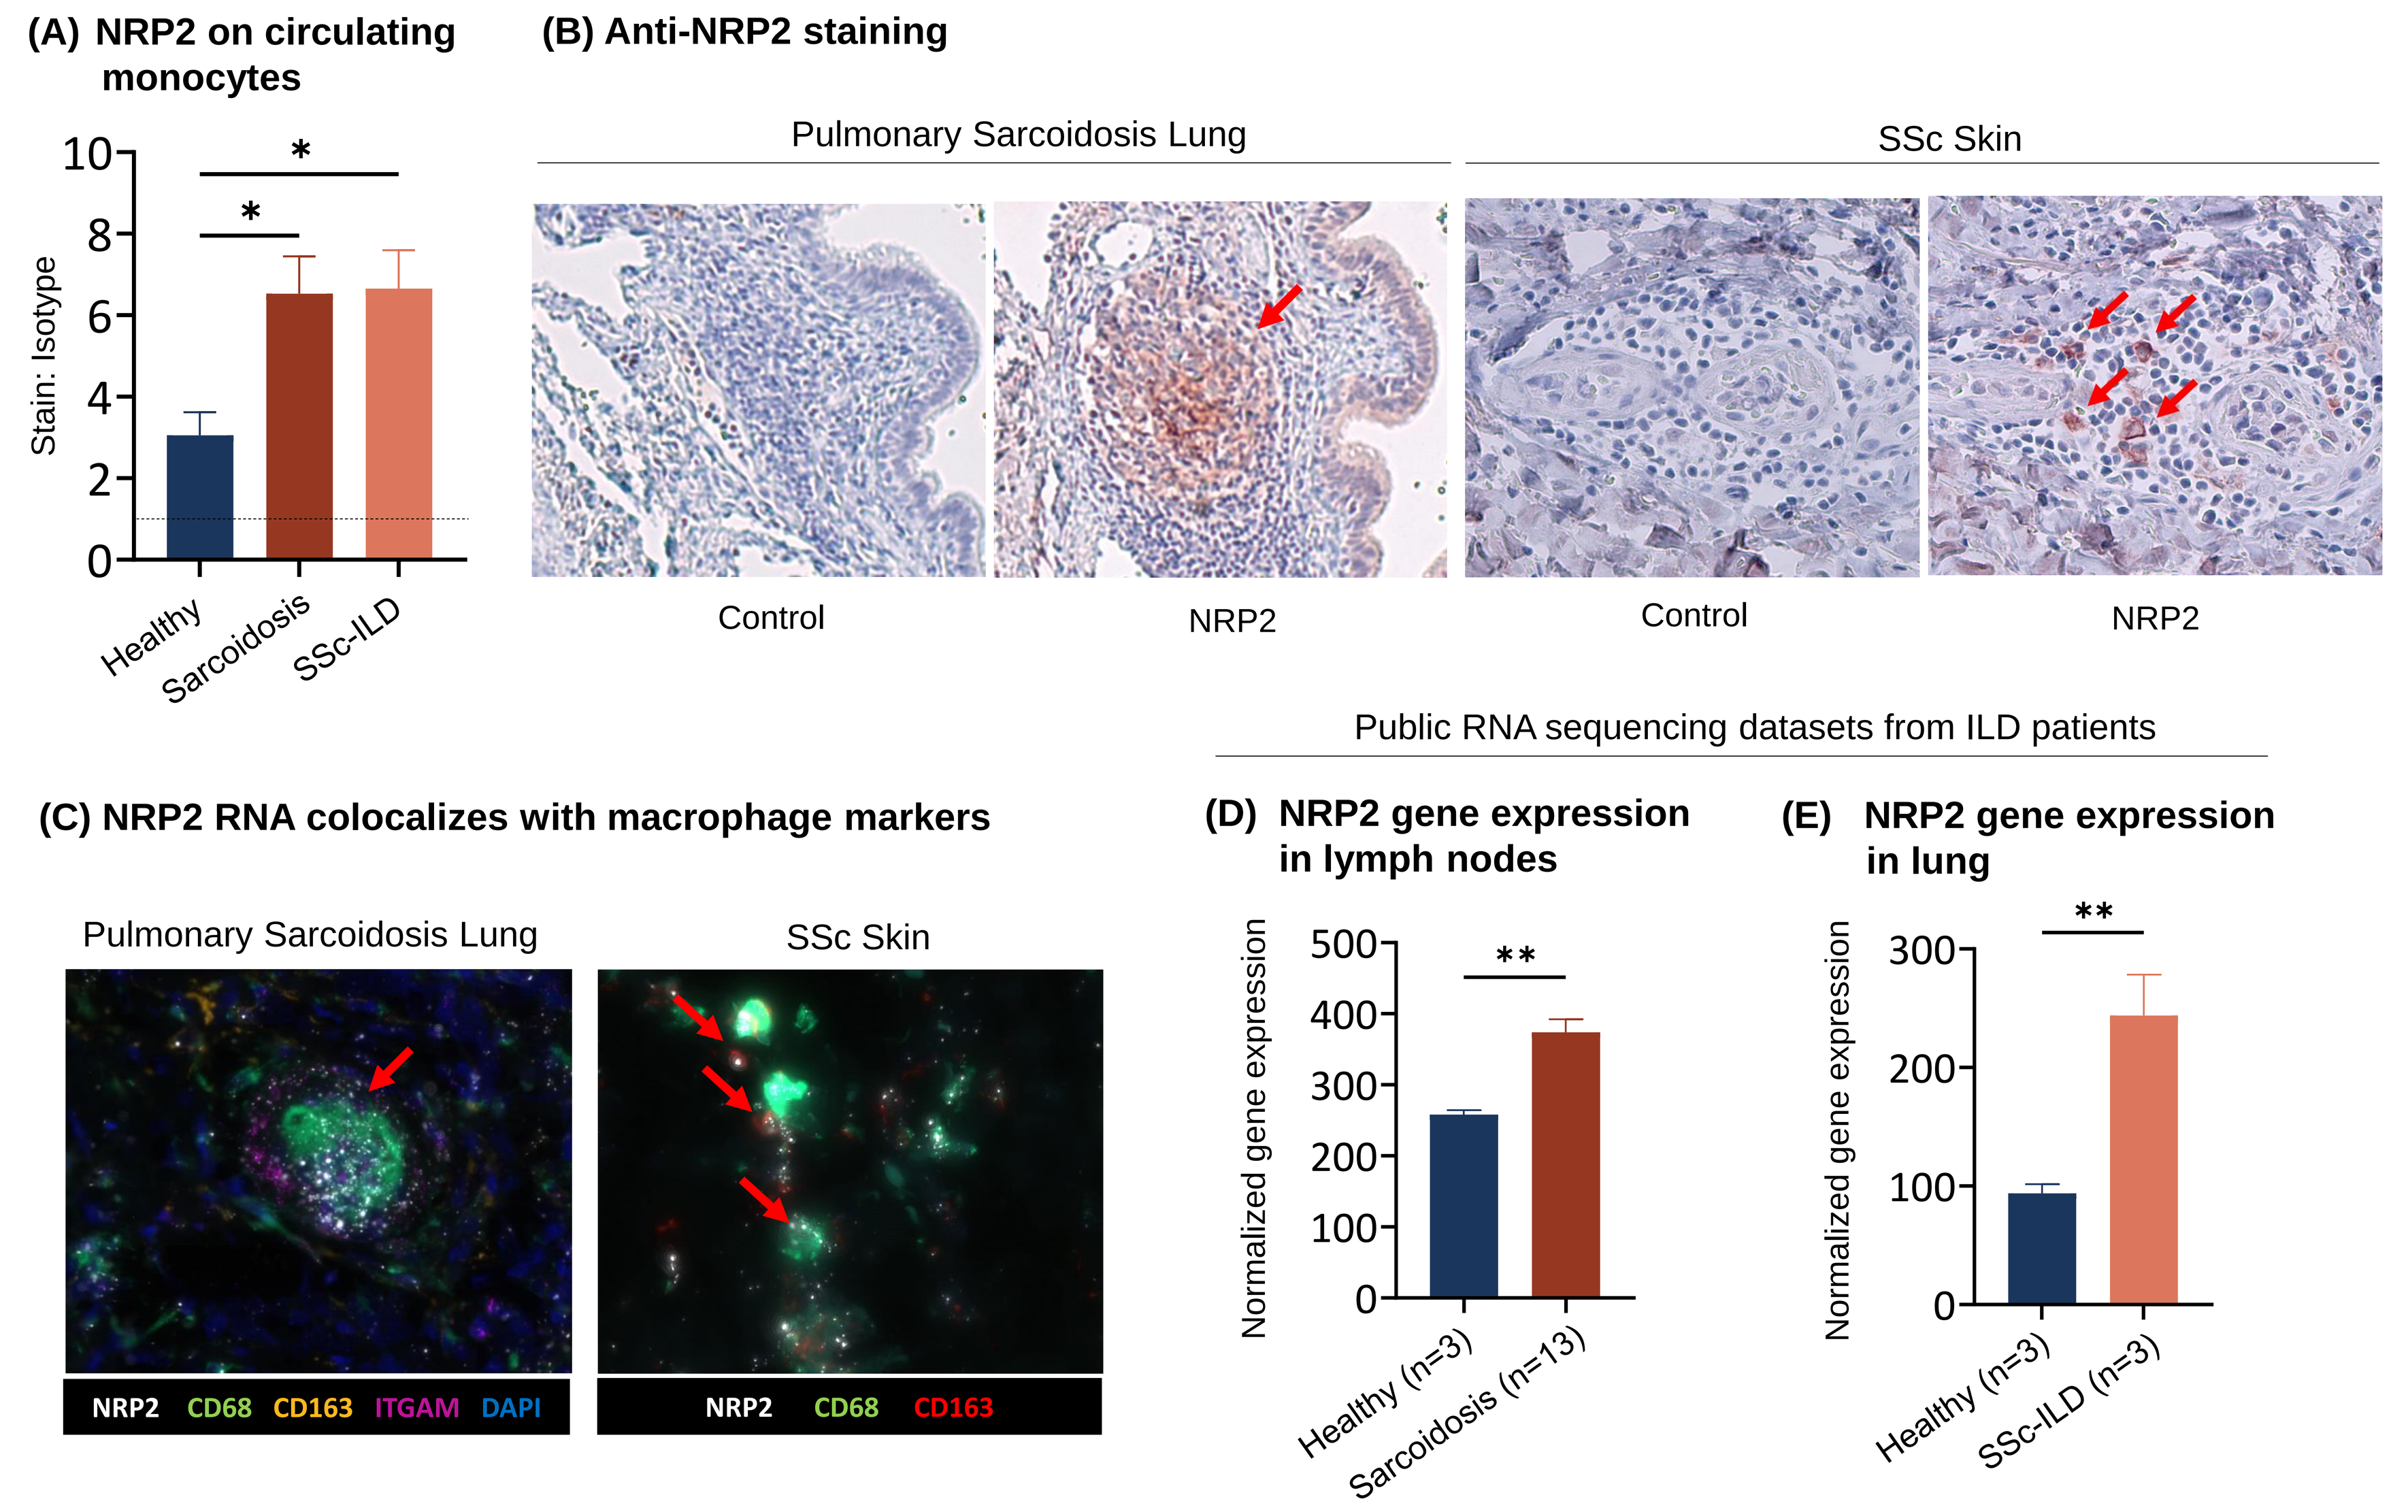
(A) NRP2 on circulating monocytes
Stain: Isotype
Healthy
Sarcoidosis
SSc-ILD
(B) Anti-NRP2 staining
Pulmonary Sarcoidosis Lung
SSc Skin
Control
NRP2
Control
NRP2
Public RNA sequencing datasets from ILD patients
(C) NRP2 RNA colocalizes with macrophage markers
Pulmonary Sarcoidosis Lung
SSc Skin
NRP2 CD68 CD163 ITGAM DAPI
NRP2 CD68 CD163
(D) NRP2 gene expression in lymph nodes
Normalized gene expression
Healthy (n=3)
Sarcoidosis (n=13)
(E) NRP2 gene expression in lung
Normalized gene expression
Healthy (n=3)
SSc-ILD (n=3)

Figure 4. (A) NRP2 expression was determined by flow cytometry on CD14+ monocytes in unstimulated PBMC from healthy donors (n=6), pulmonary sarcoidosis patients (n=7), and SSc-ILD patients (n=5). (B) NRP2 expression (brown) is detectable by IHC within granulomas of lung from a pulmonary sarcoidosis patient and skin from a scleroderma patient. (C) NRP2 RNA (white spots) co-localizes with macrophage markers CD68 (green) and CD163 (orange/red). (D,E) Public RNA sequencing datasets were utilized to compare NRP2 expression in tissues from ILD patients and controls (GSE157671, GSE166036).

Promotion of MDM Differentiation by Efzofitimod Observed in Healthy and ILD Donors

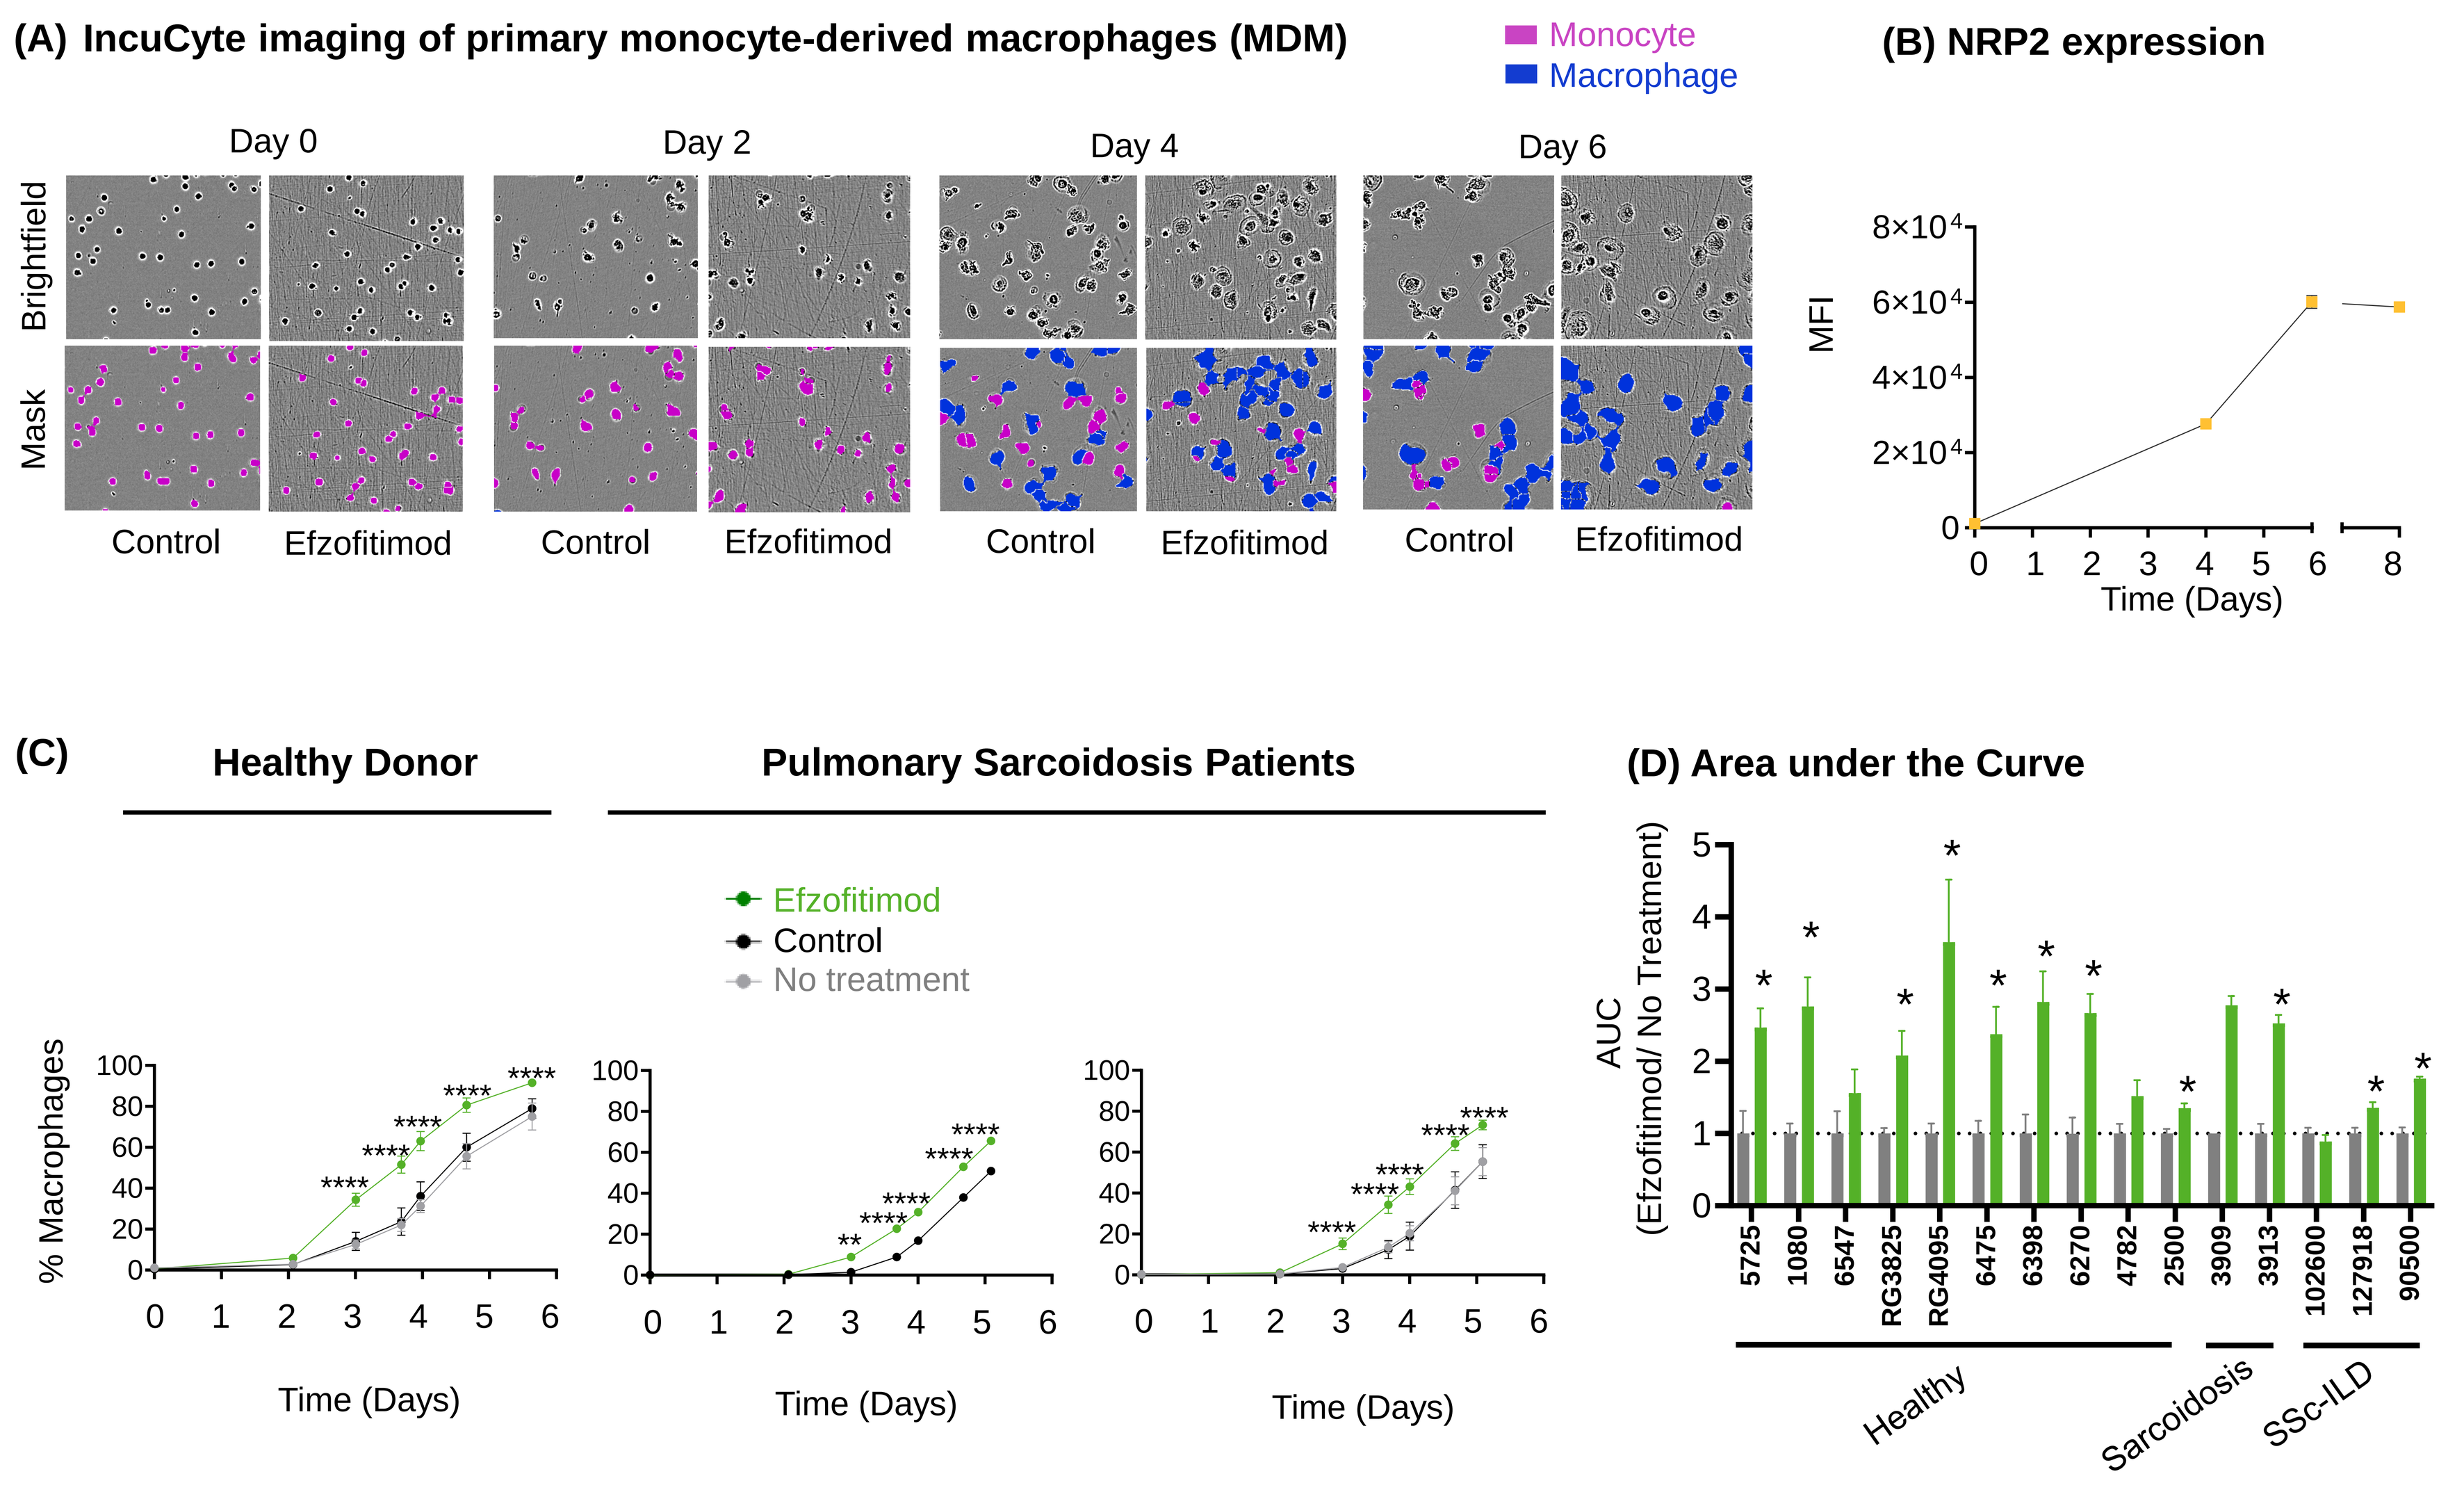
(A) IncuCyte imaging of primary monocyte-derived macrophages (MDM)
Monocyte
Macrophage
Day 0
Day 2
Day 4
Day 6
Brightfield
Mask
Control
Efzofitimod
(B) NRP2 expression
MFI
Time (Days)
(C)
Healthy Donor
Pulmonary Sarcoidosis Patients
Efzofitimod
Control
No treatment
% Macrophages
Time (Days)
(D) Area under the Curve
AUC (Efzofitimod/ No Treatment)
5725
1080
6547
RG3825
RG4095
6475
6398
6270
4782
2500
3909
3913
102600
127918
90500
Healthy
Sarcoidosis
SSc-ILD

Figure 5. (A) Images of MDM generated in the presence of efzofitimod or control Fc. Pink mask: monocyte morphology and blue mask: macrophage morphology by IncuCyte live cell imaging. (B) NRP2 upregulation during MDM differentiation by flow cytometry analysis. (C) Percentage of cells with macrophage morphology derived from primary monocytes from a healthy donor and two pulmonary sarcoidosis patients. (D) Fold change of efzofitimod over no treatment in area under the curve from days 0-4 in MDM from healthy donors and ILD patients.

Methods and Results

Novel Efzofitimod-induced Macrophage Signature Observed in MDM from Sarcoidosis Patient

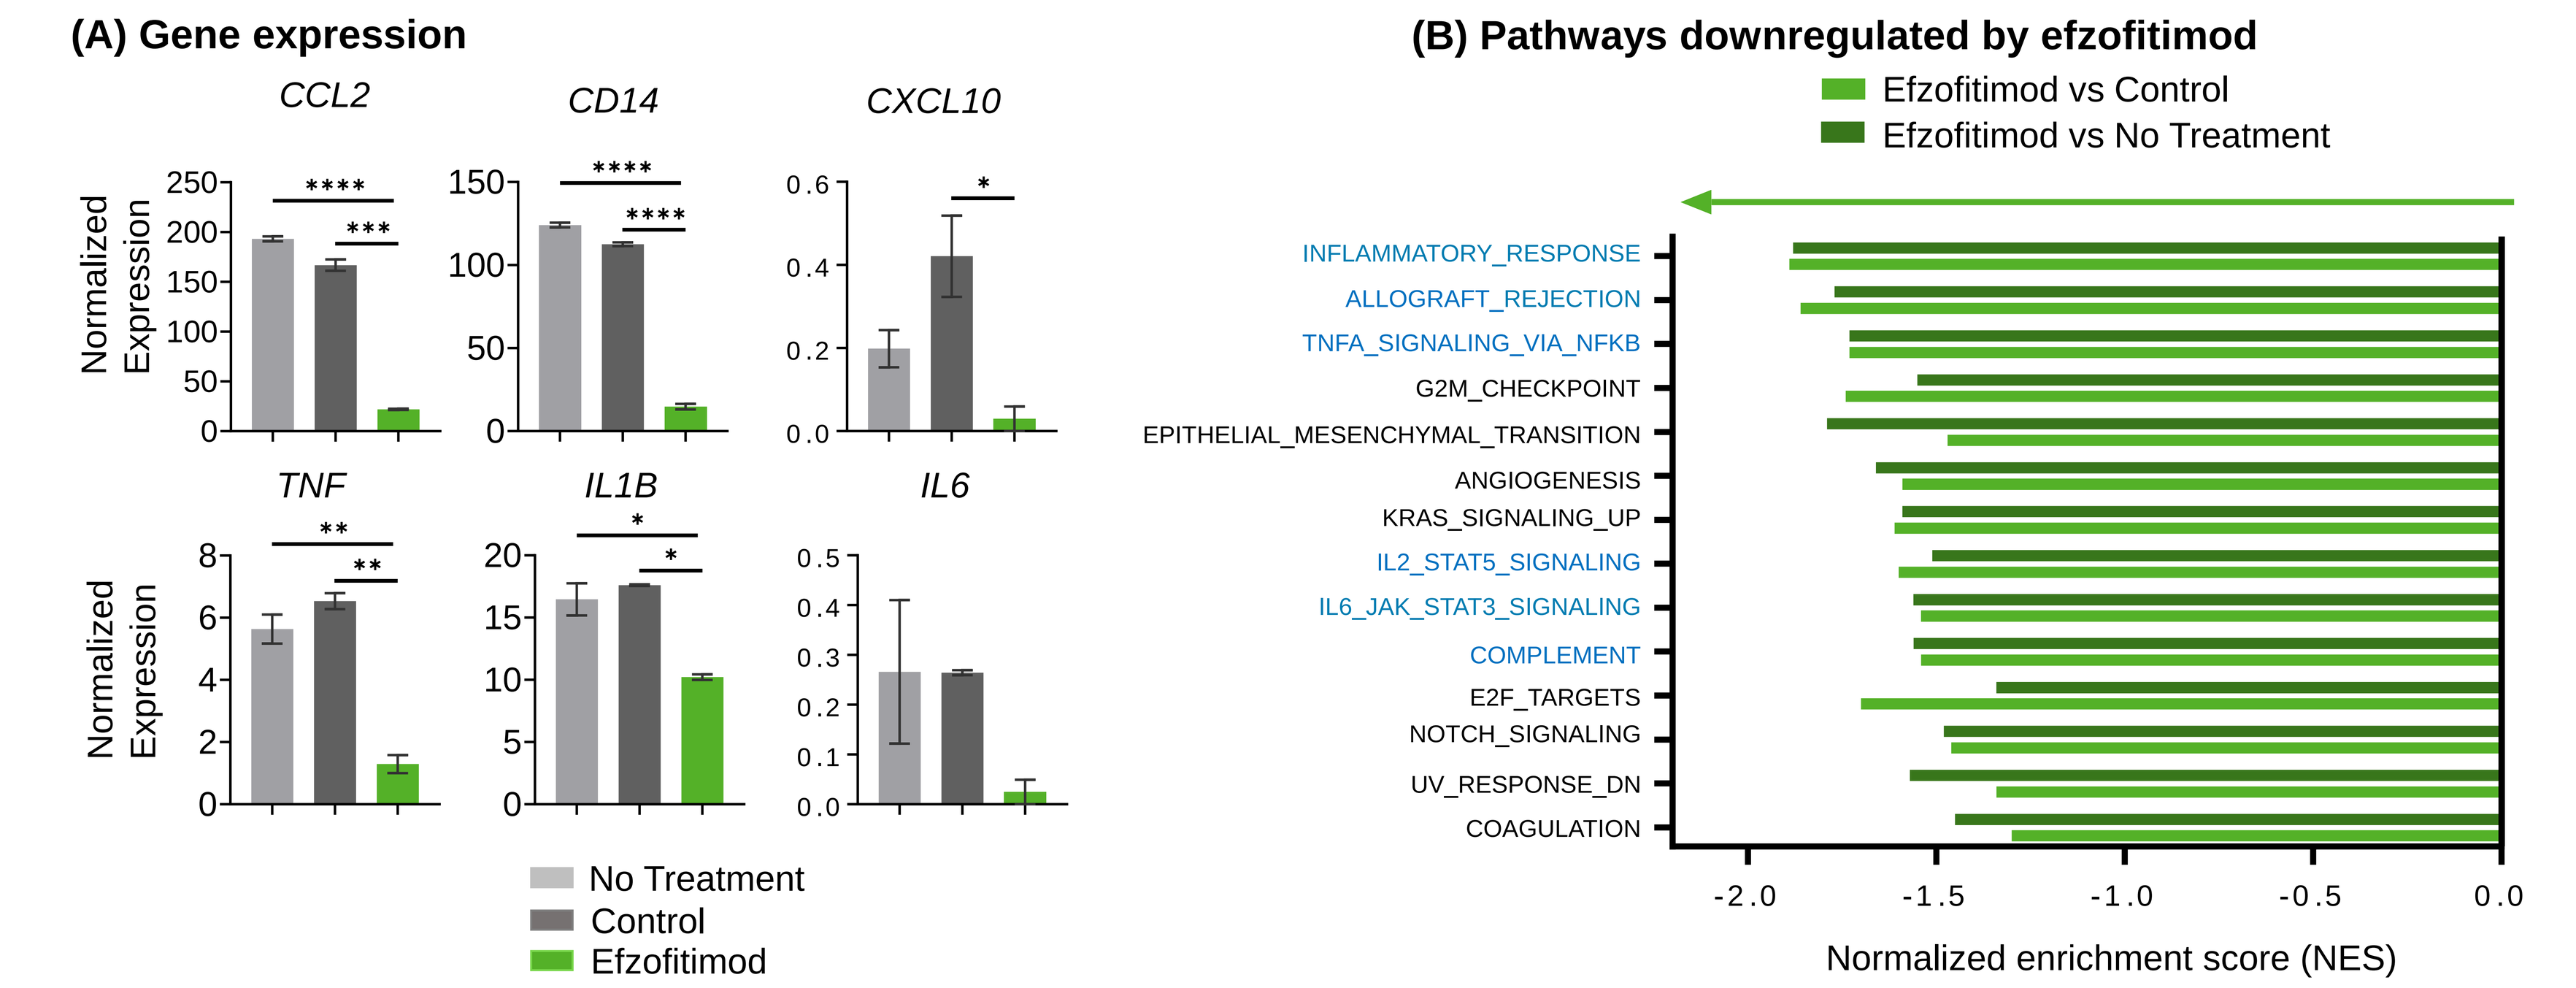
(A) Gene expression
CCL2
CD14
CXCL10
TNF
IL1B
IL6
Normalized Expression
No Treatment
Control
Efzofitimod
(B) Pathways downregulated by efzofitimod
Efzofitimod vs Control
Efzofitimod vs No Treatment
INFLAMMATORY_RESPONSE
ALLOGRAFT_REJECTION
TNFA_SIGNALING_VIA_NFKB
G2M_CHECKPOINT
EPITHELIAL_MESENCHYMAL_TRANSITION
ANGIOGENESIS
KRAS_SIGNALING_UP
IL2_STAT5_SIGNALING
IL6_JAK_STAT3_SIGNALING
COMPLEMENT
E2F_TARGETS
NOTCH_SIGNALING
UV_RESPONSE_DN
COAGULATION
Normalized enrichment score (NES)

Figure 6. MDM were generated from a pulmonary sarcoidosis patient in the presence of efzofitimod or control Fc. (A) Normalized gene expression of key inflammatory genes by RNA sequencing on day 6 of differentiation. (B) Pathways significantly downregulated by efzofitimod compared to control Fc or no treatment by gene set enrichment analysis (GSEA, 50 hallmark gene sets were analyzed). Immune pathways are highlighted in blue. Data consistent with observations from healthy donors, indicating a broad immune mechanism not specific to ILD or Sarcoidosis.

Downregulation of Inflammatory Pathways by Efzofitimod in Clinically Treated Sarcoidosis Patients

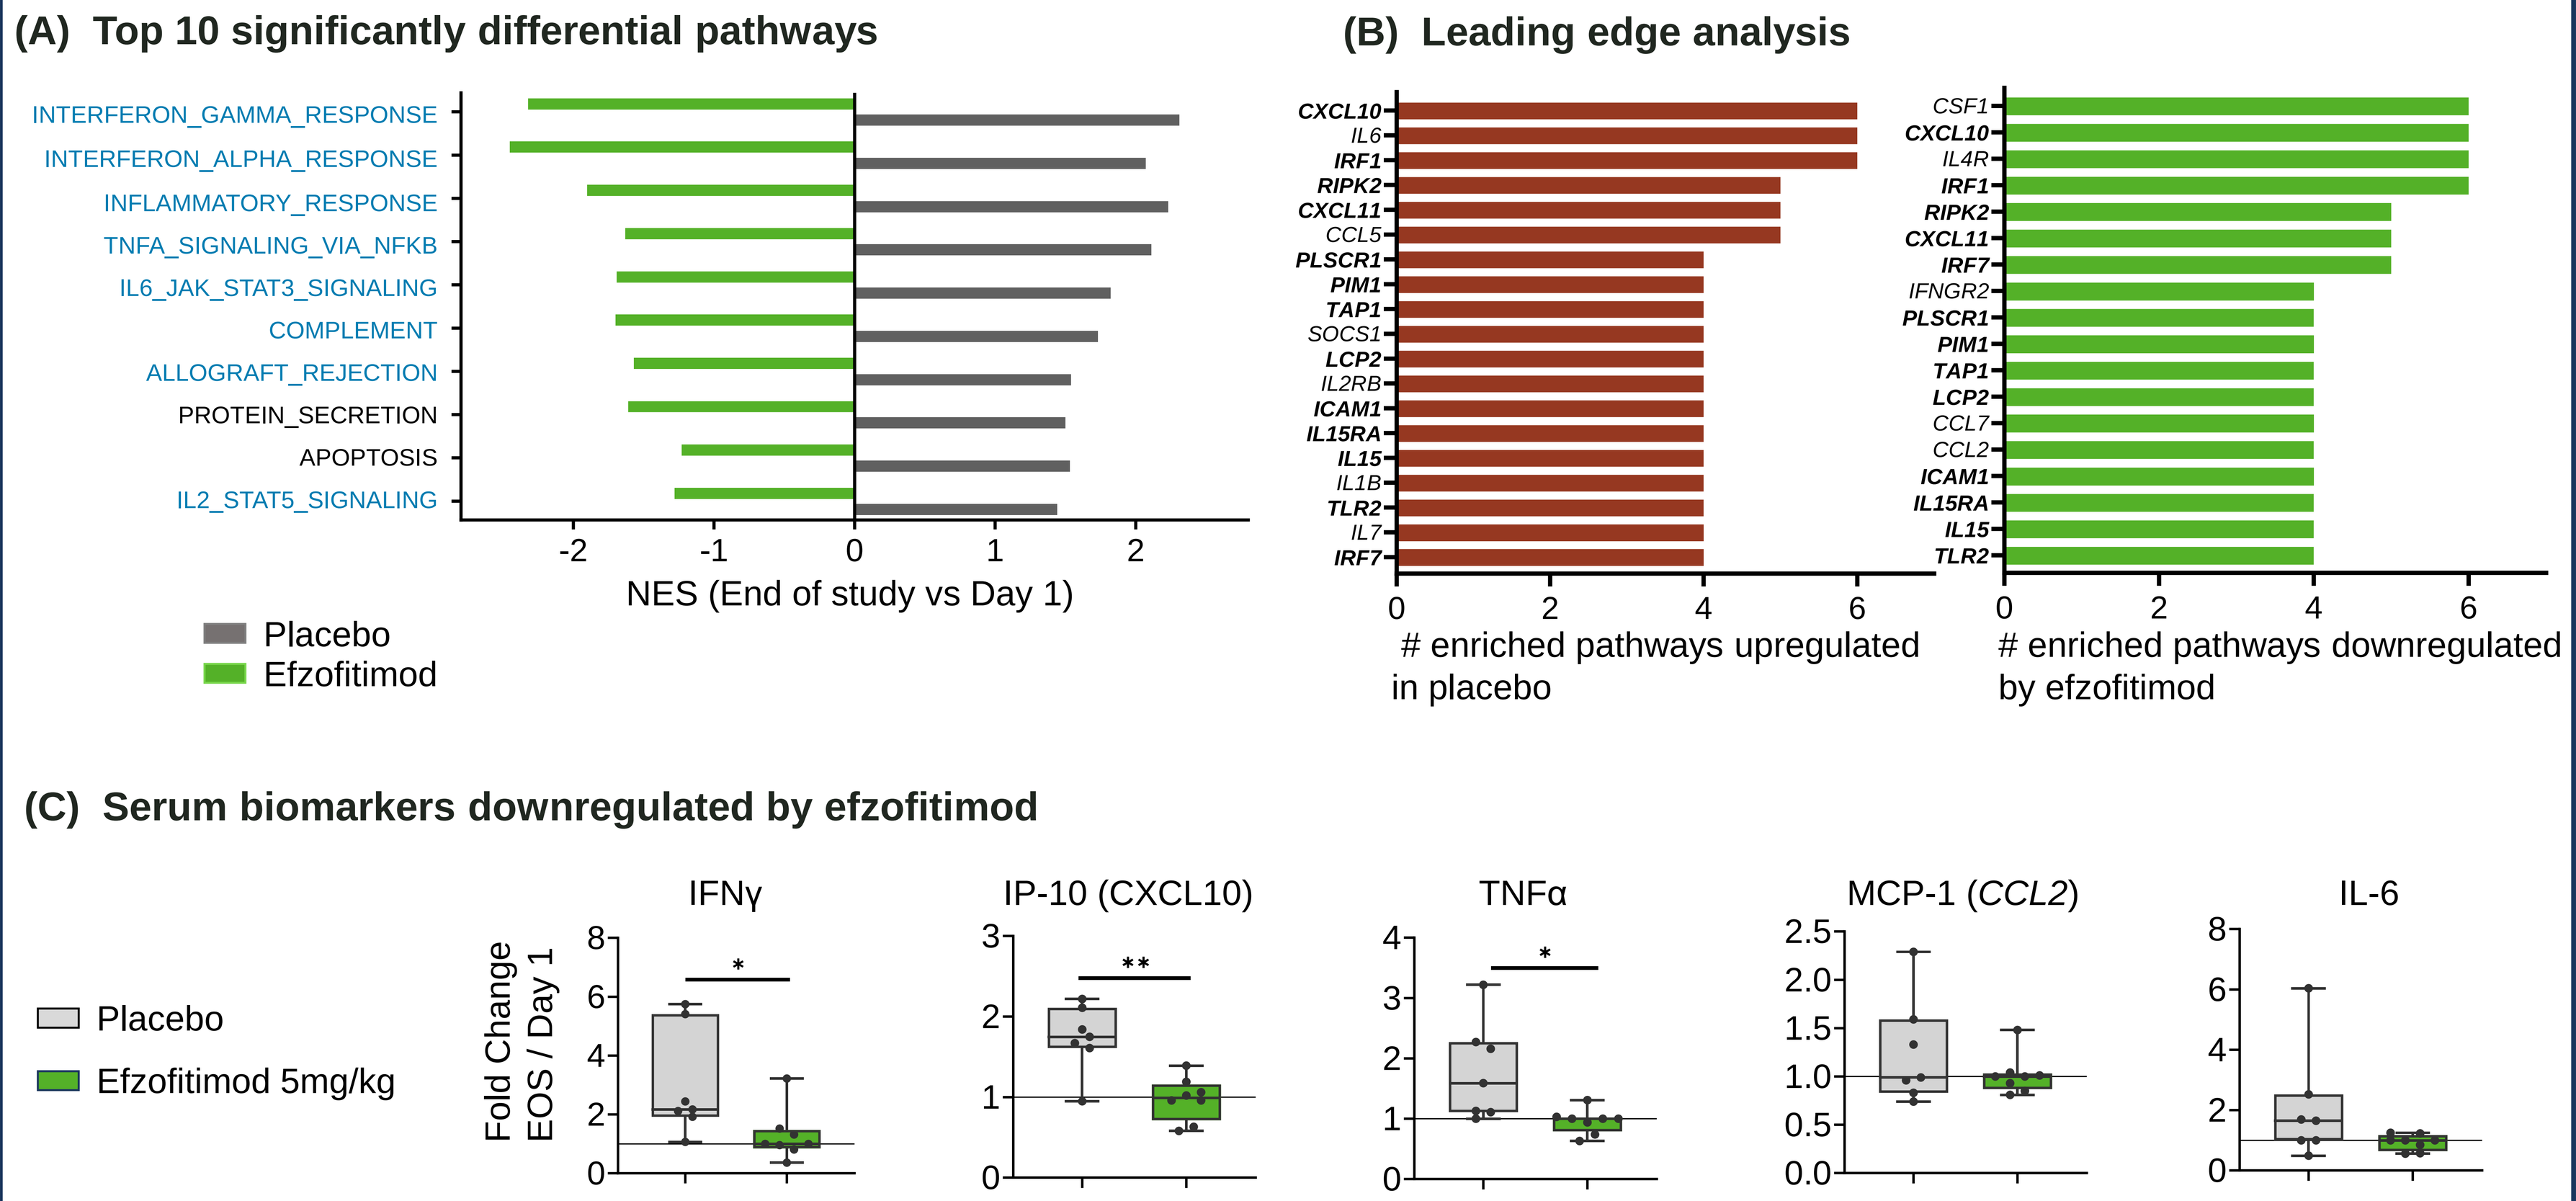
(A) Top 10 significantly differential pathways
INTERFERON_GAMMA_RESPONSE
INTERFERON_ALPHA_RESPONSE
INFLAMMATORY_RESPONSE
TNFA_SIGNALING_VIA_NFKB
IL6_JAK_STAT3_SIGNALING
COMPLEMENT
ALLOGRAFT_REJECTION
PROTEIN_SECRETION
APOPTOSIS
IL2_STAT5_SIGNALING
NES (End of study vs Day 1)
Placebo
Efzofitimod
(B) Leading edge analysis
CXCL10
IL6
IRF1
RIPK2
CXCL11
CCL5
PLSCR1
PIM1
TAP1
SOCS1
LCP2
IL2RB
ICAM1
IL15RA
IL15
IL1B
TLR2
IL7
IRF7
enriched pathways upregulated in placebo
CSF1
CXCL10
IL4R
IRF1
RIPK2
CXCL11
IRF7
IFNGR2
PLSCR1
PIM1
TAP1
LCP2
CCL7
CCL2
ICAM1
IL15RA
IL15
TLR2
enriched pathways downregulated by efzofitimod
(C) Serum biomarkers downregulated by efzofitimod
Placebo
Efzofitimod 5mg/kg
Fold Change EOS / Day 1
IFNγ
IP-10 (CXCL10)
TNFα
MCP-1 (CCL2)
IL-6

Figure 7. (A) Top 10 pathways differentially regulated at the end of study (EOS) compared to baseline (Day 1) by efzofitimod versus placebo. Immune pathways are highlighted in blue. (B) Core enriched genes by GSEA leading edge analysis. (C) Fold change in serum biomarkers at end of study compared to baseline.

Conclusions

- NRP2, the binding partner to efzofitimod, is highly expressed in cells and tissues from patients with ILD.
- NRP2 is upregulated during monocyte-derived macrophage differentiation, which corelates with efzofitimod's effect on promoting macrophages with an anti-inflammatory profile.
- Efzofitimod downregulates inflammatory genes and pathways in macrophages and PBMC from pulmonary sarcoidosis patients.
- Data support investigation of efzofitimod in ILD and other inflammatory and fibrotic diseases that involve myeloid cells in pathology.

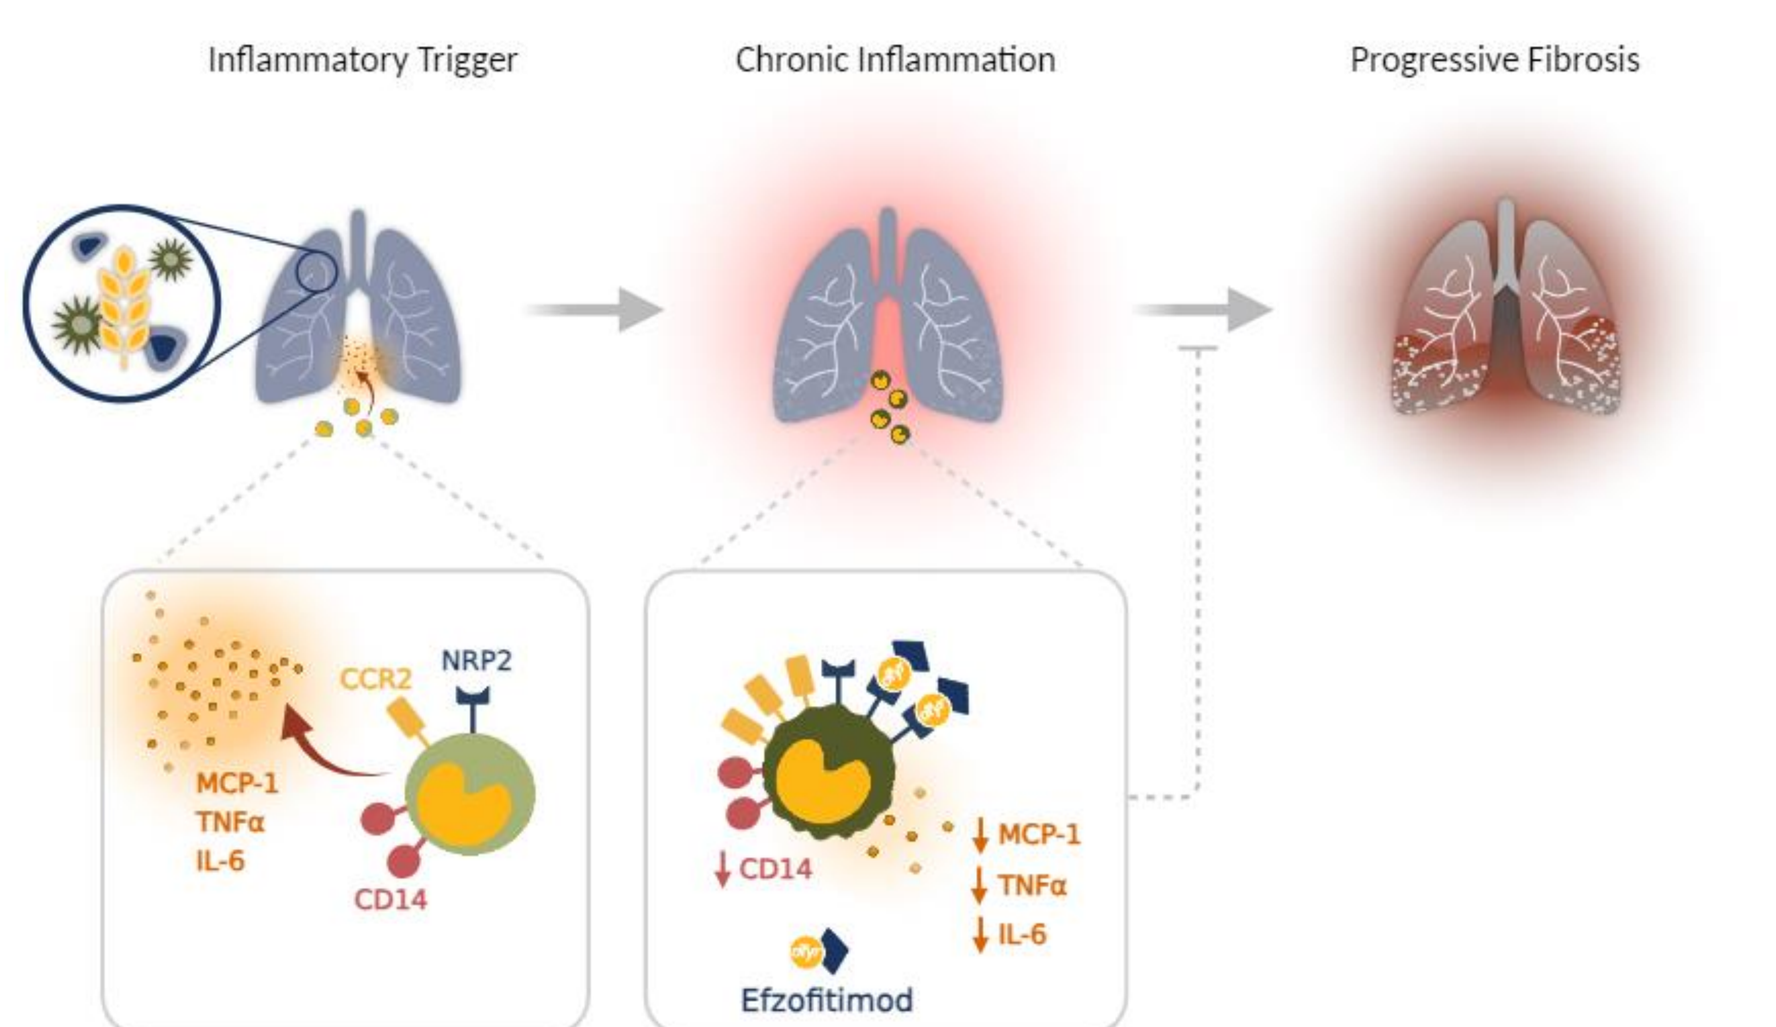
Inflammatory Trigger
Chronic Inflammation
Progressive Fibrosis
CCR2
NRP2
MCP-1
TNFα
IL-6
CD14
CD14
MCP-1
TNFα
IL-6
Efzofitimod

Disclosures:
All authors are employees of aTyr Pharma, Inc.
Funding support: This work was supported by aTyr Pharma, Inc.

References:
[1]Culver D, et. al. Efzofitimod for the Treatment of Pulmonary Sarcoidosis. *Chest*. 163 (4) (2023)
[2]Guo, Yang, et. al. New Functions of Aminoacyl-tRNA Synthetases beyond Translation. *Nat Rev Mol Cell Biol*. 11(9)(2010)
[3]Lo, Wing-Sze, et. al. Human tRNA synthetase catalytic nulls with diverse functions. *Science*. 345(6194)(2014)