4791

Immunosuppressive myeloid cells can be modulated with NRP2-targeting antibody ATYR2810 leading to enhanced anti-tumor immunity

Christoph Burkart[1], Clara Polizzi[1], John Choi[2], Max Pastenes[1], Alison Barber[1], Michael Lim[2], Leslie Nangle[1]

1. aTyr Pharma, San Diego
2. Department of Neurosurgery, Stanford School of Medicine, Stanford

Abstract

INTRODUCTION:

A major obstacle for many cancer immunotherapies is intrinsic or acquired resistance to treatment. Myeloid-derived immune cell populations such as myeloid-derived suppressor cells (MDSCs) and tumor associated macrophages (TAMs) are promoting an immunosuppressive tumor environment. This limits the effectiveness of current cancer immunotherapies. When activated, myeloid lineage immune cells express high levels of Neuropilin-2 (NRP2), a single pass transmembrane protein. NRP2 forms heterodimeric complexes with other membrane receptors such as VEGF receptors, plexins and integrins. In experimental models of immune disease, NRP2 has demonstrated anti-inflammatory effects. However, the role of NRP2 on immune cells in the context of tumor biology is largely unknown.

RESULTS:

NRP2 expression is upregulated on myeloid cells in an experimental model of acute lung injury and can be detected on myeloid cells in syngeneic tumor models. In experimental models of immune diseases like atopic dermatitis and collagen-induced arthritis, NRP2-deficient mice show a stronger immune response and worse pathology. Blocking NRP2 with a monoclonal antibody (ATYR2810) in a syngeneic, orthotopic mouse model of glioblastoma (GBM), resulted in anti-tumoral polarization of myeloid cells and a significant increase in overall survival. Additionally, in syngeneic tumor models with a large immunosuppressive myeloid population, ATYR2810 treatment improved the effectiveness of checkpoint inhibition via anti-PD-1.

CONCLUSIONS:

Here we demonstrate that NRP2 is a negative regulator of the inflammatory response and is highly expressed on immunosuppressive myeloid cells in the tumor environment. Blocking NRP2 with ATYR2810 was effective as a single therapy and when combined with checkpoint inhibition in syngeneic tumor models with a high prevalence of MDSCs. This suggests that modulating NRP2 offers a new approach to targeting and reversing the immunosuppressive function of myeloid cells in the tumor microenvironment.

Introduction

Figure 1. Neuropilin-2 is a transmembrane, pleiotropic receptor which is expressed on a subset of activated myeloid cells *in vivo*

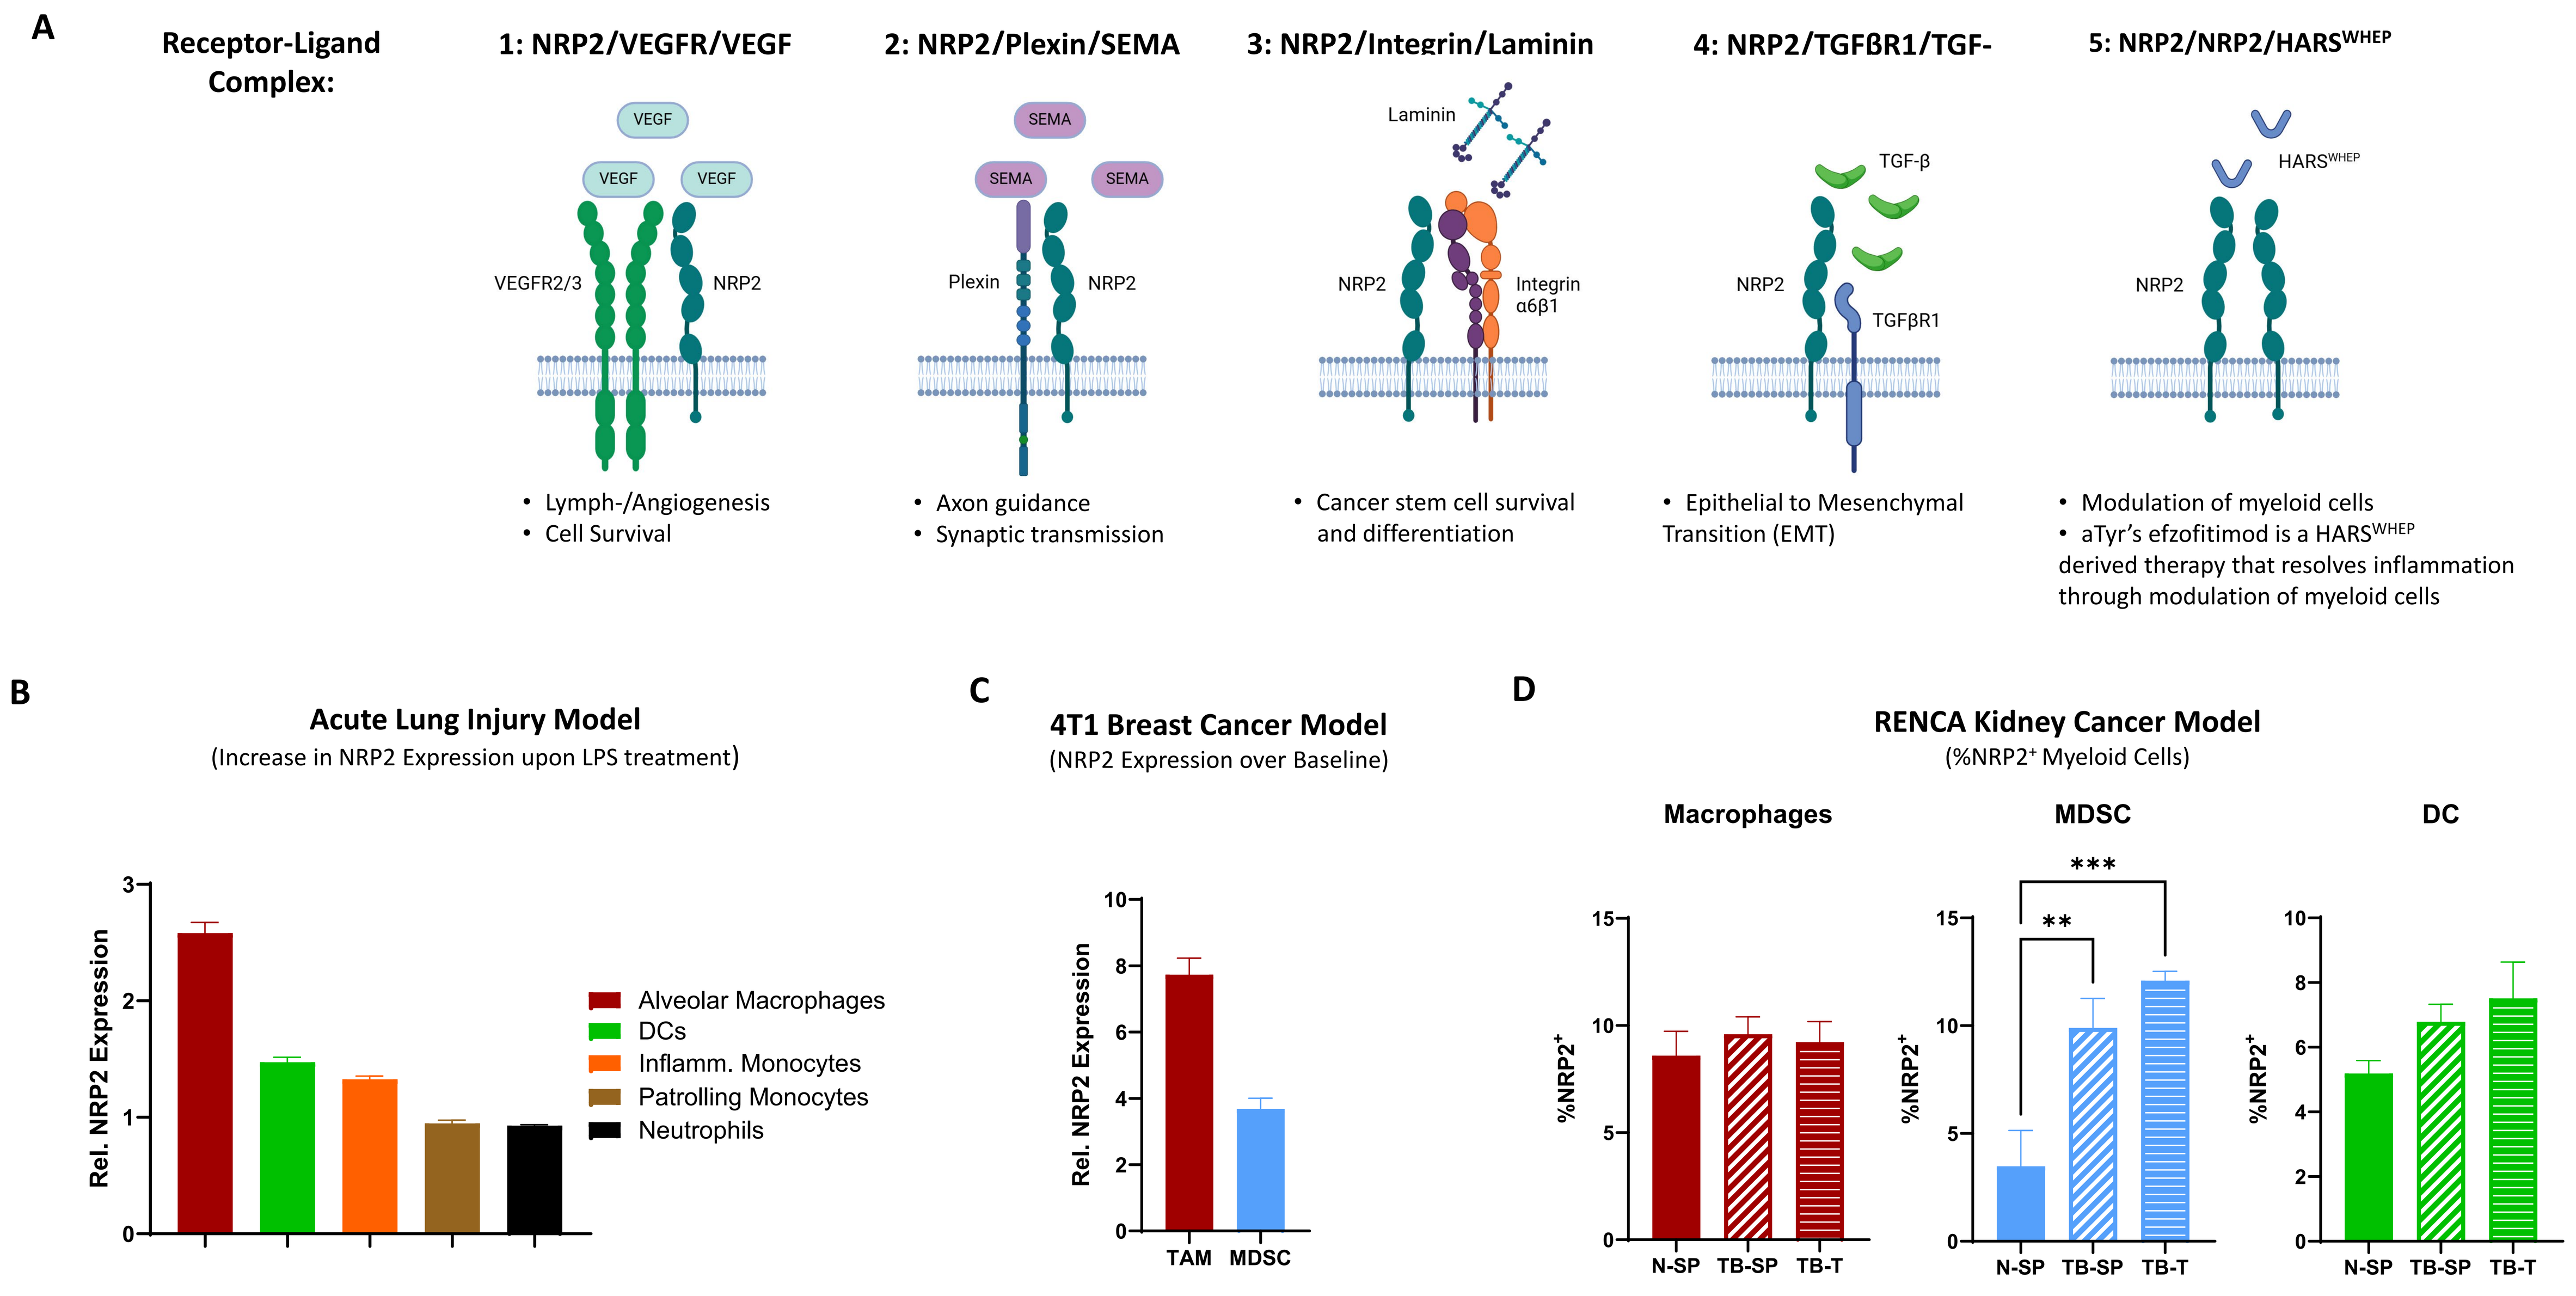
A
Receptor-Ligand Complex:
1: NRP2/VEGFR/VEGF
2: NRP2/Plexin/SEMA
3: NRP2/Integrin/Laminin
4: NRP2/TGFβR1/TGF-
5: NRP2/NRP2/HARSWHEP
VEGF
VEGFR2/3
NRP2
SEMA
Plexin
Laminin
Integrin α6β1
TGF-β
TGFβR1
HARSWHEP
• Lymph-/Angiogenesis
• Cell Survival
• Axon guidance
• Synaptic transmission
• Cancer stem cell survival and differentiation
• Epithelial to Mesenchymal Transition (EMT)
• Modulation of myeloid cells
• aTyr's efzofitimod is a HARSWHEP derived therapy that resolves inflammation through modulation of myeloid cells
B
Acute Lung Injury Model
(Increase in NRP2 Expression upon LPS treatment)
Rel. NRP2 Expression
Alveolar Macrophages
DCs
Inflamm. Monocytes
Patrolling Monocytes
Neutrophils
C
4T1 Breast Cancer Model
(NRP2 Expression over Baseline)
TAM
MDSC
D
RENCA Kidney Cancer Model
(%NRP2+ Myeloid Cells)
Macrophages
MDSC
DC
%NRP2+
N-SP
TB-SP
TB-T

- NRP2 is a cell surface receptor regulating various signaling pathways via heterodimerization[1] leading to enhanced signaling output (A1-4). Binding of the secreted N-terminal domain of histidyl-tRNA ($HARS^{WHEP}$) to NRP2 promotes differentiation of myeloid cells with decreased inflammatory profile[2] (A5).
- In a model of acute lung injury immune cells were isolated from lungs 24h post LPS administration and immunophenotyped. Myeloid cell populations such as alveolar macrophages show an increase in NRP2 surface expression in response to LPS. (B)
- Tumor-immunophenotyping of 4T1 breast cancer tumors revealed NRP2 surface expression on tumor-associated macrophages (TAMs) and myeloid-derived suppressor cells (MDSC). (C)
- In the RENCA tumor model of renal cancer the percentage of $NRP2^+$ MDSCs increased as compared to non-tumor bearing animals. (D) N-SP: naïve-spleen; TB-SP: tumor bearing - spleen; TB-T: tumor bearing - tumor

Results

Figure 2. NRP2 acts as a negative regulator of the immune response in experimental models of inflammatory disease

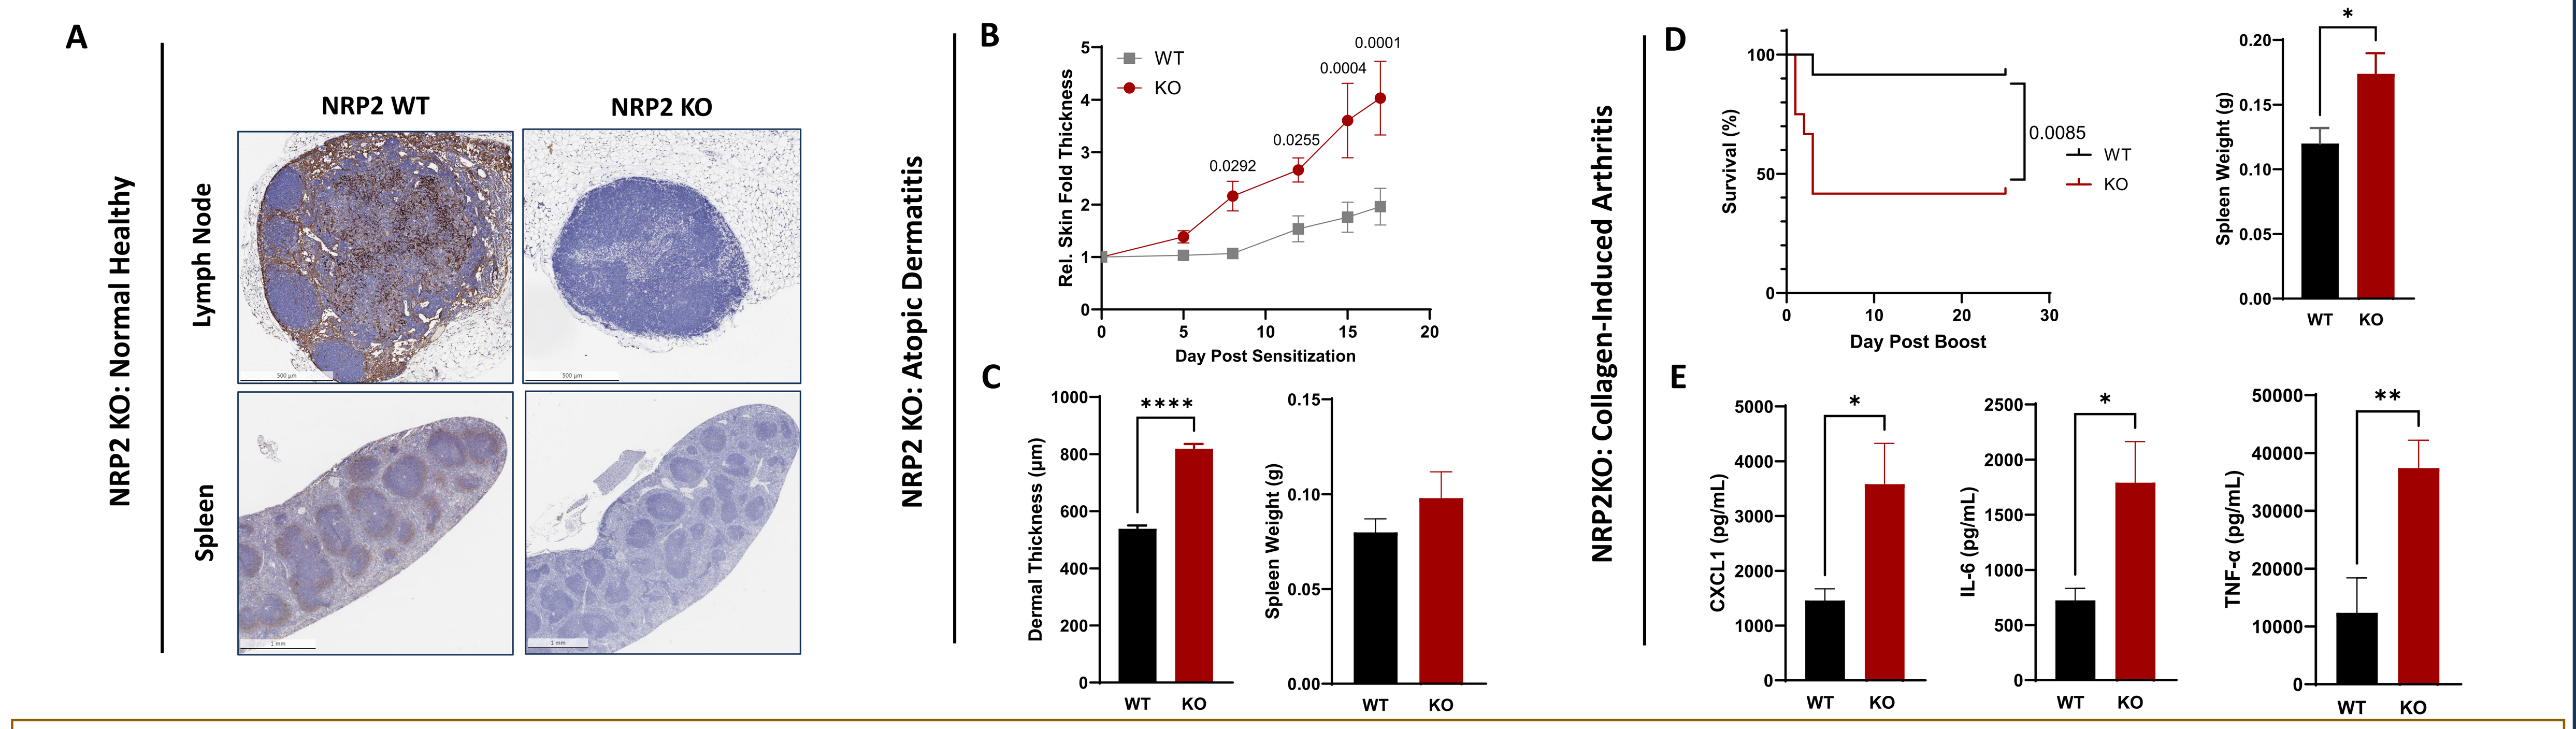
A
NRP2 KO: Normal Healthy
NRP2 WT
NRP2 KO
Lymph Node
Spleen
B
NRP2 KO: Atopic Dermatitis
Rel. Skin Fold Thickness
WT
KO
0.0292
0.0255
0.0004
0.0001
Day Post Sensitization
C
Dermal Thickness (µm)
Spleen Weight (g)
D
NRP2KO: Collagen-Induced Arthritis
Survival (%)
0.0085
Day Post Boost
E
CXCL1 (pg/mL)
IL-6 (pg/mL)
TNF-α (pg/mL)

- NRP2 IHC on healthy mouse lymph node and spleen from NRP2 WT or complete KO mice. A specific signal in wildtype tissues was observed in areas with substantial presence of myeloid cells such as macrophages and dendritic cells. Positive signal is mostly present in the medulla and cortex in the lymph node and the marginal zone of the white pulp in the spleen. (**A**)
- The oxazolone-induced atopic dermatitis model was utilized to investigate the role of NRP2 in inflammatory disease. Mice lacking NRP2 (KO) showed significantly enhanced thickening of the skin by both caliper measurements and histological analysis. In addition, a non-significant increase in spleen weights was observed indicating an enhanced immune response in the NRP2 KO animals. (**B-C**)
- NRP2 KO mice are hypersensitive to the collagen-induced arthritis model leading to increased mortality upon collagen boost and significantly larger spleens (**D**). Cytokine serum analysis at 48h post collagen boost shows a dramatic increase in pro-inflammatory cytokines suggesting the absence of an important immune regulatory mechanism in NRP2-deficient mice (**E**).

Figure 3. Targeting NRP2 with a monoclonal antibody (ATYR2810) modulates the myeloid cell population in syngeneic tumor models

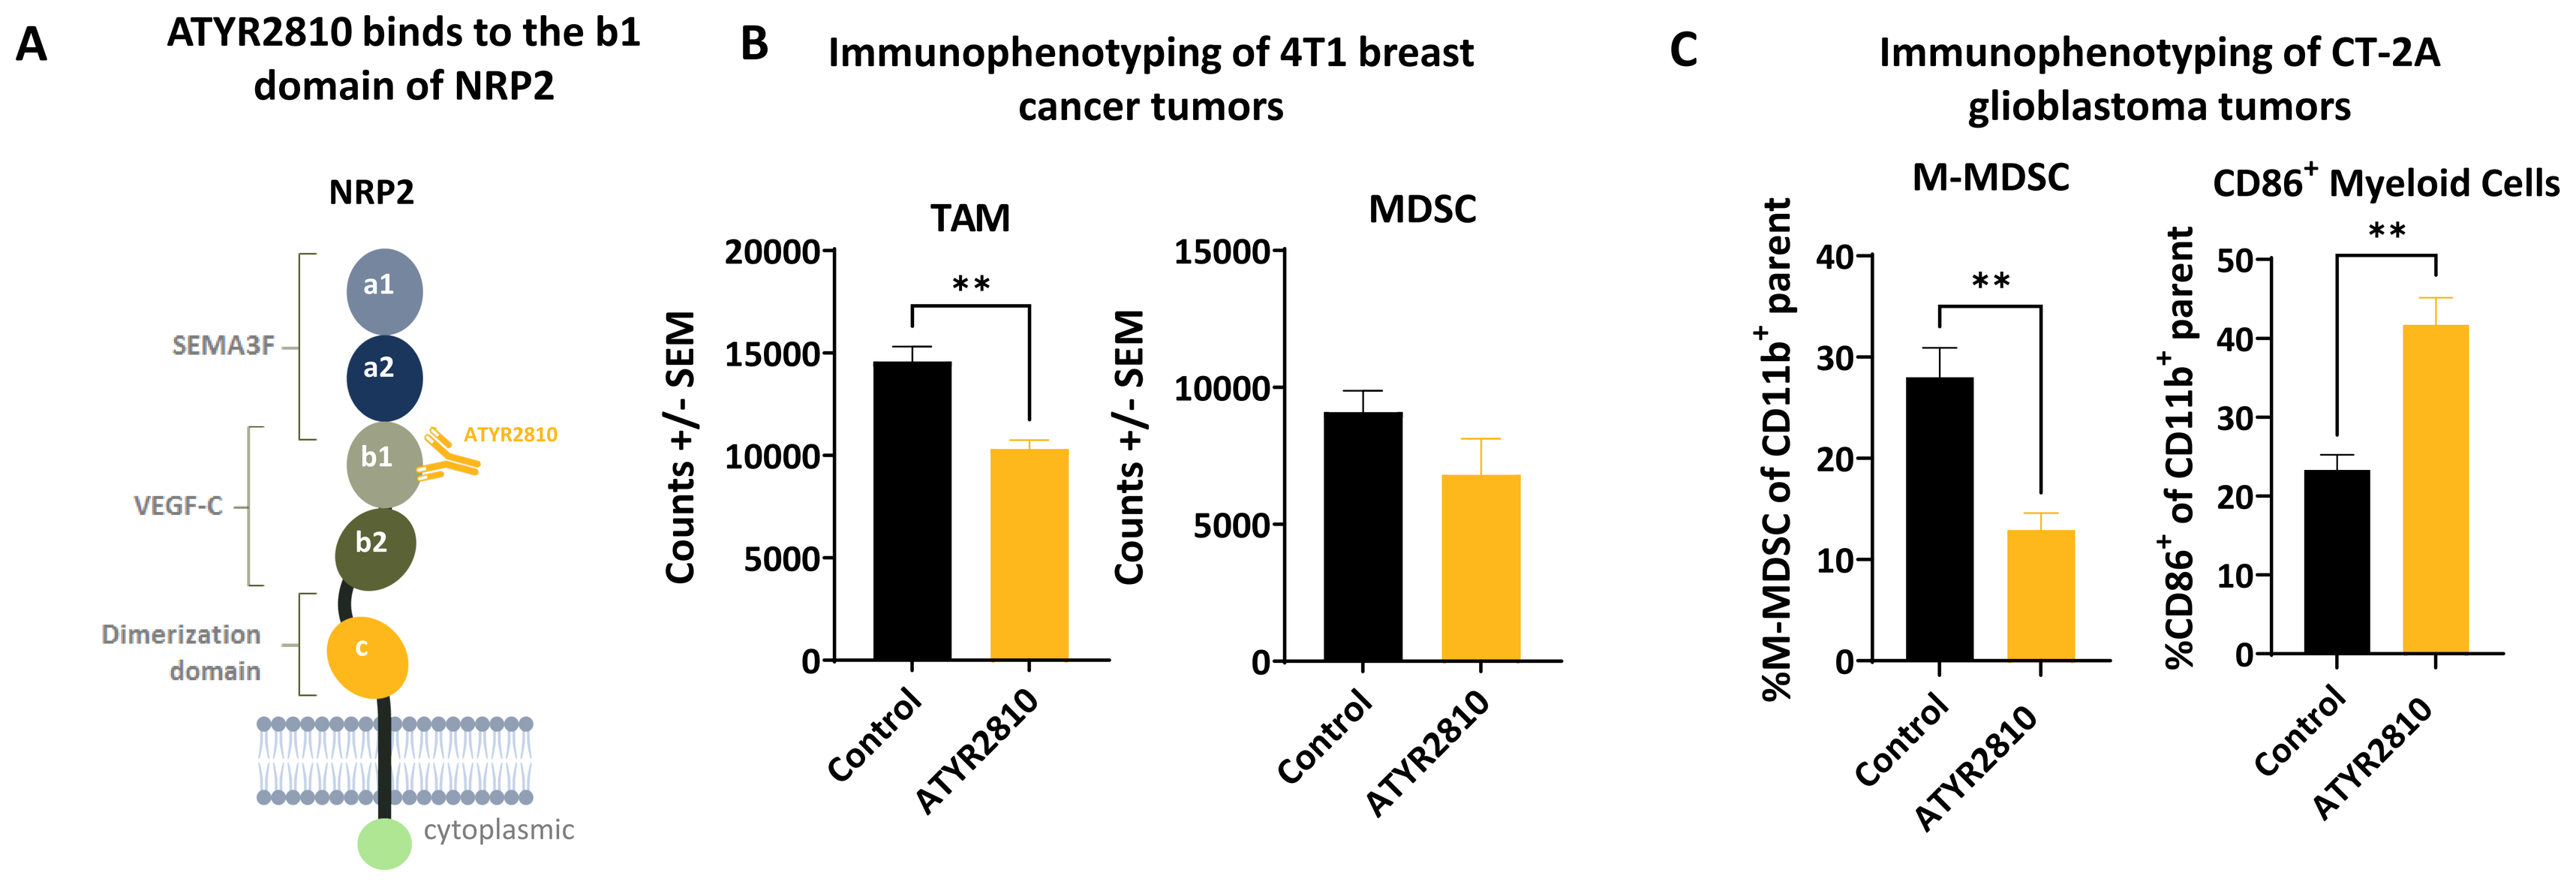
A
ATYR2810 binds to the b1 domain of NRP2
NRP2
SEMA3F
VEGF-C
Dimerization domain
a1
a2
b1
b2
c
ATYR2810
cytoplasmic
B
Immunophenotyping of 4T1 breast cancer tumors
TAM
MDSC
Counts +/- SEM
Control
ATYR2810
C
Immunophenotyping of CT-2A glioblastoma tumors
M-MDSC
CD86+ Myeloid Cells
%M-MDSC of CD11b+ parent
%CD86+ of CD11b+ parent

- The monoclonal antibody ATYR2810 binds to the b1 domain of NRP2 (**A**). This association leads to complete blockade of VEGF/NRP2 signaling.
- Single cell suspensions from 4T1 tumors with an average tumor volume of 500 mm^3 at harvest were subjected to immunophenotyping by flow cytometry. Intratumoral myeloid cell populations were reduced in ATYR2810 treated animals (**B**).
- In the orthotopic CT-2A tumor model of glioblastoma immunophenotyping revealed that ATYR2810 treatment reduces immune-inhibitory myeloid cell populations (M-MDSC) while increasing cell populations that promote anti-tumor immunity ($CD11b^+Cd86^+$) (**C**).

Figure 4. ATYR2810 increases survival and synergizes with checkpoint inhibition in syngeneic tumor models with high prevalence of MDSCs

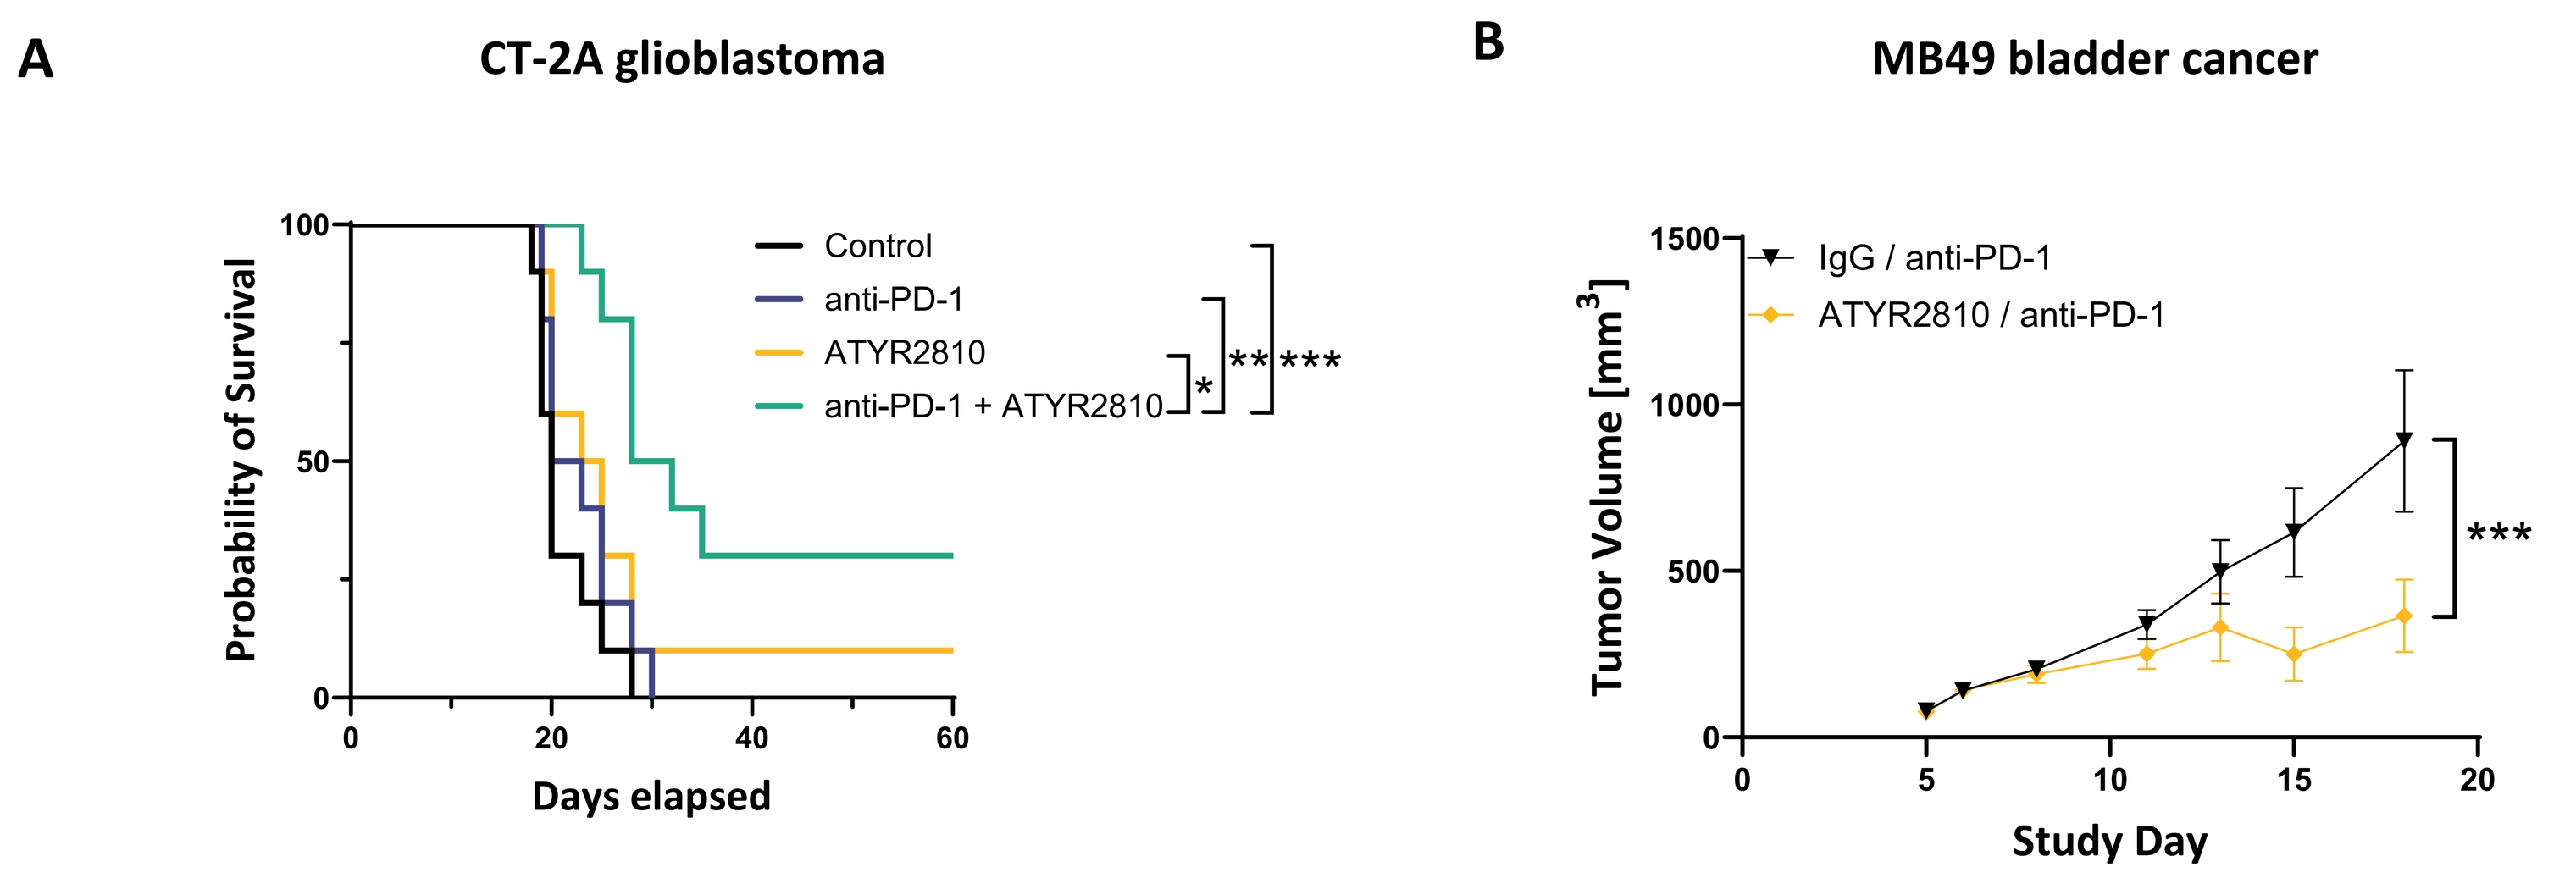
A
CT-2A glioblastoma
Probability of Survival
Control
anti-PD-1
ATYR2810
anti-PD-1 + ATYR2810
Days elapsed
B
MB49 bladder cancer
Tumor Volume [mm3]
IgG / anti-PD-1
ATYR2810 / anti-PD-1
Study Day

- The CT-2A glioblastoma and the MB49 bladder cancer model have high levels of intratumoral M-MDSCs of 20-30% [2] of the myeloid cell population.
- Mice with orthotopically implanted CT-2A tumors were treated with control, anti-PD-1 (10 mg/kg BIW), ATYR2810 (25 mg/kg BIW) or ATYR2810/anti-PD-1 combination. Combination of ATYR2810 and anti-PD-1 improved survival significantly (**A**).
- In the syngeneic, subcutaneous MB49 bladder cancer model combination of ATYR2810 (25 mg/kg BIW) and anti-PD-1 (10 mg/kg BIW) produced better tumor growth control than anti-PD-1 monotherapy (**B**).

Conclusions

- **NRP2 is upregulated on myeloid cells upon injury or during tumor development.**
- **In models of inflammatory disease, the absence of NRP2 worsens the associated pathology establishing NRP2 as an important regulator of the immune response.**
- **ATYR2810, a monoclonal antibody targeting NRP2, promotes an anti-tumor environment by reducing the number of immunosuppressive myeloid cells.**
- **Mice carrying tumors with high prevalence of MDSCs such as the CT-2A glioblastoma and the MB49 bladder cancer model benefit from anti-PD-1 and ATYR2810 combination therapy.**
- **ATYR2810 therapy may present a novel strategy to sensitize immunosuppressive tumors to immunotherapy by modulating the myeloid cell population.**

Statistical analysis: Fig. 1B - 1D: One-way ANOVA with Dunnett's multiple comparisons test; Fig. 2 + Fig. 3: Unpaired t-test; Fig 4A: Log-Rank test; Fig 4B: Mixed effects analysis. *p<0.05, **p<0.01, ***p<0.001, ****p<0.0001 significantly different from the respective control.

References:

1. Islam R, Mishra J, Bodas S, Bhattacharya S, Batra S K, Dutta S, & Datta K (2022). Role of Neuropilin-2-mediated signaling axis in cancer progression and therapy resistance. *Cancer and Metastasis Reviews*. DOI: 10.1007/s10555-022-10048-0
2. Nangle L, Xu Z, Siefker D, Burkart C, Chong YE, …., Adams R., Schimmel P. (2025). A human histidyl- tRNA synthetase splice variant therapeutic targets NRP2 to resolve lung inflammation and fibrosis. *Science Translational Medicine*. DOI: 10.1126/scitranslmed.adp4754
3. Yamamoto S, Kato M, Takeyama Y, Azuma Y, Yukimatsu N, Hirayama Y, Otoshi T, Yamasaki T, Fujioka M, Wanibuchi H, Uchida J (2023). Irradiation plus myeloid-derived suppressor cell-targeted therapy for overcoming treatment resistance in immunologically cold urothelial carcinoma. *British Journal of Cancer*. DOI: 10.1038/s41416-023-02244-8.

Funding support: This work was supported by aTyr Pharma, Inc.